Development of an IL13Rα2 x CD3 Bispecific DART® Protein for Redirected T-cell Killing of Solid Tumors

Jill Rillema, Monica Licea, Shereen Saini-Lal, Douglas H. Smith, Peter Lung, Francine Chen, Chia-Ying K. Lam, Yinhua Yang, Vatana Long, Liqin Liu, Nadia Gantt, Sergey Gorlatov, James Tamura, Ralph Alderson, Ezio Bonvini, Syd Johnson, and Paul A. Moore

MacroGenics, Inc., Rockville, MD and South San Francisco, CA

1498

http://ir.macrogenics.com/events.cfm

Abstract

Introduction: IL13Rα2 is a membrane-bound protein expressed on a number of malignant tumors, but not at significant levels in most normal tissues. Clinically tested therapies to treat cancer by targeting IL13Rα2 to date have included a ligand toxin conjugate (IL13-PE) and IL13Rα2 directed CAR-T cells. Both strategies appear to be well tolerated, although with mixed results in tumor eradication. An alternative strategy for targeting IL13Rα2 is an IL13Rα2 x CD3 Dual-Affinity Re-Targeting (DART®) bispecific molecule designed to co-engage IL13Rα2 on tumor cells and cytotoxic T cells through CD3, resulting in killing of tumor cells.

Methods: Mouse monoclonal antibodies (mAbs) were generated using standard immunization protocols followed by binding and epitope binning analyses. Cell binding was performed by flow cytometry and frozen normal and tumor tissues analyses by immunohistochemistry (IHC). In vitro studies were performed with cancer cell lines and primary human T cells or peripheral blood mononuclear cells (PBMCs). In vivo studies were performed in immune-deficient tumor-bearing mice co-implanted with activated human T cells or reconstituted with human PBMCs.

Results: A panel of mAbs that bind human and cynomolgus monkey IL13Rα2 proteins were selected, representing a range of binding characteristics and epitope diversity. IHC showed favorable normal vs. tumor tissue reactivity, with high levels of IL13Rα2 expression in glioblastoma and melanoma. The mAb panel was converted to DART molecules with an anti-CD3 arm, assessed for the ability to mediate cytotoxic T-lymphocyte (CTL) activity and cytokine release, and a lead selected for humanization and engineering into an IL13Rα2 x CD3 DART molecule incorporating an Fc domain to prolong circulating half-life. The IL13Rα2 x CD3 DART protein bound human and cynomolgus monkey IL13Rα2 (K_D = 0.64 nM and 0.5 nM, respectively) and mediated CTL activity against LOX-IMVI (melanoma) and SK-MES-1 (lung adenocarcinoma) target cells. Cytokines (IFN-γ, IL-10, and TNF-α) were released in the presence of IL13Rα2-expressing target cells and human PBMCs, but not with PBMCs alone. Administration of the IL13Rα2 x CD3 DART molecule (≤ 50 μg/kg IV for 4 consecutive days) prevented tumor growth when tumor cells (A375 and LOX-IMVI melanoma, U87 glioblastoma, or Daoy medulloblastoma cell lines) were co-mixed with activated human T cells and implanted subcutaneously (SC) in NOD/SCID/IL-2 gamma chain null (NOG) mice. Tumor eradication was also observed in NOG/MHCI KO mice reconstituted with human PBMCs and bearing established LOX-IMVI melanoma tumors following 2 weekly IV doses of 50 μg/kg IL13Rα2 x CD3 DART molecule.

Conclusion: Robust in vitro and in vivo antitumor activity for an IL13Rα2 x CD3 DART protein was demonstrated. Additional studies are underway to further characterize the molecule as a development candidate for treatment of IL13Rα2-positive cancers.

Methods

- Antibodies reactive to human IL13Rα2 were generated in mice through a whole-cell-based immunization protocol
- IL13Rα2 expression in human normal and tumor tissues was determined on frozen tissue sections
- Basic DART proteins were transiently expressed in CHO cells and purified by affinity chromatography. The Fc-bearing IL13Rα2 x CD3 DART protein was stably expressed in CHO cells and purified to homogeneity by standard protein A chromatography
- DART protein-mediated killing of target cell lines (CTL activity) in the presence of human PBMCs was determined by lactate dehydrogenase (LDH) release
- Cytokine levels in culture supernatants from CTL assays were quantitated by ELISA
- Cell-binding assays were conducted using IL13Rα2-expressing tumor cell lines and purified human or cynomolgus monkey T cells by flow cytometry
- Affinity analysis was conducted via surface plasmon resonance (SPR) using an anti-penta-His chip (1:1 binding) to capture soluble human or cynomolgus monkey IL13Rα2-His protein, followed by flowing DART proteins at 6.25–100 nM
- In vivo studies: Tumor engraftment studies (Winn's model) were conducted in NOG mice. Tumor growth inhibition studies were conducted in NOG/MHCI-/- mice reconstituted with human PBMCs

Introduction

- Antibodies derived from whole cancer-cell immunizations that display favorable normal vs. tumor differential by IHC were selected for antigen identification
- IL13Rα2 was identified as a mAb target highly expressed on tumor cells, but minimally on normal tissue
- An expanded panel of IL13Rα2 mAbs was generated through targeted immunization and characterized as follows:
 - Tumor cell and recombinant protein reactivity
 - Low normal tissue reactivity
 - Single-concentration affinity screening
 - Epitope binning
- A subset of mAbs representing different binding groups were selected for conversion to basic IL13Rα2 x CD3 DART proteins and screened for activity in CTL and cytokine-release assays
- An IL13Rα2 x CD3 DART molecule displaying optimal properties was selected for further development
 - Humanization and conversion to the Fc-bearing DART format
 - Biophysical and binding profile
 - Evaluation of in vitro CTL activity and cytokine release
 - Evaluation of in vivo activity in mouse xenograft models

IL13Rα2 Displays Favorable Normal-to-Tumor Differential Expression

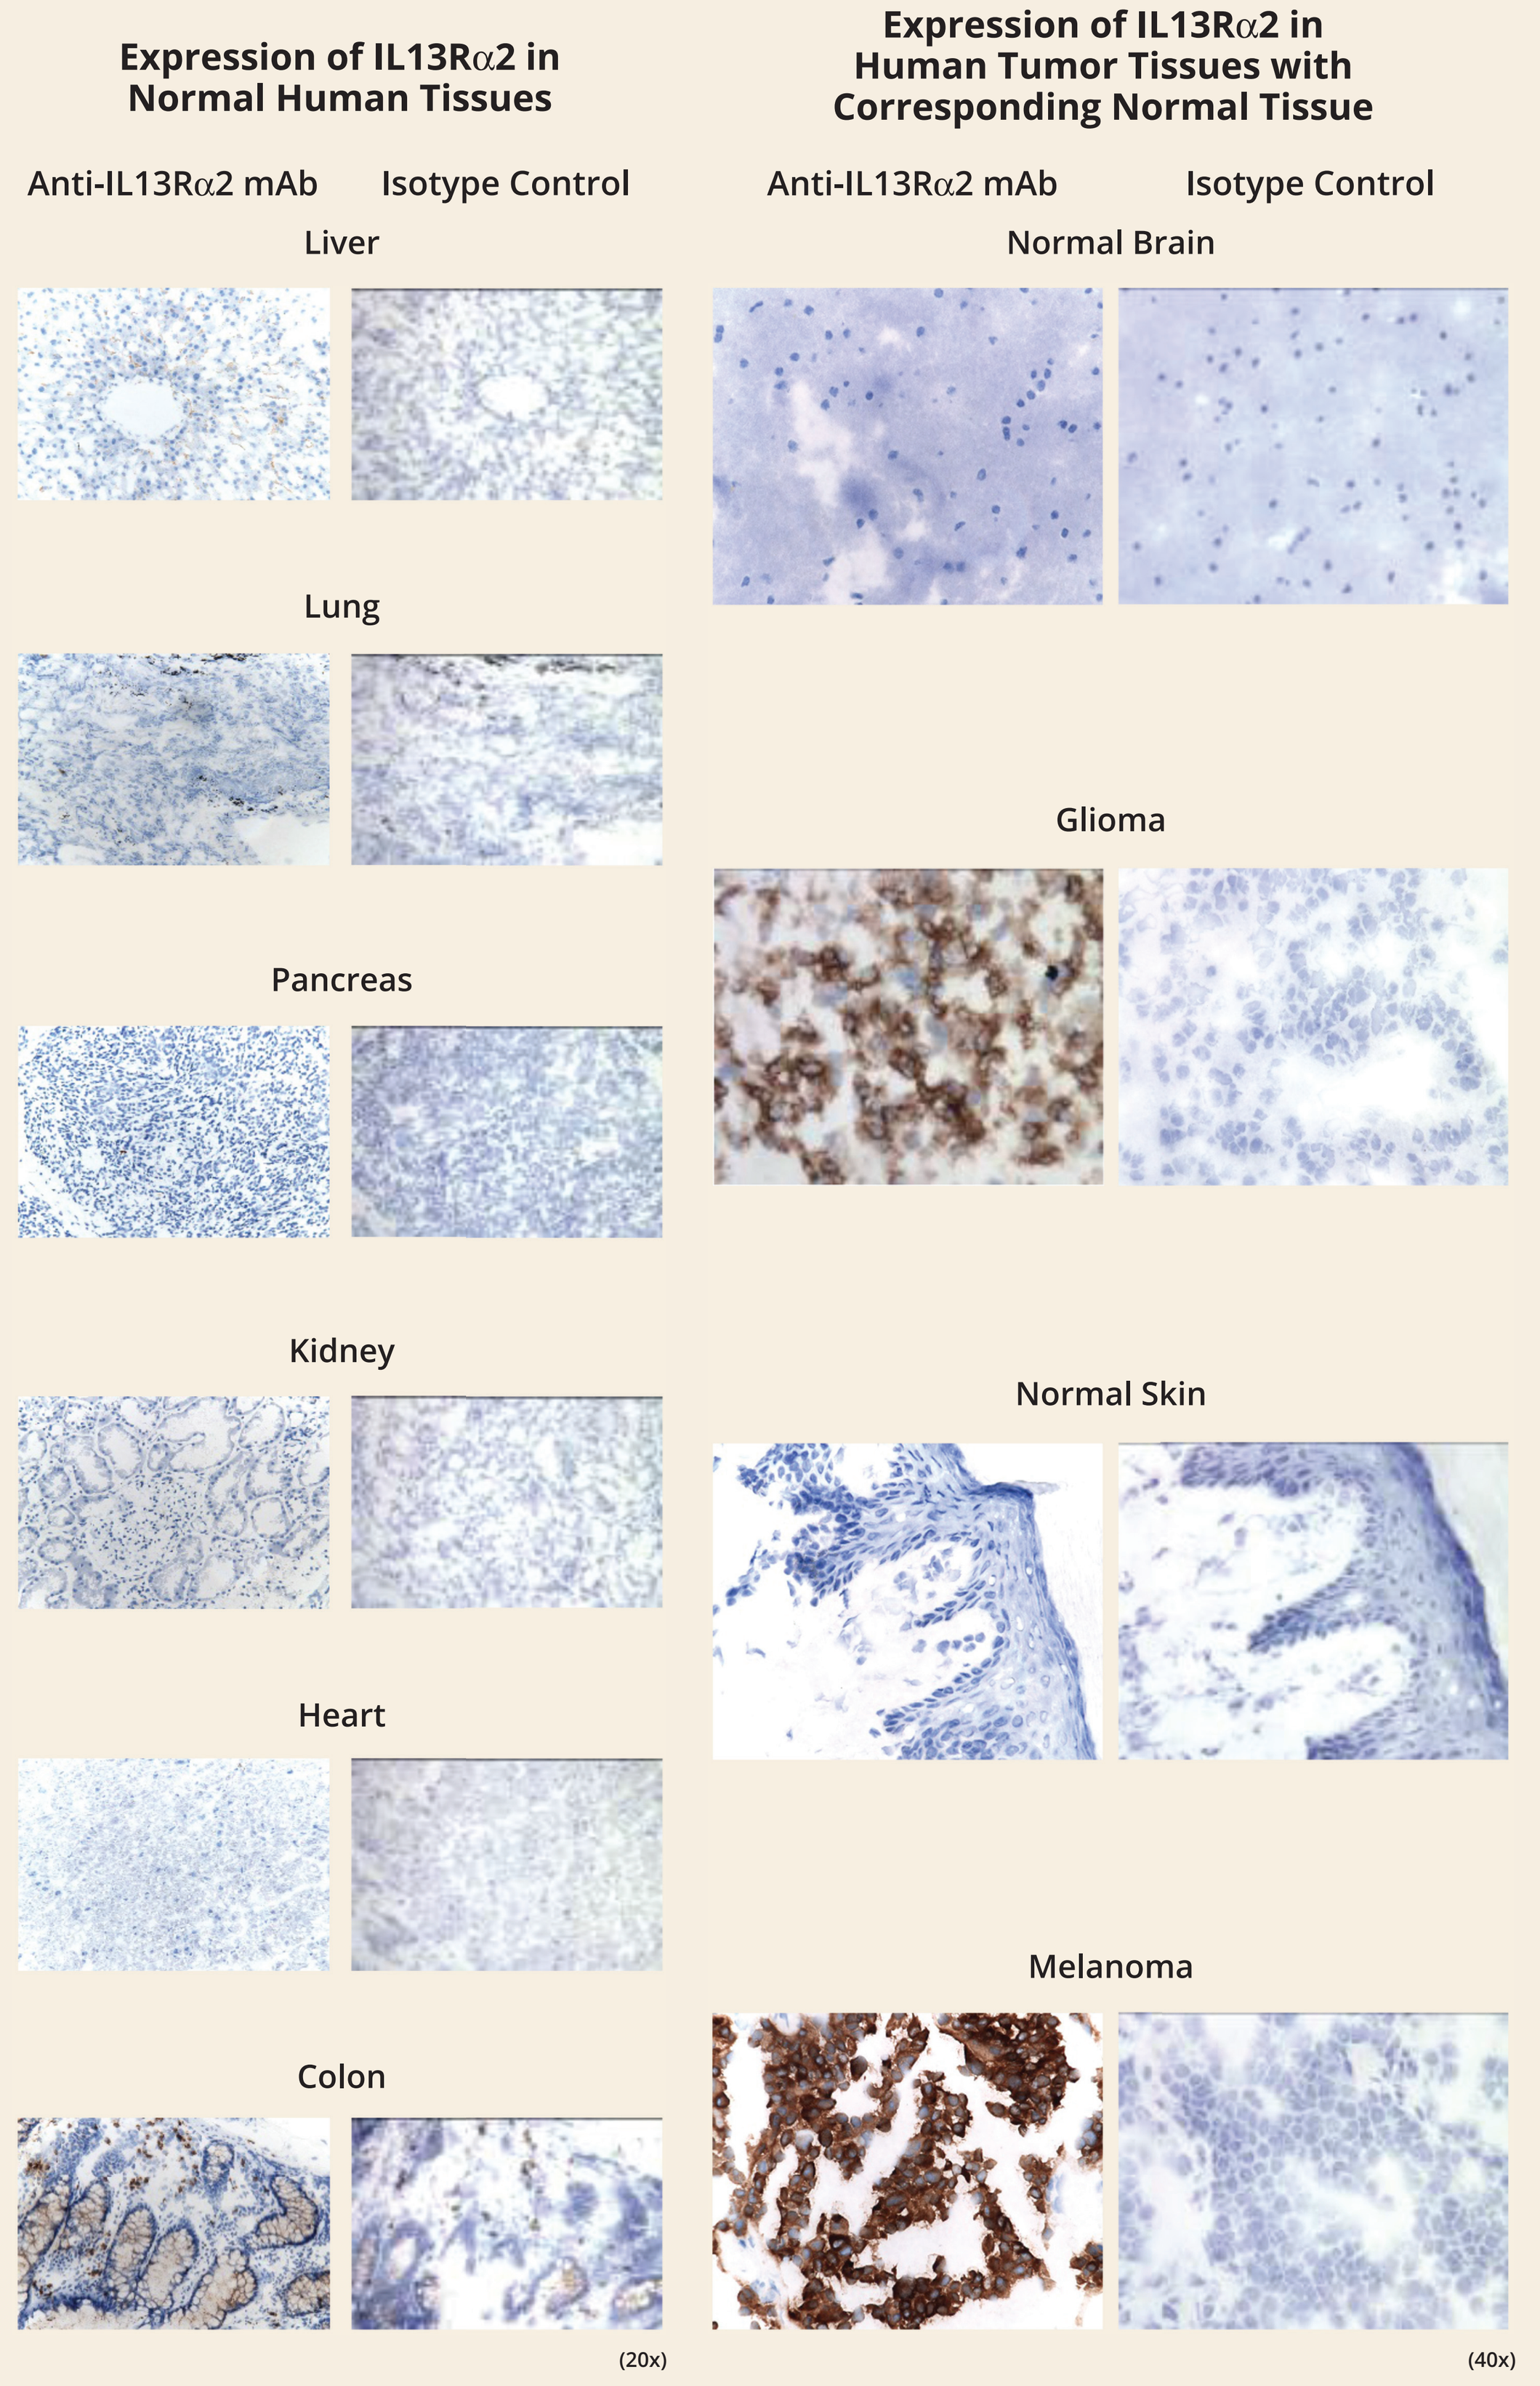
Expression of IL13Rα2 in Normal Human Tissues
Anti-IL13Rα2 mAb
Isotype Control
Liver
Lung
Pancreas
Kidney
Heart
Colon
(20x)
Expression of IL13Rα2 in Human Tumor Tissues with Corresponding Normal Tissue
Anti-IL13Rα2 mAb
Isotype Control
Normal Brain
Glioma
Normal Skin
Melanoma
(40x)

- Frozen and normal human tissue sections were stained with an anti-human IL13Rα2 mAb or isotype control antibody at 5 μg/mL
- Anti-IL13Rα2 mAb shows low reactivity to normal tissues and high reactivity in glioblastoma and melanoma

Results

Redirected T-cell Killing Through DART Proteins

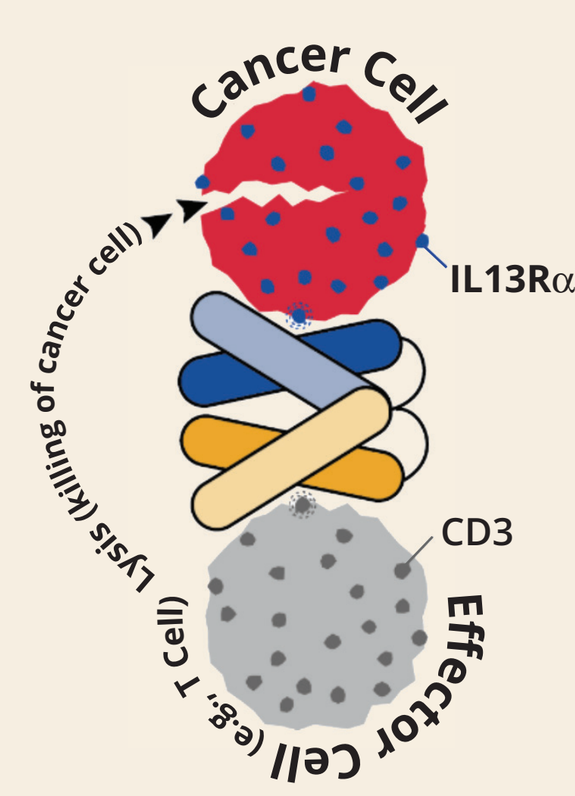
Cancer Cell
IL13Rα2
CD3
Effector Cell (e.g., T Cell)
Lysis (killing of cancer cell)

- DART proteins engage T cells through CD3 and cancer cells through tumor-associated antigens, such as IL13Rα2
- Monovalent binding to CD3 avoids target independent T-cell activation
- T-cell activation and tumor cell lysis are strictly dependent on engagement of both target antigens
- TCR & MHC-independent tumor cell recognition: virtually any T cell can kill cancer cells
- Nine IL13Rα2 antibodies were selected to represent a range of binding characteristics and epitope diversity and were used to generate a panel of IL13Rα2 x CD3 DART proteins

IL13Rα2 x CD3 DART Proteins

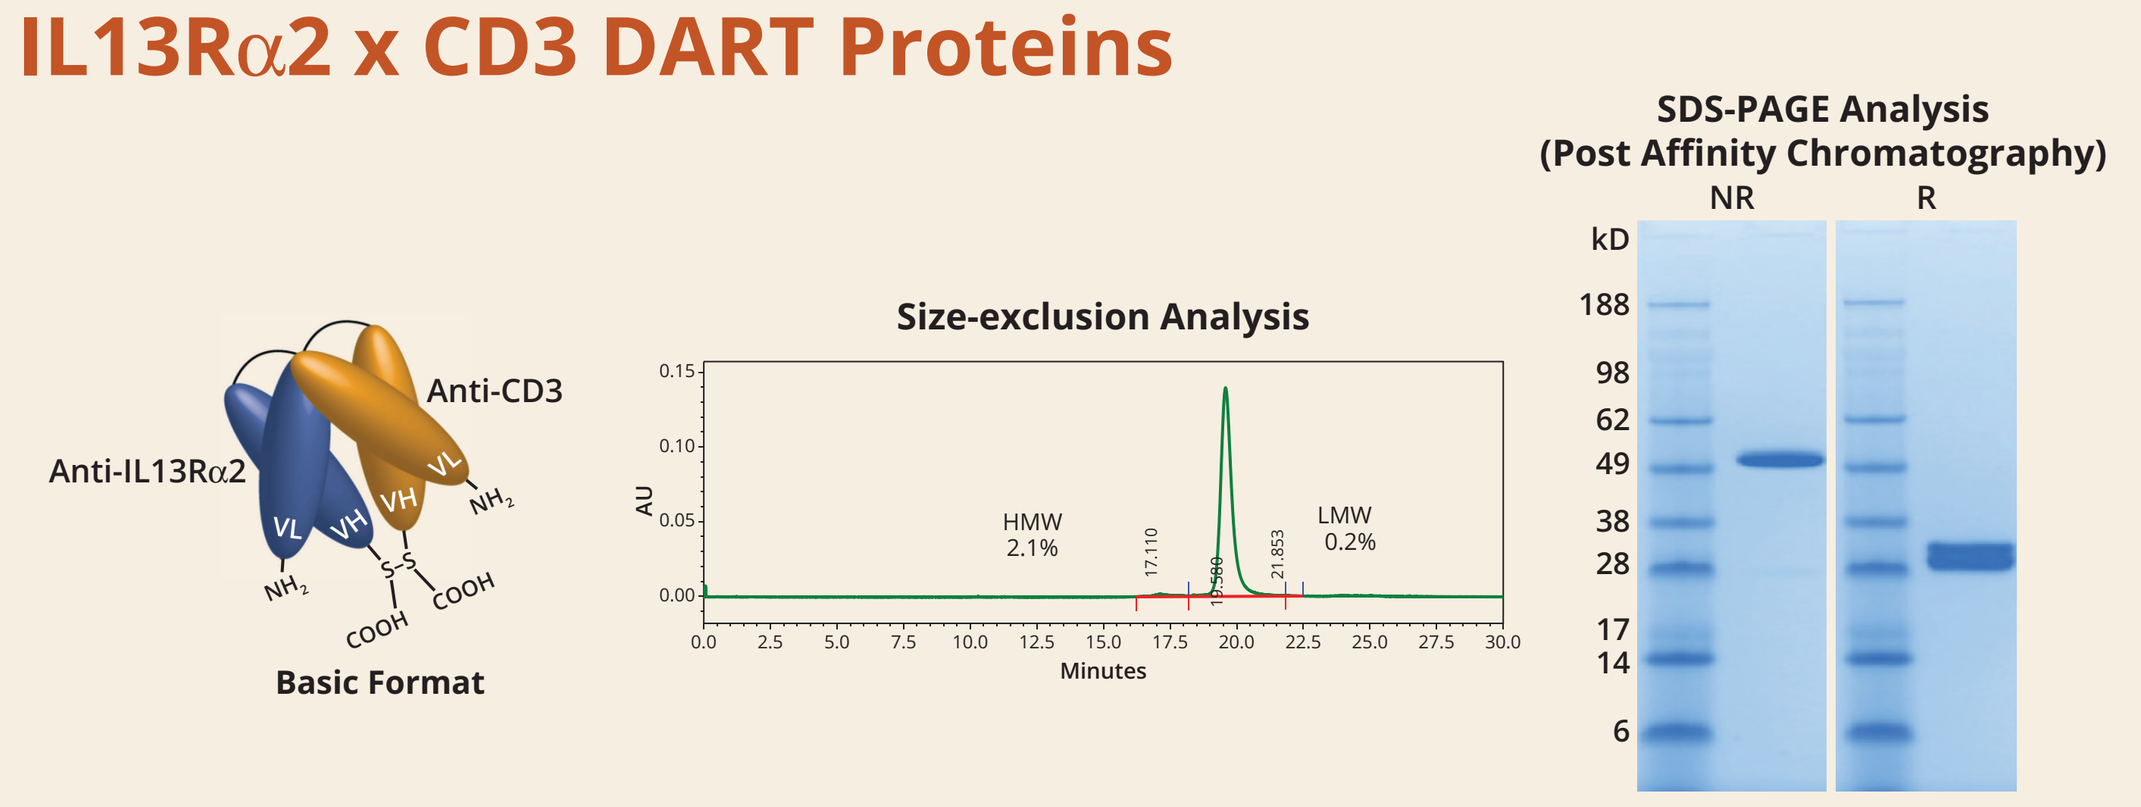
Anti-IL13Rα2
Anti-CD3
Basic Format
Size-exclusion Analysis
HMW 2.1%
LMW 0.2%
Minutes
SDS-PAGE Analysis (Post Affinity Chromatography)
NR
R
kD

- Fv regions from nine IL13Rα2 mAbs exhibiting a range of antigen binding properties were independently assembled as IL13Rα2 x CD3 DART molecules
- Purified IL13Rα2 x CD3 DART molecules were analyzed by SEC-HPLC and SDS-PAGE, evaluated for binding properties, and functionally characterized in redirected T-cell killing and cytokine release assays

Summary of Activity of IL13Rα2 x CD3 DART Proteins

DART	Binding Group	Cynomolgus Monkey Cross-reactive	CTL Assay EC_{50} (ng/mL) SKMES-1	CTL Assay EC_{50} (ng/mL) UACC-62	Cytokine Release in PBMCs Alone
MG06	1	✓	0.017	0.023	Neg
MG12	4	✓	179.2	43.34	Neg
MG16	2	✓	32.96	55.88	Neg
MG32	3	✓	8.37	1.20	Neg
MG40	1	✓	0.190	0.080	Neg
MG52	4	✓	0.051	0.010	Neg
MG55	5	✓	0.220	0.098	Neg
MG56	4	✓	0.071	0.012	Neg
MG71	5	✓	0.035	0.014	Neg

- Several IL13Rα2 x CD3 DART proteins were identified as potent mediators of CTL activity
- DART proteins did not induce cytokine release by human PBMCs alone
- MG56 was chosen for further development based on these results, as well as the low normal tissue vs. high tumor reactivity with the parental mAb by IHC

IL13Rα2 x CD3 DART Proteins Mediate CTL Activity

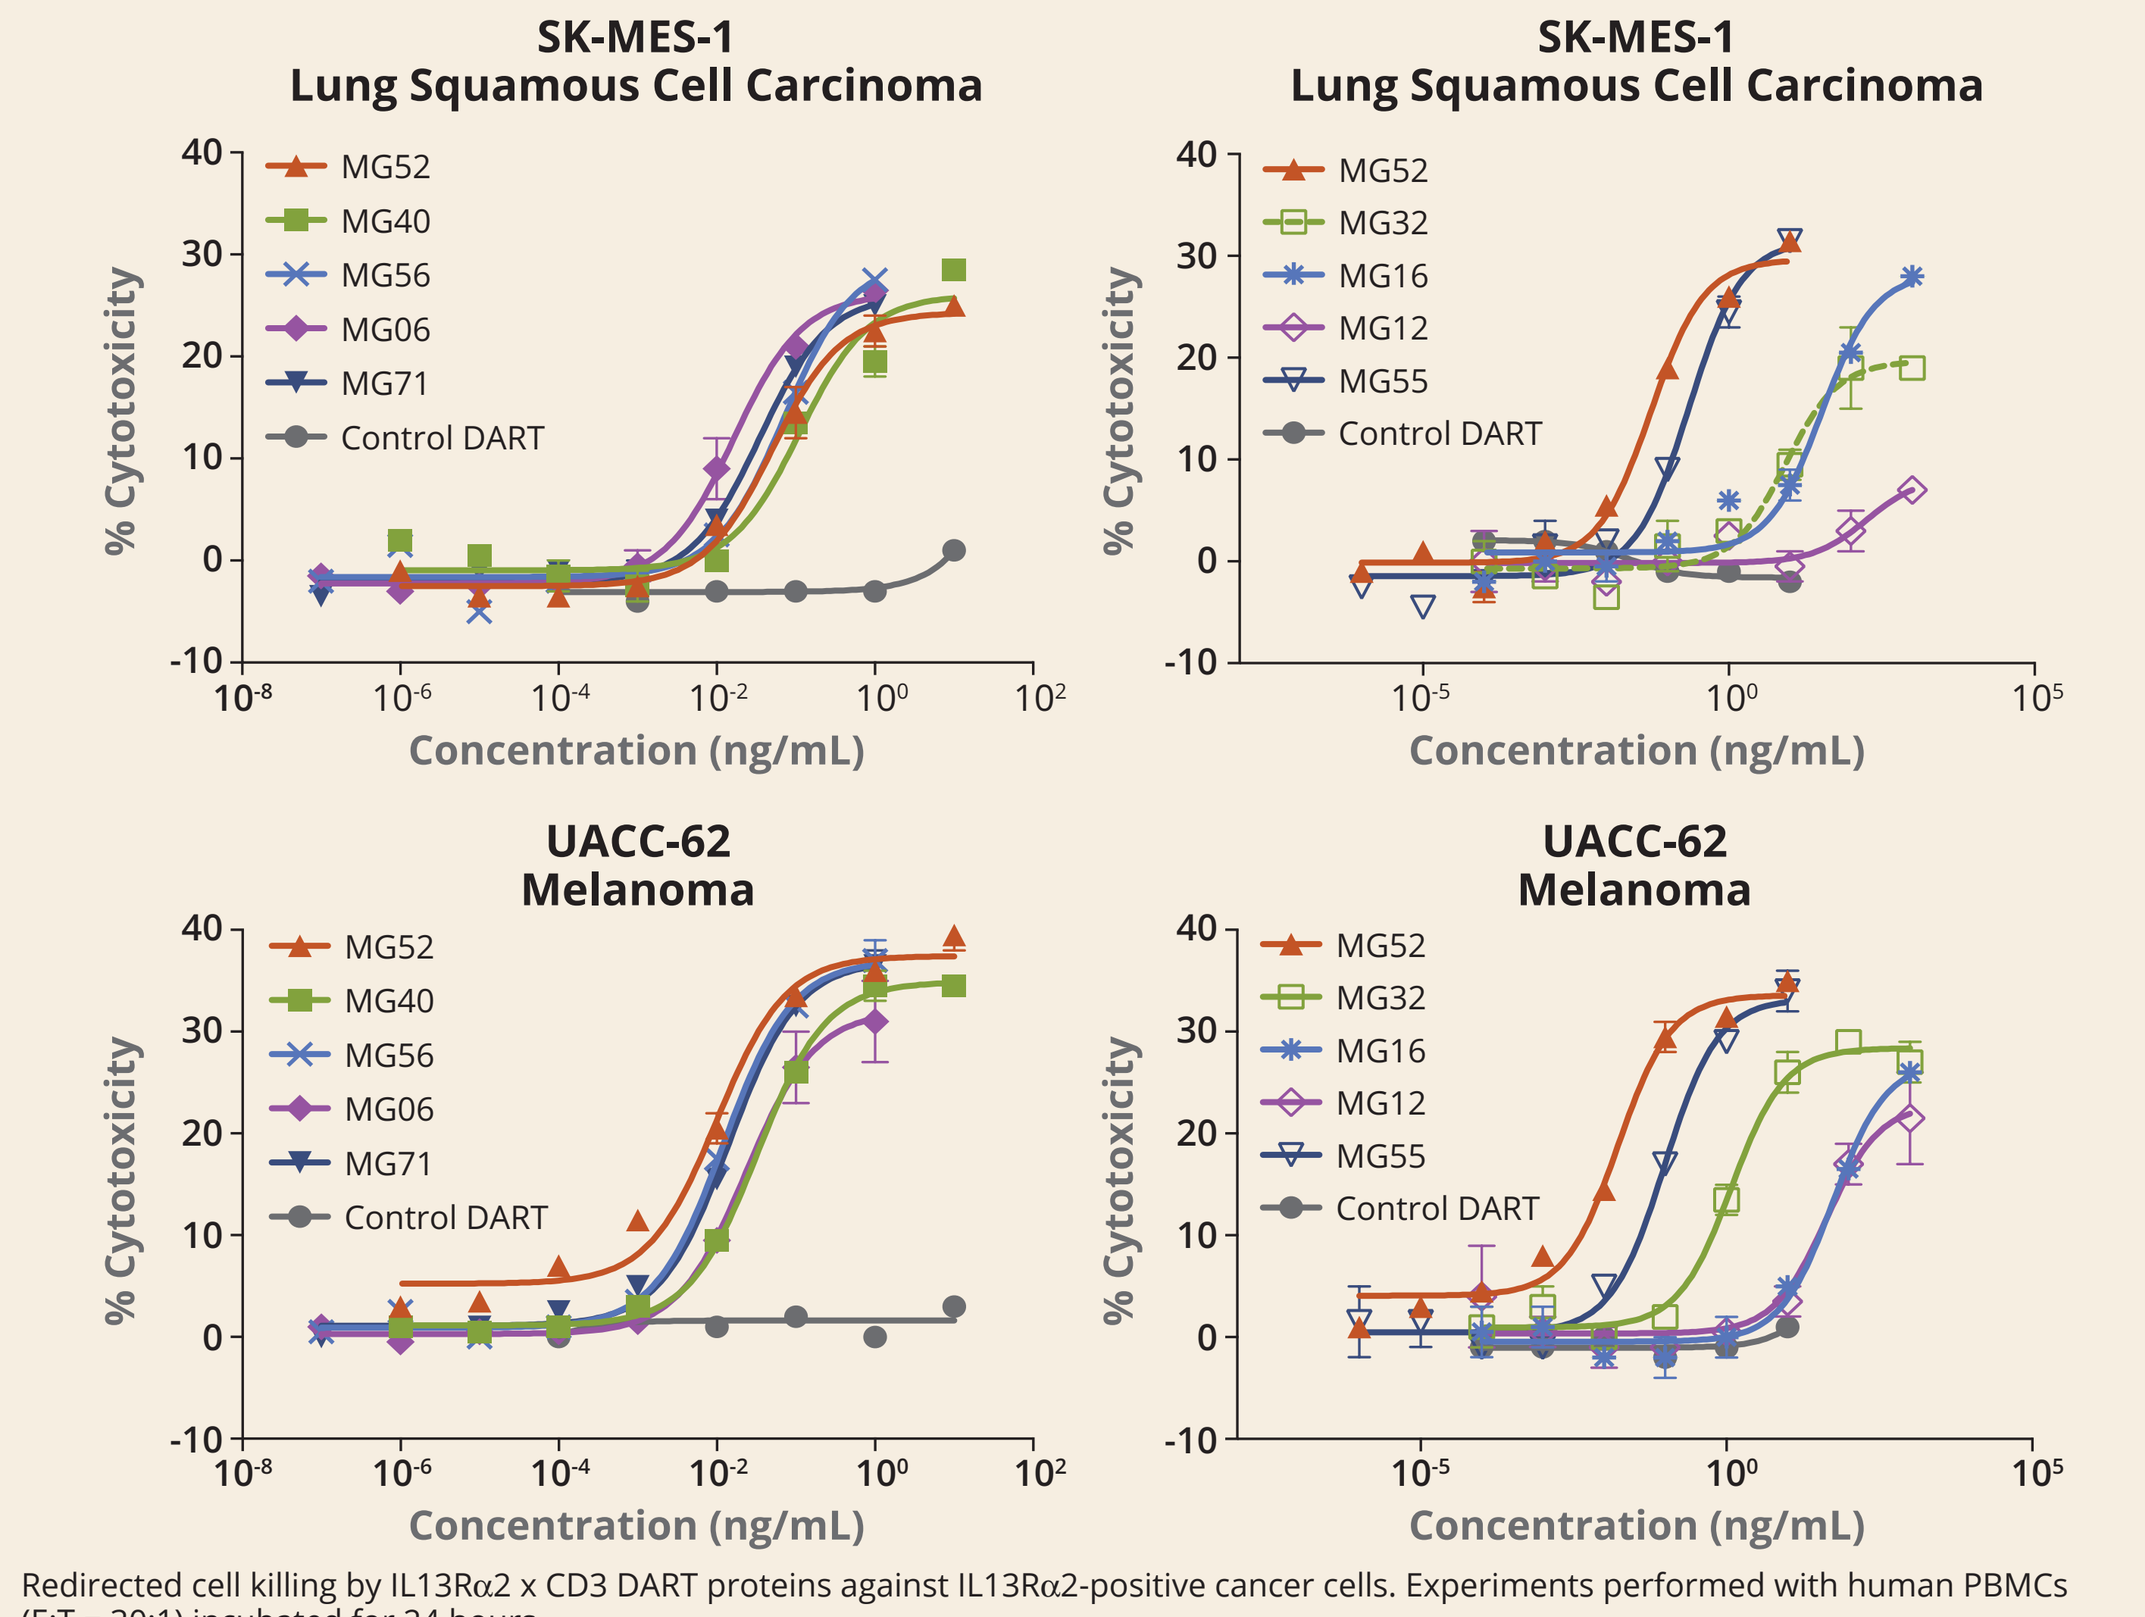
SK-MES-1 Lung Squamous Cell Carcinoma
UACC-62 Melanoma
MG52
MG40
MG56
MG06
MG71
MG32
MG16
MG12
MG55
Control DART
% Cytotoxicity
Concentration (ng/mL)

Redirected cell killing by IL13Rα2 x CD3 DART proteins against IL13Rα2-positive cancer cells. Experiments performed with human PBMCs (E:T = 30:1) incubated for 24 hours.

Humanized Fc-bearing IL13Rα2 x CD3 DART Protein Generation and Biophysical Profile

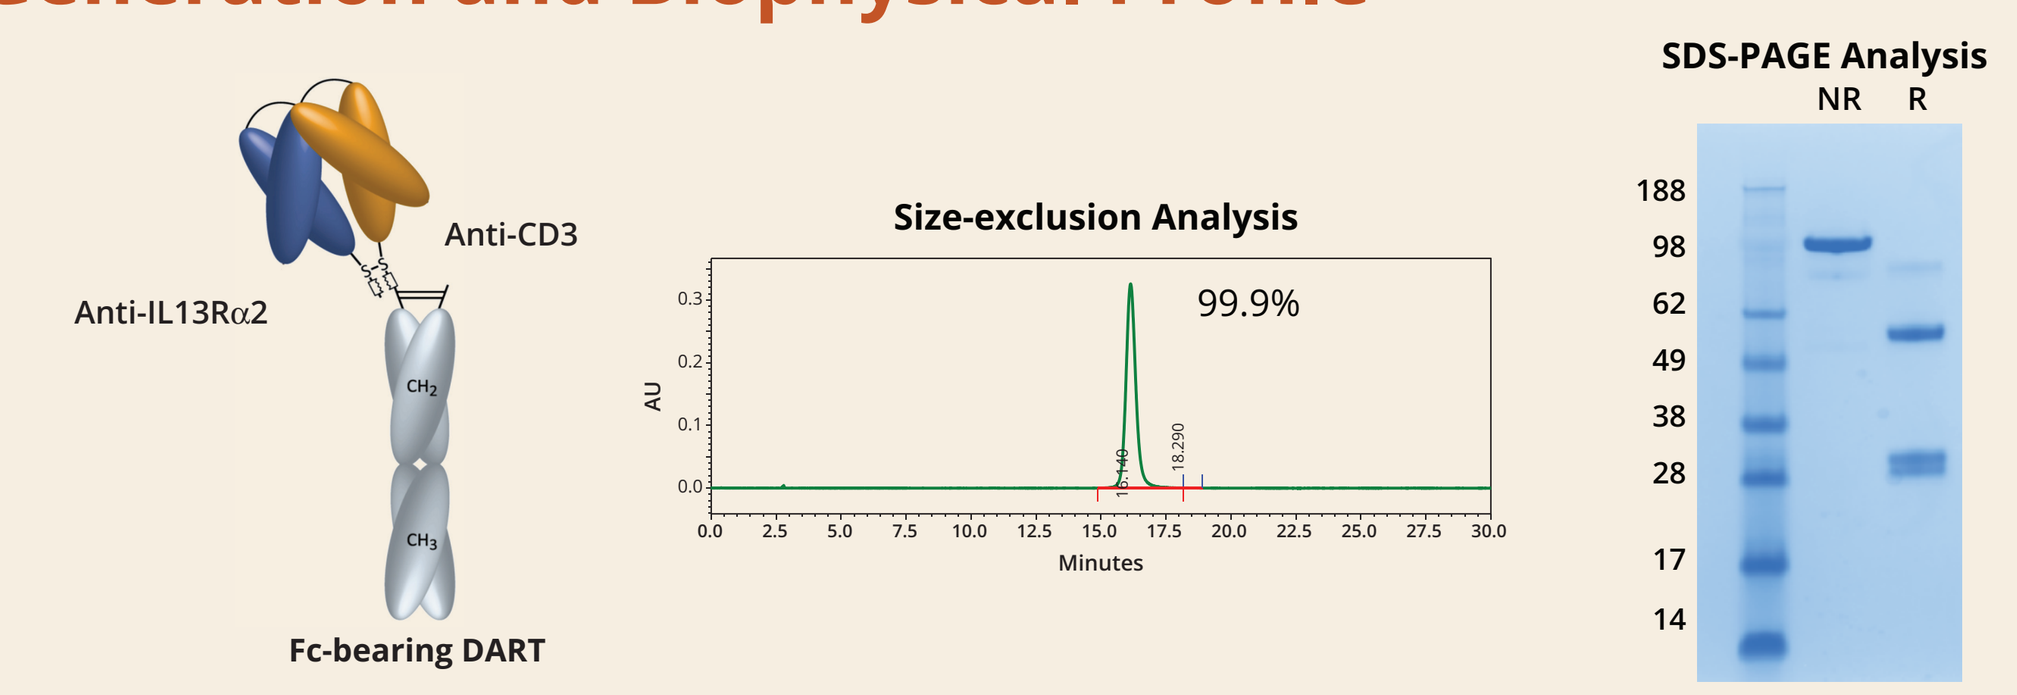
Anti-IL13Rα2
Anti-CD3
Fc-bearing DART
Size-exclusion Analysis
99.9%
Minutes
SDS-PAGE Analysis
NR
R

- Humanized variable domains specific for IL13Rα2 and CD3 were assembled as a disulfide-linked heterodimer with a modified human IgG1 Fc introduced at the terminal end of the DART protein for half-life extension
- The purified humanized IL13Rα2 x CD3 Fc-bearing DART protein was analyzed by SEC-HPLC and SDS-PAGE, evaluated for antigen binding properties by SPR and flow cytometry, as well as for functional activity in vitro and antitumor activity in mouse xenograft models

Binding Properties of a Humanized IL13Rα2 x CD3 Fc-bearing DART Protein

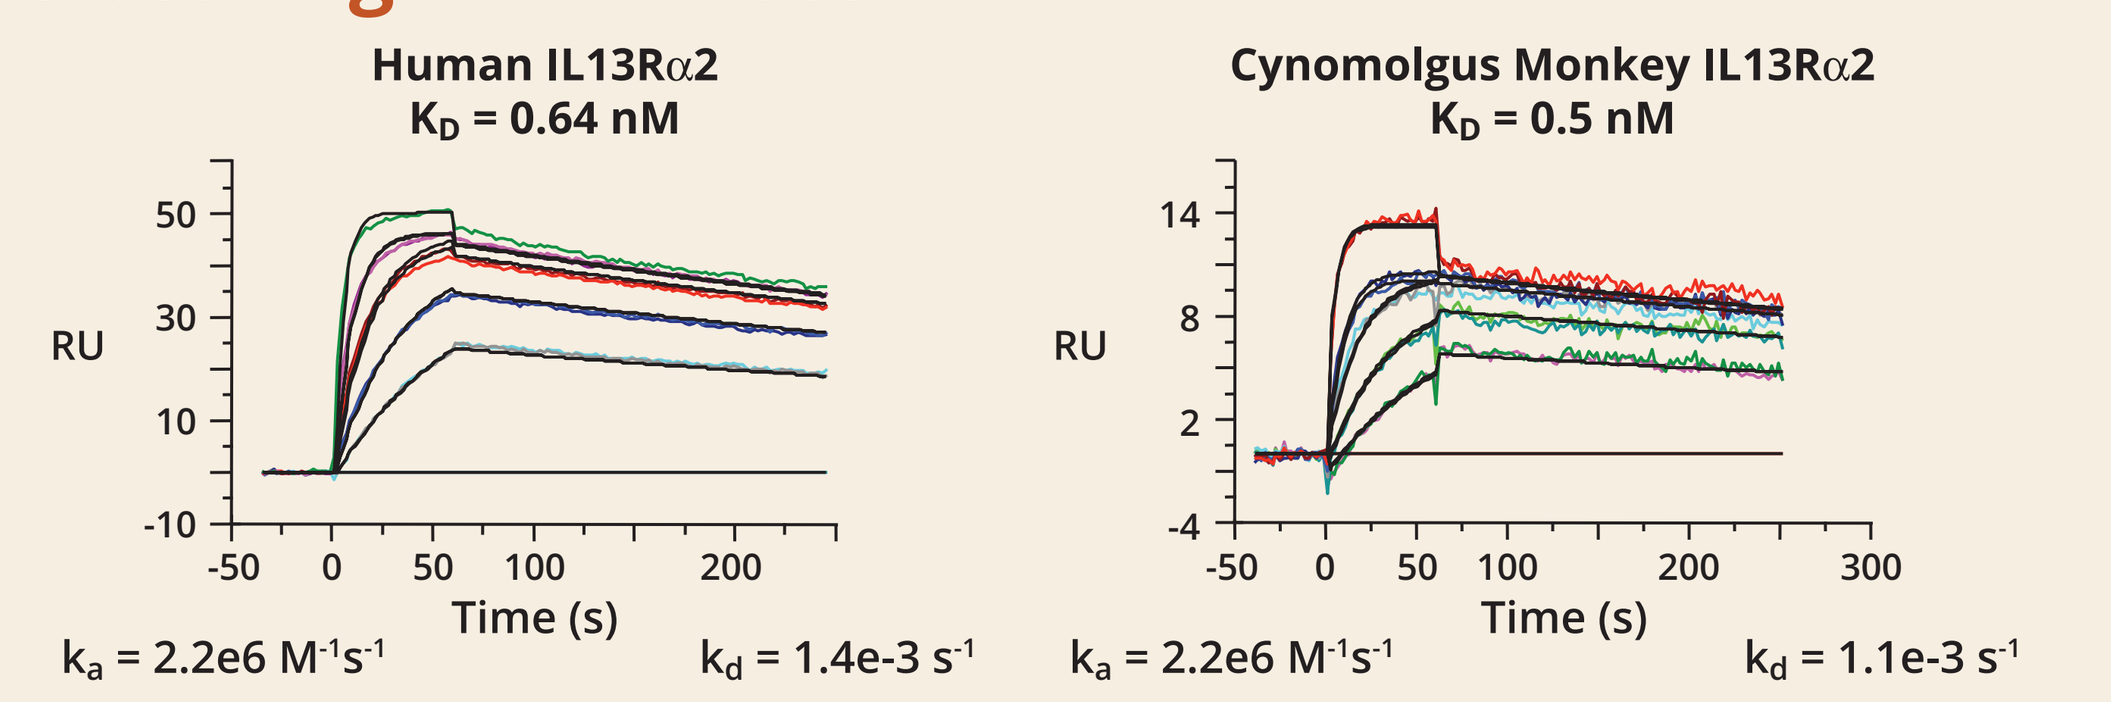
Human IL13Rα2
K_D = 0.64 nM
k_a = 2.2e6 $M^{-1}s^{-1}$
k_d = 1.4e-3 s^{-1}
Cynomolgus Monkey IL13Rα2
K_D = 0.5 nM
k_a = 2.2e6 $M^{-1}s^{-1}$
k_d = 1.1e-3 s^{-1}
RU
Time (s)

- Humanized IL13Rα2 x CD3 Fc-bearing DART protein binds with high affinity to both human and cynomolgus monkey IL13Rα2

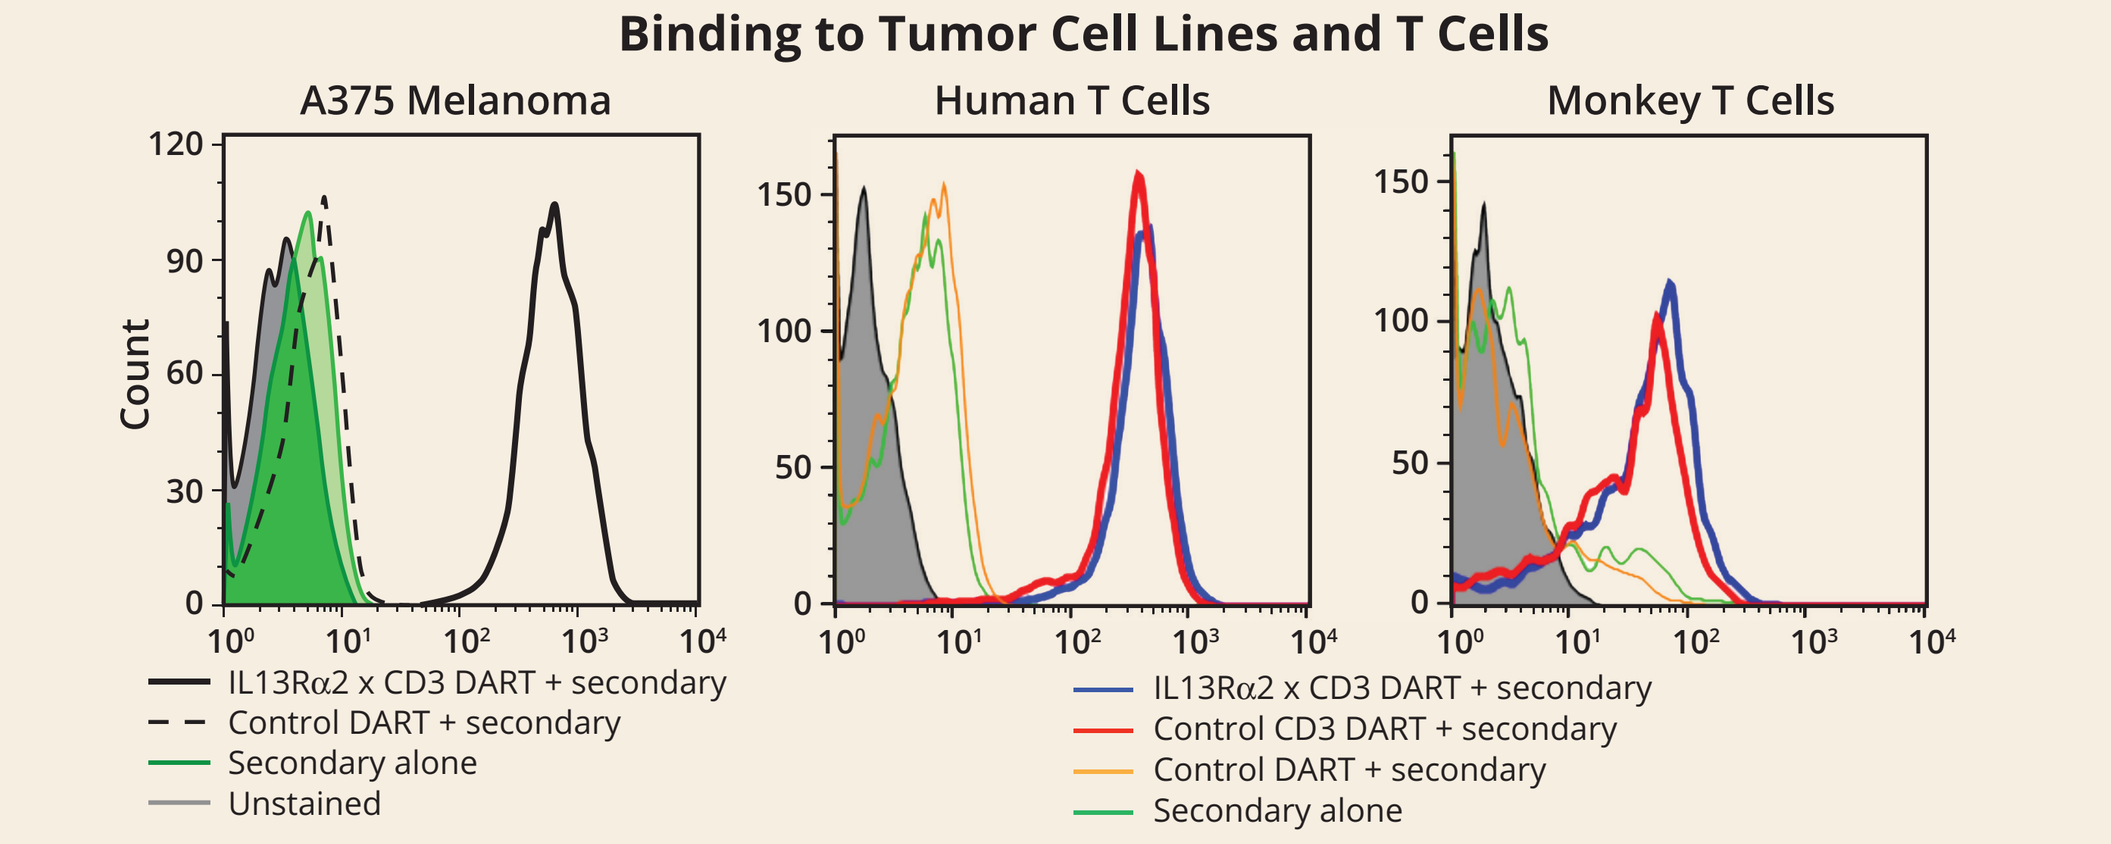
Binding to Tumor Cell Lines and T Cells
A375 Melanoma
Human T Cells
Monkey T Cells
Count
IL13Rα2 x CD3 DART + secondary
Control DART + secondary
Secondary alone
Unstained
Control CD3 DART + secondary

- Humanized IL13Rα2 x CD3 Fc-bearing DART protein binds target-expressing cancer cell lines
- Humanized IL13Rα2 x CD3 Fc-bearing DART protein binds human and cynomolgus monkey T cells

Humanized IL13Rα2 x CD3 Fc-bearing DART Protein Mediates CTL Activity

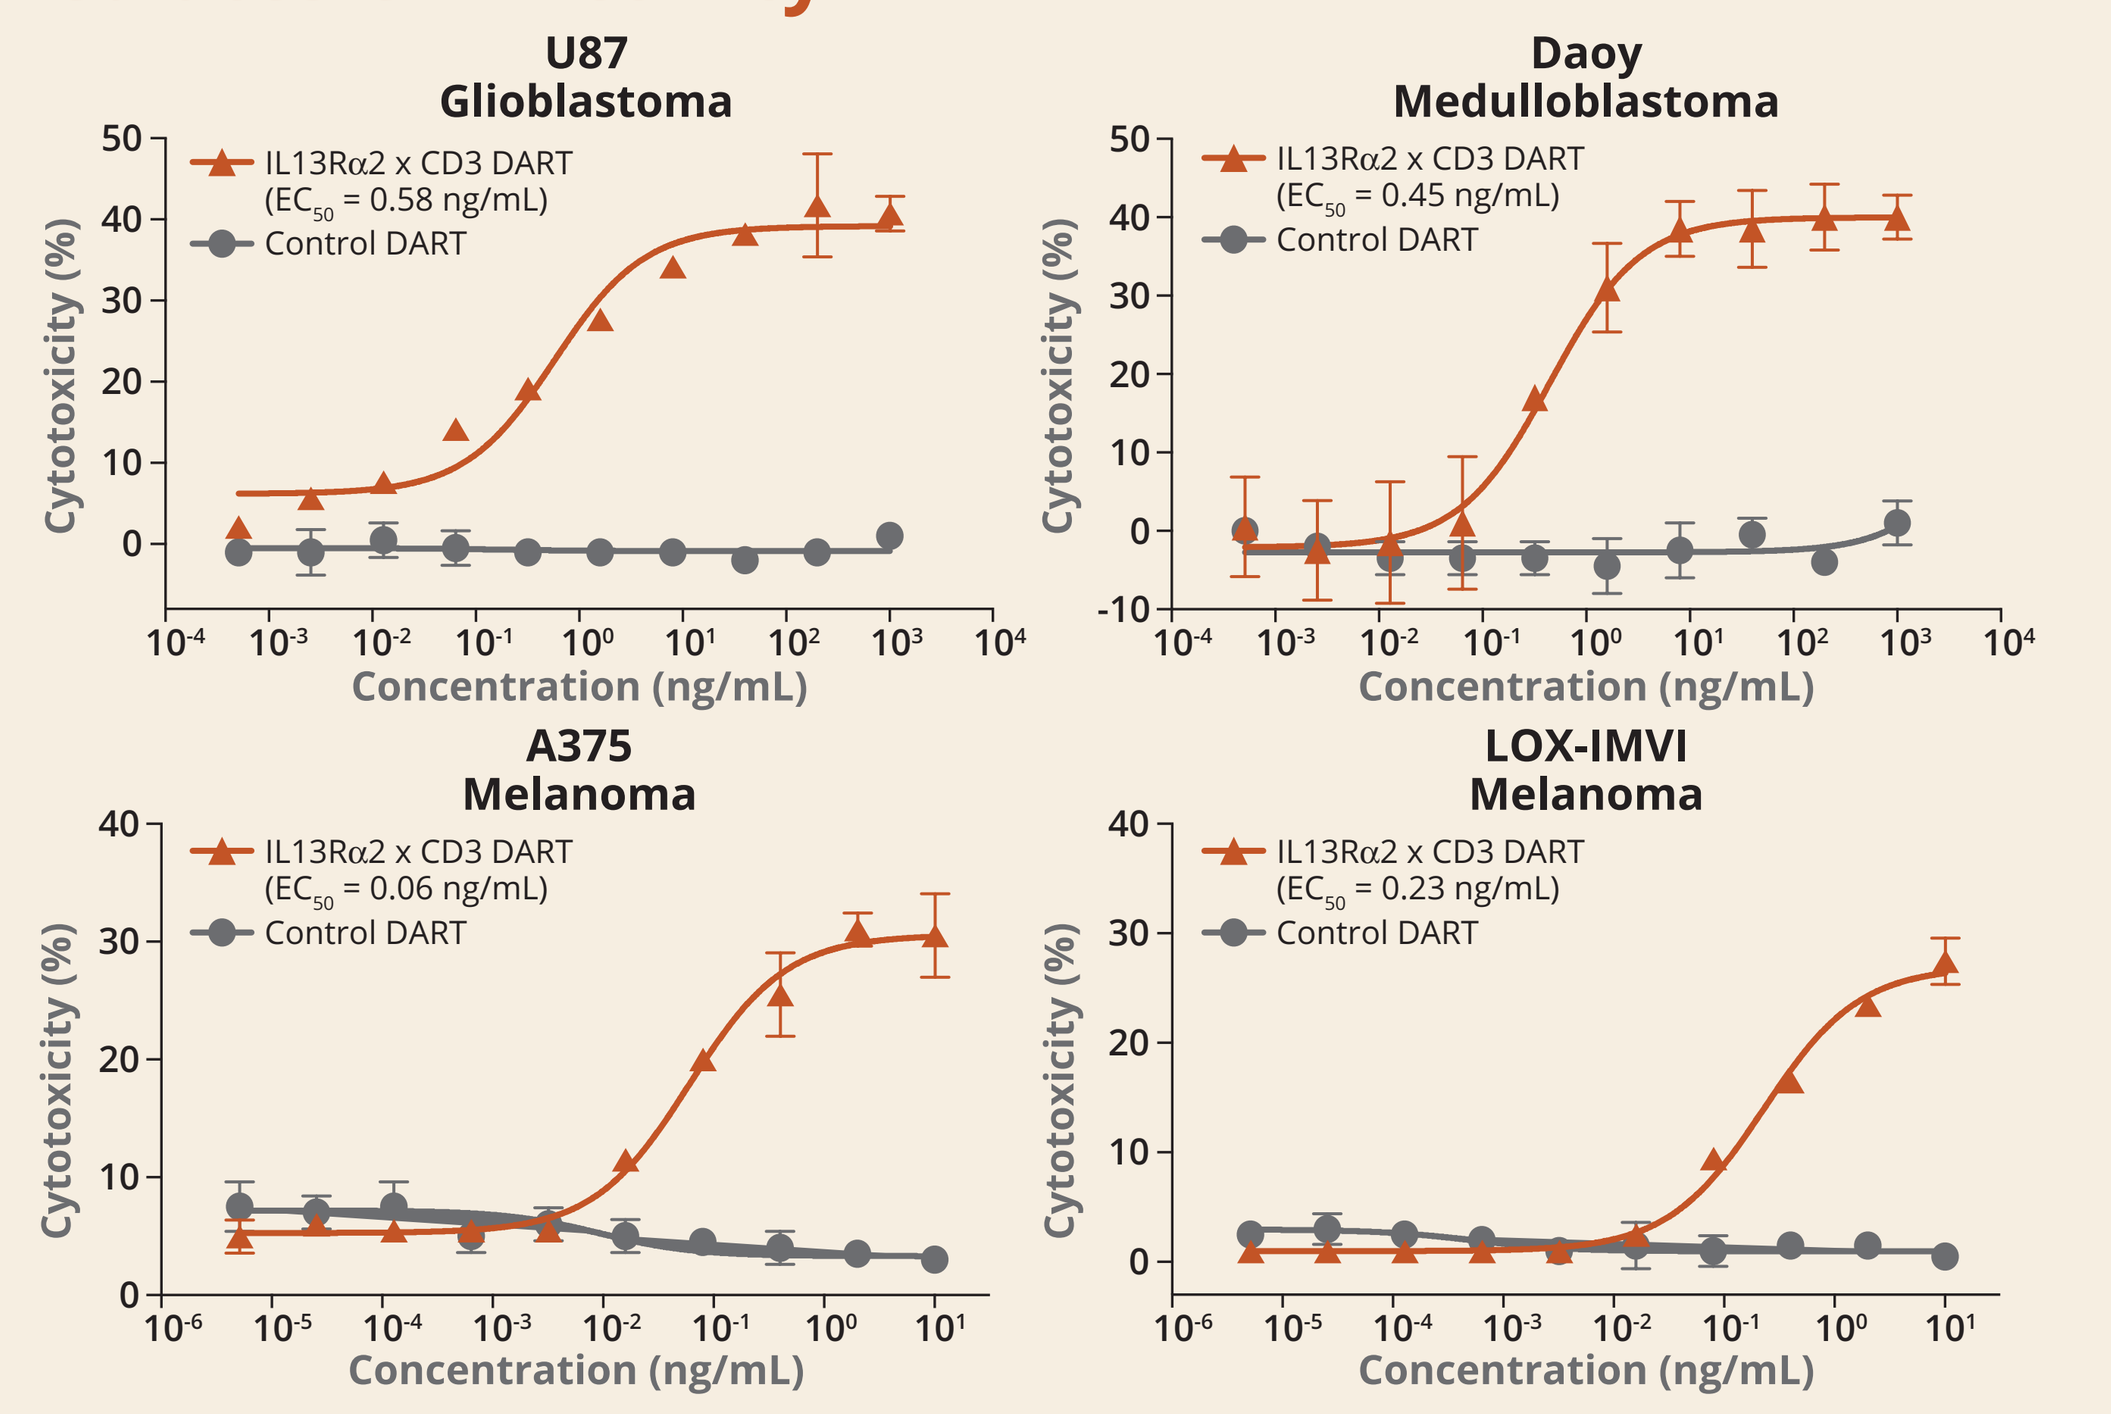
U87 Glioblastoma
IL13Rα2 x CD3 DART (EC_{50} = 0.58 ng/mL)
Daoy Medulloblastoma
IL13Rα2 x CD3 DART (EC_{50} = 0.45 ng/mL)
A375 Melanoma
IL13Rα2 x CD3 DART (EC_{50} = 0.06 ng/mL)
LOX-IMVI Melanoma
IL13Rα2 x CD3 DART (EC_{50} = 0.23 ng/mL)
Control DART
Cytotoxicity (%)
Concentration (ng/mL)

Redirected cell killing by IL13Rα2 x CD3 Fc-bearing DART protein against IL13Rα2-expressing cancer cell lines. Experiments performed with human PBMCs (E:T = 30:1) incubated for 24 hours.

- Humanized IL13Rα2 x CD3 Fc-bearing DART protein exhibits sub-ng/mL (EC_{50}) activity against a panel of target-expressing cancer cell lines

Humanized IL13Rα2 x CD3 Fc-bearing DART Protein Mediates Antitumor Activity In Vivo

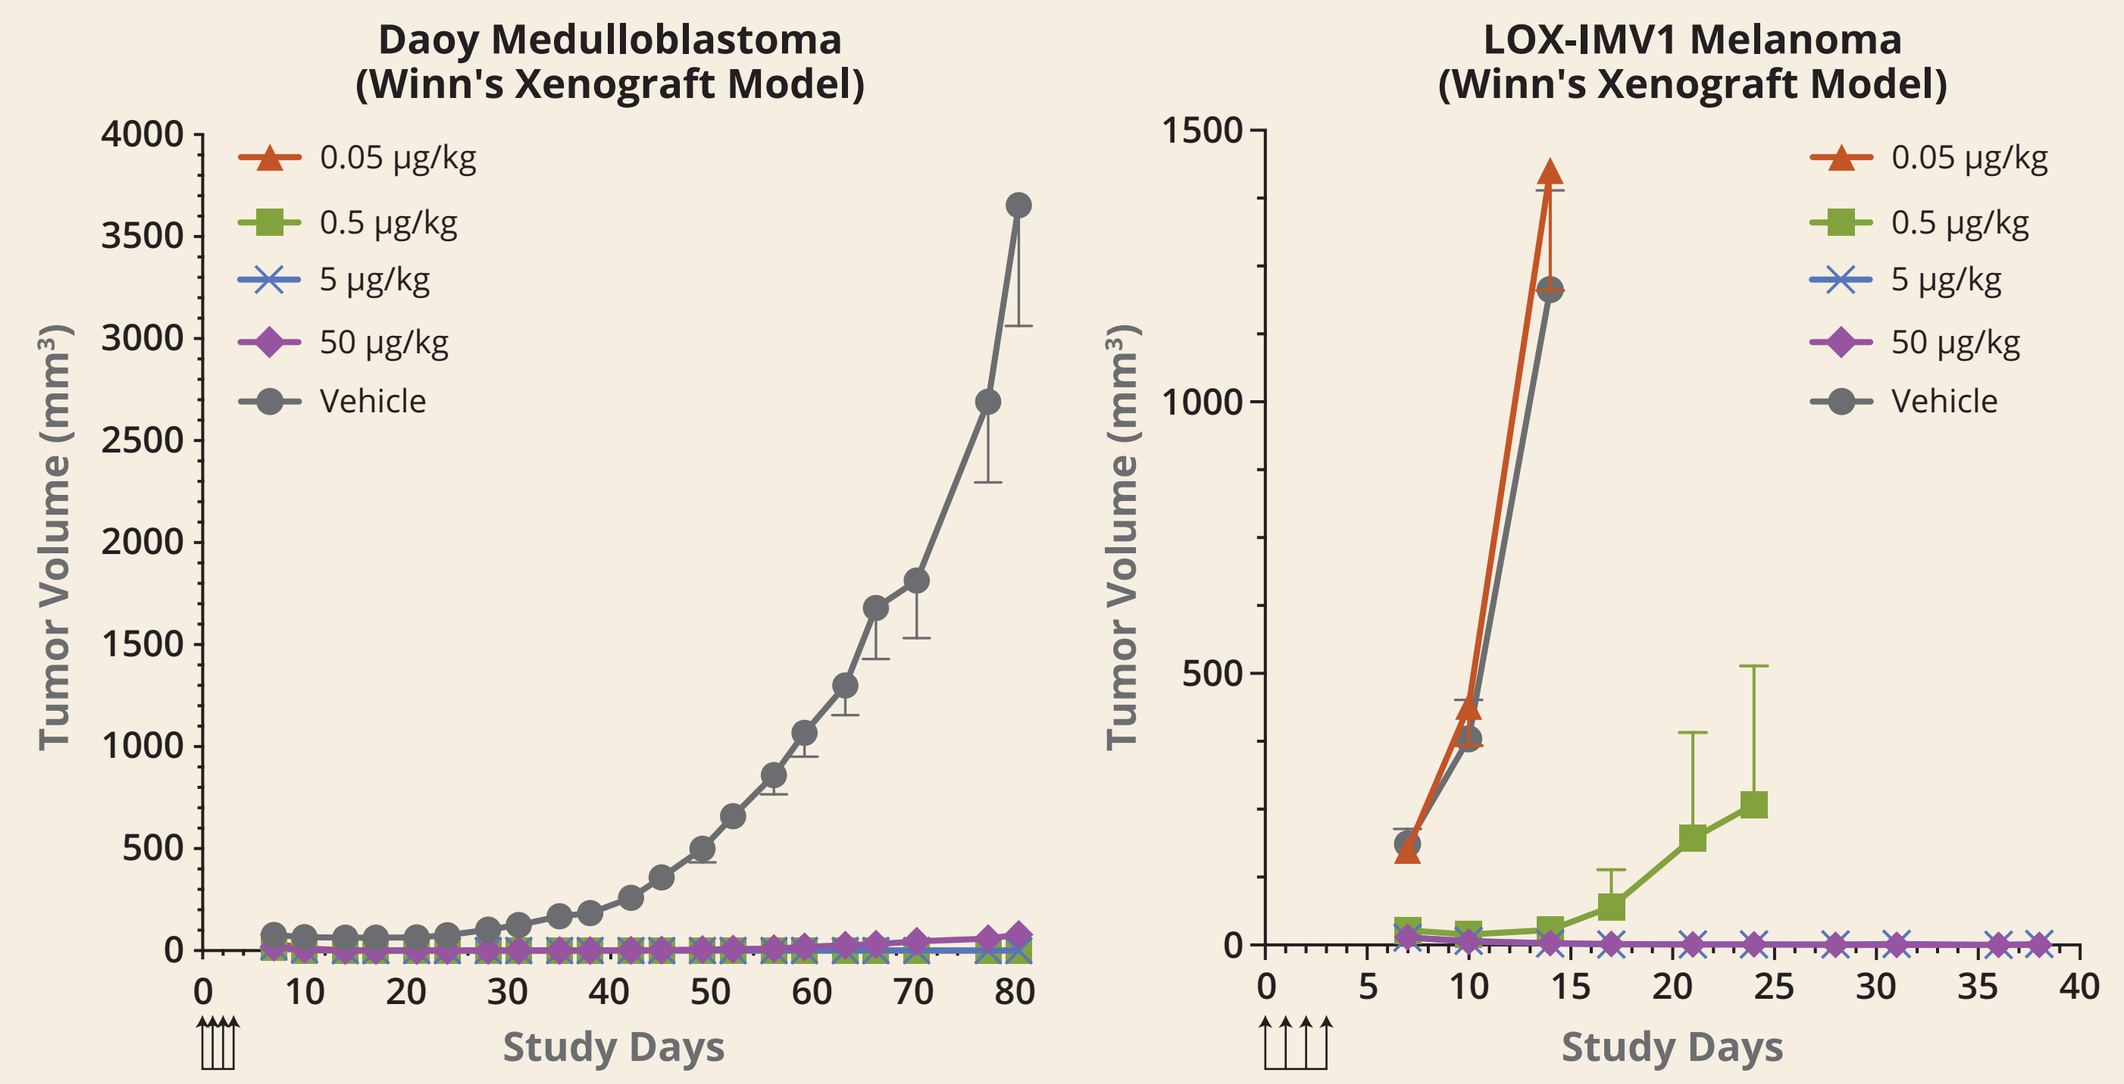
Daoy Medulloblastoma (Winn's Xenograft Model)
LOX-IMV1 Melanoma (Winn's Xenograft Model)
0.05 μg/kg
0.5 μg/kg
5 μg/kg
50 μg/kg
Vehicle
Tumor Volume (mm³)
Study Days

NOG mice (N = 7/group) were implanted SC with activated human T cells and target cells (E:T = 1:5) on Day 0. IL13Rα2 x CD3 DART protein was administered IV once daily for 4 days starting on Day 0 as indicated by arrows.

- Comparable data were observed with other IL13Rα2-expressing cell lines, including U87 glioblastoma and A375 melanoma

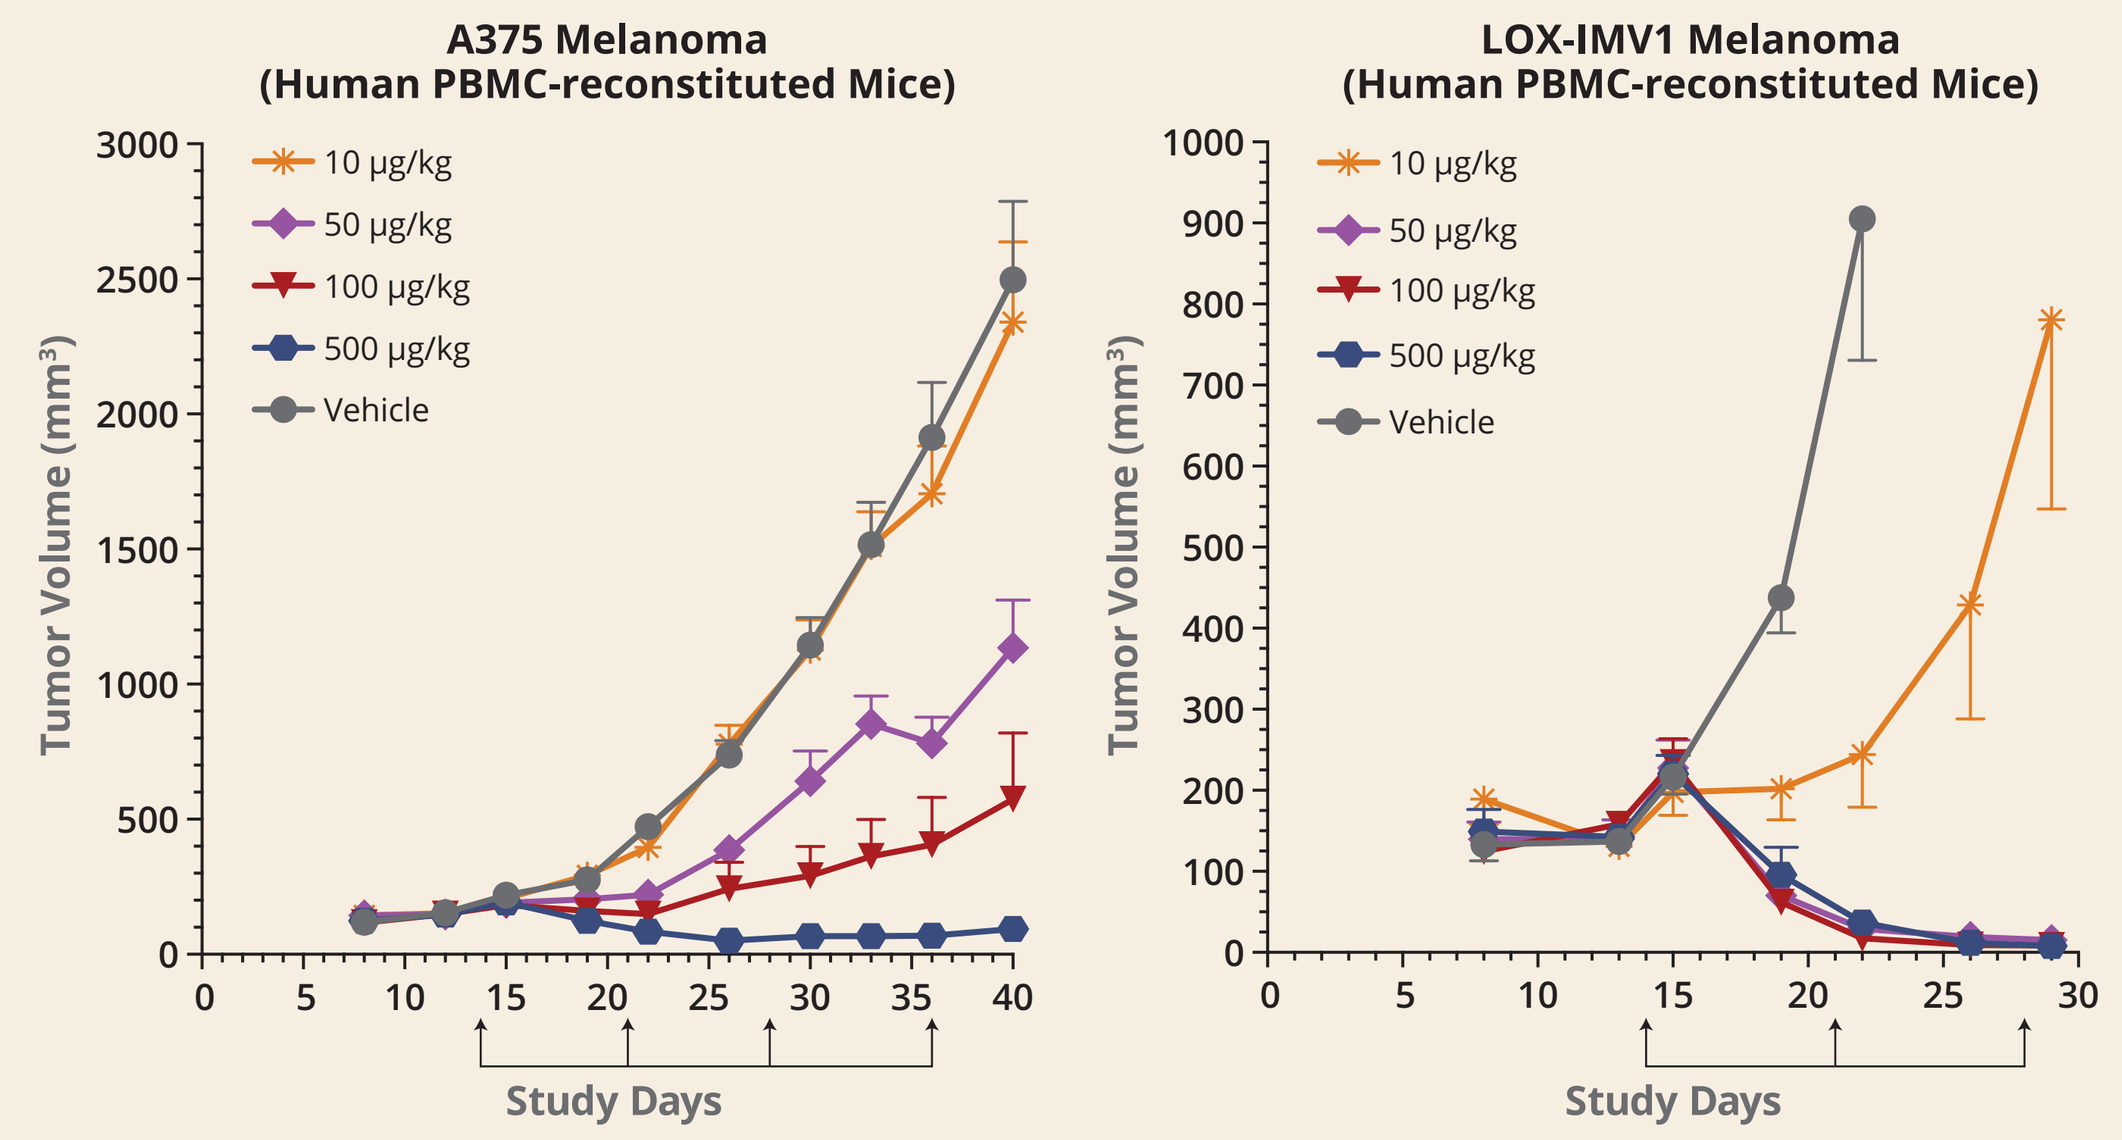
A375 Melanoma (Human PBMC-reconstituted Mice)
LOX-IMV1 Melanoma (Human PBMC-reconstituted Mice)
10 μg/kg
50 μg/kg
100 μg/kg
500 μg/kg
Vehicle
Tumor Volume (mm³)
Study Days

NOG/MHCI-/- mice were implanted with human PBMCs (10 x 10⁶) IP and target cells (5 x 10⁶) ID on Day 0.
Following randomization for A375 model, mice (N = 7/group) were first treated IV on Day 14 and at weekly intervals for a total of 4 doses as indicated by arrows.
For LOX-IMVI model, mice (N = 7/group) were first treated IV on Day 14 and at weekly intervals for a total of 3 doses as indicated by arrows.

Humanized IL13Rα2 x CD3 Fc-bearing DART Protein Mediates Target-dependent Cytokine Release

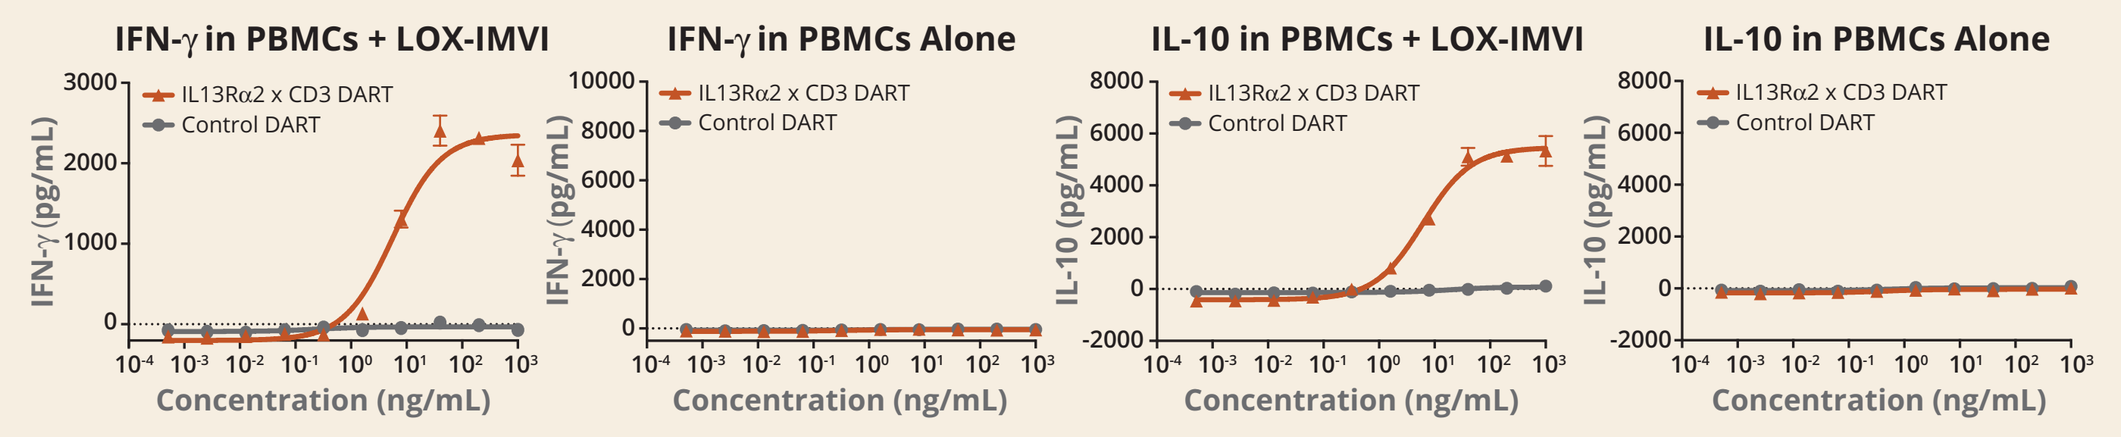
IFN-γ in PBMCs + LOX-IMVI
IFN-γ in PBMCs Alone
IL-10 in PBMCs + LOX-IMVI
IL-10 in PBMCs Alone
IL13Rα2 x CD3 DART
Control DART
Concentration (ng/mL)

Human PBMCs were incubated for 24 hours with increasing DART protein concentrations in the presence or absence of LOX-IMVI target cells (E:T = 30:1). At the end of incubation, cytokine release was measured by ELISA. Shown are data for two representative cytokines.

- No DART protein-mediated cytokine release was observed in the absence of target cells

Conclusions

- Antibodies generated against the human IL13Rα2 extracellular domain demonstrate minimal reactivity with normal human tissues and high-tumor reactivity, including melanoma and glioblastoma
- Screening of IL13Rα2 x CD3 DART proteins identified a subset with potent in vitro CD3-mediated killing responses
- A lead IL13Rα2 mAb was humanized and engineered as an IL13Rα2 x CD3 Fc-bearing DART molecule that displays high affinity binding to target antigens, cynomolgus monkey cross-reactivity, and potent antitumor activity in vitro and in vivo

These data support further development of IL13Rα2 x CD3 DART protein as a potential therapeutic for cancer

Presented at the 2016 American Association for Cancer Research Annual Meeting, April 16–20, 2016, New Orleans, Louisiana

©2016 MacroGenics, Inc. All rights reserved.