Co-targeting of PD-1 and CTLA-4 Inhibitory Pathways with Bispecific DART® and TRIDENT™ Molecules

Gurunadh R. Chichili, Kalpana Shah, Ross La Motte-Mohs, Alex Berezhnoy, Daorong Liu, Jessica Hill, Leilei He, Christine Shoemaker, Jennifer G. Brown, Kurt Stahl, Hua Li, Jonathan Li, Sergey Gorlatov, Tim Gaynutdinov, James Tamura, Valentina Ciccarone, Ralph Alderson, Ezio Bonvini, Paul Moore, Syd Johnson

MacroGenics, Inc., Rockville, MD and South San Francisco, CA

3637

http://ir.macrogenics.com/events.cfm

Abstract

Introduction: Immunotherapy with the combination of monoclonal antibodies (mAbs) that block PD-1 (nivolumab) and CTLA-4 (ipilimumab) has shown clinical benefit beyond that observed with either mAb alone. We have developed PD-1 x CTLA-4 bispecific proteins aimed at inducing antitumor immunity through simultaneous blockade of both checkpoint molecules. Two proteins, a tetravalent PD-1 x CTLA-4 bispecific DART® protein (bivalent for both PD-1 and CTLA-4) and a trivalent PD-1 x CTLA-4 TRIDENT™ protein (bivalent for PD-1 but monovalent for CTLA-4) were engineered; the TRIDENT protein was designed to promote CTLA-4 blockade through avidity when bound to PD-1 on cells coexpressing both receptors, thus favoring PD-1 over CTLA-4 blockade in cells that do not coexpress both molecules.

Methods: Proteins were engineered from humanized PD-1 and CTLA-4 mAb sequences and demonstrated favorable biophysical properties. Binding assays were performed by ELISA or flow cytometry. Coligation of PD-1 and CTLA-4 was assessed in an enzyme-fragment complementation assay (DiscoverX). T-cell activation was tested in reporter cells, staphylococcus enterotoxin B-stimulated PBMCs or MLR assays.

Results: Both DART and TRIDENT proteins showed equivalent potency in binding immobilized recombinant PD-1 or PD-1-expressing cell lines, inhibition of PD-1 interaction with PD-L1 or PD-L2 as well as reversal of PD-1/PD-L1 mediated T-cell signal inhibition. In all assays, both formats showed activities that were comparable to those of the precursor PD-1 mAb. With respect to CTLA-4, the DART protein showed a minor potency loss in binding to CTLA-4 expressing cells, inhibition of CTLA-4/B7 interaction and reversal of T-cell signal inhibition compared to the precursor mAb. The TRIDENT protein, however, showed substantial lower potency than the DART protein in all CTLA-4 assays, consistent with the monovalent nature of the CTLA-4 arm. Importantly, in cells that coexpress both receptors, DART and TRIDENT proteins show comparable co-engagement of PD-1 and CTLA-4, as shown by enzyme-fragment complementation, suggesting that anchoring through PD-1 compensates for the decreased CTLA-4 avidity of the TRIDENT molecule when both target receptors are expressed. Similarly, in T-cell coactivation assays, both DART and TRIDENT proteins showed a comparable increase in IFN-γ response that recapitulated that observed with the combination of the individual parental mAbs.

Conclusions: Both PD-1 x CTLA-4 DART and TRIDENT bispecific molecules block PD-1 and CTLA-4 pathways, with the TRIDENT protein demonstrating a PD-1-biased binding preference, consistent with its design intended to reduce CTLA-4 blockade in the absence of PD-1 coexpression. Both molecules showed comparable T-cell activation activity. Further development of bispecific PD-1 x CTLA-4 molecules for cancer treatment is warranted.

Introduction

PD-1 x CTLA-4 Dual Checkpoint Targeting Strategy

- PD-1 and CTLA-4 are clinically validated T-cell co-inhibitory molecules with complementary mechanisms of action
- Coordinated PD-1 and CTLA-4 blockade has shown combinatorial antitumor activity in the clinic
 - PD-1 blockade reverses T-cell inhibition at tumor sites
 - CTLA-4 blockade results in polyclonal activation/expansion
- Increased antitumor activity with anti-CTLA-4 addition is associated with increased adverse event profile
- Challenge: Maintain PD-1 ligand blockade with tunable CTLA-4 binding blockade

PD-1 x CTLA-4 DART
Bivalent PD-1 & CTLA-4 arms
Equivalent PD-1 & CTLA-4 Blockade

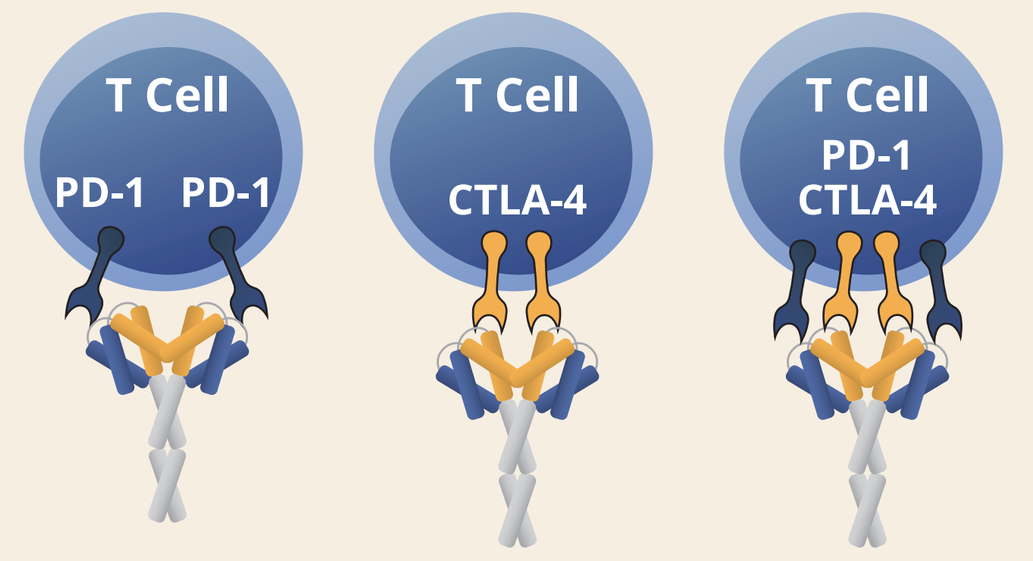
T Cell
PD-1 PD-1
T Cell
CTLA-4
T Cell
PD-1
CTLA-4

PD-1 x CTLA-4 TRIDENT
Bivalent PD-1 & monovalent CTLA-4 arms
CTLA-4 Blockade Biased Toward PD-1 Coexpression

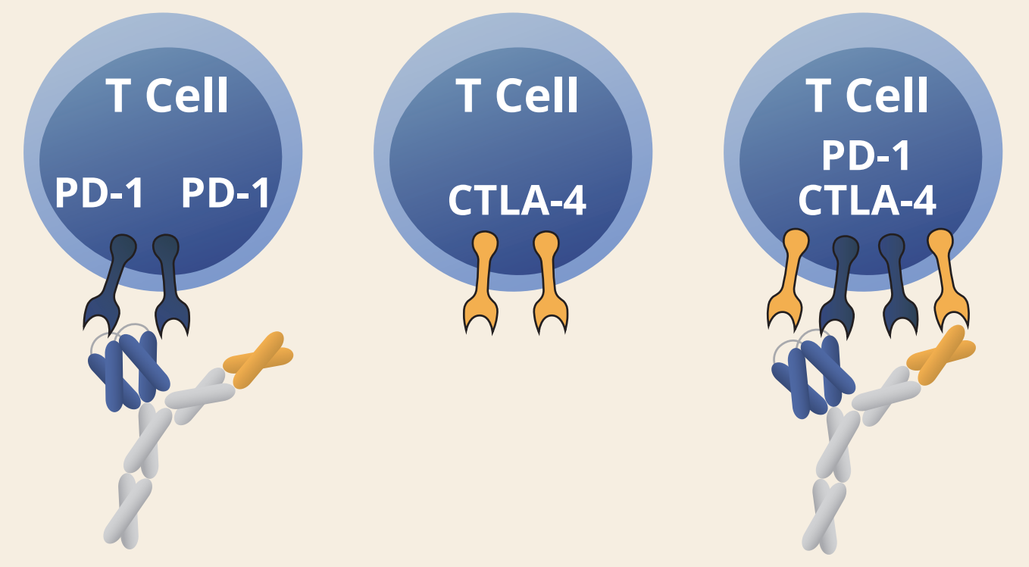
T Cell
PD-1 PD-1
T Cell
CTLA-4
T Cell
PD-1
CTLA-4

Bispecific PD-1 x CTLA-4 Checkpoint Inhibitor Molecules

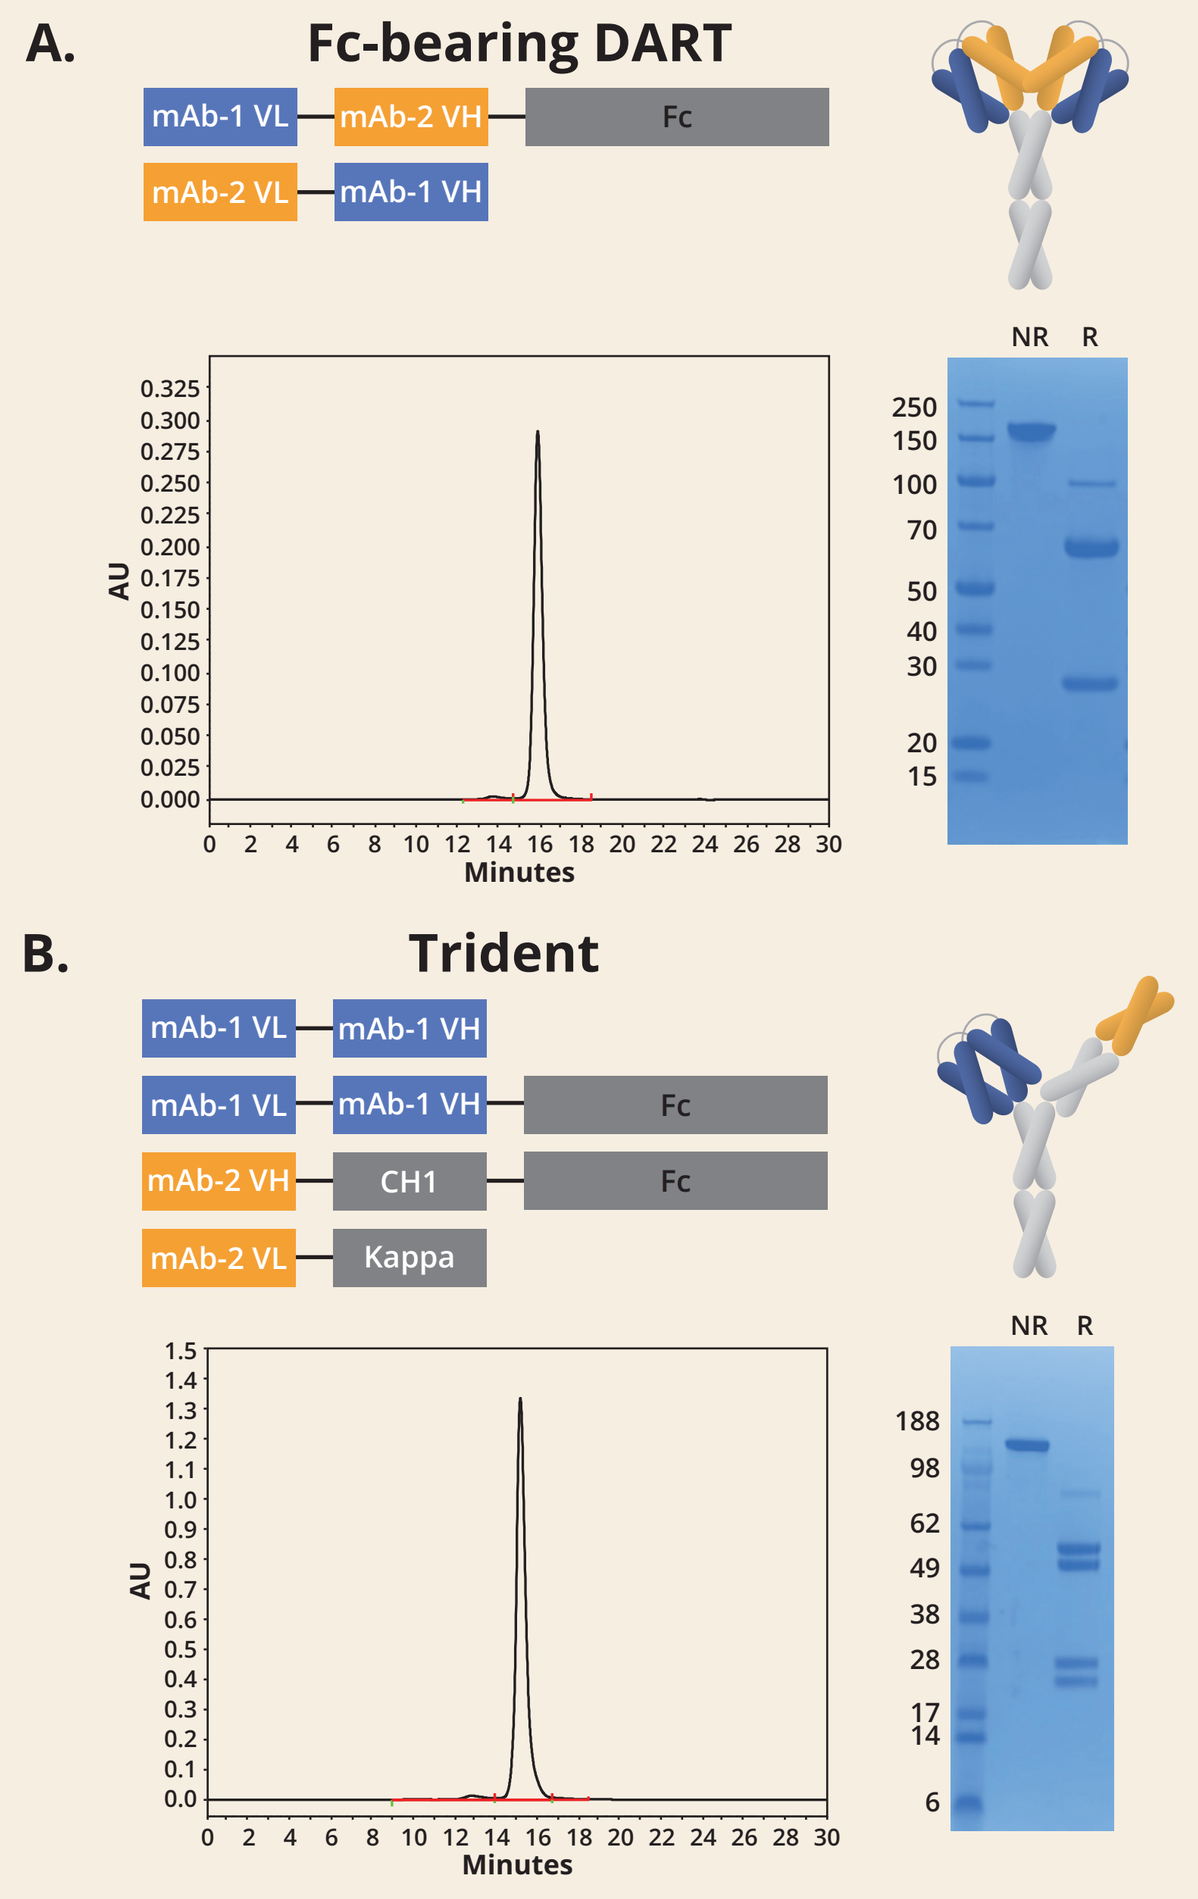
A.
Fc-bearing DART
mAb-1 VL
mAb-2 VH
Fc
mAb-2 VL
mAb-1 VH
AU
Minutes
NR
R
B.
Trident
mAb-1 VL
mAb-1 VH
mAb-1 VL
mAb-1 VH
Fc
mAb-2 VH
CH1
Fc
mAb-2 VL
Kappa

PD-1 mAb (proprietary mAb, MGA012) binding affinity ~1 nM; CTLA-4 mAb binding affinity ~5 nM.

A. PD-1 x CTLA-4 DART is an Fc-bearing (IgG4) DART protein bispecific and bivalent for PD-1 and CTLA-4. Size-exclusion chromatography (bottom left) and SDS-PAGE (bottom right).
B. PD-1 x CTLA-4 TRIDENT is designed with the same mAb sequences as in the DART molecule, but contains monovalent CTLA-4 binding arm. Size-exclusion chromatography (bottom left) and SDS-PAGE (bottom right).

Results

PD-1 x CTLA-4 DART and TRIDENT Binding Properties

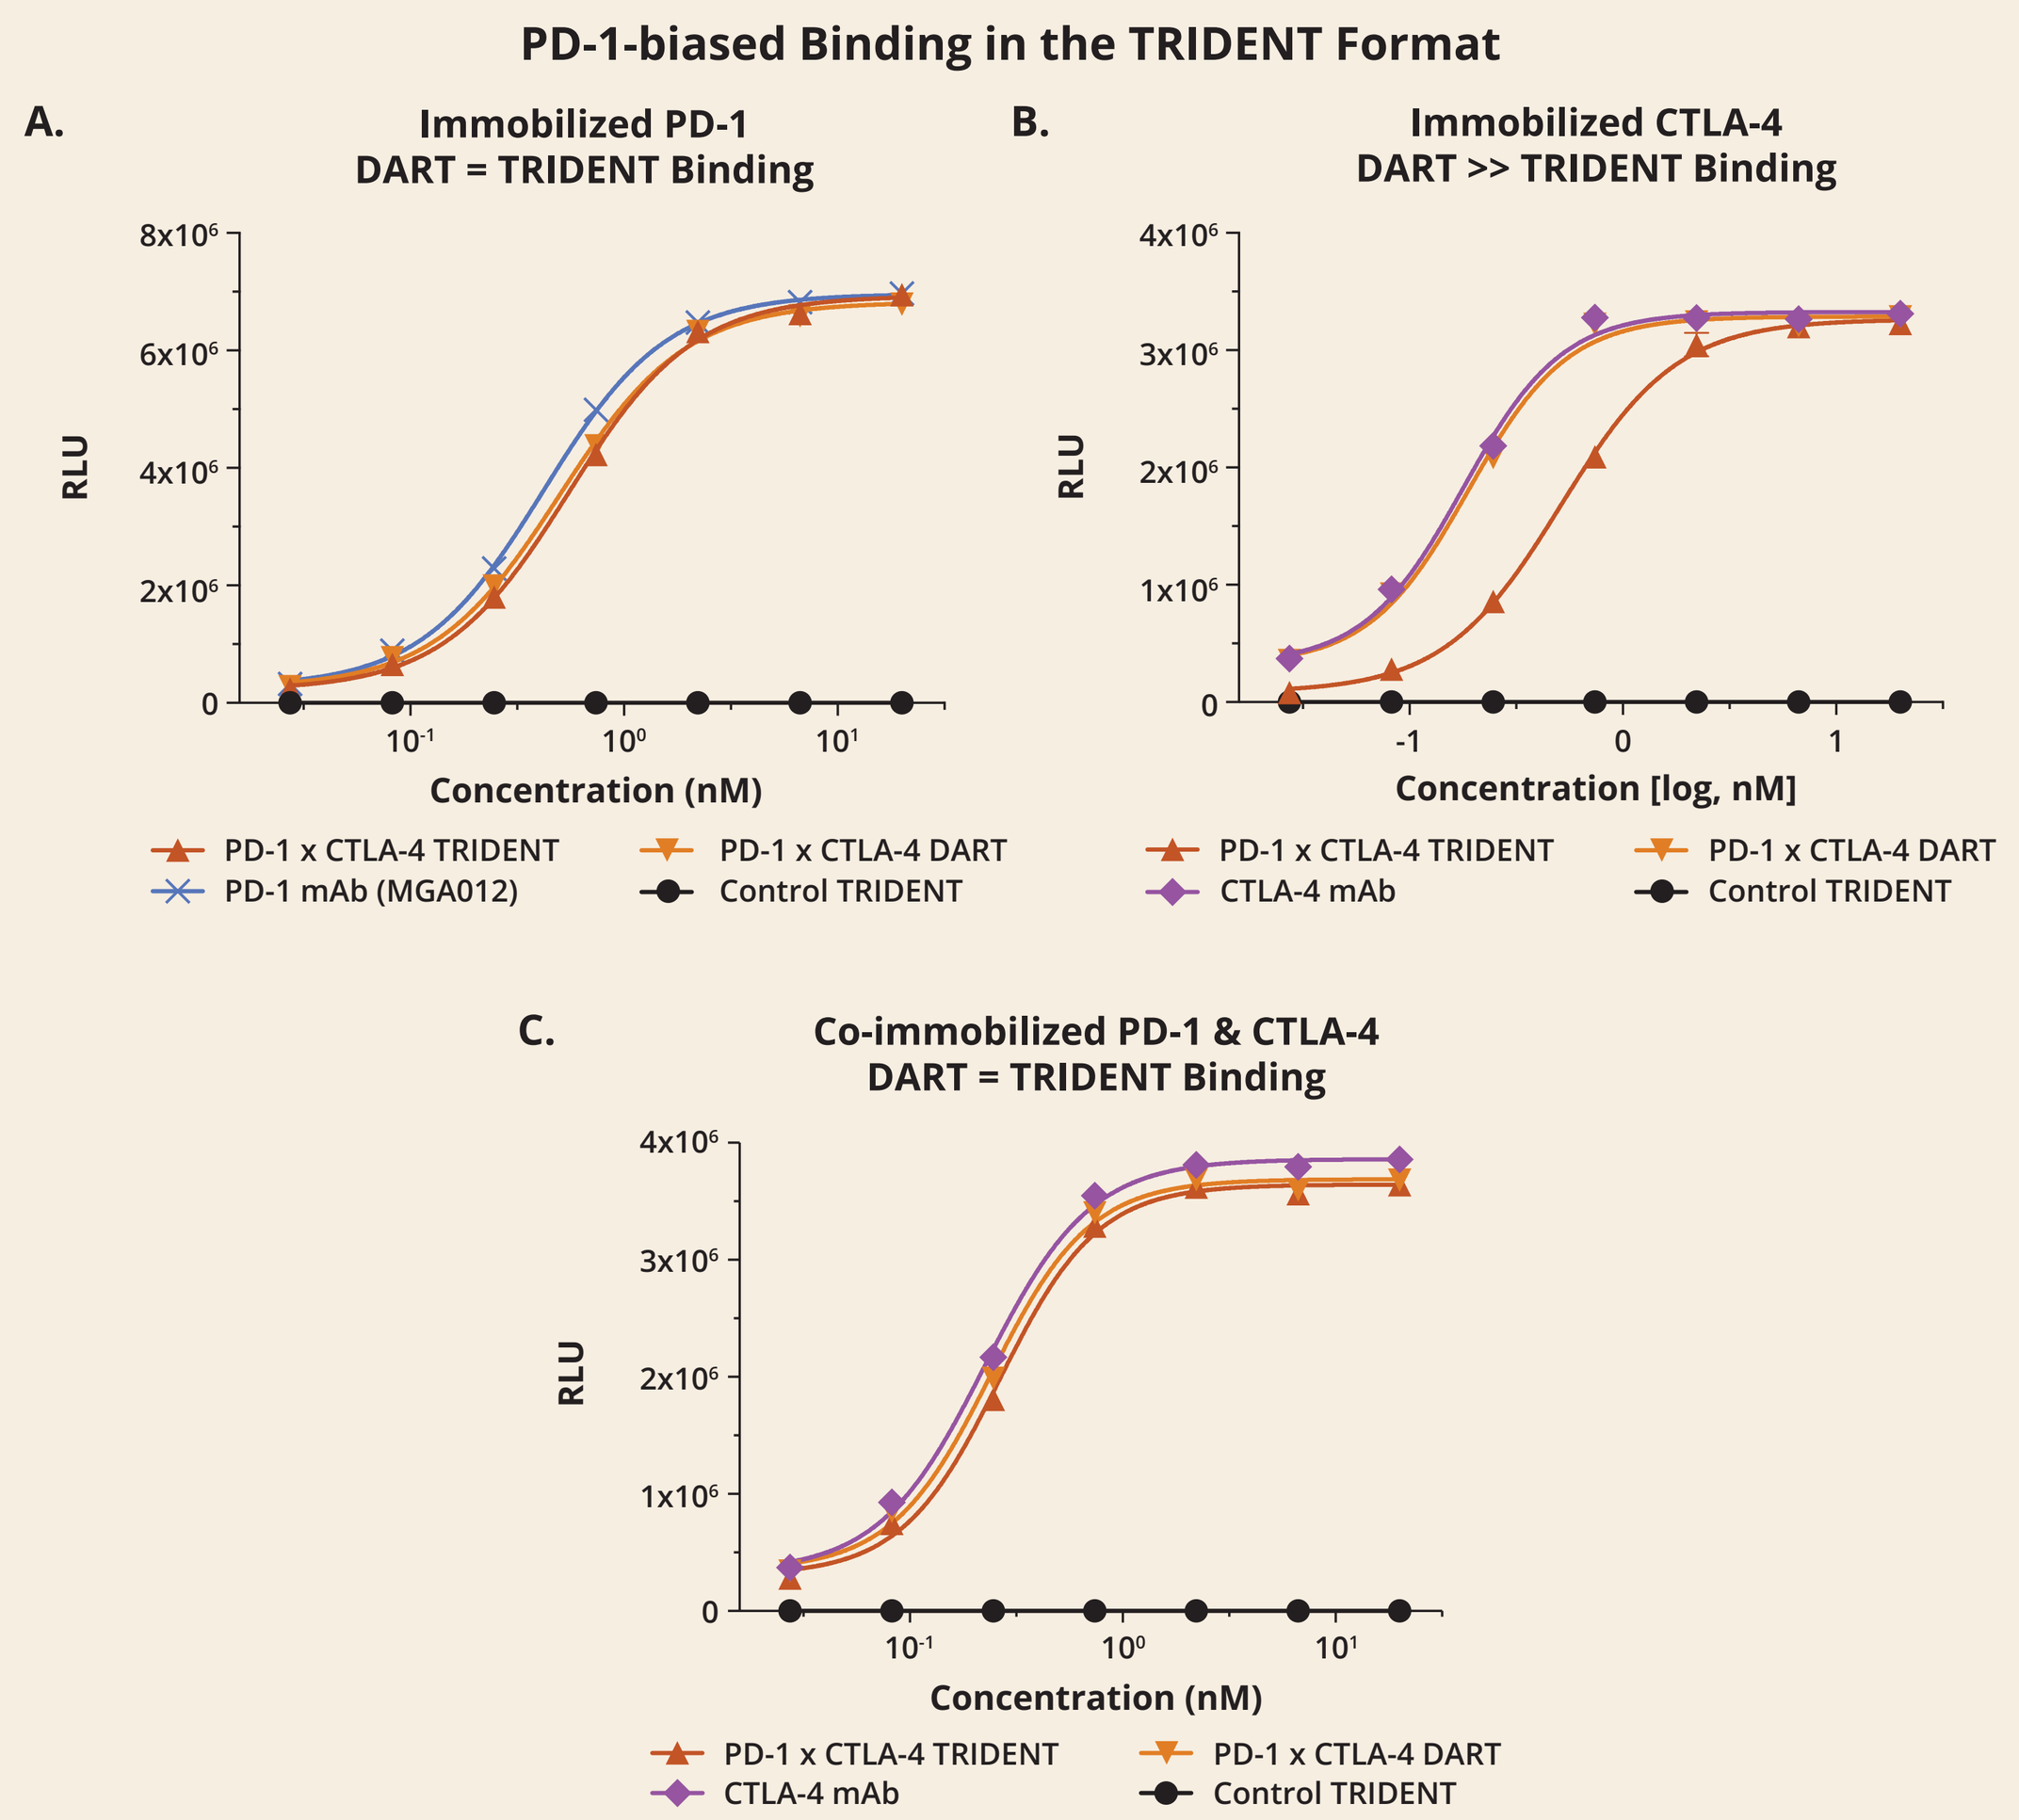
PD-1-biased Binding in the TRIDENT Format
A. Immobilized PD-1
DART = TRIDENT Binding
B. Immobilized CTLA-4
DART >> TRIDENT Binding
C. Co-immobilized PD-1 & CTLA-4
DART = TRIDENT Binding
RLU
Concentration (nM)
Concentration [log, nM]
PD-1 x CTLA-4 TRIDENT
PD-1 x CTLA-4 DART
PD-1 mAb (MGA012)
CTLA-4 mAb
Control TRIDENT

A-B. ELISA-based binding of PD-1 x CTLA-4 DART and TRIDENT proteins to plate-immobilized recombinant human PD-1 **(A)** or human CTLA-4 **(B)** proteins detected by anti-human IgG HRP.
C. Binding of PD-1 x CTLA-4 DART and TRIDENT molecules to co-immobilized PD-1 and CTLA-4 detected by ELISA.

PD-1 x CTLA-4 DART and TRIDENT Molecules Bind to Cell-surface Expressed PD-1 or CTLA-4

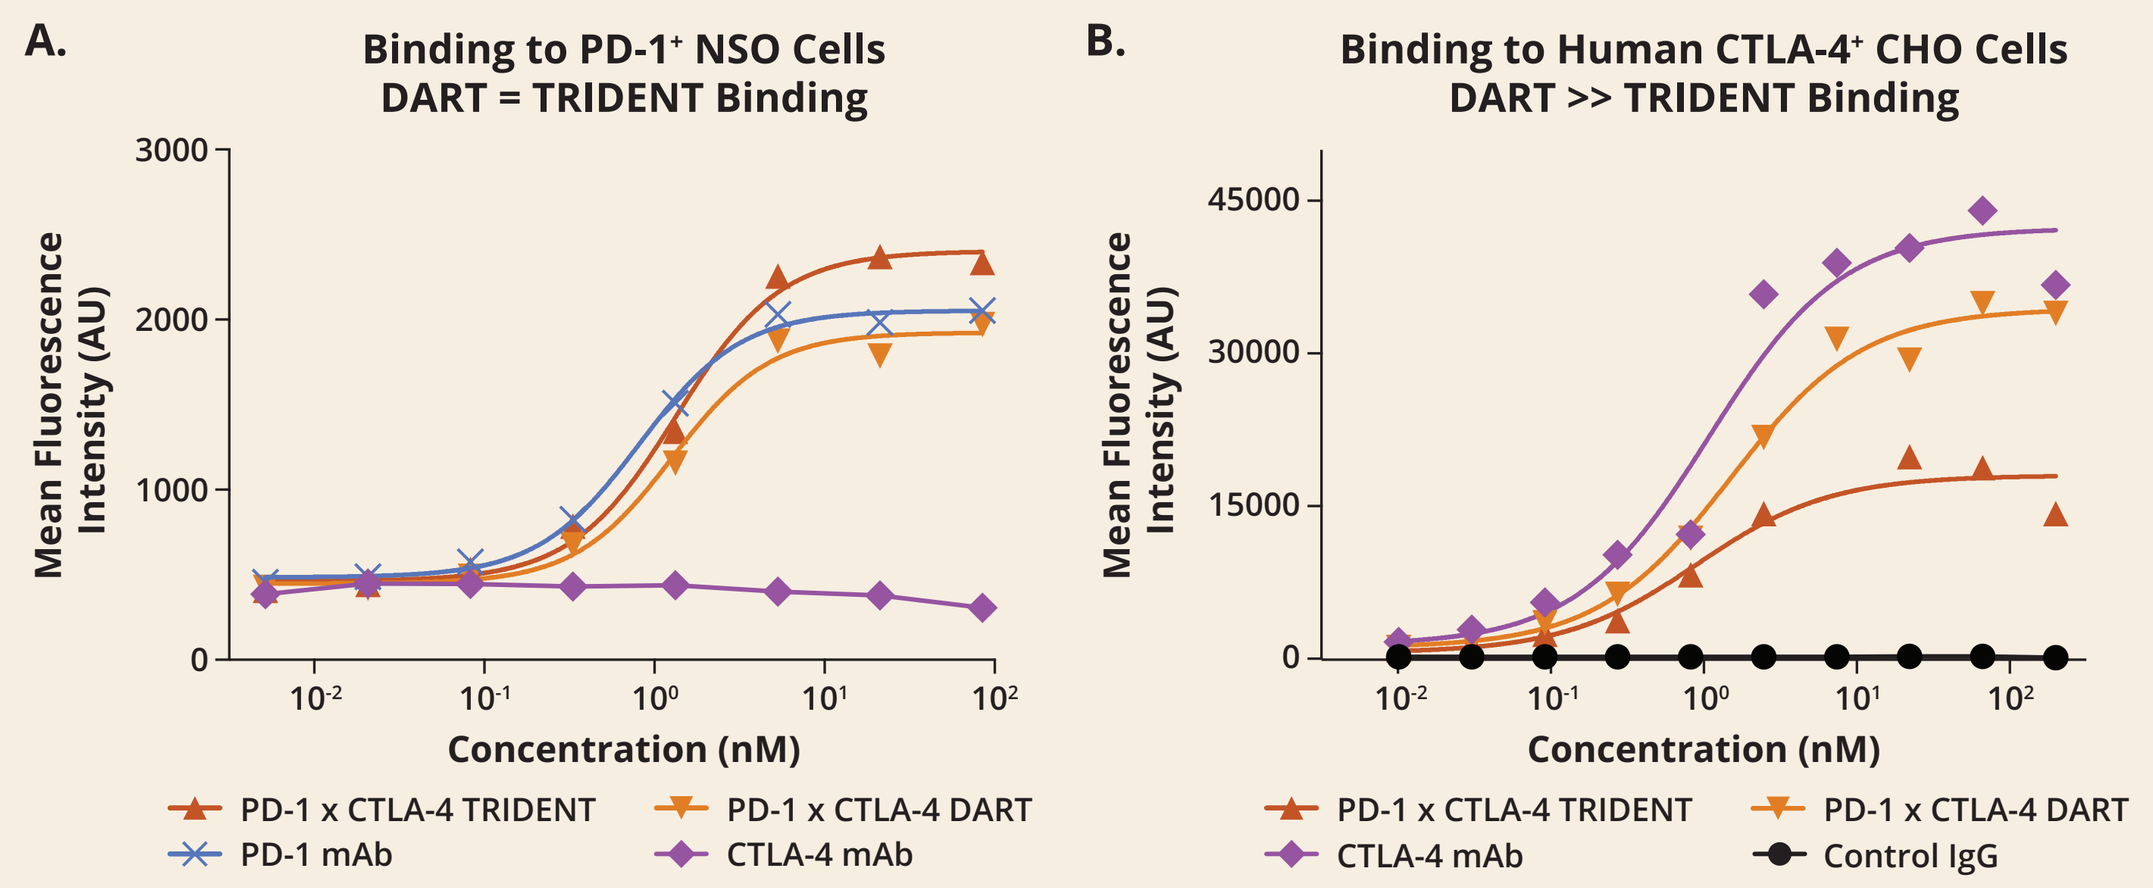
A. Binding to PD-1+ NSO Cells
DART = TRIDENT Binding
B. Binding to Human CTLA-4+ CHO Cells
DART >> TRIDENT Binding
Mean Fluorescence Intensity (AU)
Concentration (nM)
PD-1 x CTLA-4 TRIDENT
PD-1 x CTLA-4 DART
PD-1 mAb
CTLA-4 mAb
Control IgG

Flow cytometric analyses of PD-1 x CTLA-4 DART or TRIDENT binding to cells expressing PD-1 **(A)** or CTLA-4 **(B)** detected using anti-human IgG.

Bispecific Co-engagement of PD-1 and CTLA-4 by DART and TRIDENT Molecules

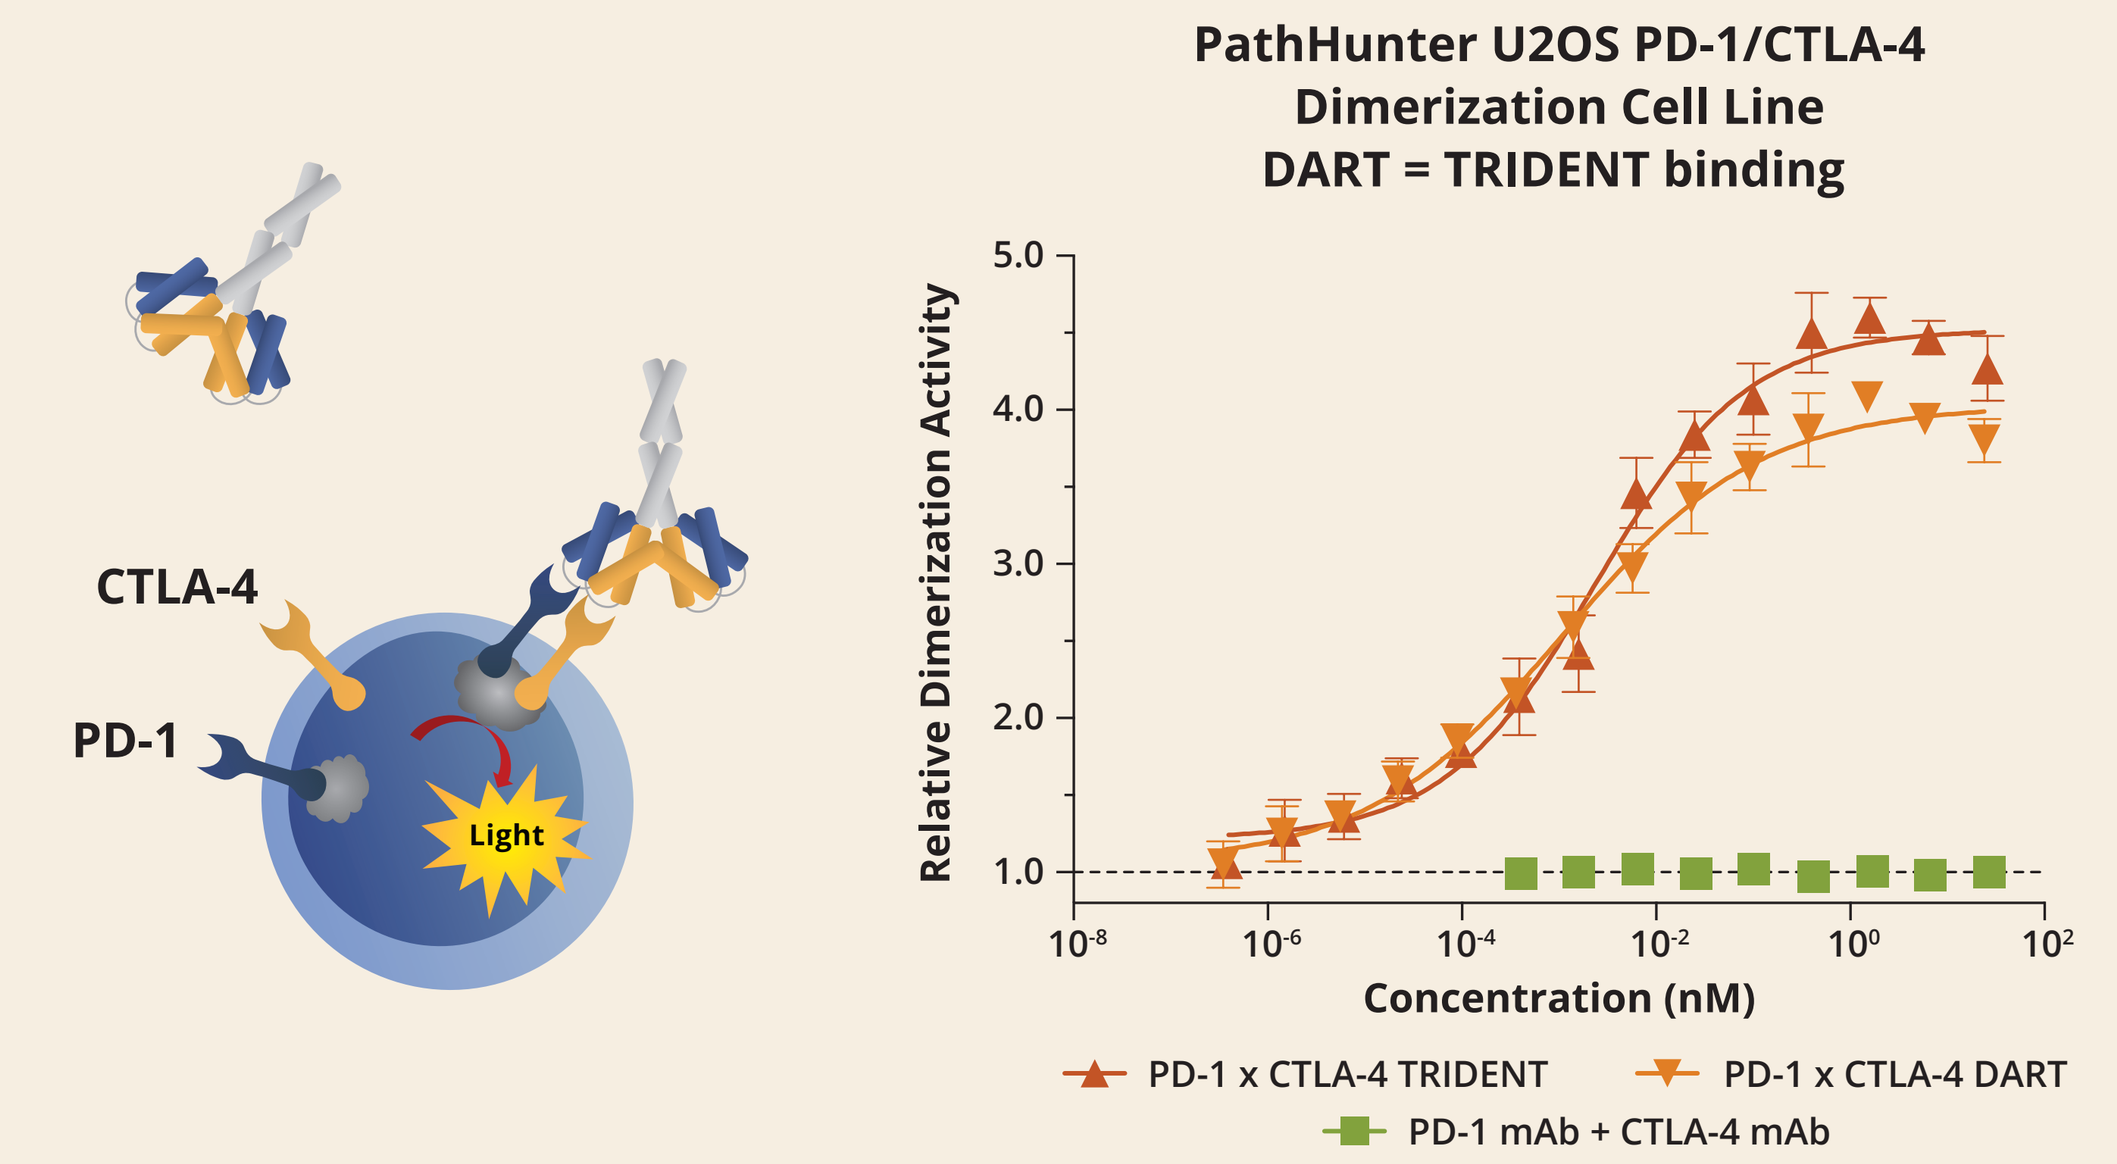
CTLA-4
PD-1
Light
PathHunter U2OS PD-1/CTLA-4
Dimerization Cell Line
DART = TRIDENT binding
Relative Dimerization Activity
Concentration (nM)
PD-1 x CTLA-4 TRIDENT
PD-1 x CTLA-4 DART
PD-1 mAb + CTLA-4 mAb

Indicated molecules were evaluated using enzyme fragment complementation assay employing PathHunter® U2OS PD-1/CTLA-4 dimerization cell Line (DiscoverX).

PD-1 x CTLA-4 DART and TRIDENT Molecules Inhibit Ligand Binding: Differential Inhibition of CTLA-4 Ligand Binding

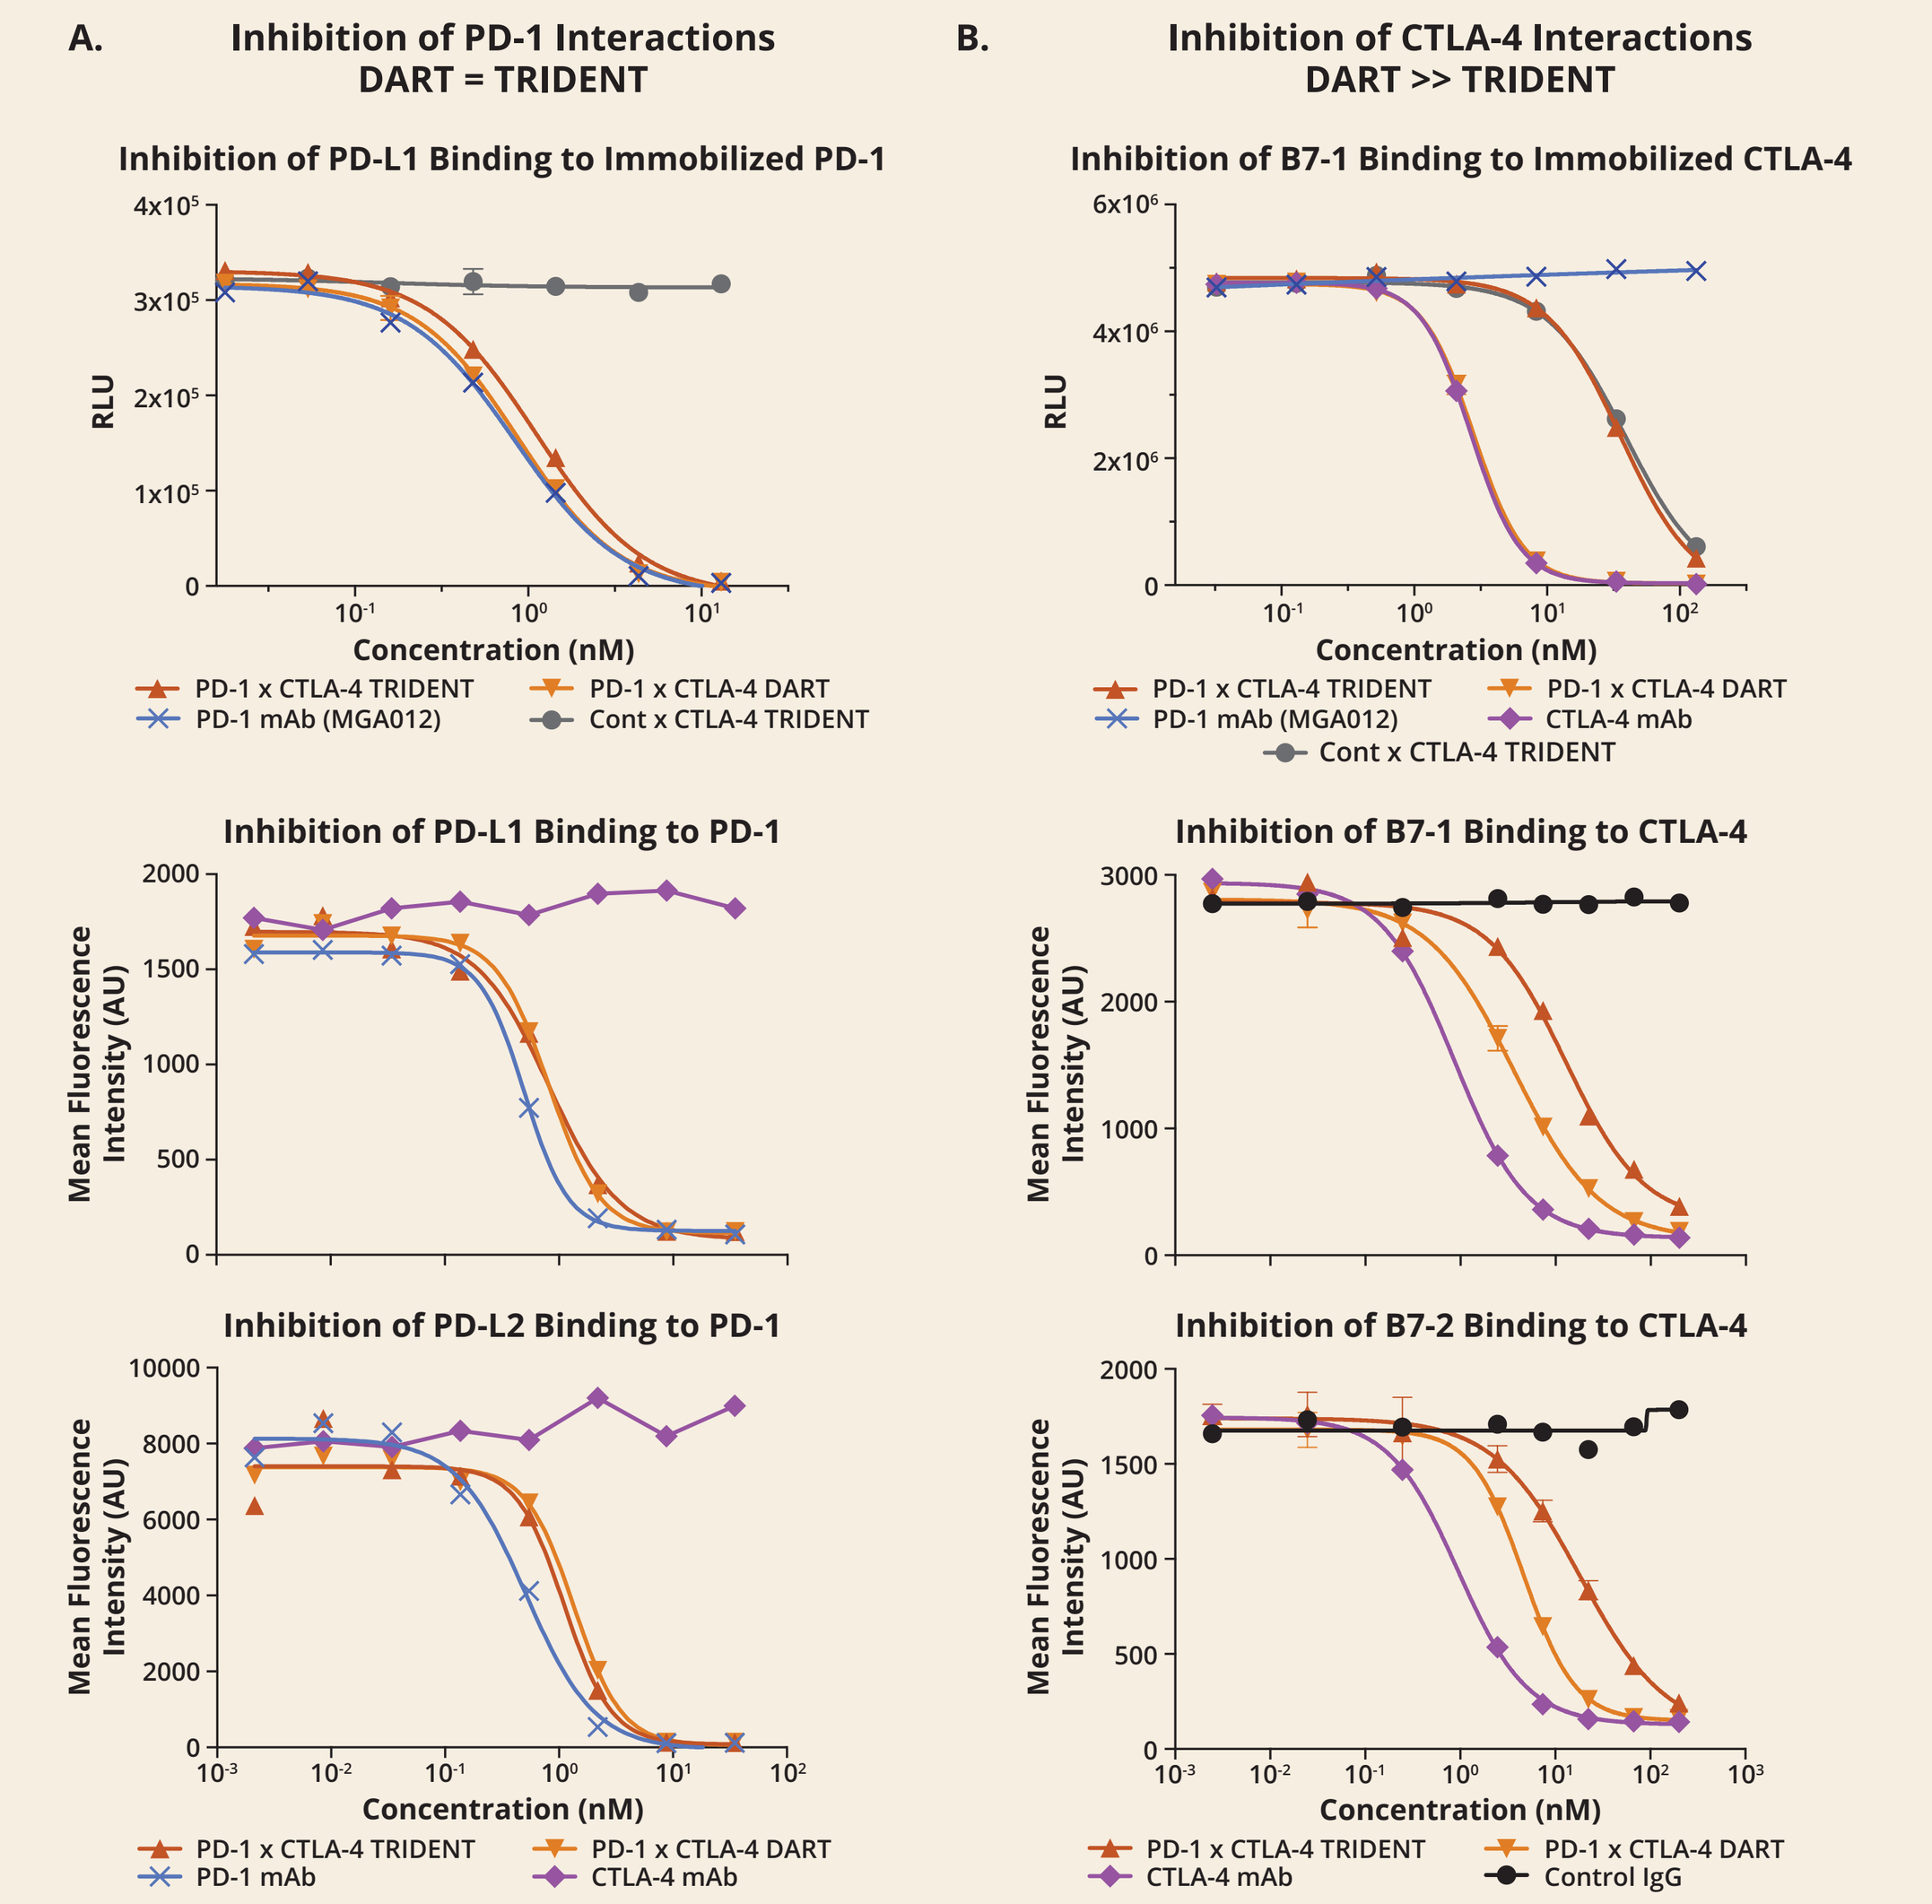
A. Inhibition of PD-1 Interactions
DART = TRIDENT
B. Inhibition of CTLA-4 Interactions
DART >> TRIDENT
Inhibition of PD-L1 Binding to Immobilized PD-1
Inhibition of B7-1 Binding to Immobilized CTLA-4
Inhibition of PD-L1 Binding to PD-1
Inhibition of B7-1 Binding to CTLA-4
Inhibition of PD-L2 Binding to PD-1
Inhibition of B7-2 Binding to CTLA-4
RLU
Mean Fluorescence Intensity (AU)
Concentration (nM)
PD-1 x CTLA-4 TRIDENT
PD-1 x CTLA-4 DART
PD-1 mAb (MGA012)
Cont x CTLA-4 TRIDENT
CTLA-4 mAb
Control IgG

A. Inhibition of PD-L1 ligand binding by ELISA and Flow cytometric analysis of soluble PD-L1 or PD-L2 binding to NS0-PD-1+ cells in the presence of titrating concentrations of the indicated mAbs, PD-1 x CTLA-4 DART and TRIDENT molecules.
B. Inhibition of B7-1 ligand binding by ELISA and Flow cytometric analyses of soluble B7-1-his or B7-2-his binding to CTLA-4+ Jurkat cells in the presence of titrating concentrations of the indicated mAbs, PD-1 x CTLA-4 DART and TRIDENT molecules.

PD-1 x CTLA-4 DART and TRIDENT Molecules Reverse Inhibition of TCR Signaling: Differential Release of CTLA-4 Inhibition

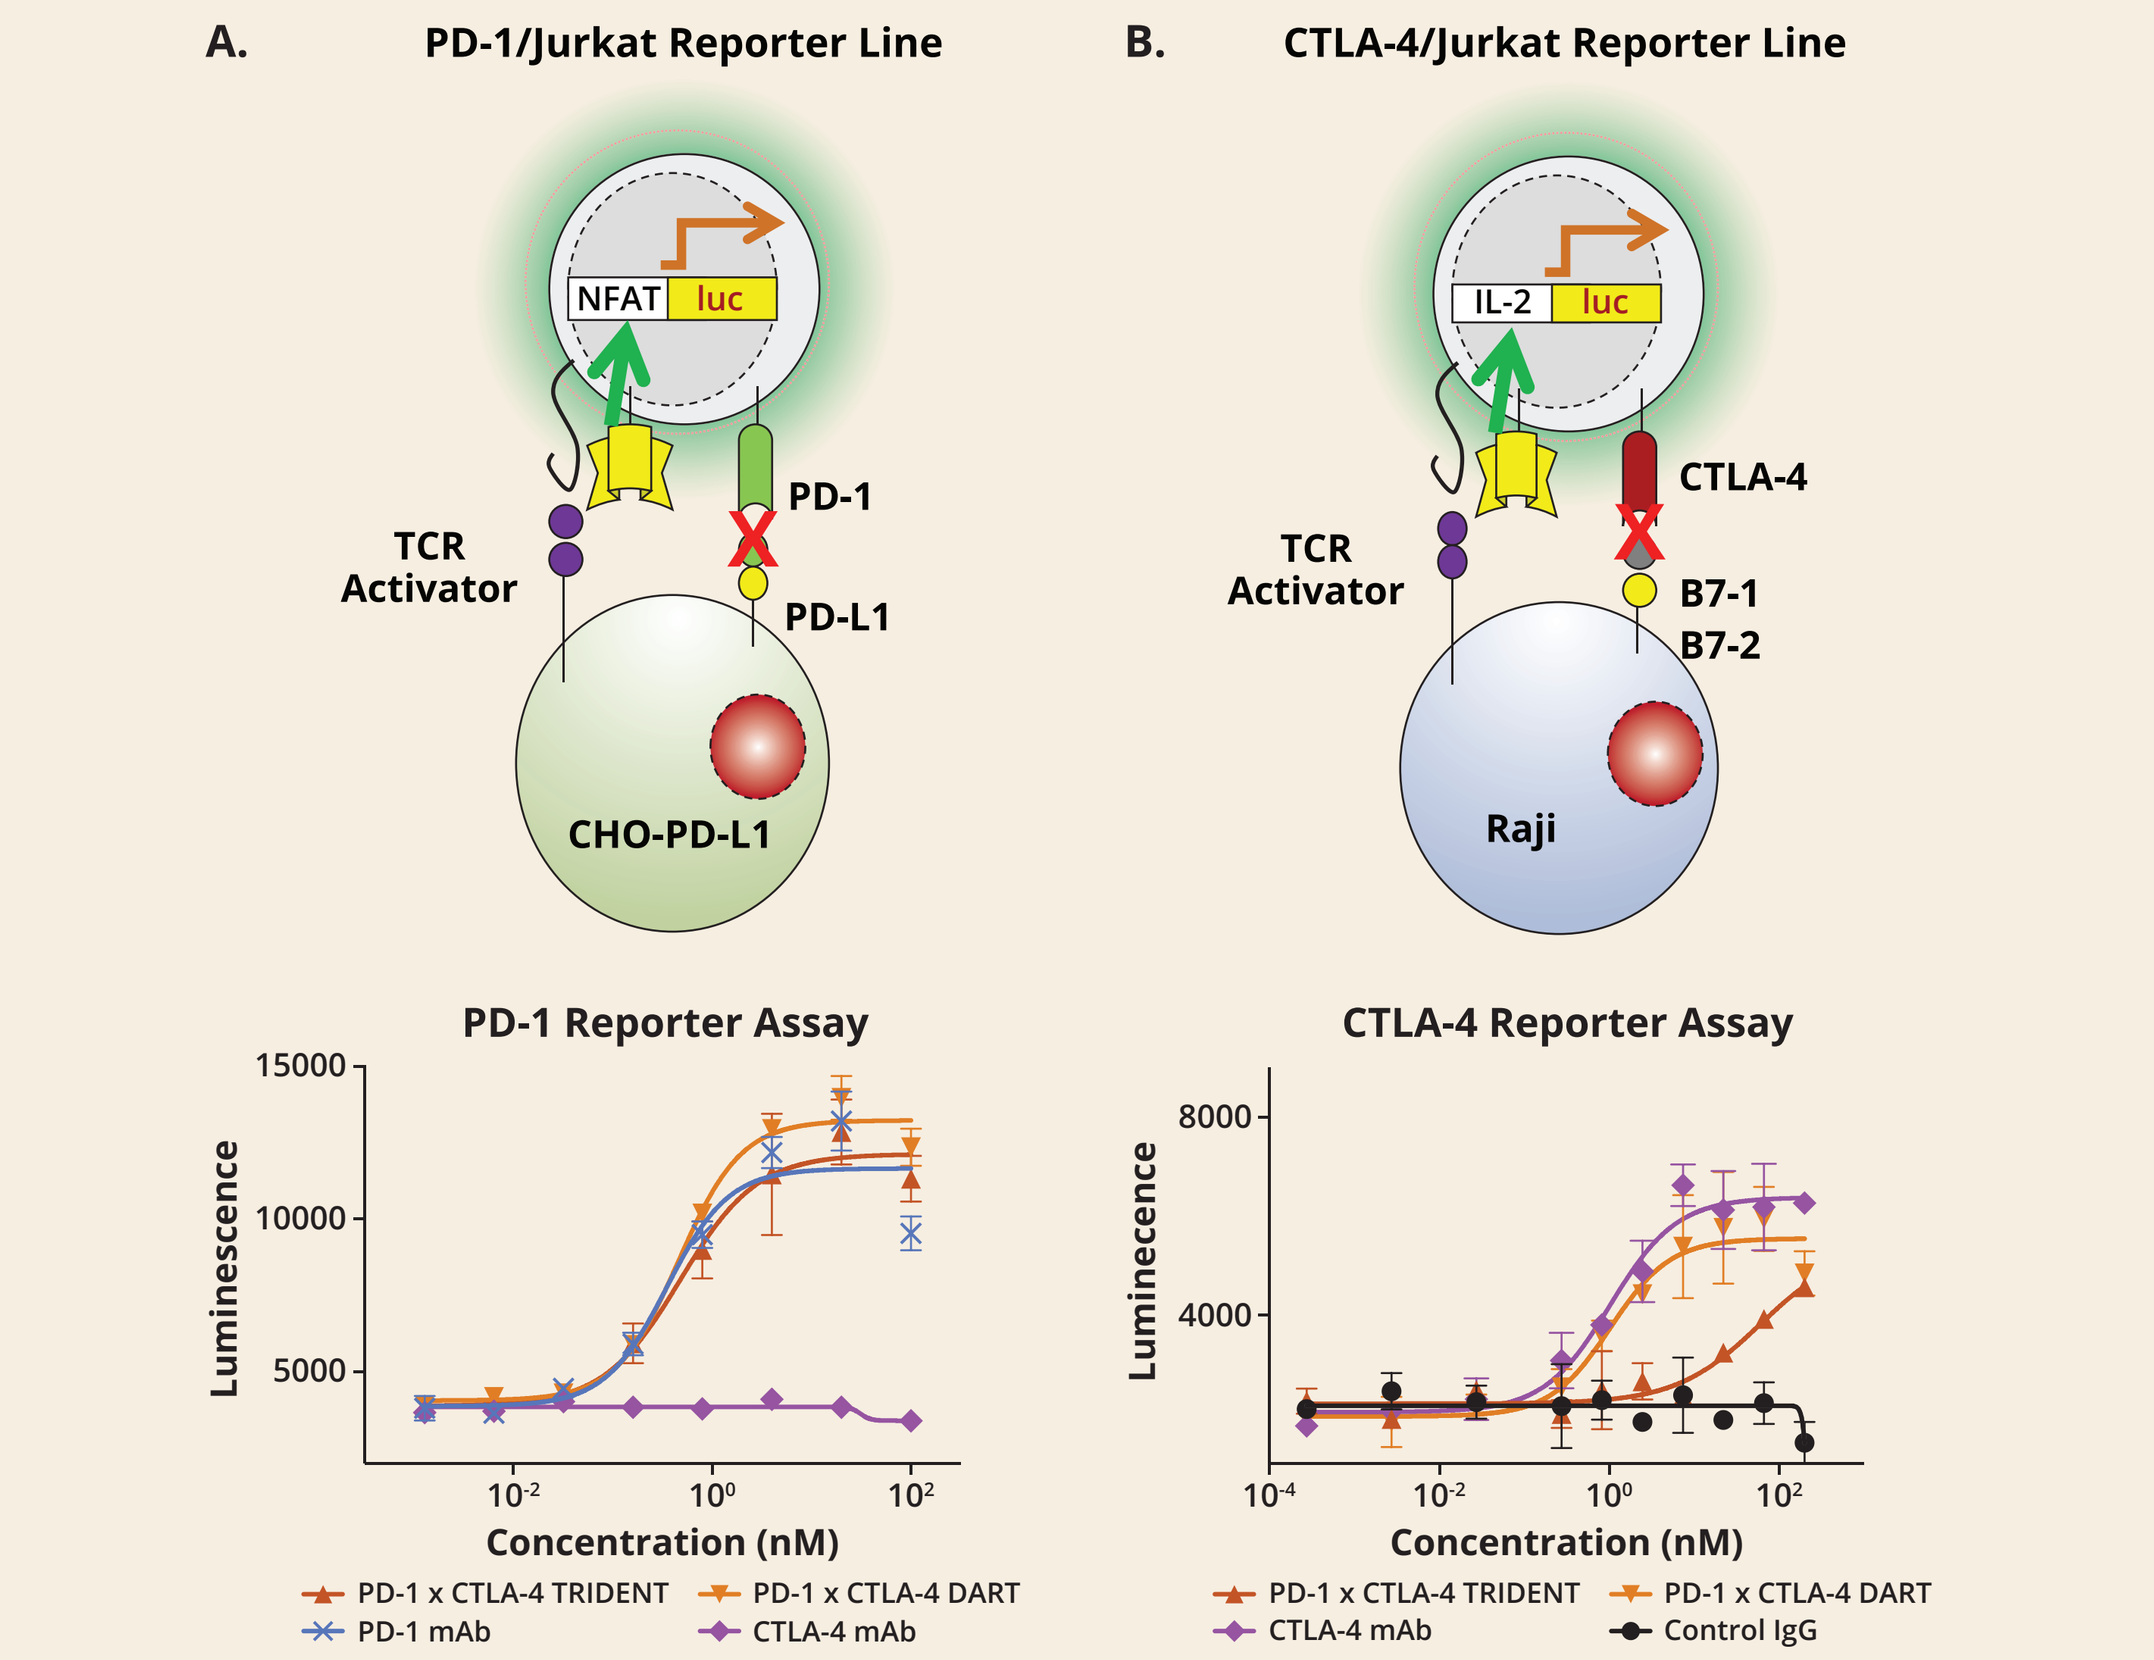
A. PD-1/Jurkat Reporter Line
B. CTLA-4/Jurkat Reporter Line
NFAT
luc
IL-2
luc
PD-1
PD-L1
TCR Activator
CHO-PD-L1
CTLA-4
B7-1
B7-2
Raji
PD-1 Reporter Assay
CTLA-4 Reporter Assay
Luminescence
Concentration (nM)
PD-1 x CTLA-4 TRIDENT
PD-1 x CTLA-4 DART
PD-1 mAb
CTLA-4 mAb
Control IgG

A. The indicated molecules were tested in the Promega PD-1/PD-L1 assay system employing Jurkat-PD-1+ cells (transduced with an NF-AT luciferase reporter) cultured with a CHO-PD-L1 cell line that expresses a TCR activator. The luminescence represents the release of PD-1-mediated suppression of the NF-AT-driven luciferase gene upon TCR stimulation.
B. The indicated molecules were tested in the Promega CTLA-4/B7-1&B7-2 assay system employing Jurkat-CTLA-4+ cells (transduced with an IL-2 promoter-driven luciferase reporter) cultured with a Raji cell line that expresses a TCR activator. The luminescence represents the release of CTLA-4-mediated suppression of the IL-2 promoter-driven luciferase gene upon TCR stimulation.

PD-1 x CTLA-4 DART and TRIDENT Molecules Augment T-cell Response to SEB Stimulation

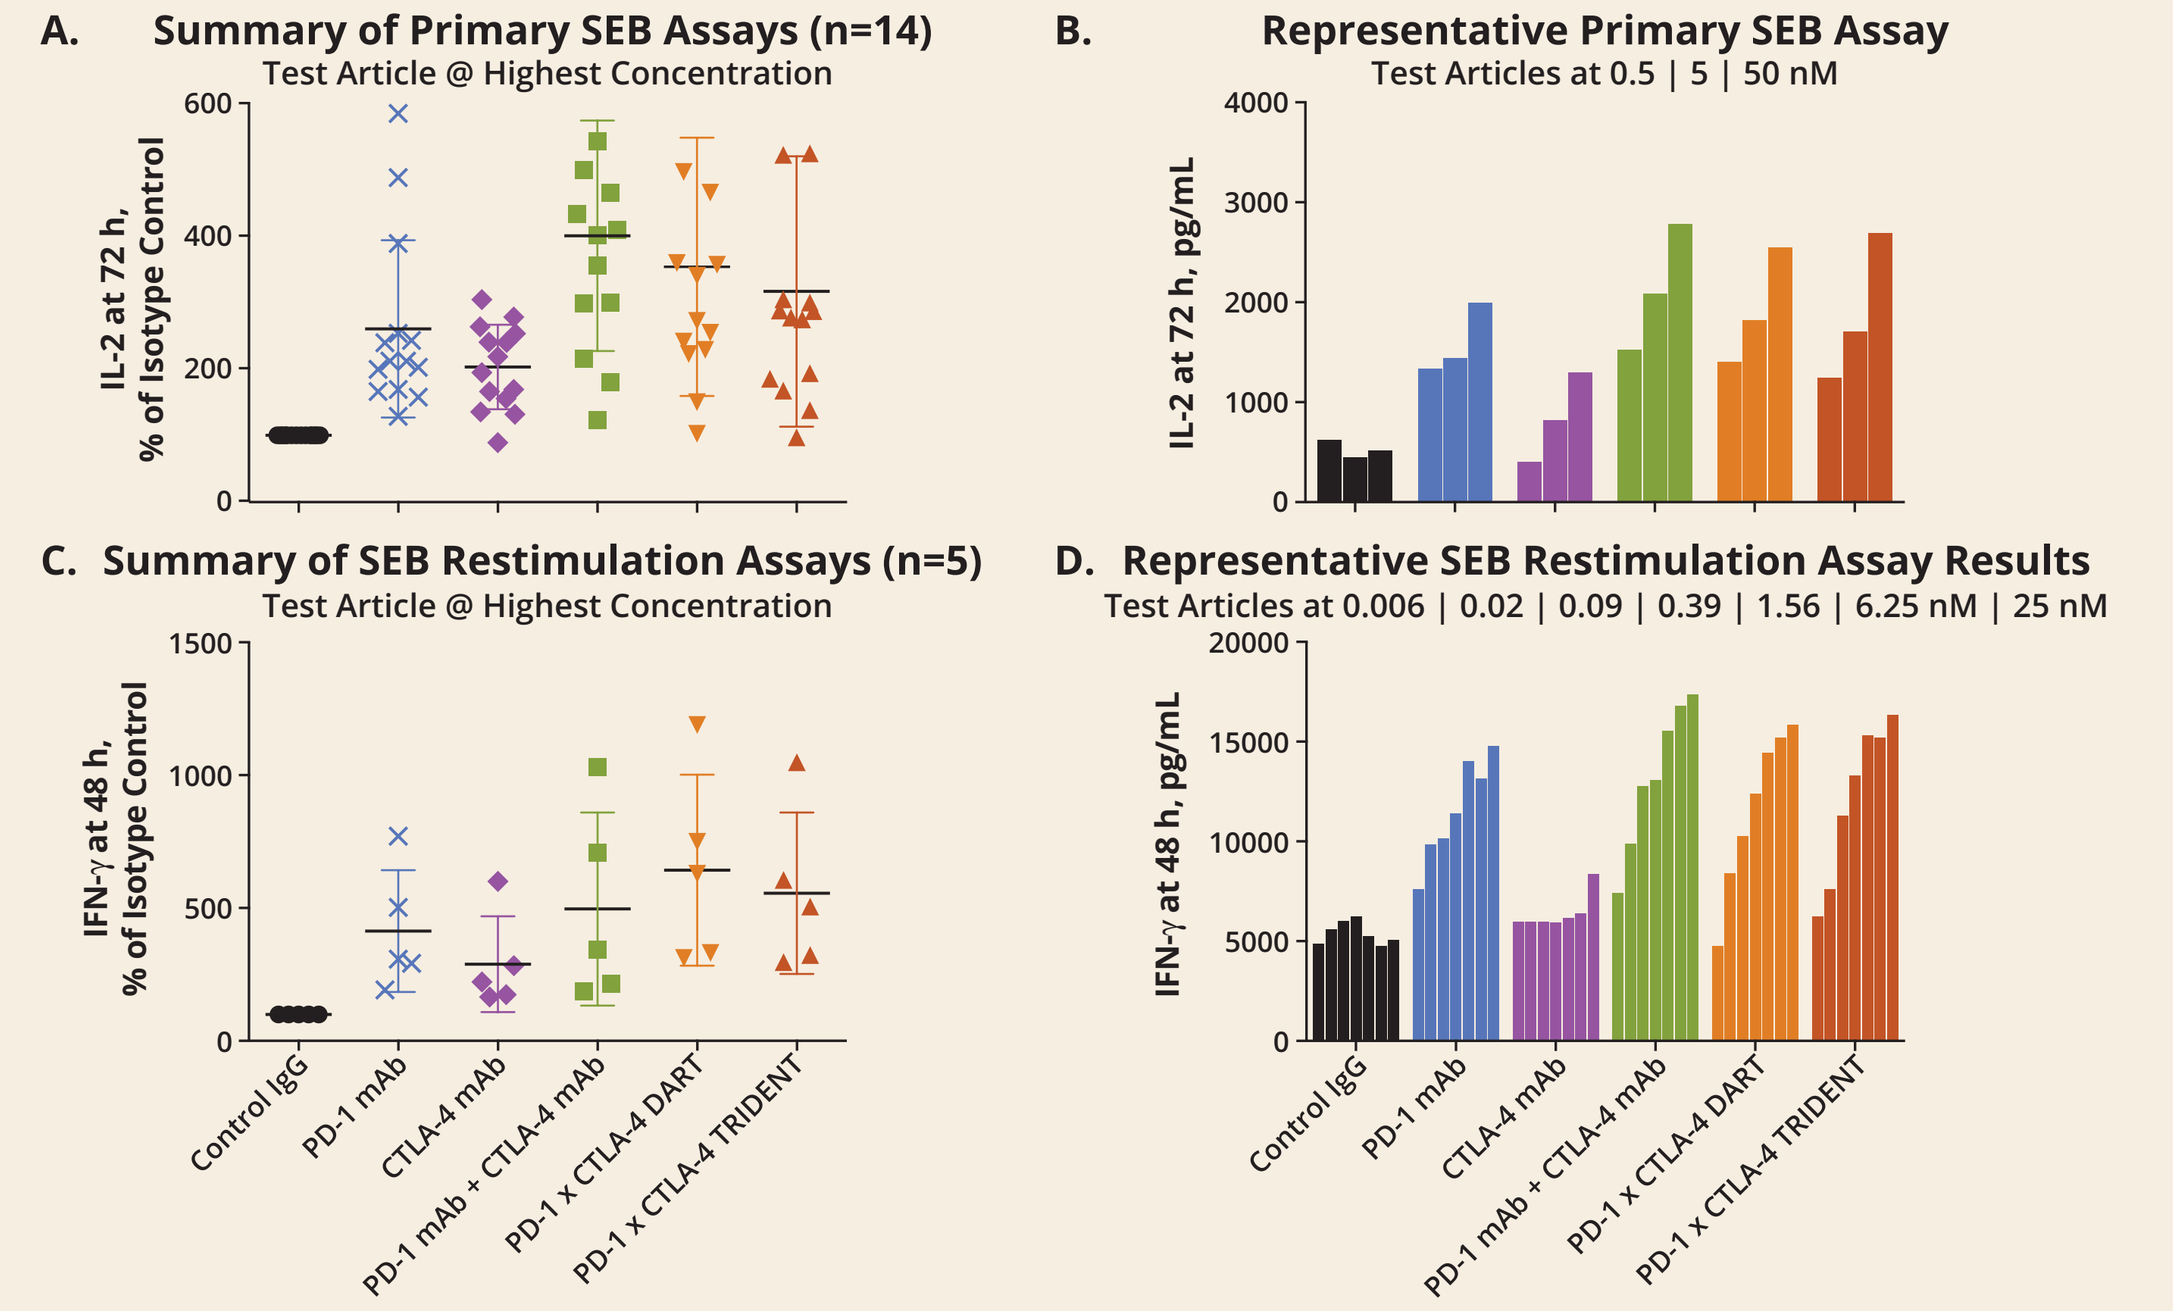
A. Summary of Primary SEB Assays (n=14)
Test Article @ Highest Concentration
B. Representative Primary SEB Assay
Test Articles at 0.5 | 5 | 50 nM
C. Summary of SEB Restimulation Assays (n=5)
Test Article @ Highest Concentration
D. Representative SEB Restimulation Assay Results
Test Articles at 0.006 | 0.02 | 0.09 | 0.39 | 1.56 | 6.25 nM | 25 nM
IL-2 at 72 h, % of Isotype Control
IL-2 at 72 h, pg/mL
IFN-γ at 48 h, % of Isotype Control
IFN-γ at 48 h, pg/mL
Control IgG
PD-1 mAb
CTLA-4 mAb
PD-1 mAb + CTLA-4 mAb
PD-1 x CTLA-4 DART
PD-1 x CTLA-4 TRIDENT

A-B. Human PBMCs from healthy donors were stimulated with 500 ng/mL SEB for 72 h in the presence of indicated molecules. IL-2 levels were measured in supernatants. Molarity refers to the concentration of individual components, whether used alone or in combination (e.g., 50 nM PD-1 mAb + 50 nM CTLA-4 mAb proteins were compared to 50 nM PD-1 x CTLA-4 bispecifics).
C-D. Human PBMCs from healthy donors were stimulated with SEB for 48 h, washed, and restimulated with 0.5 ng/mL SEB. IFN-γ secretion was determined by ELISA. Molarity refers to the concentration of individual components, whether used alone or in combination.

PD-1 x CTLA-4 DART and TRIDENT Molecules Enhance the MLR Response

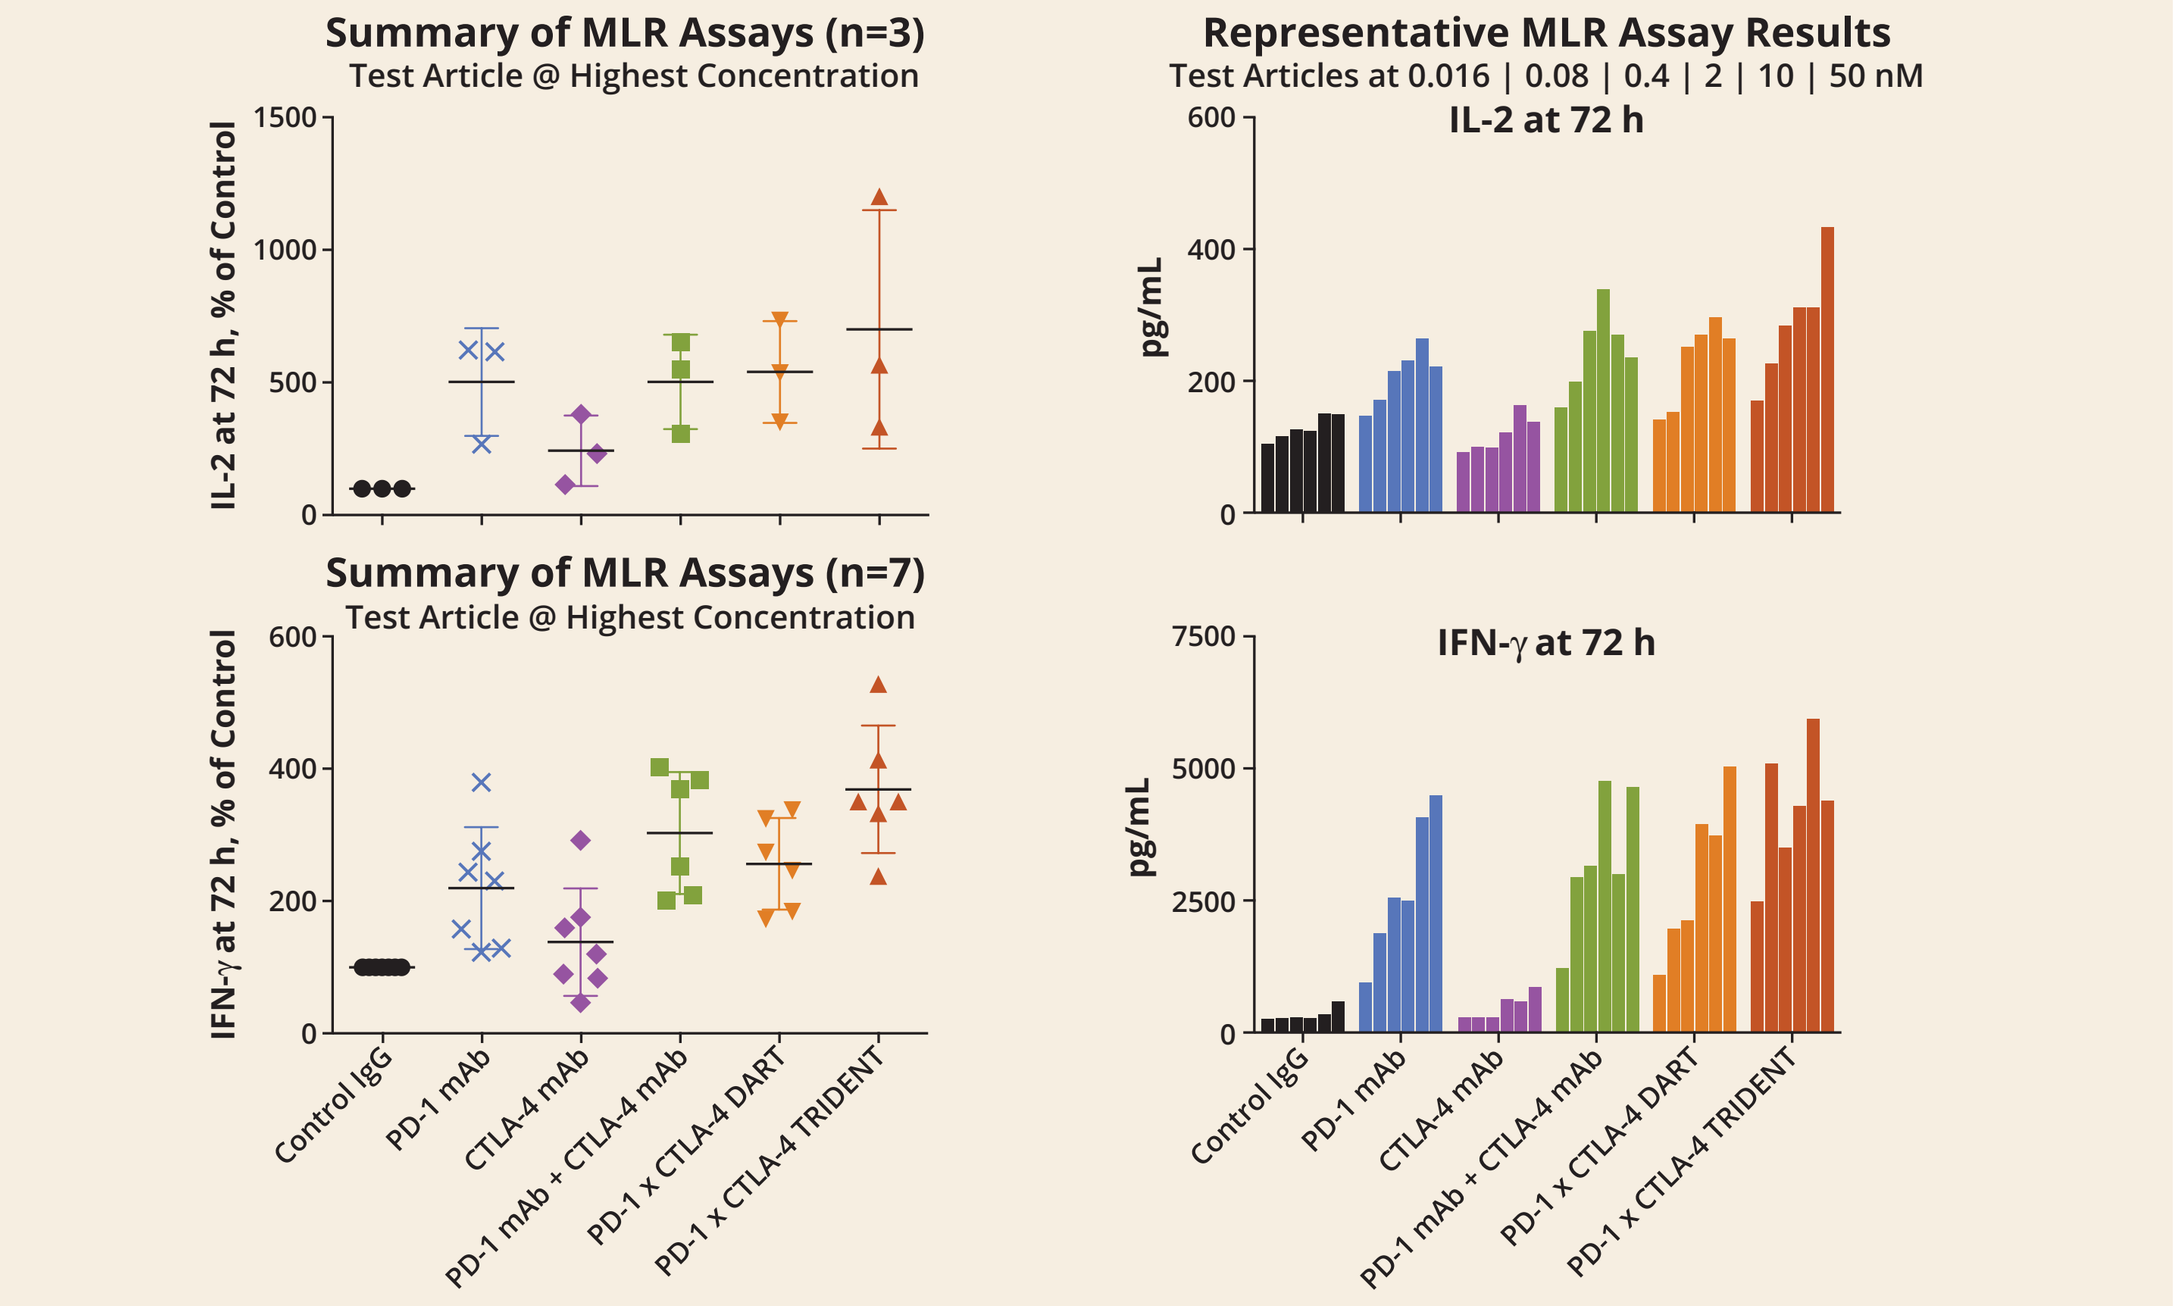
Summary of MLR Assays (n=3)
Test Article @ Highest Concentration
Representative MLR Assay Results
Test Articles at 0.016 | 0.08 | 0.4 | 2 | 10 | 50 nM
IL-2 at 72 h
Summary of MLR Assays (n=7)
Test Article @ Highest Concentration
IFN-γ at 72 h
IL-2 at 72 h, % of Control
IFN-γ at 72 h, % of Control
pg/mL
Control IgG
PD-1 mAb
CTLA-4 mAb
PD-1 mAb + CTLA-4 mAb
PD-1 x CTLA-4 DART
PD-1 x CTLA-4 TRIDENT

Mixed lymphocyte reaction (MLR) between monocyte derived DCs generated by culturing with GM-CSF and IL-4 for 7 days and freshly isolated CD4+ T cells from different donors. After 72 h, IL-2 and IFN-γ in the culture supernatants were measured by ELISA.

Conclusions

- PD-1 x CTLA-4 DART and TRIDENT molecules bind to and inhibit ligand interaction with their targets
 - TRIDENT protein with a monovalent CTLA-4 binding arm shows biased binding favoring PD-1 interaction
- DART and TRIDENT molecules co-engage their targets in an equivalent fashion when PD-1 and CTLA-4 are coexpressed on the same cell
- DART and TRIDENT molecules augment T-cell activation (cytokine release) to levels comparable to those observed with the individual PD-1 and CTLA-4 mAbs
- Further development (nonhuman primate toxicology) of bispecific PD-1 x CTLA-4 molecules is in progress

Presented at the American Association for Cancer Research Annual Meeting 2017, April 1–5, 2017, Washington, DC

©2017 MacroGenics, Inc. All rights reserved.