A 5T4 x CD3 Bispecific DART® Molecule with Extended Half-life for T-cell Immunotherapy of Cancers

Ling Huang, Gurunadh Chichili, Ralph Alderson, Francine Chen, Jennifer G. Brown, Hua Li, Valentina Ciccarone, James Tamura, Daorong Liu, Liqin Liu, Syd Johnson, Ezio Bonvini, and Paul Moore

MacroGenics, Inc., Rockville, MD and South San Francisco, CA

4608

http://ir.macrogenics.com/events.cfm

Abstract

Introduction: 5T4 (trophoblast glycoprotein), an oncofetal antigen involved in embryo development, is expressed on the cell surface of multiple cancers. Several 5T4-directed interventions have been reported, including a cancer vaccine and an antibody-drug conjugate. A superantigen-driven redirected cell killing modality was also developed, but obstacles, such as immunogenicity, remain to a successful 5T4 therapeutic molecule. Using the DART platform, we have developed a bispecific Fc-bearing 5T4 x CD3 protein designated "5T4 x CD3 DART", designed to redirect T cells to target 5T4-expressing tumors.

Methods: 5T4 x CD3 DART, a humanized Fc-bearing DART molecule, was stably expressed in CHO cells and purified to homogeneity via a standard antibody-purification platform. In vitro characterization and functional studies were performed with 5T4-positive tumor cell lines and human T cells. In vivo studies were performed in immune-deficient tumor-bearing mice co-implanted with activated human T cells or reconstituted with human peripheral blood mononuclear cells (PBMCs). Pharmacokinetic studies were performed in both human FcRn transgenic mice and cynomolgus monkeys.

Results: 5T4 x CD3 DART demonstrated bispecific binding properties to both human and cynomolgus monkey antigens. In redirected cytolysis studies, 5T4 x CD3 DART-mediated lysis of 5T4-positive pancreatic, lung, renal, triple-negative breast, and ovarian cancer cell lines with EC_{50} values ranging between 0.03 and 0.08 ng/mL. 5T4 x CD3 DART displayed favorable pharmacokinetics with a prolonged circulating half-life in human FcRn transgenic mice and cynomolgus monkeys. Tumor clearance studies in NOD/SCID mice implanted subcutaneously with activated human T cells and tumor cells demonstrated robust inhibition of tumor growth upon intravenous (IV) administration of 5T4 x CD3 DART at doses as low as 0.8 μg/kg, but not with a CD3-binding control DART protein. In addition, human PBMC-reconstituted NOD/SCID/IL2rγ chain null mice were implanted intradermally with renal and pancreatic tumor cell lines or orthotopically with a triple negative breast cancer line and, after tumor establishment, were treated with 5T4 x CD3 DART. Antitumor activity was observed at doses as low as 4 μg/kg. Immunohistochemical analysis of tumor xenografts confirmed tumor clearance was associated with T-cell recruitment into the tumor mass.

Conclusion: In summary, 5T4 x CD3 DART displays robust antitumor activity against several cancer cell lines in vitro and in vivo together with a favorable pharmacokinetic profile and merits further consideration as a potential treatment for 5T4-positive cancers.

Introduction

- 5T4 (trophoblast glycoprotein), an oncofetal antigen involved in embryo development[1], is expressed on the cell surface of multiple cancers with limited expression in normal adult tissues[2]
- Several 5T4-directed interventions have been evaluated clinically, including a cancer vaccine and an antibody-drug conjugate
- A superantigen-driven redirected cell killing modality was also developed[3], but was thwarted by immunogenicty
- Using the DART platform, we have developed a bispecific Fc-bearing 5T4 x CD3 protein designated "5T4 x CD3 DART", designed to redirect T cells to target 5T4-expressing tumors

Expression of 5T4 in Cancer Tissues

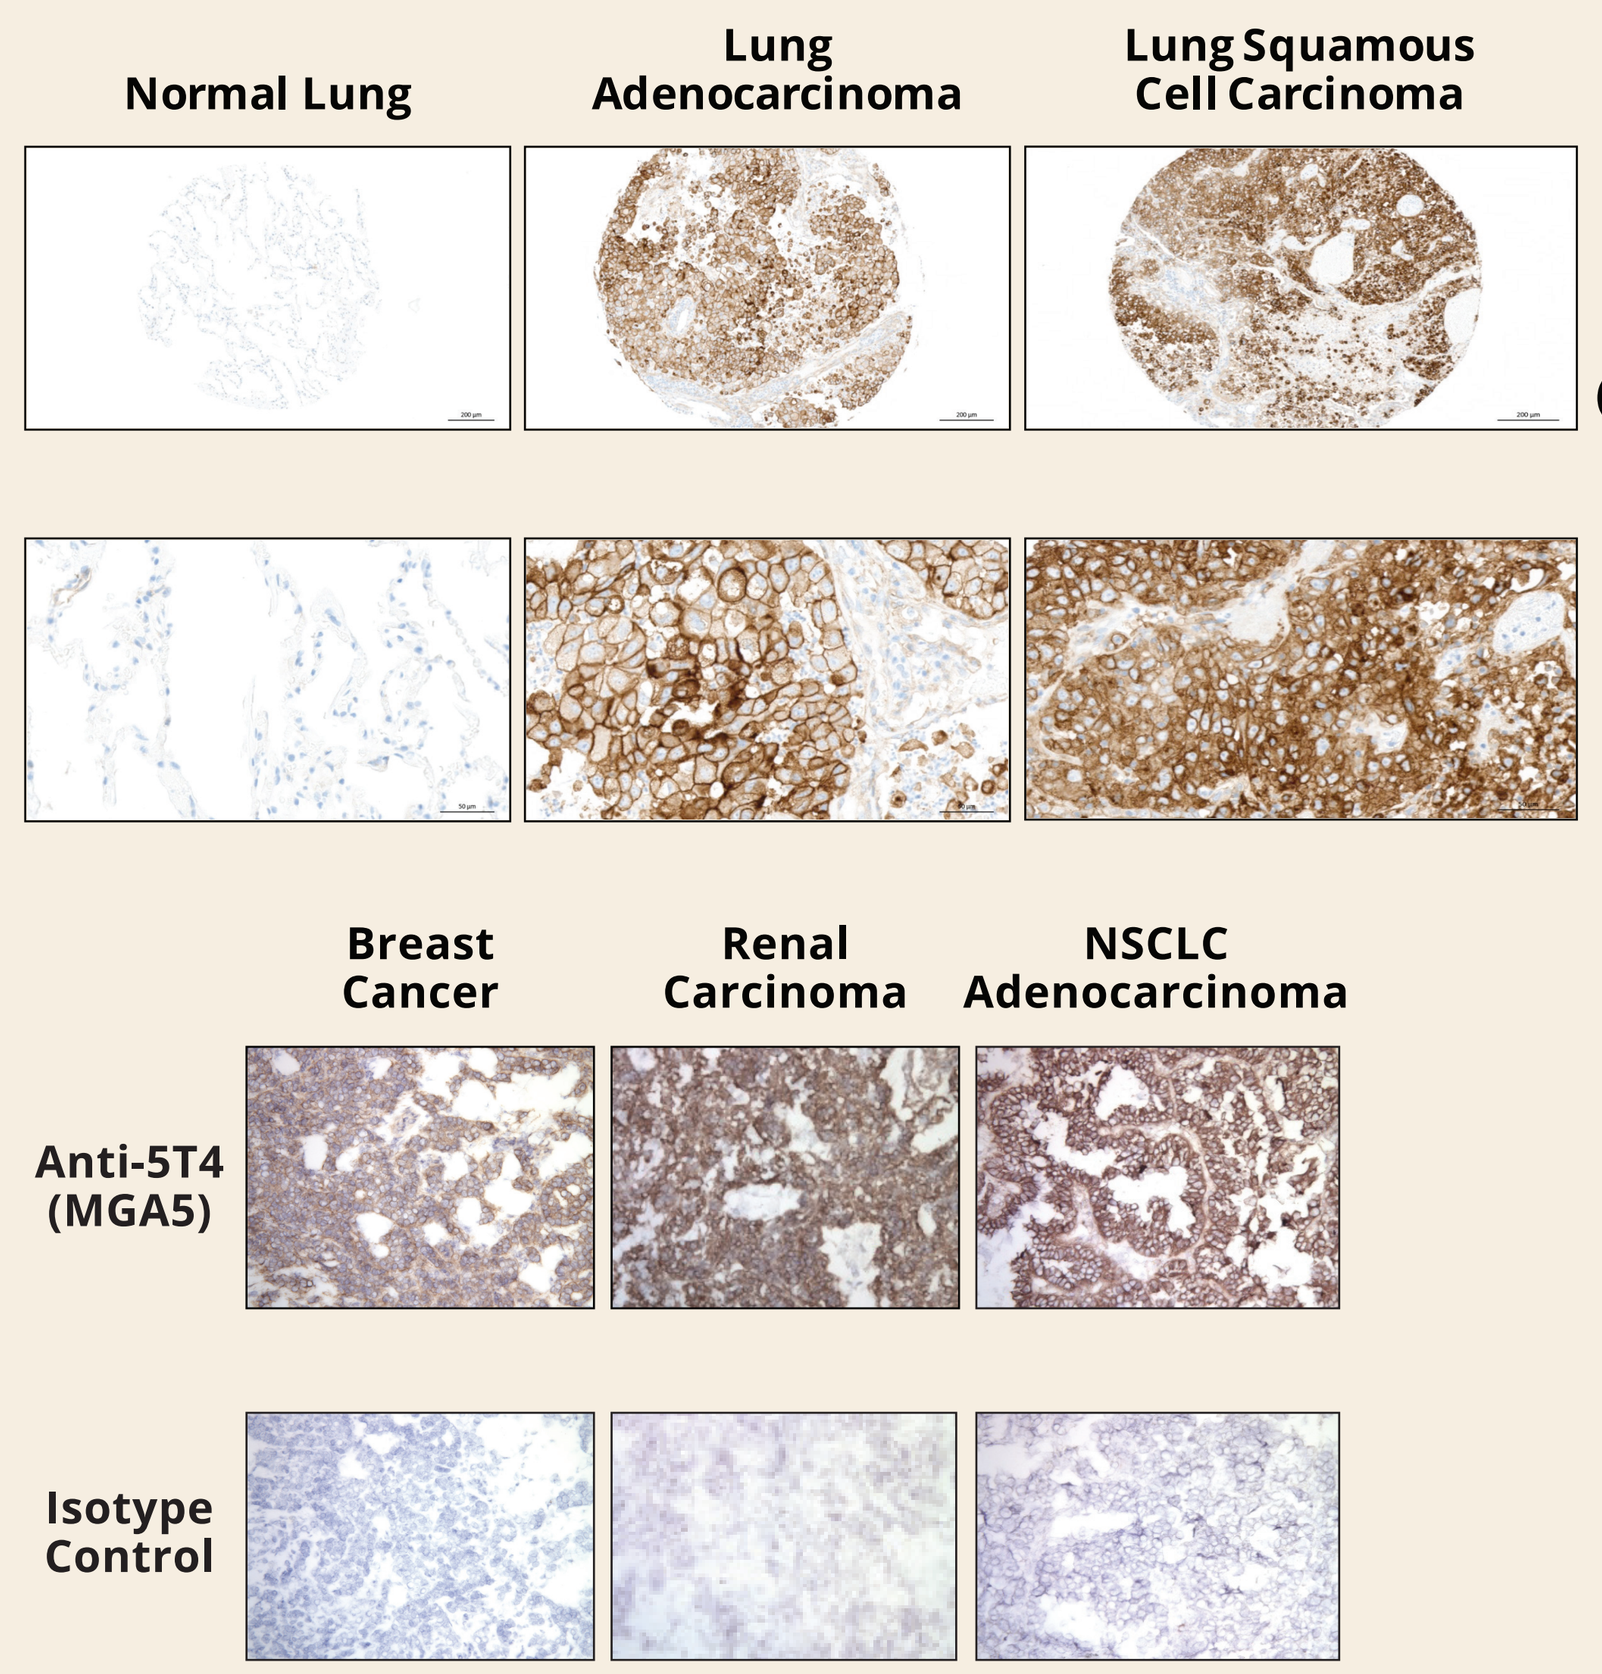
A.
Normal Lung
Lung Adenocarcinoma
Lung Squamous Cell Carcinoma
Positivity (membrane only)
Adenocarcinoma: 22/38 (58%)
Squamous: 31/37 (84%)
B.
Breast Cancer
Renal Carcinoma
NSCLC Adenocarcinoma
Anti-5T4 (MGA5)
Isotype Control

A. IHC of FFPE tissues on independent TMAs with anti-5T4 Ab EPR5529 (abcam®) confirmed favorable normal tumor differential and high penetrance of expression in lung cancer.
B. Evaluation of a panel of anti-5T4 mAbs generated at MG demonstrating exhibited broad tumor reactivity identified MGA5, the parental mAb humanized and incorporated into the 5T4 x CD3 DART.

5T4 x CD3 DART

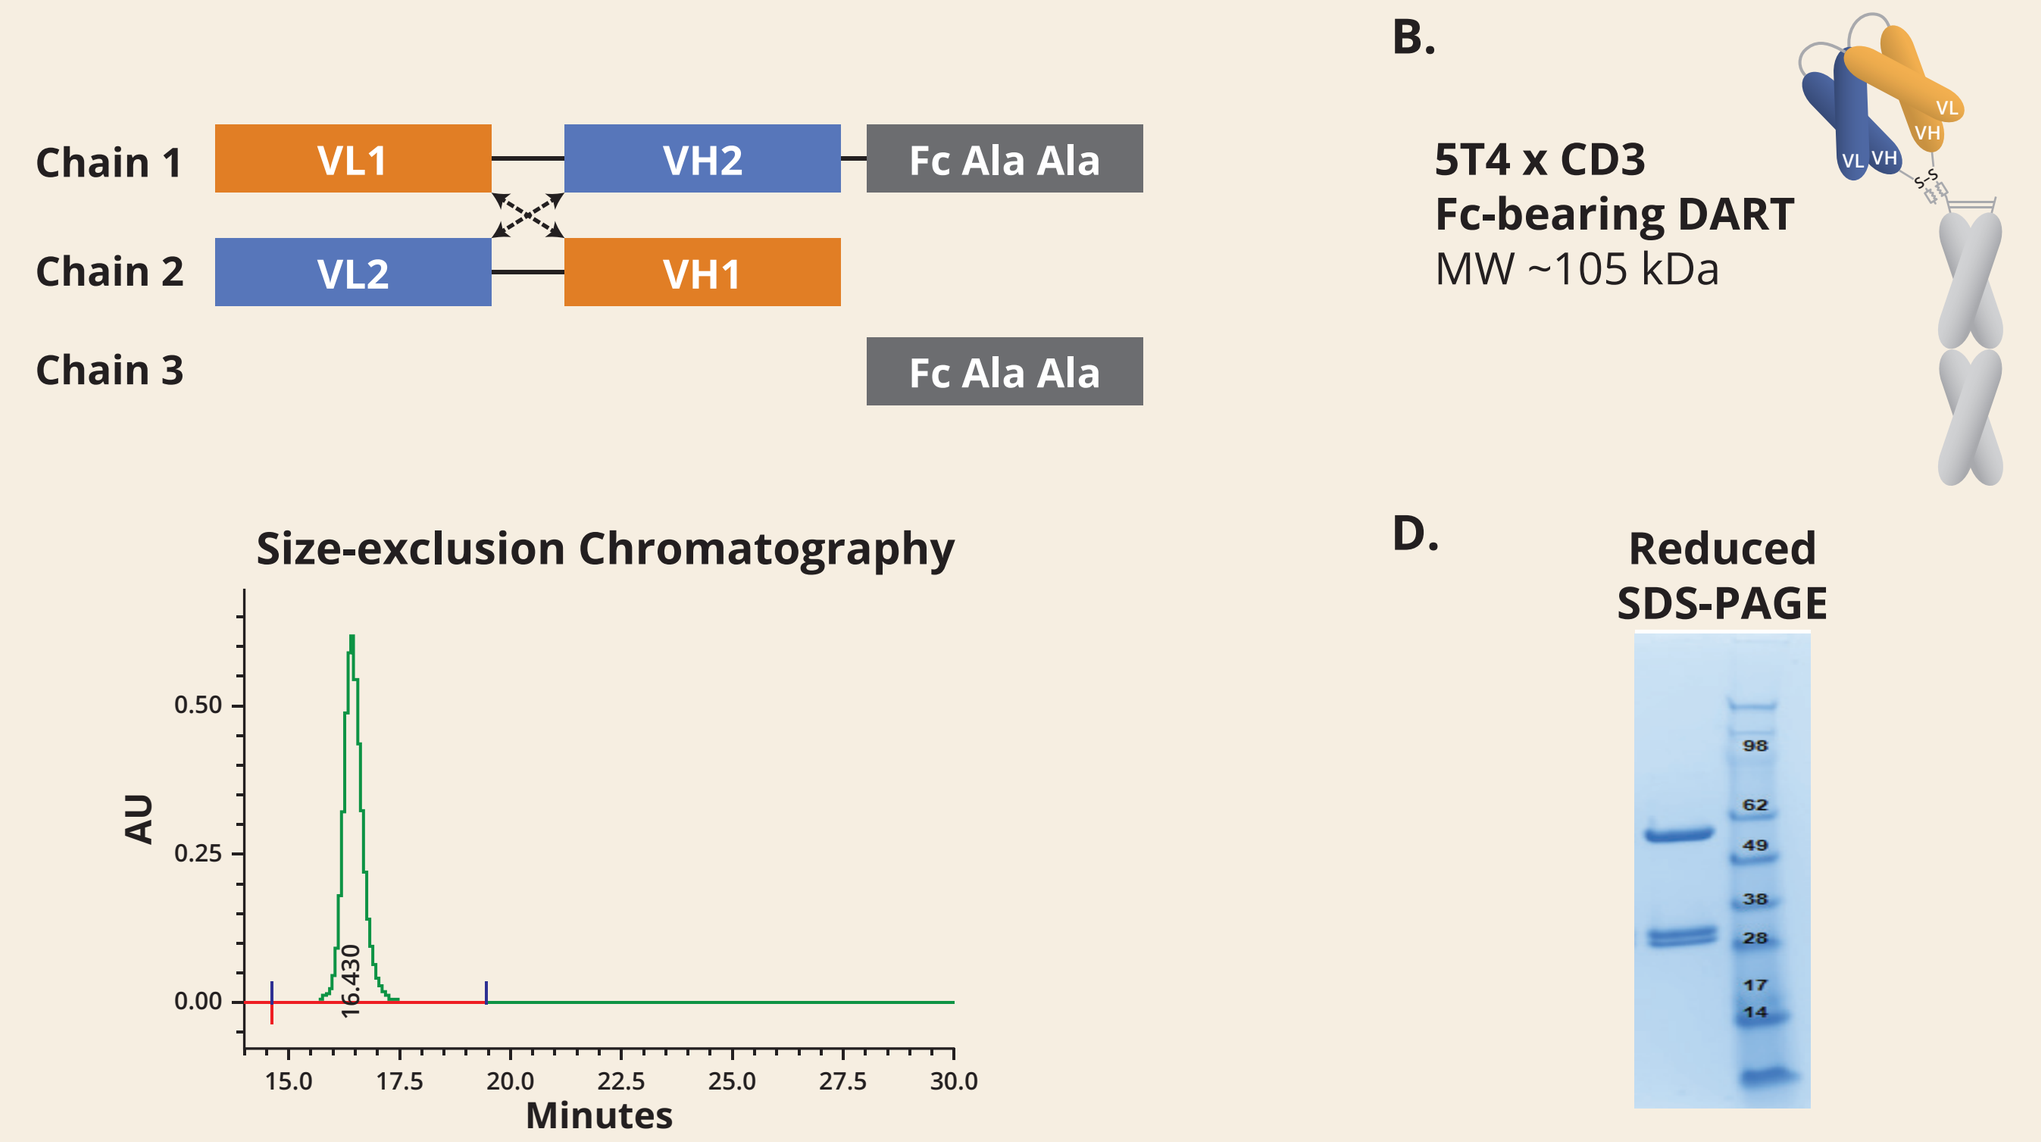
A.
Chain 1
VL1
VH2
Fc Ala Ala
Chain 2
VL2
VH1
Chain 3
Fc Ala Ala
B.
5T4 x CD3
Fc-bearing DART
MW ~105 kDa
C.
Size-exclusion Chromatography
AU
16.430
Minutes
D.
Reduced SDS-PAGE

- Humanized anti-5T4 and anti-CD3 variable domains were assembled through disulfide-linked heterodimer to form the DART protein
- Modified Fc, lacking effector function, was introduced for half-life improvement
- DART protein was generated in CHO cells
- Purified protein was subjected to SEC and reduced SDS-PAGE

Results

Binding Properties of 5T4 x CD3 DART

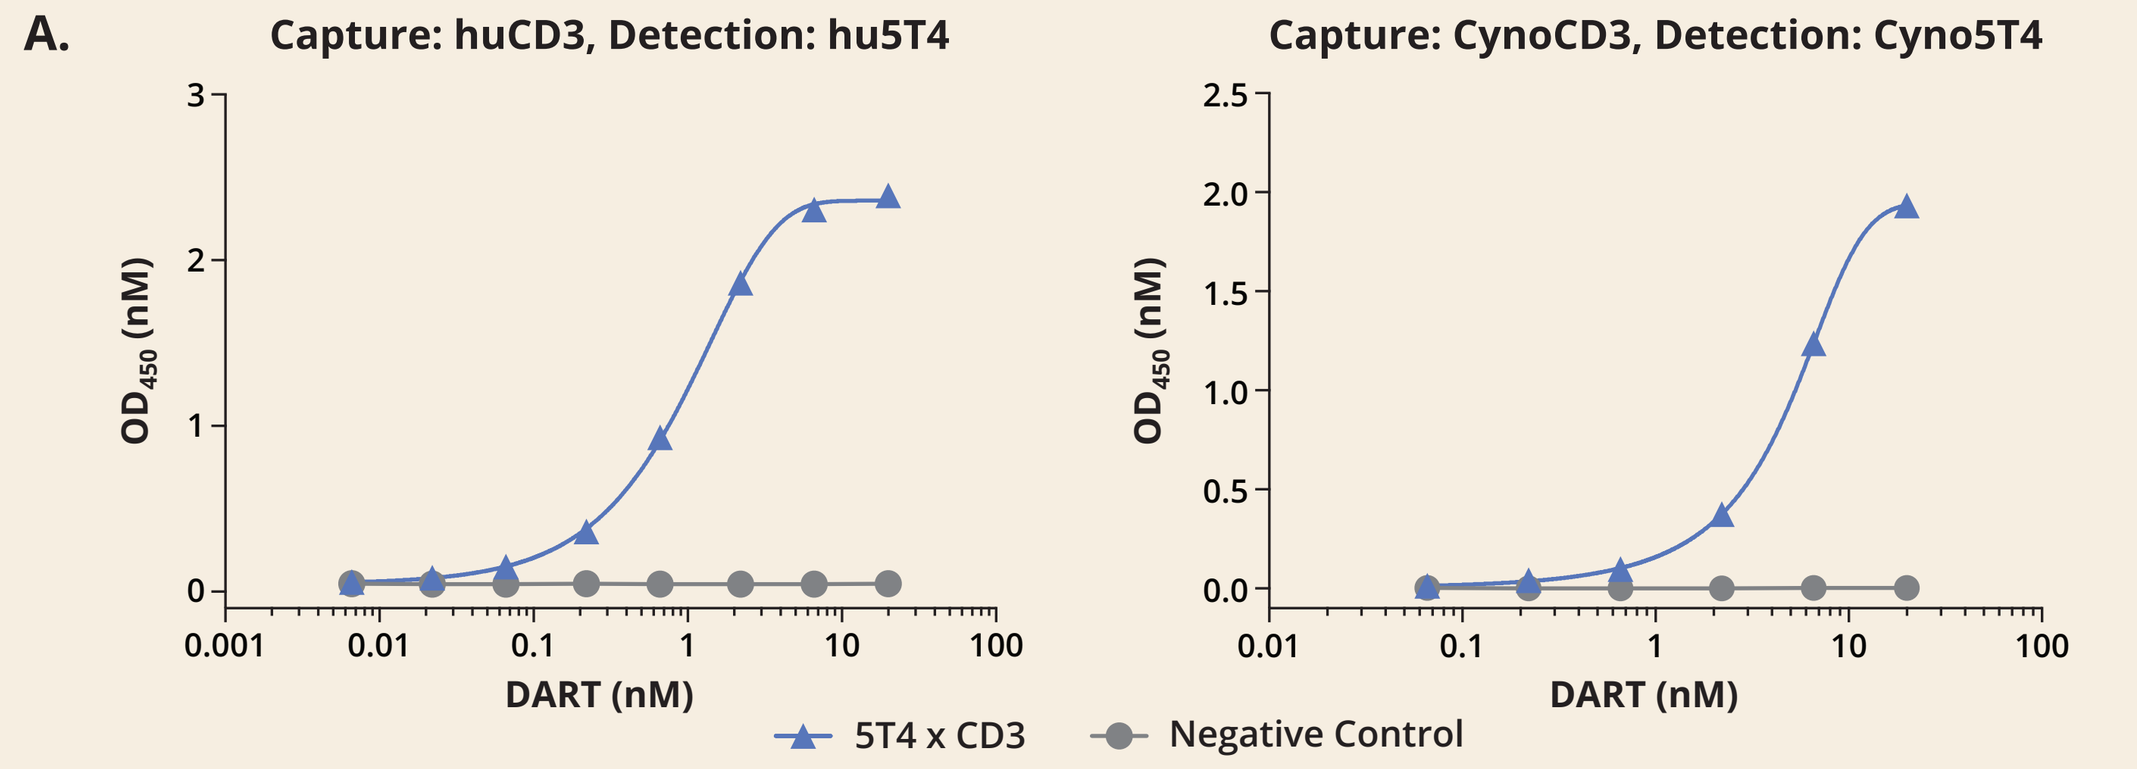
A.
Capture: huCD3, Detection: hu5T4
Capture: CynoCD3, Detection: Cyno5T4
OD450 (nM)
DART (nM)
5T4 x CD3
Negative Control

B.

Binding of 5T4 x CD3 DART to Human and Cynomolgus Monkey CD3 and 5T4			
Antigen	Association Rate Constant (k_a) ($M^{-1}S^{-1}$)	Dissociation Rate Constant (k_d) (S^{-1})	Equilibrium Dissociation Constant (K_D) (nM)
Human CD3	2.1 x 10^5	2.6 x 10^{-3}	12.4
Cyno CD3	1.9 x 10^5	2.2 x 10^{-3}	11.6
Human 5T4	3.5 x 10^6	1.0 x 10^{-3}	0.29
Cyno 5T4	2.5 x 10^6	15 x 10^{-3}	6.0

A. Bispecific ELISA demonstrates simultaneous engagement of both target antigens by 5T4 x CD3 DART.
B. Equilibrium dissociation constants (K_D) for the binding of 5T4 x CD3 DART to human and cynomolgus (Cyno) monkey CD3 and 5T4 determined by surface plasmon resonance analysis.

5T4 x CD3 DART-mediated CTL Activity Against Tumor Cell Lines

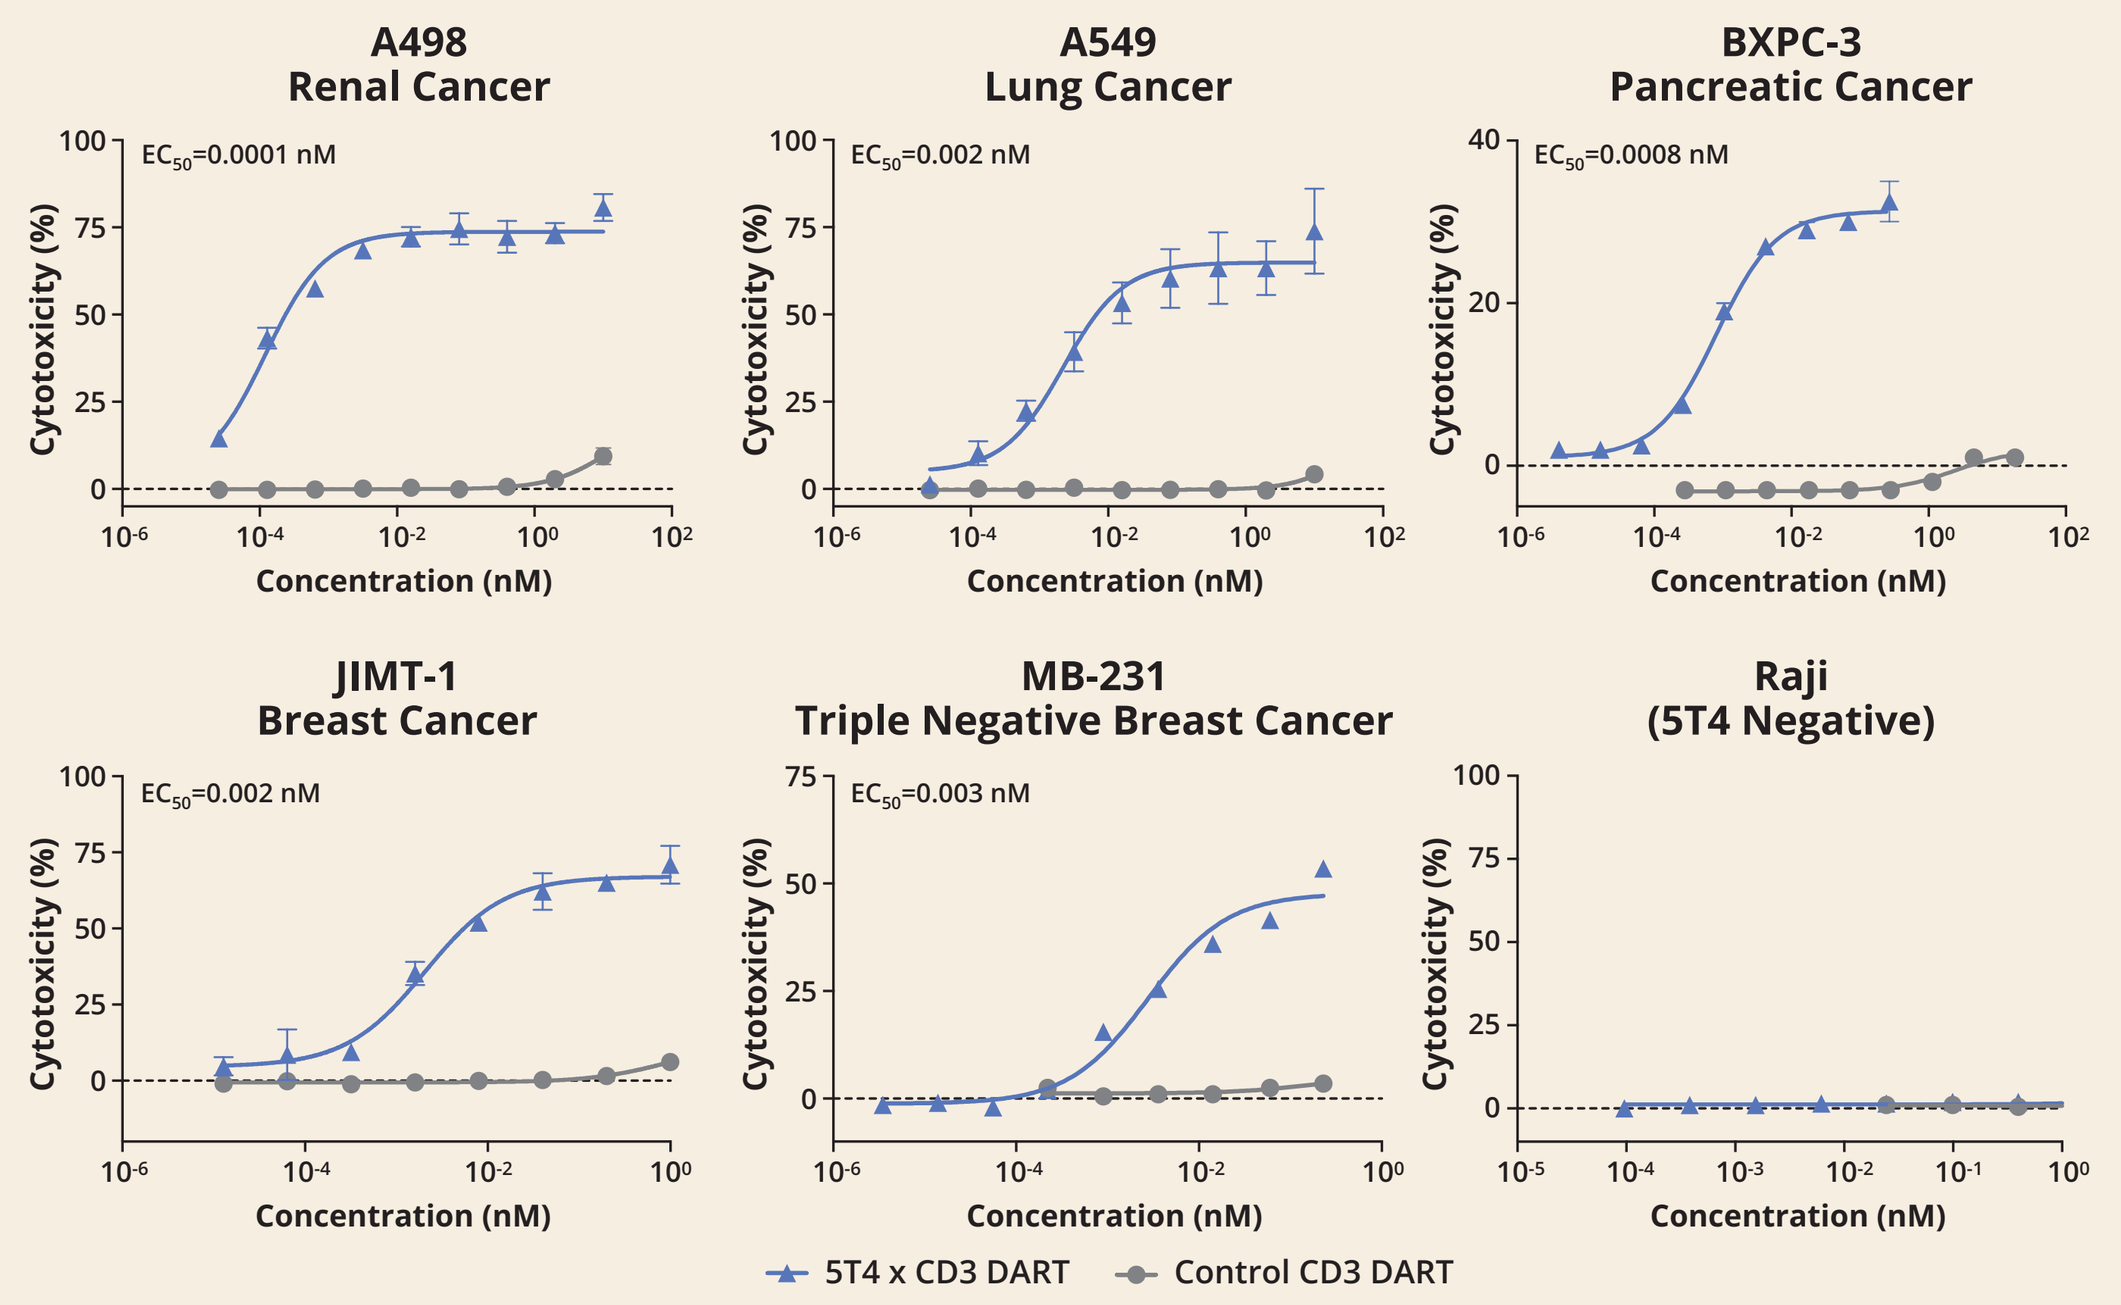
A498 Renal Cancer
EC_{50}=0.0001 nM
A549 Lung Cancer
EC_{50}=0.002 nM
BXPC-3 Pancreatic Cancer
EC_{50}=0.0008 nM
JIMT-1 Breast Cancer
EC_{50}=0.002 nM
MB-231 Triple Negative Breast Cancer
EC_{50}=0.003 nM
Raji (5T4 Negative)
Cytotoxicity (%)
Concentration (nM)
5T4 x CD3 DART
Control CD3 DART

Redirected cell killing (LDH method) by 5T4 x CD3 DART against 5T4-positive cancer cells. Experiments performed with human T cells (E:T=10:1) incubated for 24 hours.

T-cell Activation by 5T4 x CD3 DART Requires Target Engagement

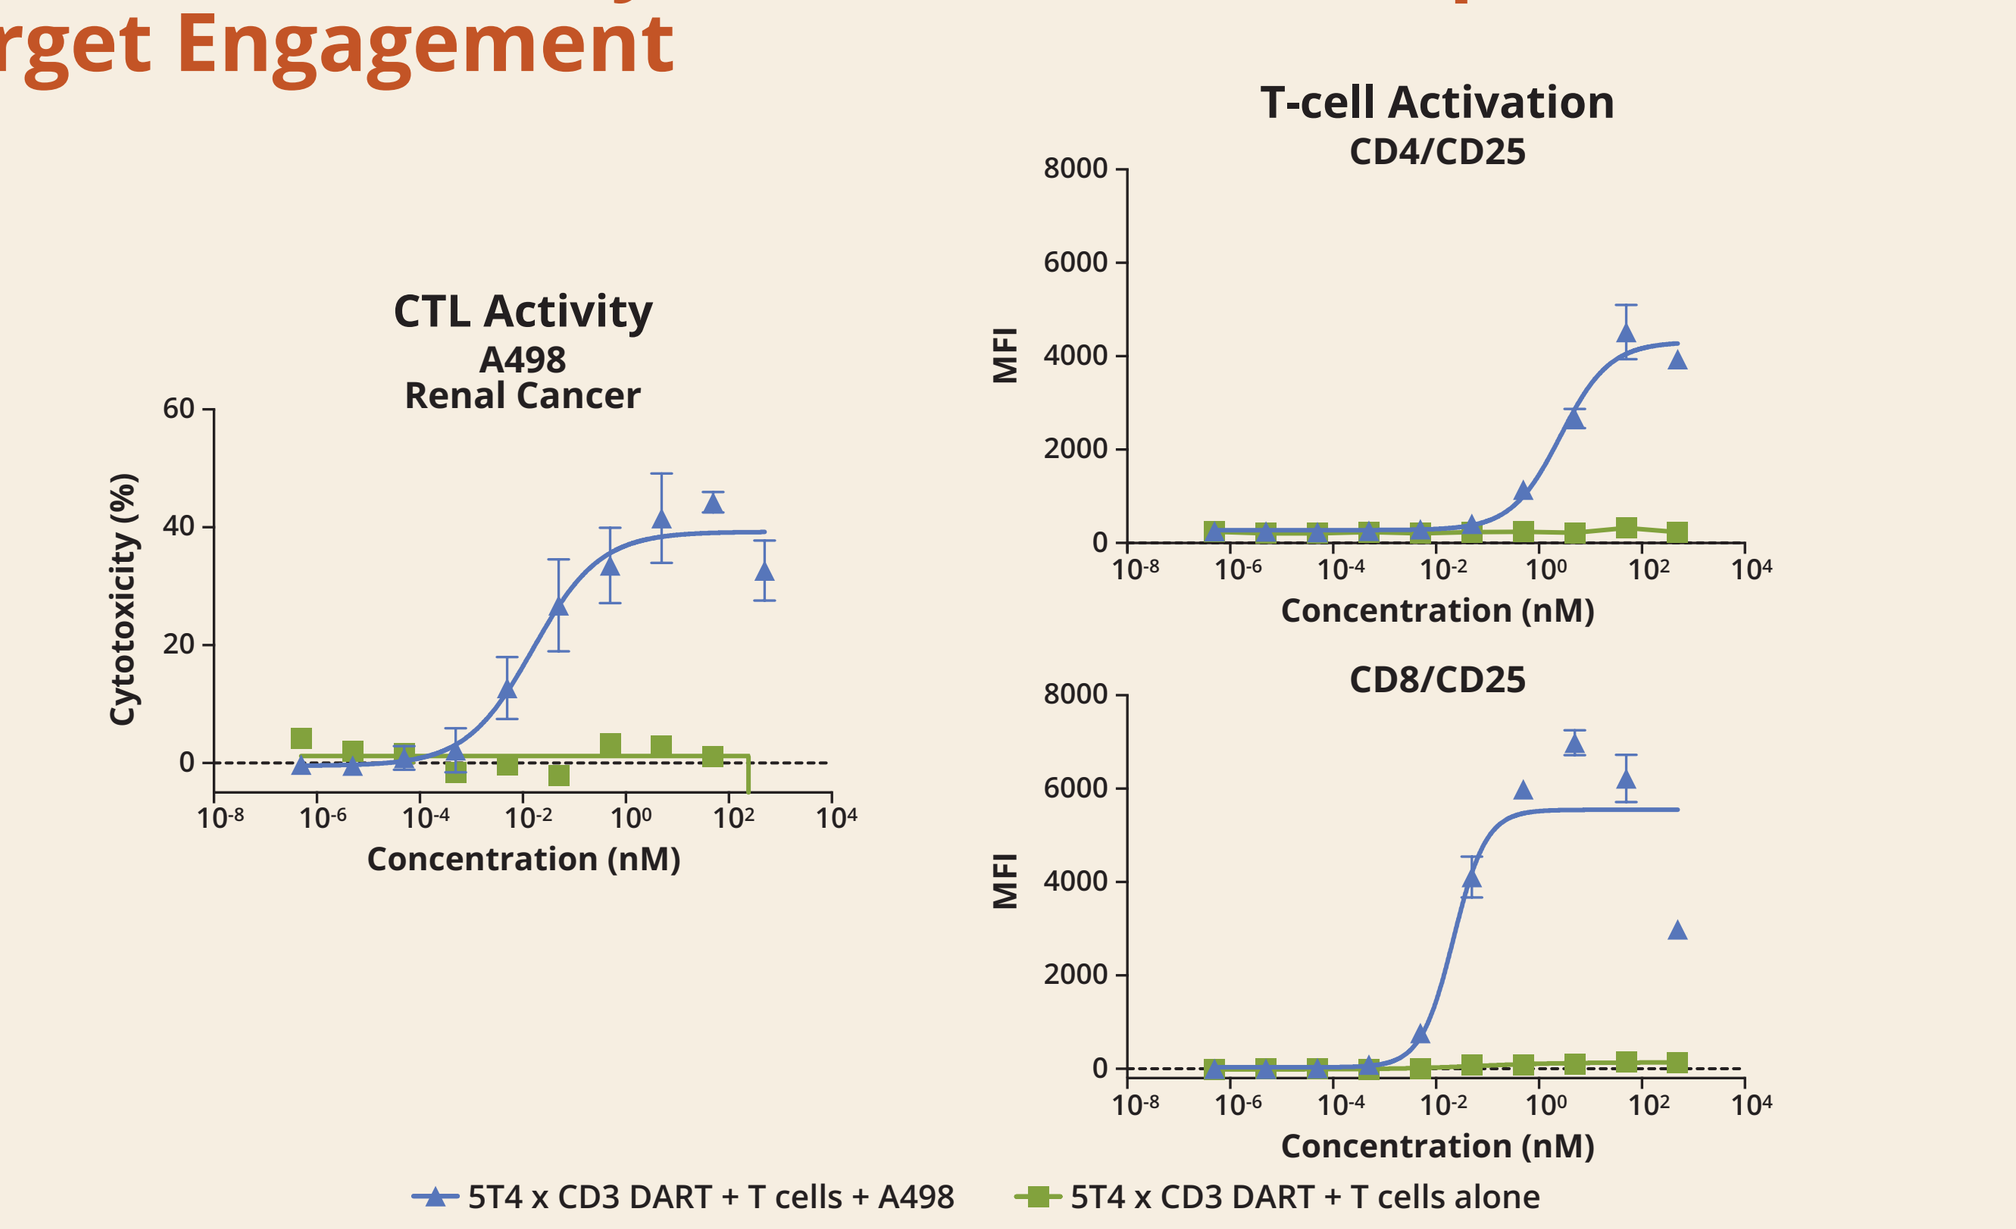
CTL Activity
A498
Renal Cancer
Cytotoxicity (%)
T-cell Activation
CD4/CD25
CD8/CD25
MFI
Concentration (nM)
5T4 x CD3 DART + T cells + A498
5T4 x CD3 DART + T cells alone

Redirected cytotoxicity (left panel) and T-cell activation (right panels) by 5T4 x CD3 DART in the absence or presence of target cells (Renal carcinoma cells A498). Human primary T cells were incubated with 5T4 x CD3 DART in the presence or absence of target cells (E:T=5:1) for 24 h. T-cell activation markers were measured by FACS analysis. Cytotoxicity was determined by LDH release assay.

Regression of Established A498 RCC in Human PBMC-reconstituted Mice

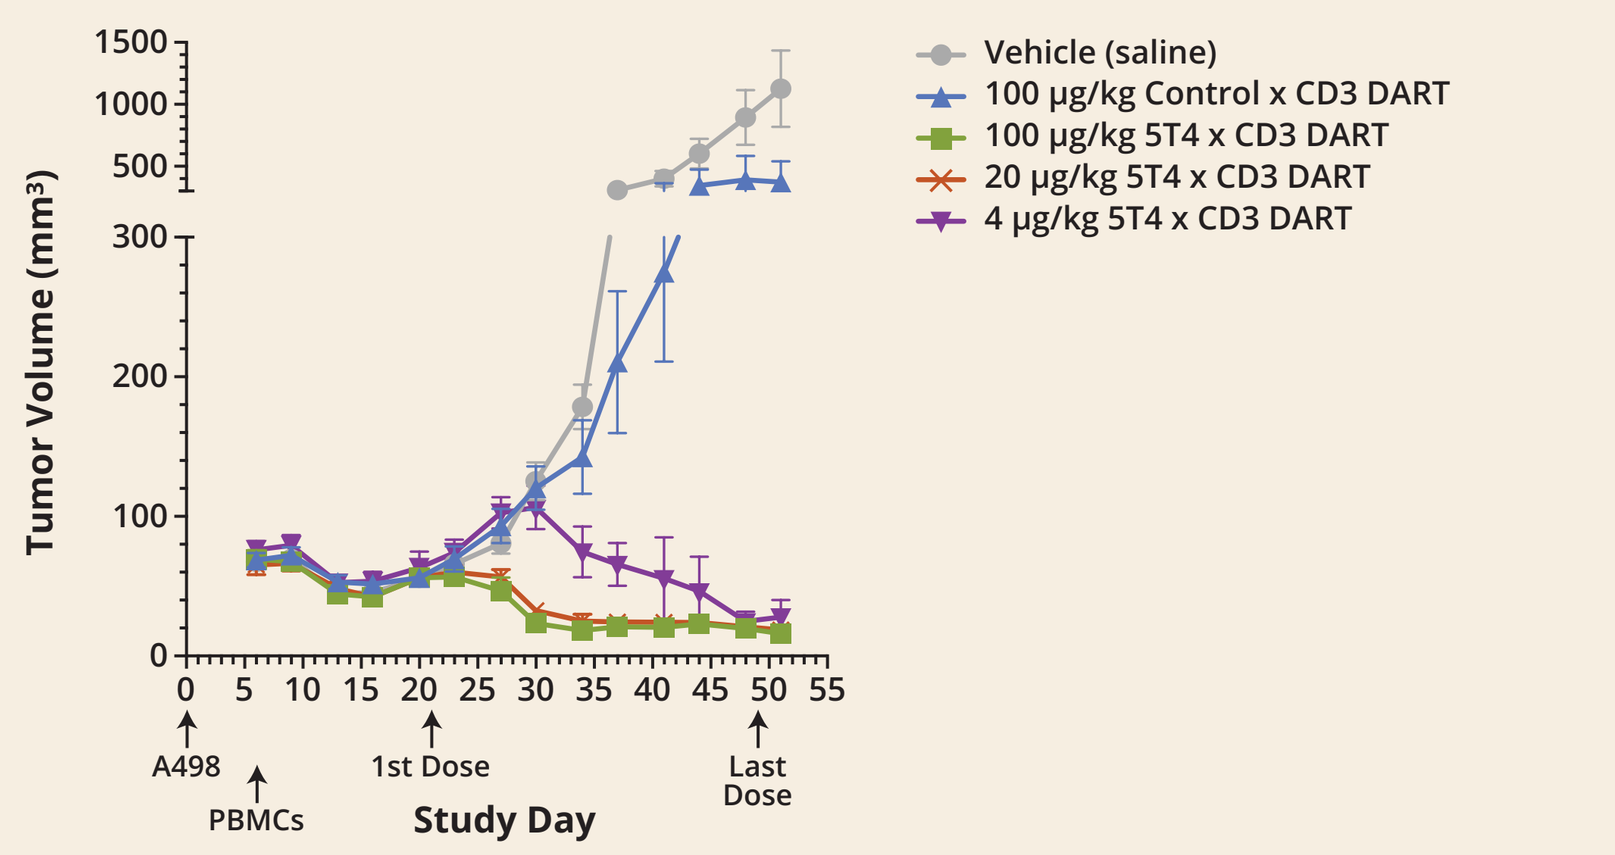
Vehicle (saline)
100 μg/kg Control x CD3 DART
100 μg/kg 5T4 x CD3 DART
20 μg/kg 5T4 x CD3 DART
4 μg/kg 5T4 x CD3 DART
Tumor Volume (mm³)
A498
PBMCs
1st Dose
Last Dose
Study Day

NOD/SCID mice (n=8/group) were injected with tumor cells on Day 0 (intradermal, A498 cells 5 x 10^6), reconstituted with PBMCs (10 x 10^6) on Day 6 (IP), and treated as indicated starting on Day 21 (IV, Q4Dx9 schedule).

Antitumor Activity in an Orthotopic TNBC Model

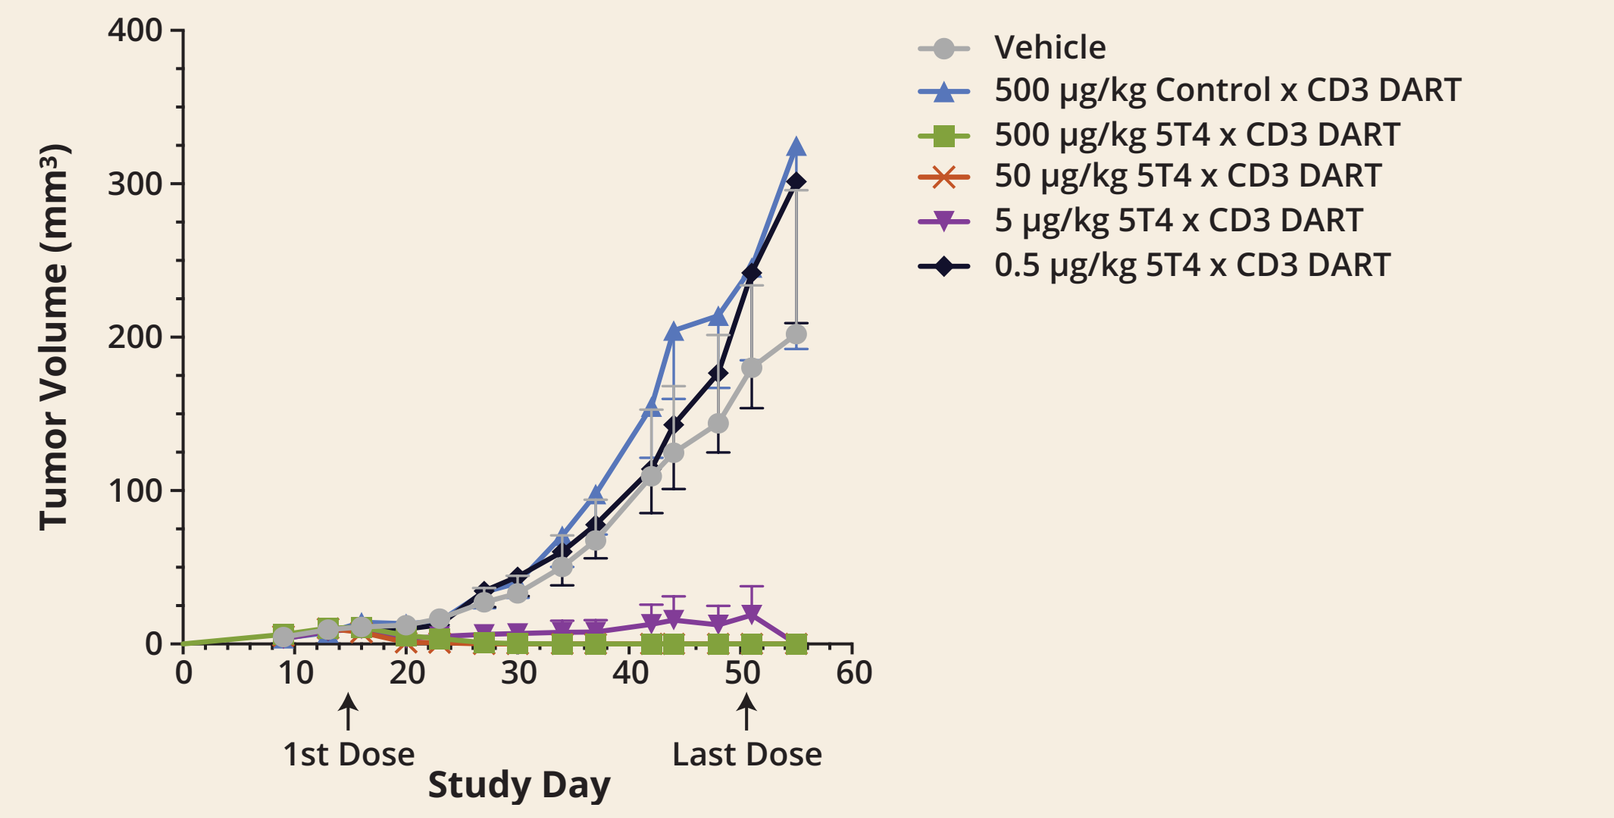
Vehicle
500 μg/kg Control x CD3 DART
500 μg/kg 5T4 x CD3 DART
50 μg/kg 5T4 x CD3 DART
5 μg/kg 5T4 x CD3 DART
0.5 μg/kg 5T4 x CD3 DART
Tumor Volume (mm³)
1st Dose
Last Dose
Study Day

$MHCI^{-/-}$ mice were reconstituted with PBMCs (10 x 10^6) (IP) and implanted orthotopically with TNBC MB-231 cells (1 x 10^6) (OT) on Day 0. Following randomization, mice (n=7/group) were first treated (IV) on Day 15 and at weekly intervals thereafter for a total of 6 doses.

Antitumor Activity in an Orthotopic Pancreatic Cancer Model

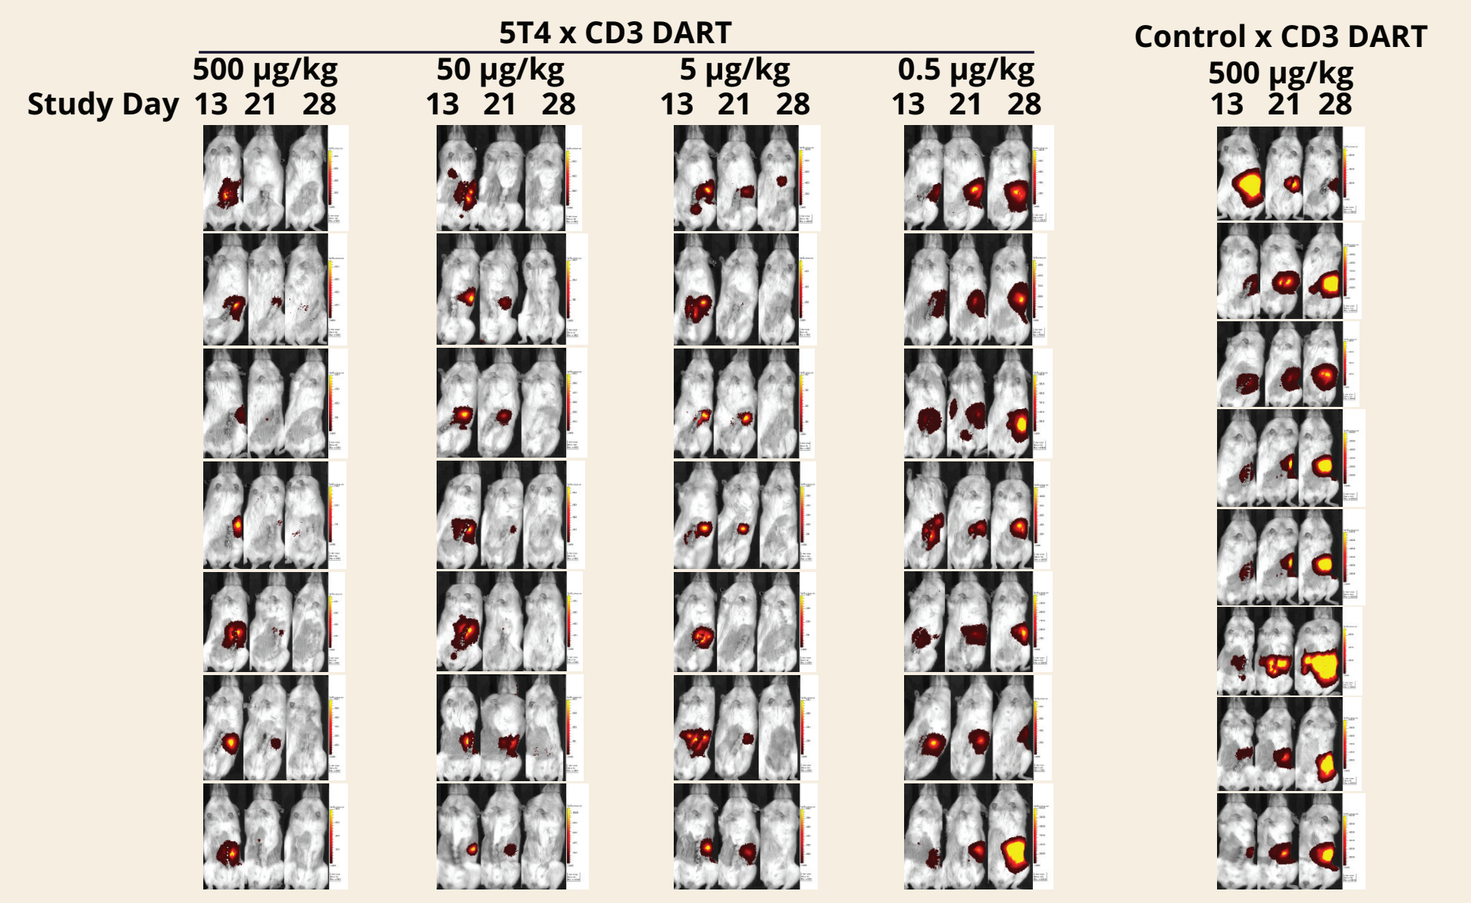
5T4 x CD3 DART
Control x CD3 DART
Study Day
500 μg/kg
50 μg/kg
5 μg/kg
0.5 μg/kg
13 21 28

$MHCI^{-/-}$ mice were reconstituted with PBMCs (10 x 10^6) (IP) on Day 0 and implanted BXPC3 cells (1 x 10^6) (OT) on Day 8. Following randomization, mice (n=7/group) were first treated (IV) on Day 15 and at weekly intervals thereafter for a total of 6 doses.

5T4 x CD3 DART Promotes T-cell Recruitment into Lung Cancer Mass

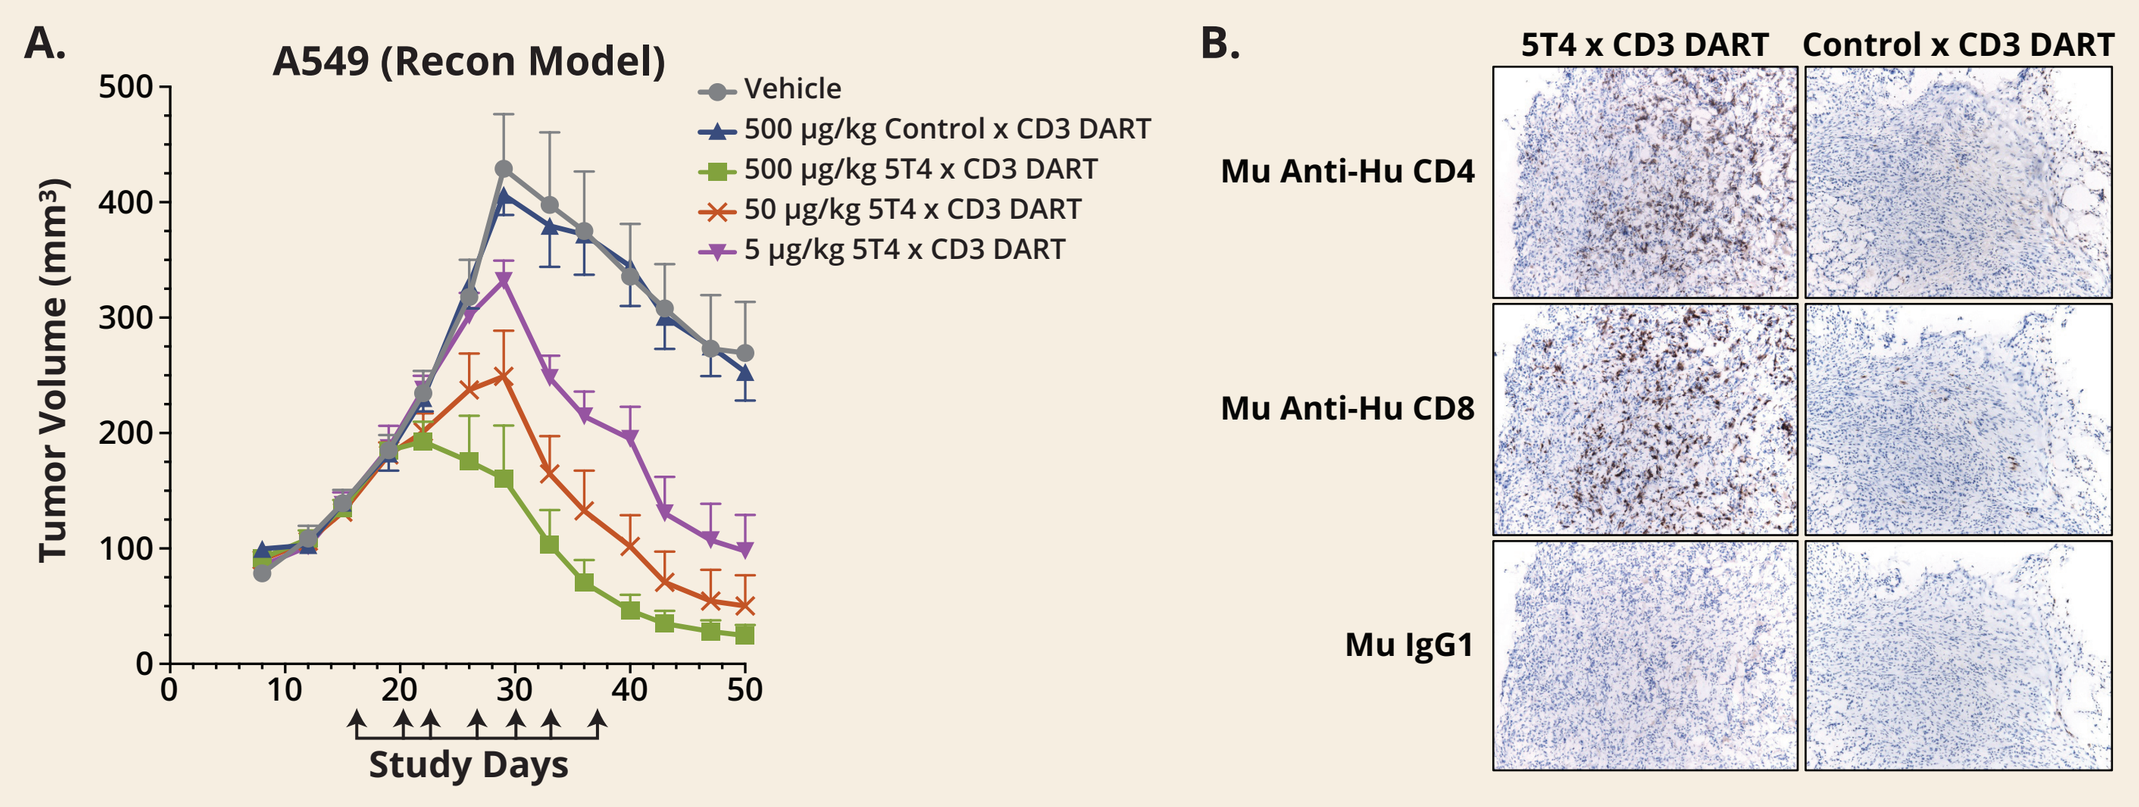
A.
A549 (Recon Model)
Vehicle
500 μg/kg Control x CD3 DART
500 μg/kg 5T4 x CD3 DART
50 μg/kg 5T4 x CD3 DART
5 μg/kg 5T4 x CD3 DART
Tumor Volume (mm³)
Study Days
B.
5T4 x CD3 DART
Control x CD3 DART
Mu Anti-Hu CD4
Mu Anti-Hu CD8
Mu IgG1

A. $B2M^{-/-}$ mice (n = 7/group) were reconstituted with PBMCs (10 x 10^6) (IP) on Day 0 and implanted A549 lung cancer cells (5 x 10^6) (ID) on Day 1. 5T4 x CD3 or control DART was administered IV every 3 or 4 days for total 7 doses starting on Day 16 (arrows).
B. IHC using mouse anti-human CD4 or CD8 mAb in A549 lung cancer mass from mice treated with 5T4 x CD3 (50 μg/kg) or control (500 μg/kg) DART (**A**) after 72 hours.

5T4 x CD3 DART Displays Prolonged PK in Cynomolgus Monkeys

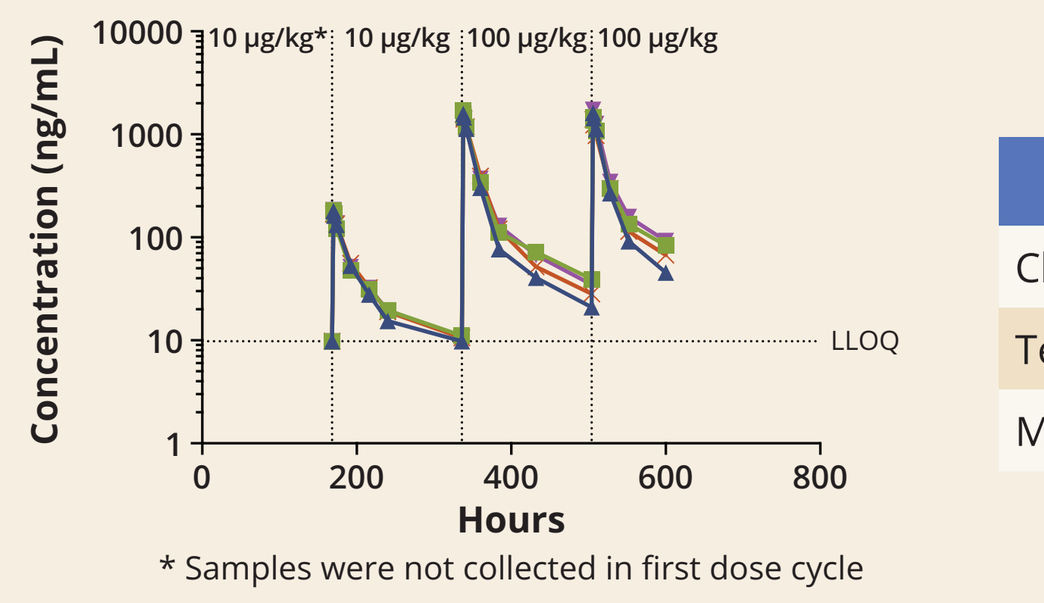
10 μg/kg*
10 μg/kg
100 μg/kg
100 μg/kg
Concentration (ng/mL)
LLOQ
Hours

* Samples were not collected in first dose cycle

PK Parameters (Mean ± SEM) (10 μg/kg)	
Clearance	1.7 ± 0.52 mL/h/Kg
Terminal $t_{1/2}$	4.3 ± 0.04 days
MRT	4.0 ± 0.58 days

- 4 weekly doses with dose escalation at 3rd and 4th week of 5T4 x CD3 DART via 2-hour IV infusion were well tolerated
- Drug exposure increased proportionally with dose escalation

Tissue Cross-reactivity of 5T4 x CD3 DART

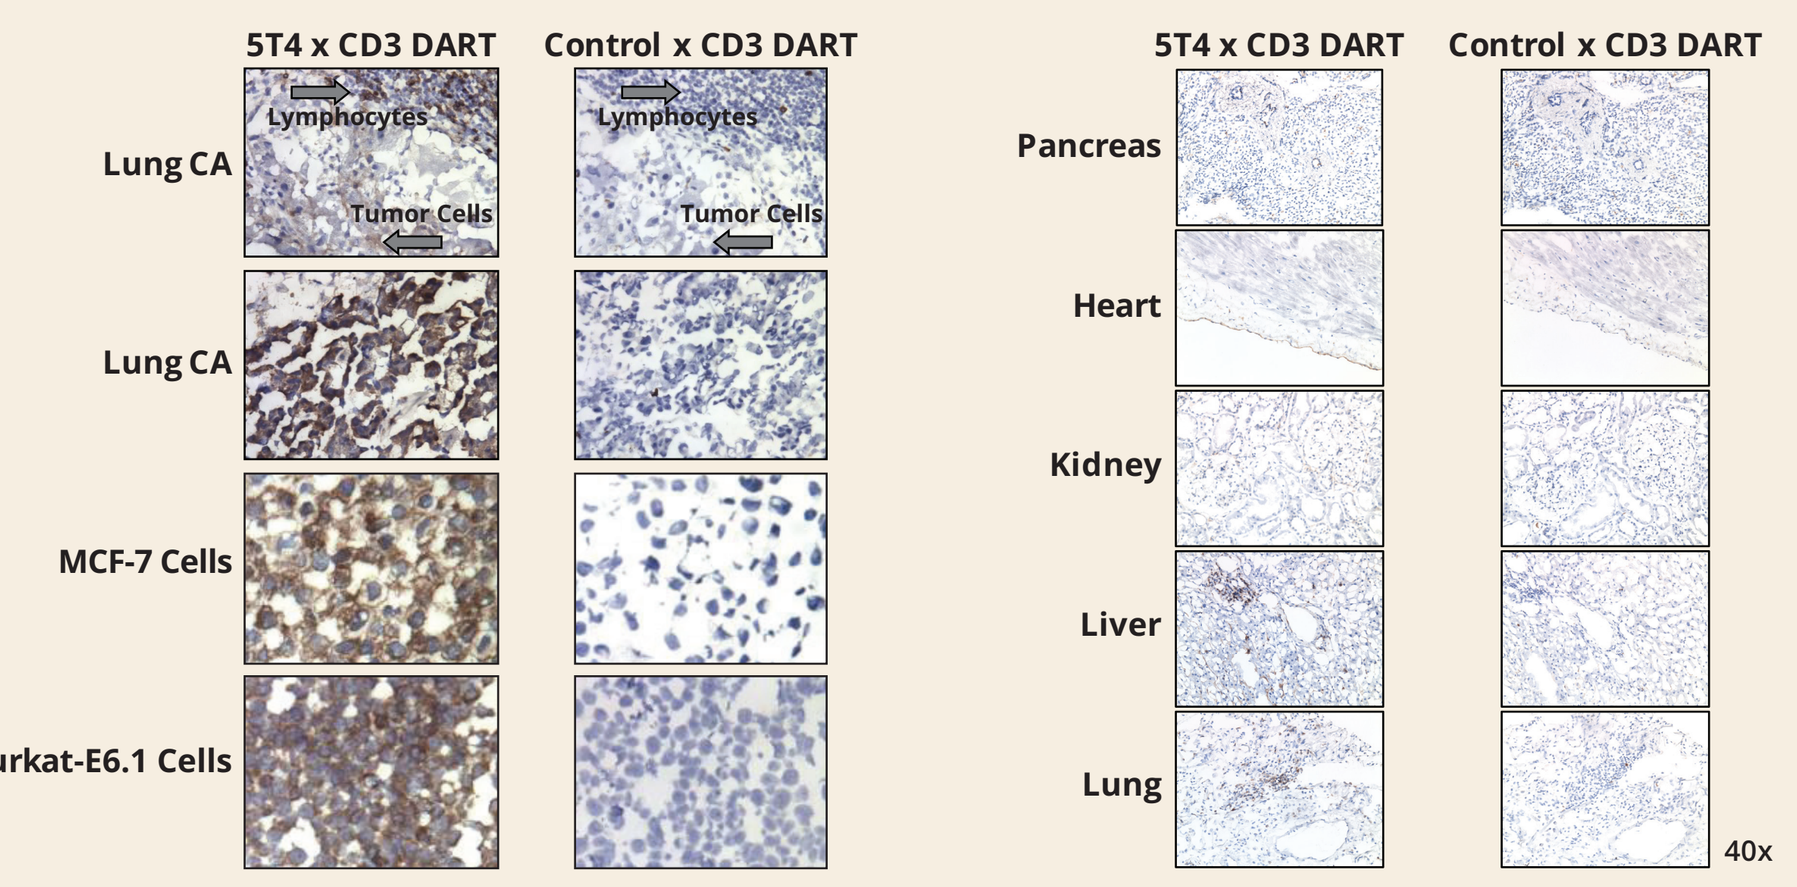
5T4 x CD3 DART
Control x CD3 DART
Lung CA
Lymphocytes
Tumor Cells
MCF-7 Cells
Jurkat-E6.1 Cells
Pancreas
Heart
Kidney
Liver
Lung
40x

- 5T4 x CD3 DART exhibits strong tumor vs. normal tissue reactivity

Conclusions

- The oncofetal trophoblast glycoprotein 5T4 is overexpressed in a range of tumors
 - 5T4 shows limited adult tissue expression with strong tumor/normal differential
- A bispecific Fc-bearing 5T4 x CD3 DART was generated to specifically target T cells to 5T4-expressing tumors
- 5T4 x CD3 DART exhibited:
 - Dose-dependent redirected cell killing against a wide range of solid tumor cell lines in vitro
 - T–cell activation strictly dependent on co-engagement of 5T4 x CD3 DART with 5T4-expressing target cells
 - Efficacy in multiple established tumor models (renal, pancreatic, and triple negative breast cancer) in human PBMC-reconstituted mice
 - 5T4 x CD3 DART promoted T-cell recruitment to tumor xenograft in mouse model
 - 4 weekly doses with dose escalation up to 100 μg/kg were well tolerated in cynomolgus monkeys
 - Extended circulating half-life in nonhuman primates (~4.3 days) supports feasibility of convenient clinical dosing

These data support further investigation of 5T4 x CD3 DART as a potential candidate for the treatment of 5T4-expressing cancers

References

1. Ward, C. M. J. Cell Sci. (2003), 116:4533-4542. **2.** Southall, P.J. et. al. Br. J. Cancer (1990), 61: 89-95.
3. Eisen, T. et. al. Curr Oncol Rep (2014) 16:370.

Acknowledgment

We thank Sharad Sharma for providing cynomolgus monkeys PK analysis, Anushka De Costa and Nicholas Yee-Toy for their assist in IHC images, and Arash Adami and Judy Downey for their help in generating the poster.

Presented at the American Association for Cancer Research Annual Meeting 2017, April 1–5, 2017, Washington, DC

©2017 MacroGenics, Inc. All rights reserved.