Target Validation, Antibody Discovery and Preclinical Data Supporting ADAM9 as an Antibody-Drug Conjugate Therapeutic Target for Solid Tumors

38

Juniper A. Scribner, Bhaswati Barat, Stuart W. Hicks, Nicholas C. Yoder, Thomas Son, Lusiana Widjaja, Gundo Diedrich, Sergey Gorlatov, Jeff Hooley, Ann Easton, Peter Lung, Anushka De Costa, Francine Chen, Michael Chiechi, Pam Li, Monica Licea, Timothy E. Hotaling, Michael Spliedt, Valentina Ciccarone, Nadia Gantt, James Tamura, Megan E. Fuller, Molly A. McShea, Scott Koenig, Syd Johnson, Paul A. Moore, Ezio Bonvini, Deryk Loo

MacroGenics, Inc., Rockville, MD and South San Francisco, CA; ImmunoGen, Inc., Waltham, MA

http://ir.macrogenics.com/events.cfm

Abstract

Introduction: A target-unbiased approach based on intact cell immunizations with fetal progenitor cells and cancer stem cells, followed by an immunohistochemistry (IHC) screen for cancer-specific candidates, led to the identification of anti-ADAM9 (a disintegrin and metalloproteinase) mAbs with highly differential tumor-versus-normal tissue binding. ADAM9 is a cell surface protein overexpressed in multiple tumors, with a possible role in promotion and progression of cancer through multiple mechanisms, including modulation of adhesion and migration as well as processing of tumorigenic and pro-angiogenic factors. In this preclinical study, we performed target/mAb validation and evaluated the therapeutic potential of anti-ADAM9 antibody-drug conjugates (ADCs) toward ADAM9-expressing solid cancers.

Methods: IHC was performed with anti-ADAM9 mAbs to confirm and extend available data of human normal and tumor tissue expression. Epitope mapping studies were conducted to define epitope-specificity. mAbs were also screened to identify those that efficiently internalized into tumor cells. *In vitro* cellular processing studies were performed to further evaluate the mAbs as ADC candidates. Selected mAbs were converted to ADCs via chemical conjugation to potent anti-microtubule (DM4) or DNA alkylating (DGN549) agents; *in vitro* cytotoxicity studies were conducted with tumor cell lines representing human cancer types that overexpress ADAM9. A lead mAb was then selected for humanization and affinity maturation to yield a development candidate.

Results: Anti-ADAM9 mAbs exhibited strong reactivity toward the tumor epithelium of solid cancers, including pancreatic, kidney, prostate, bladder, breast, colon, lung, and ovarian cancer, but limited reactivity toward normal tissues. Anti-ADAM9 mAbs were efficiently internalized and processed by tumor cell lines, including lines with only modest ADAM9 expression. Anti-ADAM9 ADCs exhibited specific, dose-dependent cytotoxicity toward ADAM9-positive cancer cell lines *in vitro*, with IC_{50} values in the sub-nanomolar range. Humanization and affinity maturation of the lead mAb yielded a development candidate that retains potent antitumor activity toward ADAM9-positive tumor cell lines and equivalent, high affinity binding to both human and cynomolgus monkey ADAM9.

Conclusion: ADAM9 is a cell surface antigen that is overexpressed on a wide range of solid tumors. Anti-ADAM9 mAbs that were strongly reactive with representative tumors exhibited high affinity for the antigen and were efficiently internalized and processed by ADAM9-bearing tumor cells. Anti-ADAM9 ADCs demonstrated dose-dependent cytotoxicity *in vitro* toward a panel of ADAM9-positive tumor cell lines. Our findings demonstrate that an ADC targeting ADAM9 may serve as a potential therapeutic for ADAM9-expressing solid tumors.

Background

- Intact cell immunizations of mice with viable human fetal progenitor cells or tumor initiating/cancer stem-like cells were used to generate a panel of mAbs reactive with cancer cells
- An IHC screen for cancer specific mAbs identified a panel with high tumor-versus-normal differential
- Immunoprecipitation/mass spectrometry analysis of the panel identified mAbs that had specificity towards ADAM9

Objectives:

- Validate ADAM9 as a therapeutic target
- Evaluate the therapeutic potential of anti-ADAM9 antibody-drug conjugates

Introduction

Antibody/Target Discovery Platform

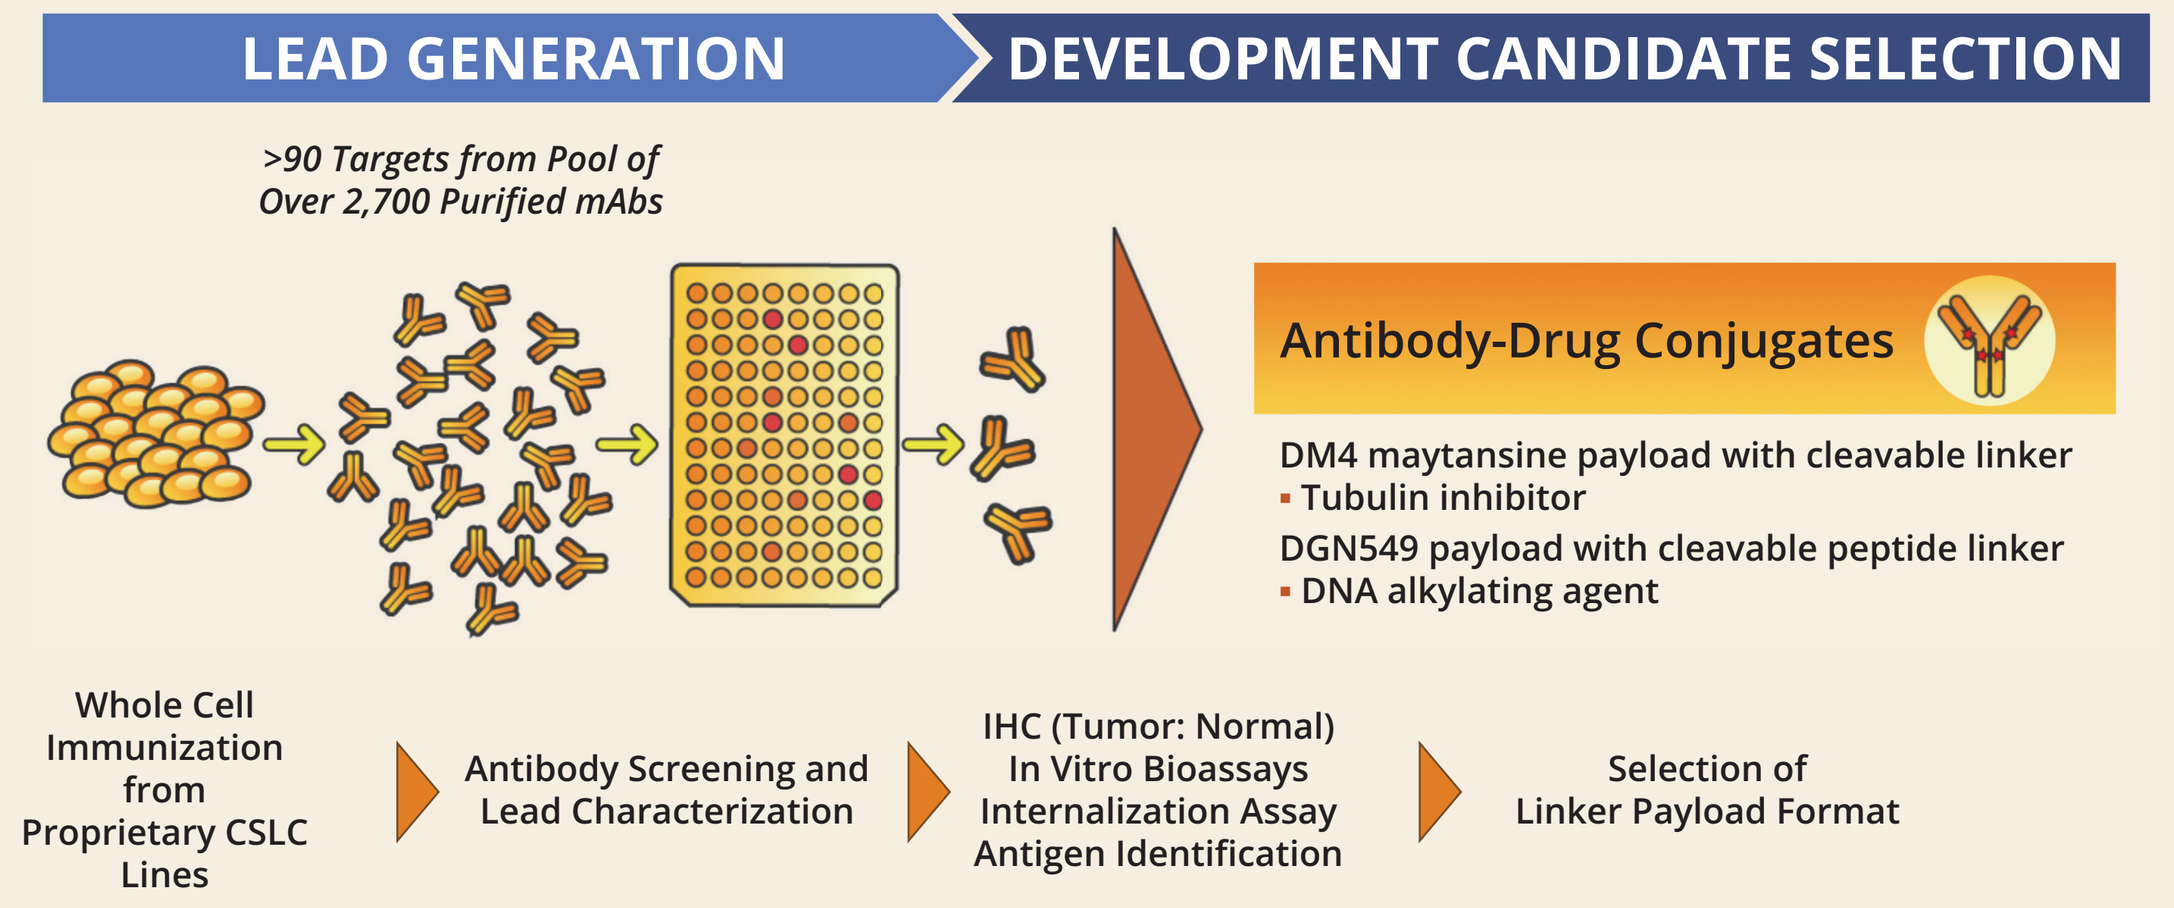
LEAD GENERATION
DEVELOPMENT CANDIDATE SELECTION
>90 Targets from Pool of Over 2,700 Purified mAbs
Antibody-Drug Conjugates
DM4 maytansine payload with cleavable linker
Tubulin inhibitor
DGN549 payload with cleavable peptide linker
DNA alkylating agent
Whole Cell Immunization from Proprietary CSLC Lines
Antibody Screening and Lead Characterization
IHC (Tumor: Normal) In Vitro Bioassays Internalization Assay Antigen Identification
Selection of Linker Payload Format

Immunizations with intact viable fetal progenitor and tumor initiating/cancer stem-like cells were performed as previously described (Loo D, et al. Mol Cancer Ther. 2007 Mar;6(3):856-65).

IHC screening on Frozen Sections of Human Tissue Specimens

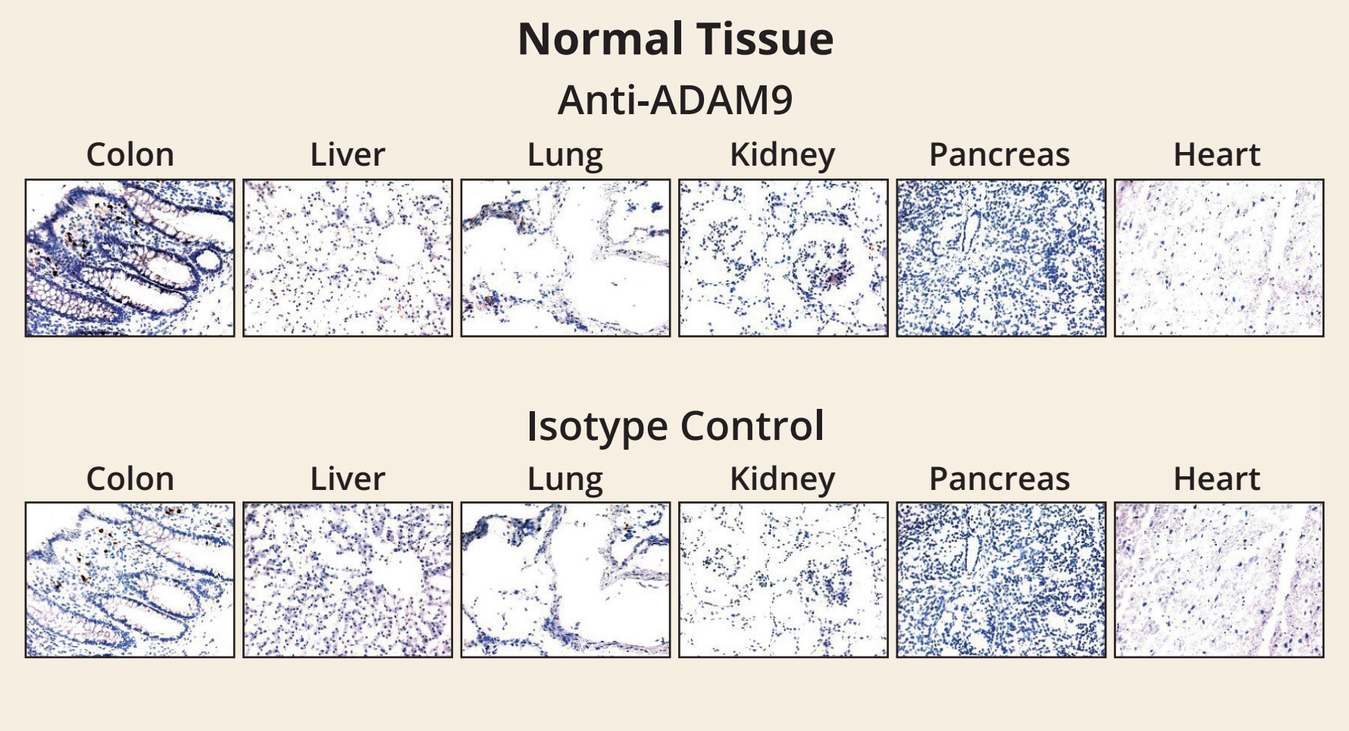
Normal Tissue
Anti-ADAM9
Colon
Liver
Lung
Kidney
Pancreas
Heart
Isotype Control

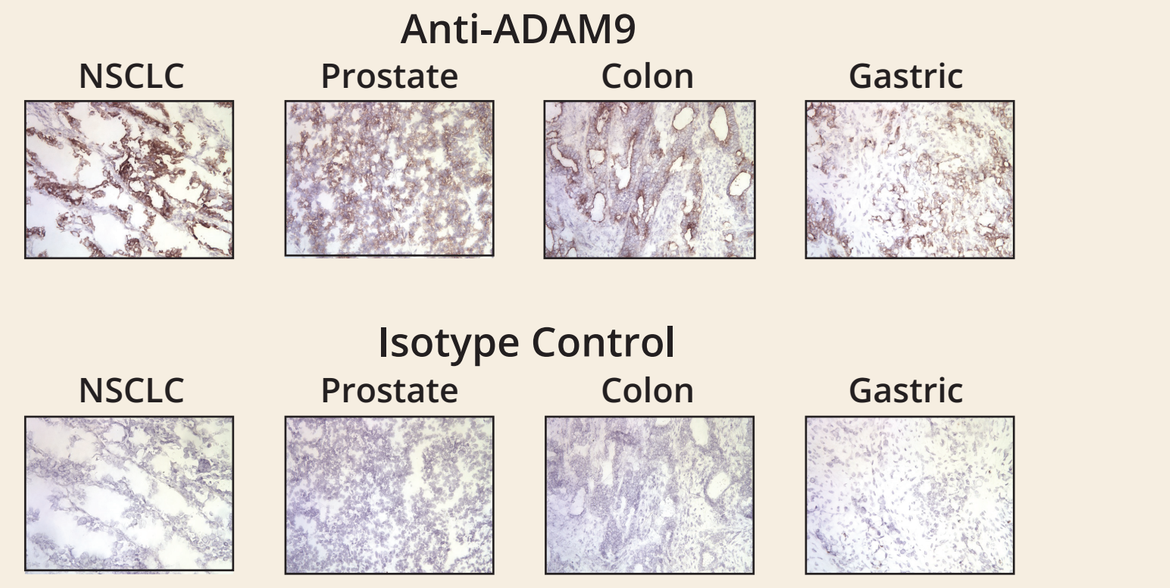
Tumor Tissue
Anti-ADAM9
NSCLC
Prostate
Colon
Gastric
Isotype Control

Fab-ZAP Internalization Assay ADAM9 mAbs

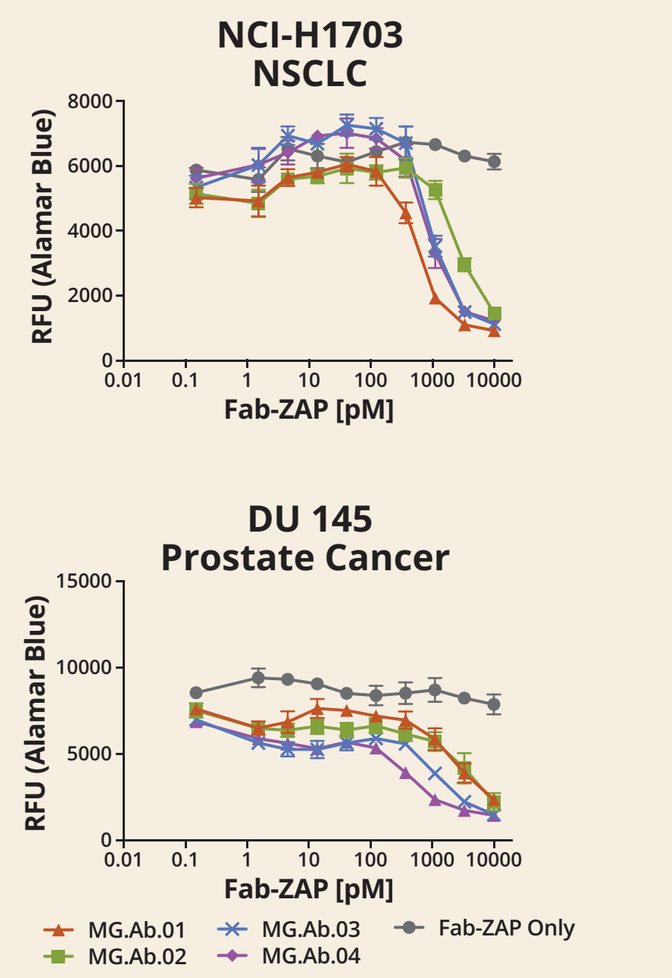
NCI-H1703 NSCLC
DU 145 Prostate Cancer
RFU (Alamar Blue)
Fab-ZAP [pM]
MG.Ab.01
MG.Ab.02
MG.Ab.03
MG.Ab.04
Fab-ZAP Only

NCI-H1703 NSCLC

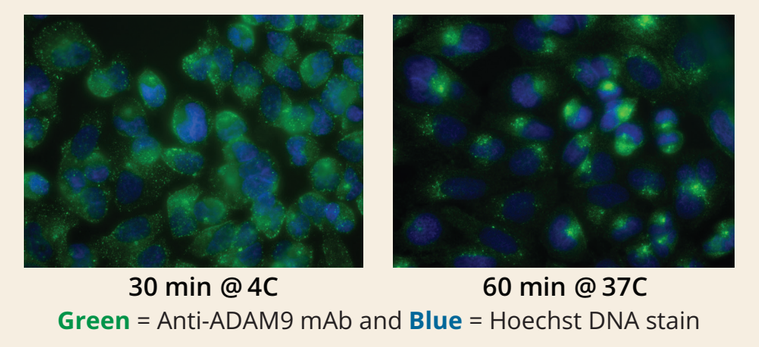
30 min @4C
60 min @37C
Green = Anti-ADAM9 mAb and Blue = Hoechst DNA stain

Internalization assay was performed in a 5 day assay using a saporin-conjugated anti-mouse Fab at 10:1 Fab-ZAP:Test mAb ratio. Internalization imaged by fluorescence microscopy with an Alexafluor488 conjugated anti-ADAM9 mAb.

- Tumor-specific, internalizing ADAM9 mAbs identified

ADAM9 – A Disintegrin and Metalloproteinase 9

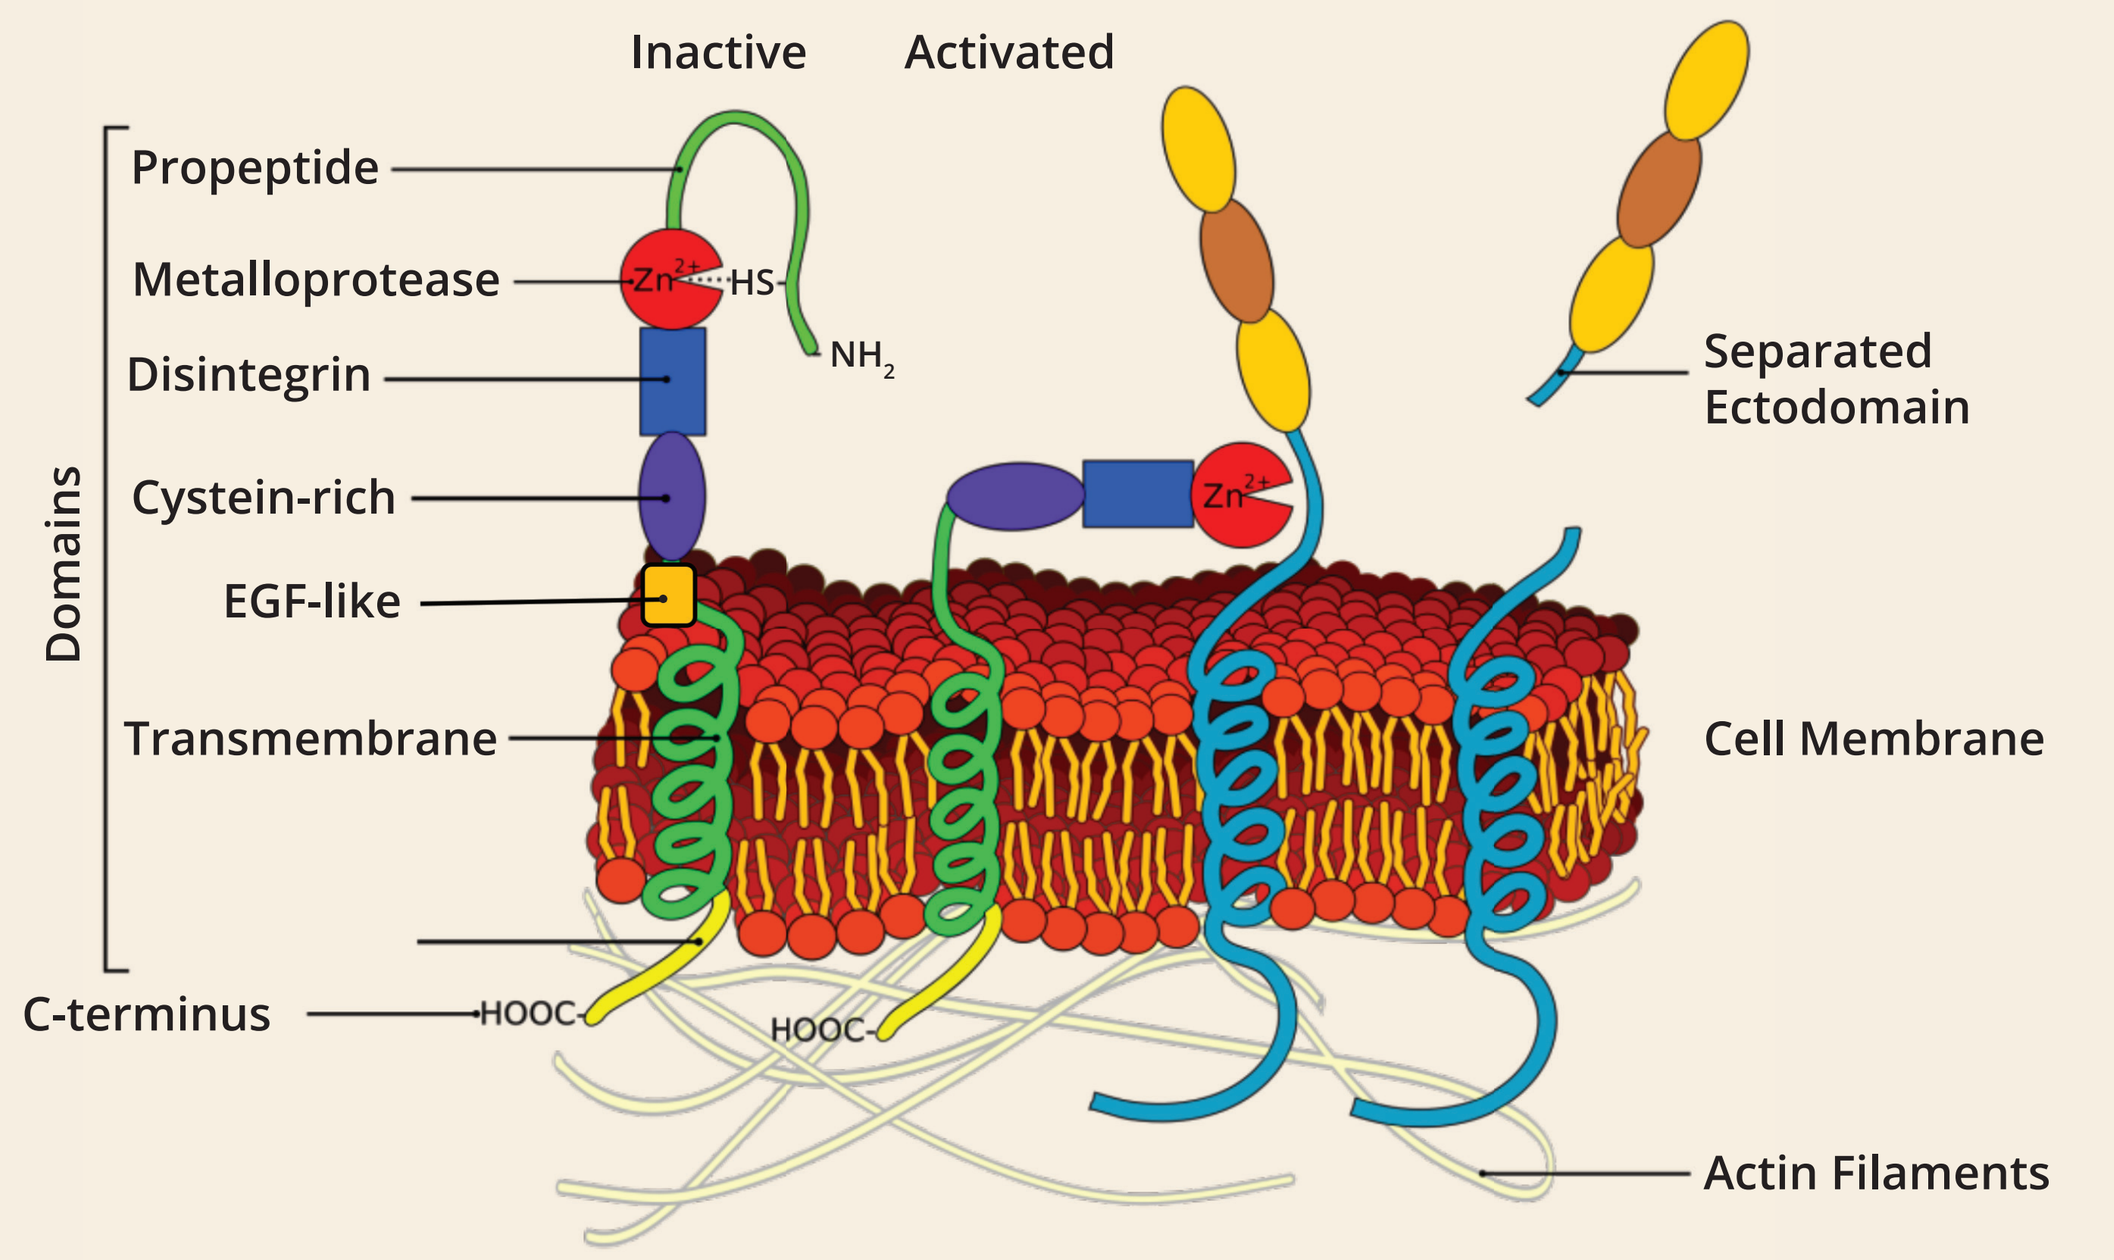
Sheddase
Membrane Protein
Inactive
Activated
Domains
Propeptide
Metalloprotease
Disintegrin
Cystein-rich
EGF-like
Transmembrane
C-terminus
HOOC
HS
NH2
Zn2+
Separated Ectodomain
Cell Membrane
Actin Filaments

- Role in the protein ectodomain shedding of membrane-bound molecules
- Regulates processing of tumorigenic and pro-angiogenic growth factors
- Evidence suggests a role in migration, adhesion, invasion and metastasis
- Overexpressed in multiple carcinomas, including NSCLC, colon, gastric, breast, liver, pancreatic, myeloma, prostate, melanoma, liver, ovarian, and esophageal cancers
 - Overexpression in prostate, breast and pancreatic cancers correlates with poor prognosis

Results

ADAM9 Is Highly Expressed on Range of Cancers

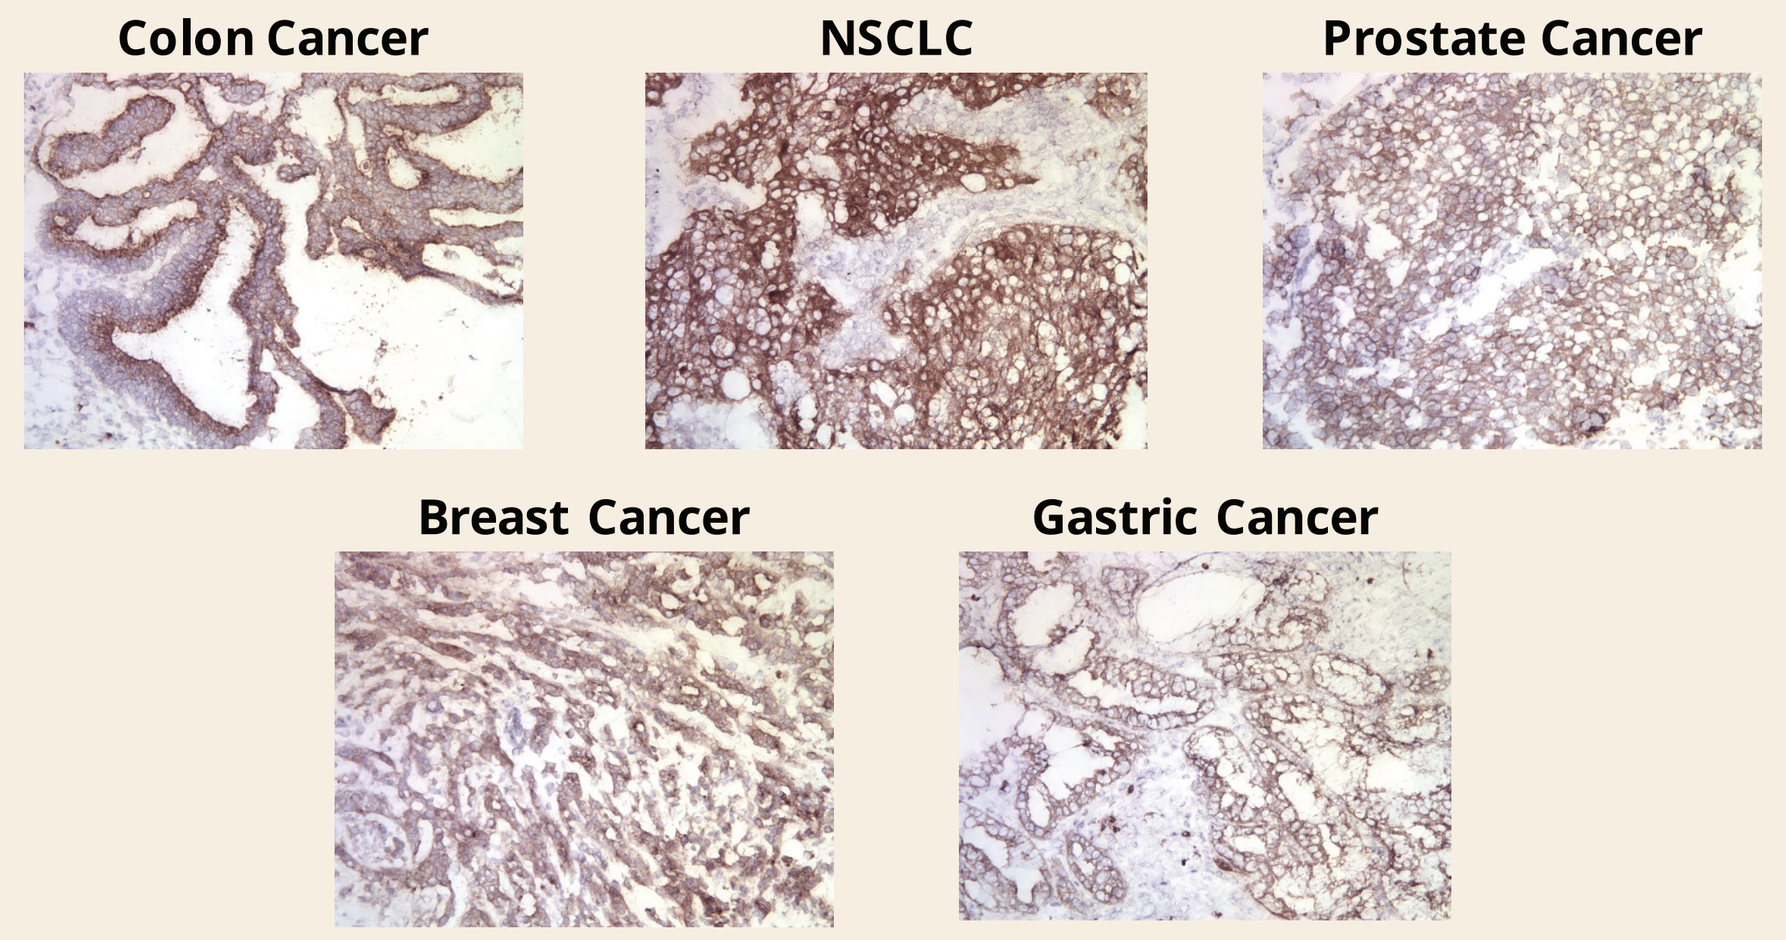
Colon Cancer
NSCLC
Prostate Cancer
Breast Cancer
Gastric Cancer

Cancer Type	IHC Summary of Tumor Samples Screened: ADAM9 Positive	
Colon Cancer	25/28	89%
Pancreatic Cancer	14/17	82%
Prostate Cancer	37/48	77%
Gastric Cancer	9/12	75%
Lung Cancer	38/53	72%
Kidney Cancer	4/10	40%
Ovarian Cancer	4/12	33%
Breast Cancer	7/25	28%

ADAM9 expression also observed in limited number of specimens from bladder, uterine, head and neck, and thyroid cancer. IHC staining was performed on frozen human tissue specimens with MG.Ab.01 mAb.

ADAM9 mAbs Bin Into Two Binding Groups

Cross Competition ELISA

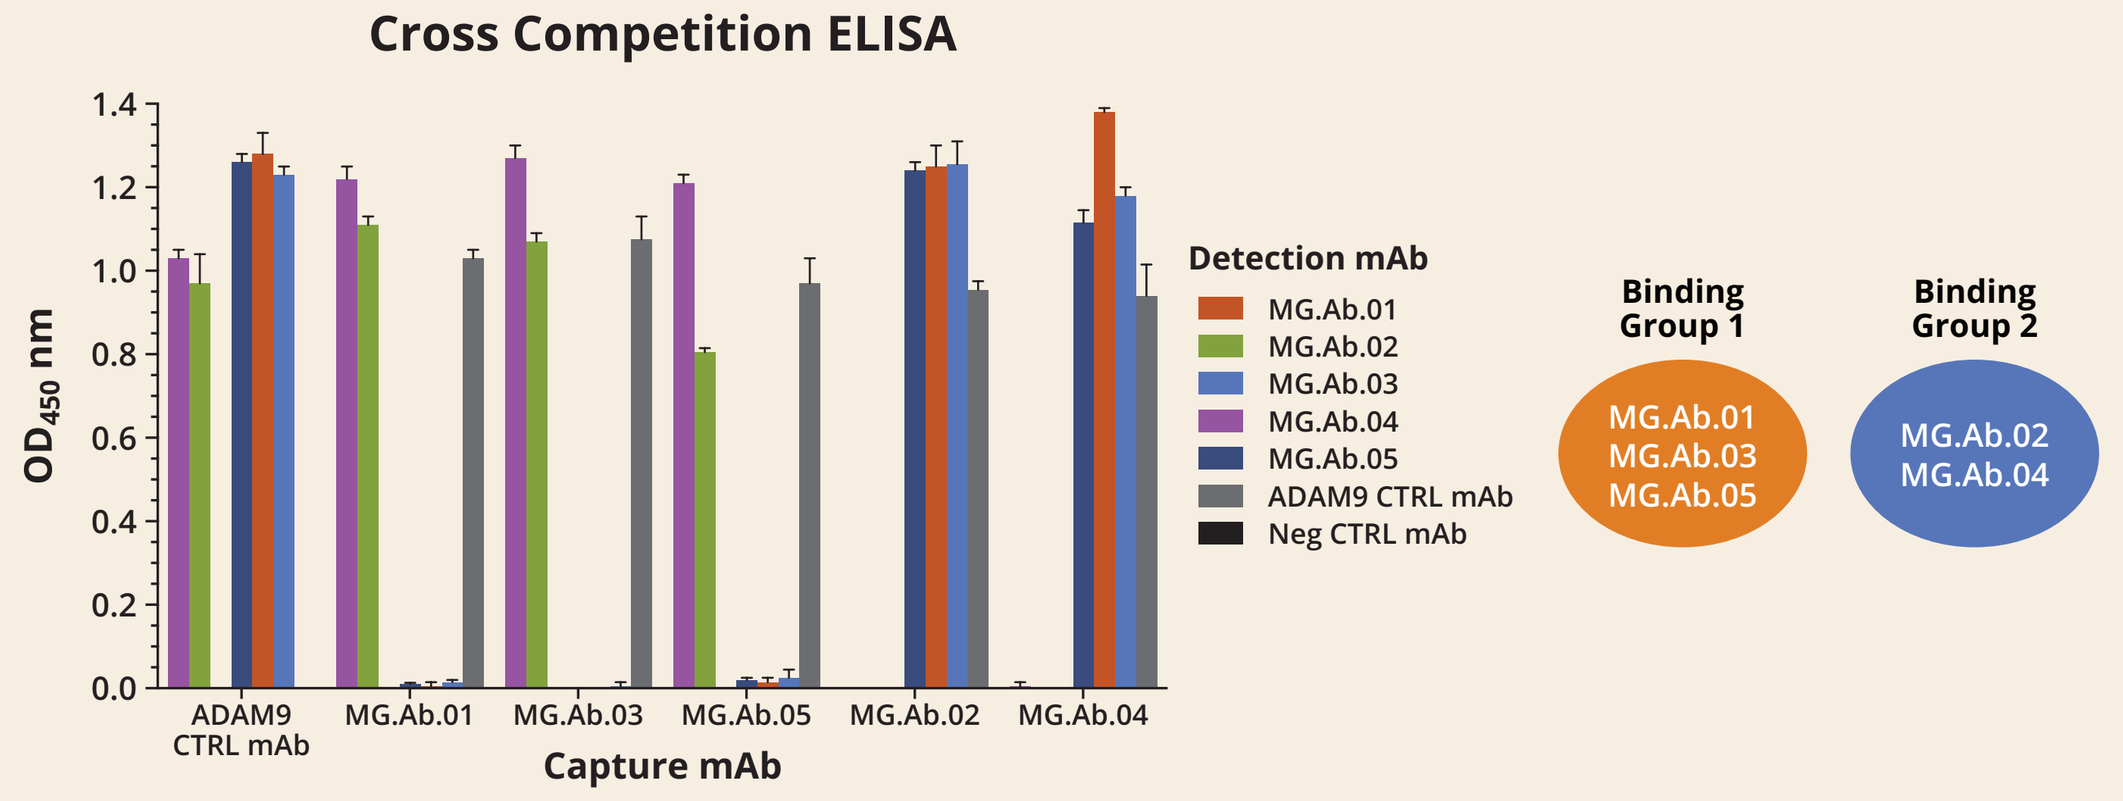
OD_{450} nm
Capture mAb
ADAM9 CTRL mAb
MG.Ab.01
MG.Ab.03
MG.Ab.05
MG.Ab.02
MG.Ab.04
Detection mAb
ADAM9 CTRL mAb
Neg CTRL mAb
Binding Group 1
MG.Ab.01 MG.Ab.03 MG.Ab.05
Binding Group 2
MG.Ab.02 MG.Ab.04

- Two non-overlapping binding groups were identified

ADAM9 ADCs Exhibit Potent In Vitro Cytotoxicity

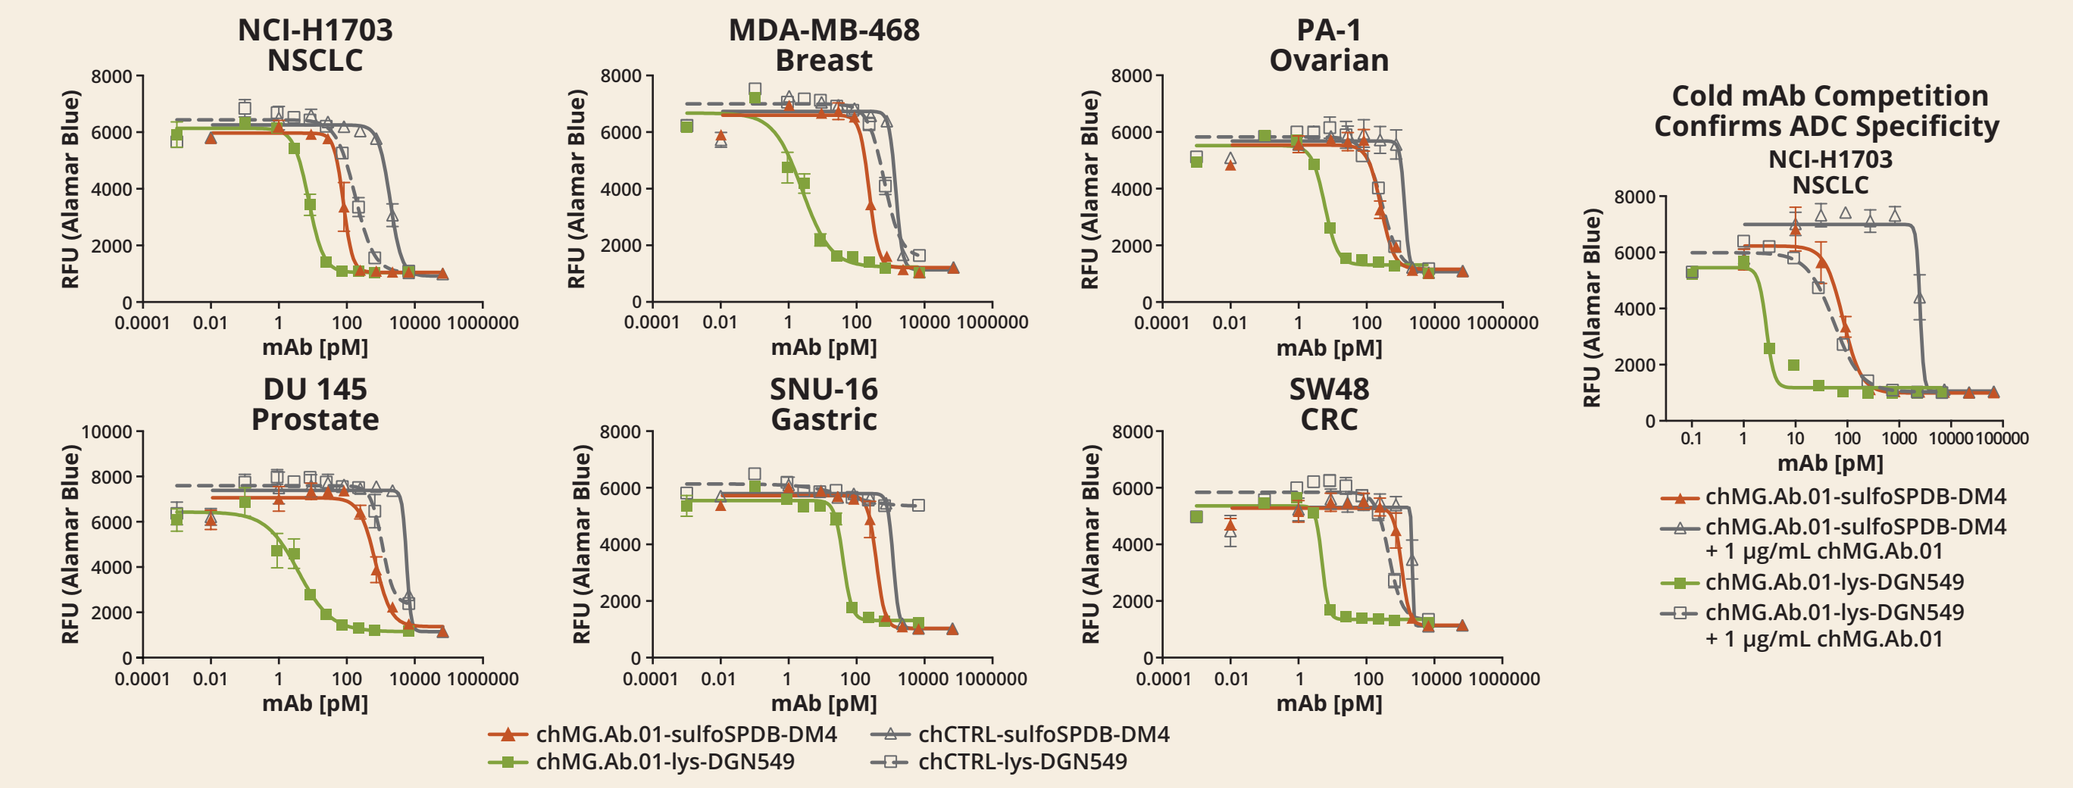
NCI-H1703 NSCLC
MDA-MB-468 Breast
PA-1 Ovarian
DU 145 Prostate
SNU-16 Gastric
SW48 CRC
Cold mAb Competition Confirms ADC Specificity
RFU (Alamar Blue)
mAb [pM]
chMG.Ab.01-sulfoSPDB-DM4
chMG.Ab.01-lys-DGN549
chCTRL-sulfoSPDB-DM4
chCTRL-lys-DGN549
chMG.Ab.01-sulfoSPDB-DM4 + 1 µg/mL chMG.Ab.01
chMG.Ab.01-lys-DGN549 + 1 µg/mL chMG.Ab.01

Summary Table of IC_{50} Values

IC_{50} (pM)	Non-Small Cell Lung Cancer	Breast Cancer	Ovarian Cancer	Prostate Cancer	Gastric Cancer	Colorectal Cancer
	NCI-H1703	MDA-MB-468	PA-1	DU 145	SNU-16	SW48
Antibody Binding Capacity*	4.12E5	8.24E4	6.88E4	1.38E5	2.05E5	1.84E4
chMG.Ab.01-sulfoSPDB-DM4	80	225	265	702	385	1091
chMG.Ab.01-lys-DGN549	8	2	6	4	41	5

*Antibody Binding Capacity determined by Bangs QFACS Kit.
7 day *in vitro* cytotoxicity was quantified using Alamar Blue reagent.

- Activity observed against a range of ADAM9-positive tumor lines

Humanized ADAM9 mAb Retains Binding Affinity

SPR Analysis

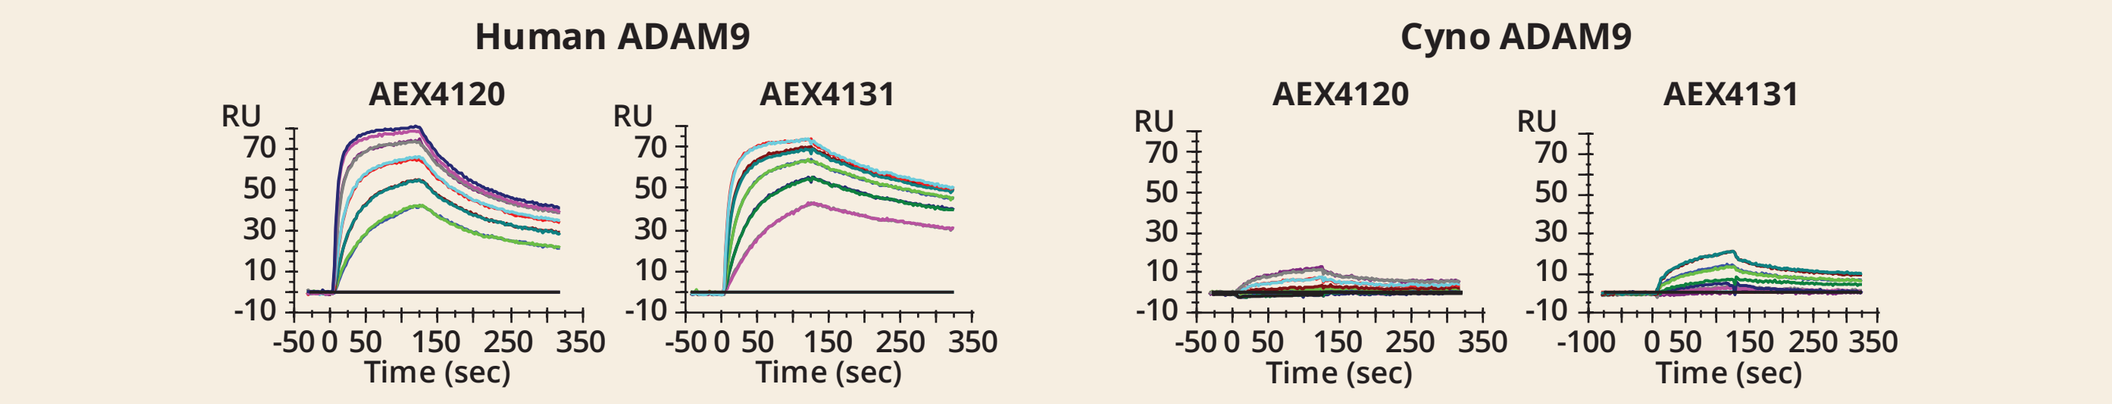
Human ADAM9
Cyno ADAM9
AEX4120
AEX4131
RU
Time (sec)

Summary of SPR Analysis

mAb	Variation	Human ADAM9 K_D, nM	Human ADAM9 k_a, 1/Ms	Human ADAM9 k_d, 1/s	Cyno ADAM9 K_D, nM	Cyno ADAM9 k_a, 1/Ms	Cyno ADAM9 k_d, 1/s
AEX4120	chimeric	**3.7**	1.7E6	6.2E-3	**ND**	ND	ND
AEX4131	humanized	**1.5**	1.5E6	2.2E-3	**117**	1.2E5	0.014

The chMG.Ab.01 (AEX4120) mAb was humanized by replacing mouse variable region heavy chain (VH) and mouse variable region light chain (VL) framework sequences with human germline framework sequences. Binding of human and cyno ADAM9-His (6.25-100nM) to mAb variants captured on Fab2 GAH Fc surface (normalized;1:1 Binding fit). ND: not determined.

Maturation Improves Affinity to Cyno Monkey ADAM9

SPR Analysis

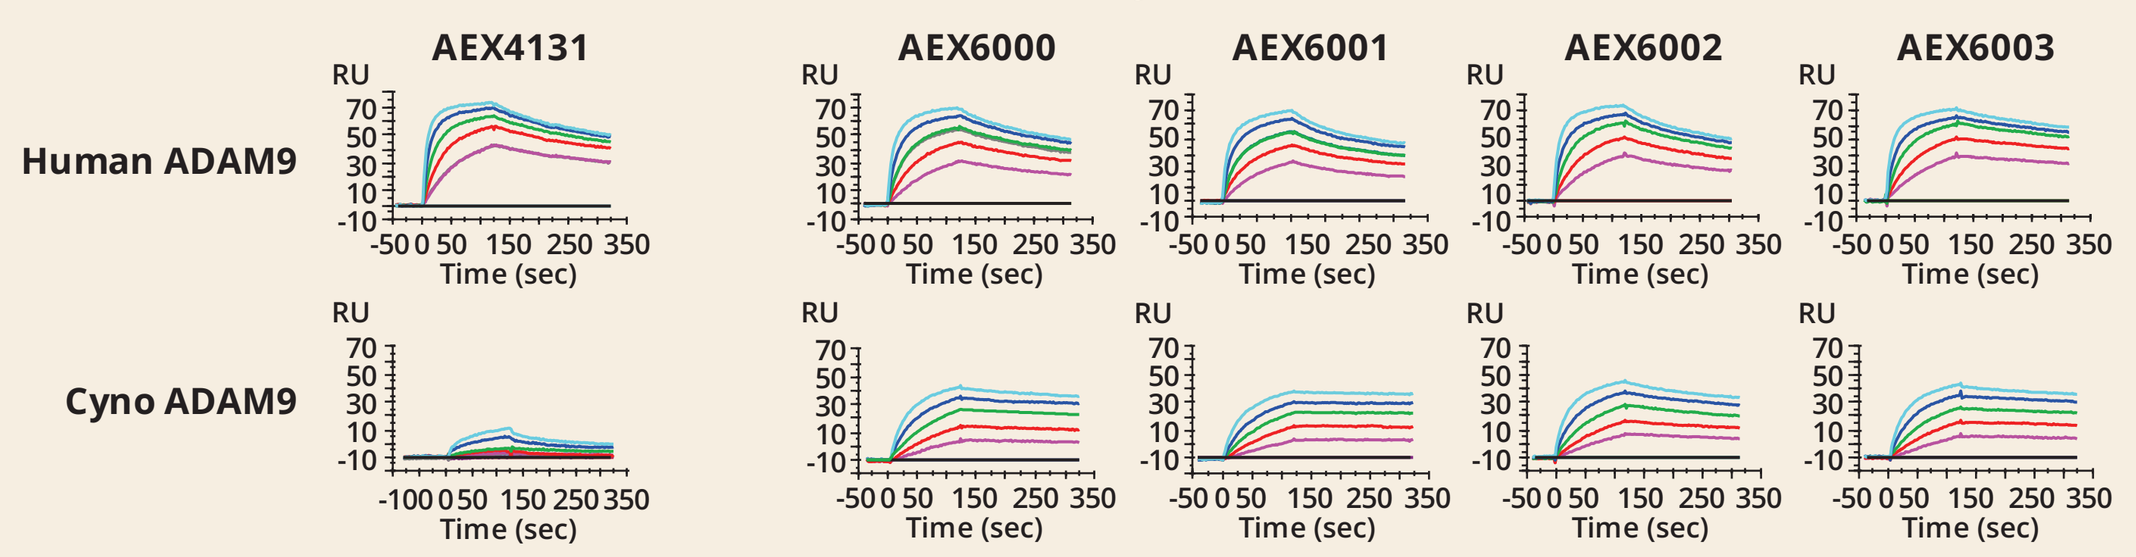
Human ADAM9
Cyno ADAM9
AEX4131
AEX6000
AEX6001
AEX6002
AEX6003
RU
Time (sec)

Summary of SPR Analysis

mAb	Variation	Human ADAM9 K_D, nM	Human ADAM9 k_a, 1/Ms	Human ADAM9 k_d, 1/s	Cyno ADAM9 K_D, nM	Cyno ADAM9 k_a, 1/Ms	Cyno ADAM9 k_d, 1/s
AEX4131	humanized	**1.5**	1.5E6	2.2E-3	**117**	1.2E5	0.014
AEX6000	affinity matured	**2.5**	9.4E5	2.3E-3	**1.3**	4.1E5	5.3E-4
AEX6001	affinity matured	**3.5**	9.1E5	3.2E-3	**0.3**	4.1E5	1.4E-4
AEX6002	affinity matured	**2.6**	1.0E6	2.6E-3	**2.4**	5.0E5	1.2E-3
AEX6003	affinity matured	**1.0**	1.0E6	9.7E-4	**1.1**	4.8E5	5.4E-4

The Fab mutagenic library was generated from AEX4131. VH/VL was screened for improved binding to cyno ADAM9 by phage display. VH/VL of selected clones were reformatted into mammalian IgG. Binding of human and cyno ADAM9-His (6.25-100nM) to mAb variants captured on Fab2 GAH Fc surface (normalized;1:1 Binding fit).

Flow Binding

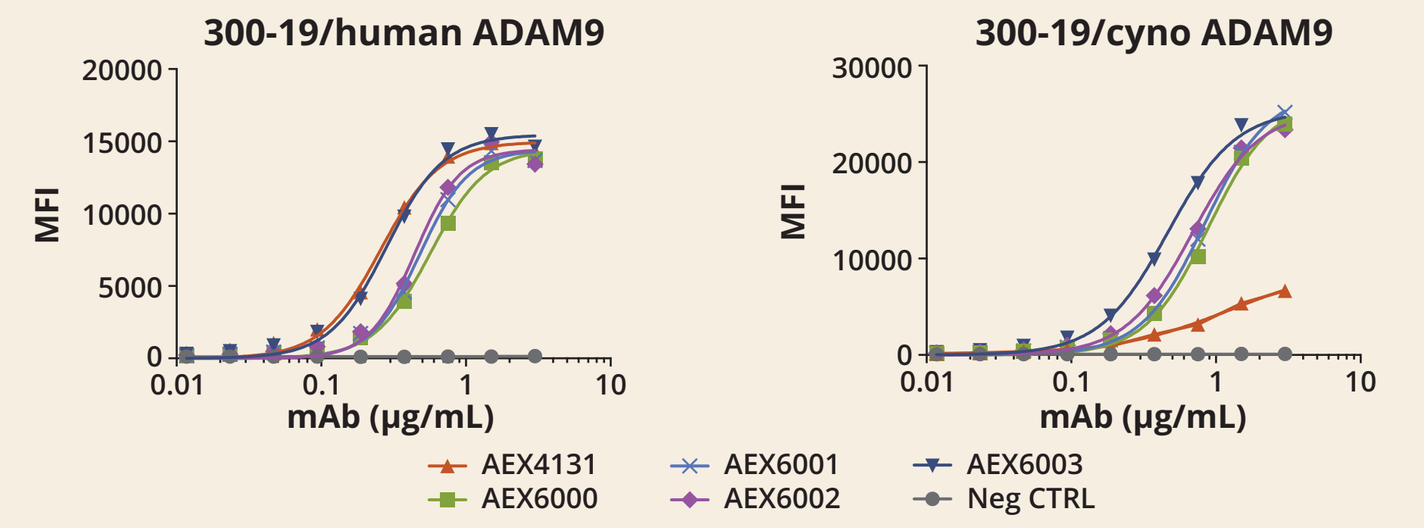
300-19/human ADAM9
300-19/cyno ADAM9
MFI
mAb (µg/mL)
AEX4131
AEX6000
AEX6001
AEX6002
AEX6003
Neg CTRL

- Affinity matured ADAM9 mAb (AEX6003):
 - Maintains binding to human ADAM9
 - Exhibits equivalent affinity for human and cyno ADAM9

Affinity Matured ADAM9 ADCs Retain Activity

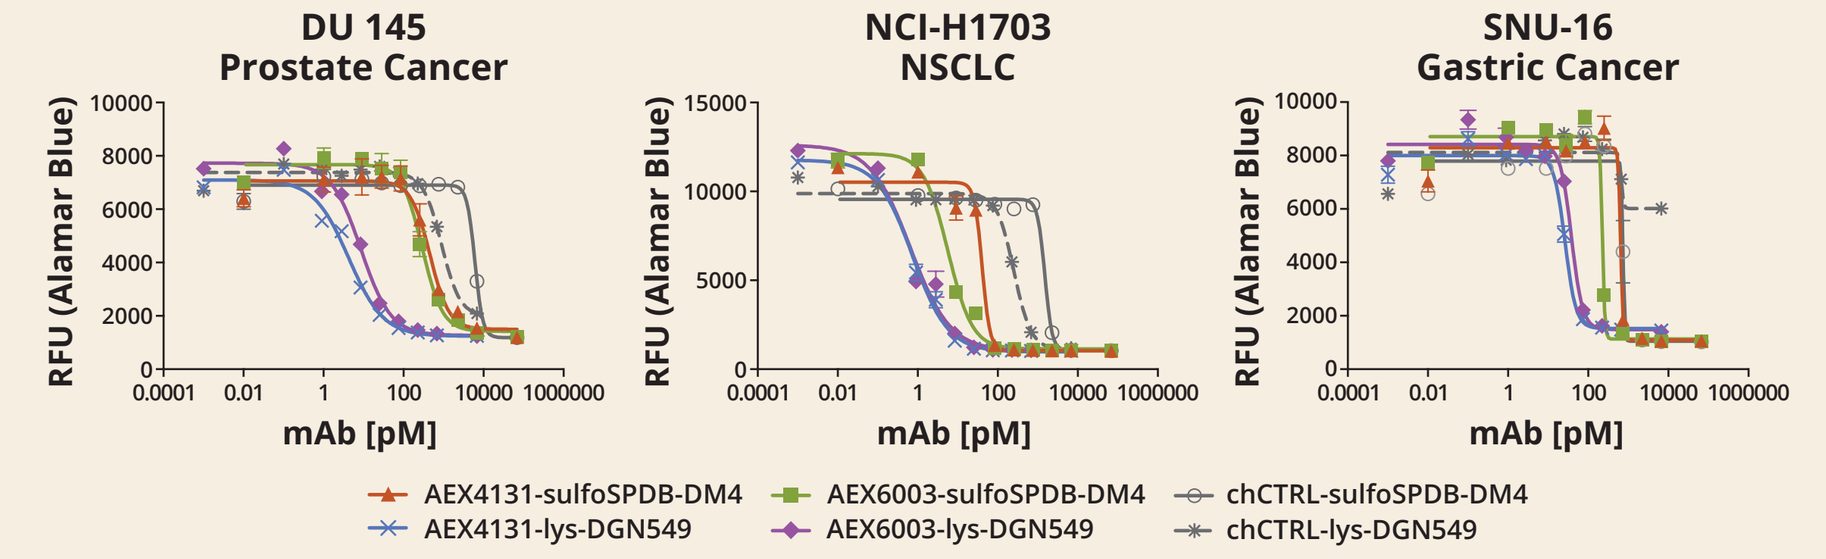
DU 145 Prostate Cancer
NCI-H1703 NSCLC
SNU-16 Gastric Cancer
RFU (Alamar Blue)
mAb [pM]
AEX4131-sulfoSPDB-DM4
AEX4131-lys-DGN549
AEX6003-sulfoSPDB-DM4
AEX6003-lys-DGN549
chCTRL-sulfoSPDB-DM4
chCTRL-lys-DGN549

Summary Table of IC_{50} Values

IC_{50} (pM)		Prostate Cancer	Non-Small Cell Lung Cancer	Gastric Cancer
mAb	Variation	DU145	NCI-H1703	SNU-16
AEX4131-sulfoSPDB-DM4	humanized	451	40	649
AEX6003-sulfoSPDB-DM4	affinity matured	288	6	227
AEX4131-lys-DGN549	humanized	4	1	26
AEX6003-lys-DGN549	affinity matured	9	1	38

7 day *in vitro* cytotoxicity was quantified using Alamar Blue reagent.

- Affinity matured AEX6003 ADCs retain potency toward ADAM9-positive tumor cells

ADAM9 mAbs are Rapidly Internalized and Processed

Antibody Internalization

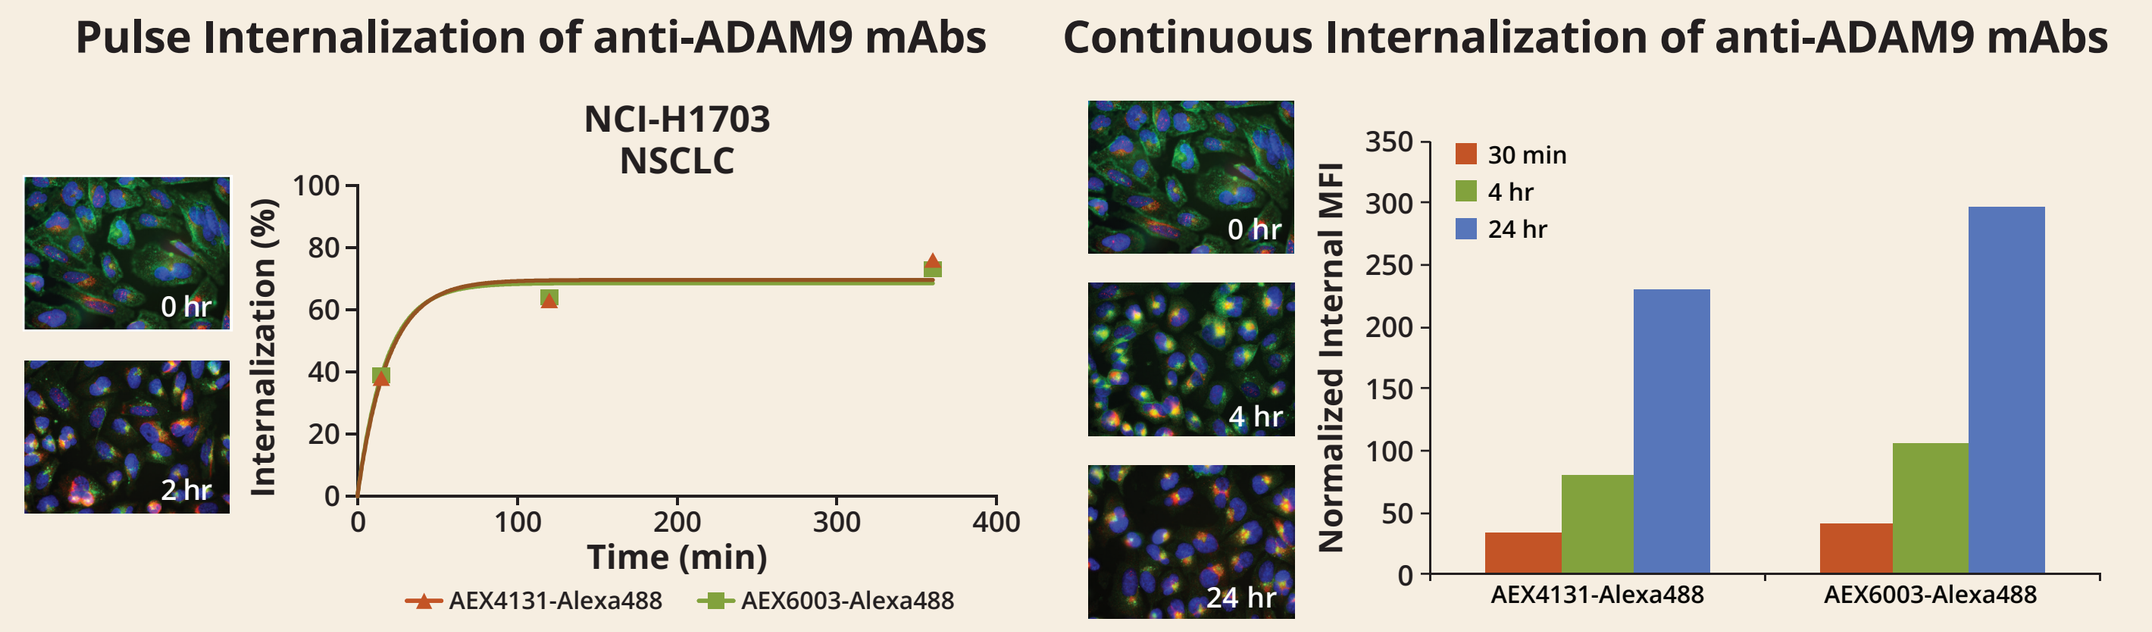
Pulse Internalization of anti-ADAM9 mAbs
Continuous Internalization of anti-ADAM9 mAbs
NCI-H1703 NSCLC
0 hr
2 hr
4 hr
24 hr
Internalization (%)
Time (min)
AEX4131-Alexa488
AEX6003-Alexa488
Normalized Internal MFI
30 min
4 hr
24 hr

Internalization imaged by fluorescence microscopy with an Alexafluor488 conjugated anti-ADAM9 mAb
Green = Anti-ADAM9 mAb; Blue = Hoechst DNA stain; Red = Anti-LAMP-1 lysosome marker

Pulse Exposure

Anti-ADAM9 Conjugates	% Internalized 15 min	% Internalized 2 hr	% Internalized 6 hr
AEX4131-Alexa488	38	63	76
AEX6003-Alexa488	39	64	73

Cells incubated at 37°C for the time indicated prior to quenching with anti-A488 mAbs.

Continuous Exposure

Anti-ADAM9 Conjugates		Normalized MFI 30 min	Normalized MFI 4 hr	Normalized MFI 24 hr
AEX4131-Alexa488	Internal	34	81	230
	Total	64	111	258
AEX6003-Alexa488	Internal	41	106	298
	Total	79	138	333

Cells continuously incubated at 37°C for the time indicated prior to quenching with anti-A488 mAbs.

- Evidence for rapid internalization and ADAM9 receptor recycling and reinternalization

Antibody Processing

Anti-ADAM9 mAbs Processed per Cell

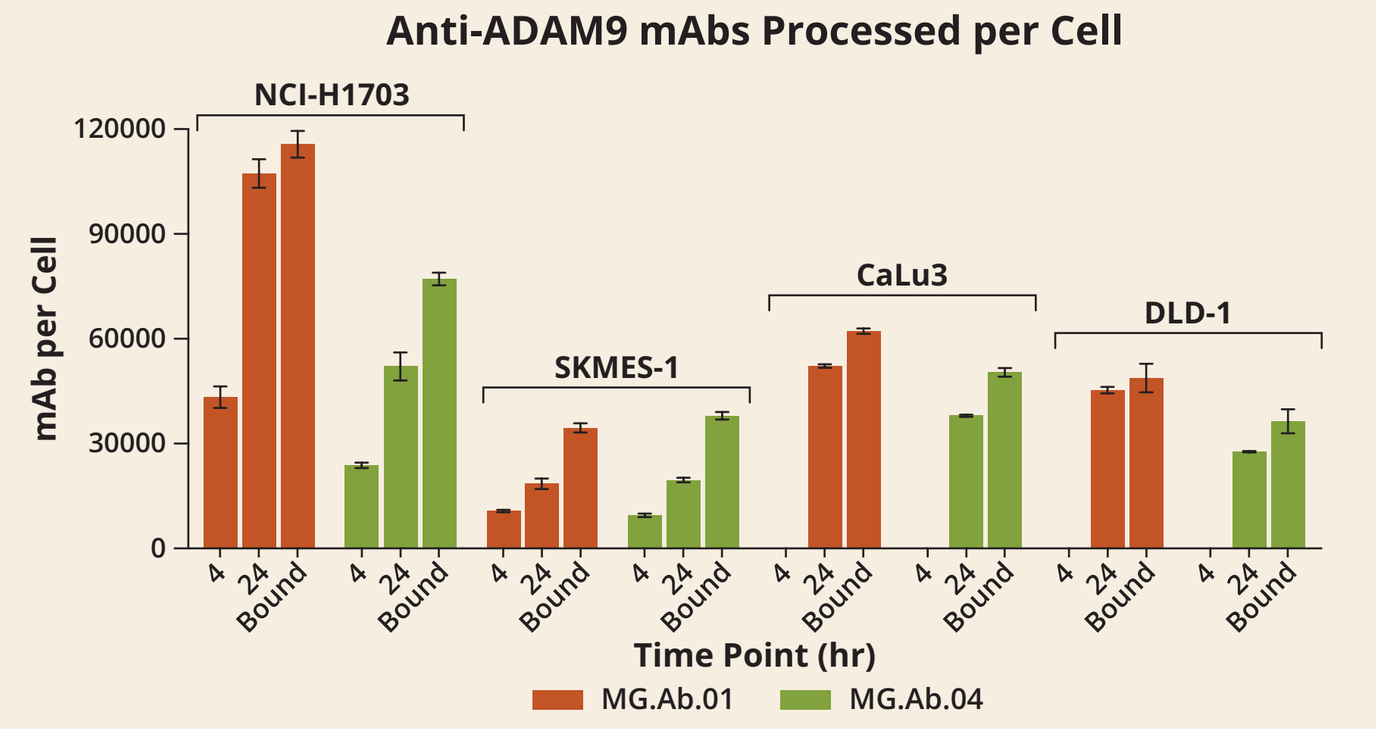
mAb per Cell
NCI-H1703
SKMES-1
CaLu3
DLD-1
4
24
Bound
Time Point (hr)
MG.Ab.01
MG.Ab.04

Bound: Total mAb bound per cell at 0 hr (Antibody binding capacity)

Pulse Processing Rates

Cell Line	Time (hr)	MG.Ab.01 (% Processed)	MG.Ab.04 (% Processed)
NCI-H1703	4	38	31
	24	93	68
SK-MES-1	4	31	25
	24	54	52
Calu-3	4	ND	ND
	24	84	75
DLD-1	4	ND	ND
	24	92	73

Cells were pulse-treated with tritium-labeled mAbs then processed catabolite and intact mAbs were quantified. ND: Not determined.

- Antibody binding capacity and processing rate contribute to total antibody processed

Conclusions

- ADAM9 is a cell surface antigen that is over-expressed on a wide range of solid tumor indications
- Anti-ADAM9 mAbs were efficiently internalized and processed by ADAM9-expressing tumor cells
- ADAM9 mAbs conjugated to a microtubule inhibitor (DM4) or a DNA alkylating agent (DGN549) demonstrated dose-dependent cytotoxicity *in vitro* toward a panel of ADAM9-positive tumor cell lines
- A humanized and affinity optimized lead development candidate (AEX6003) was engineered:
 - High-affinity binding to human and cynomolgus monkey ADAM9
 - Potent in vitro cytotoxicity toward ADAM9-expressing tumor cell lines

DM4- and DGN549-based ADAM9 ADCs may serve as potential therapeutics targeting ADAM9-expressing solid tumors

Presented at the American Association for Cancer Research Annual Meeting 2017, April 1–5, 2017, Washington, DC

©2017 MacroGenics, Inc. All rights reserved.