1165

Combinatorial Anti-Tumor Activity in Animal Models of a Novel CD123 x CD3 Bispecific DART® Molecule (MGD024) with Cytarabine, Venetoclax or Azacitidine Supports Combination Therapy in Acute Myeloid Leukemia

Ralph F. Alderson, PhD, Ling Huang*, Xiaoyu Zhang*, Haiquan Li*, Thomas Kaufman*, Gundo Diedrich, PhD*, Paul Moore, PhD* and Ezio Bonvini, MD

MacroGenics, Inc., Rockville, Maryland, USA

bonvinie@macrogenics.com

Abstract

Introduction: Notwithstanding recent progress, acute myeloid leukemia (AML) remains an incurable disease, particularly in patients (pts) with relapsing/refractory disorder or ineligible for intensive induction therapy (unfit pts). CD123, the IL-3 receptor alpha chain, is expressed by both leukemic blasts and leukemic stem cells and is a suitable therapeutic target in AML[1]. Flotetuzumab, a continuous infusion CD123 x CD3 DART molecule, has shown preliminary single-agent activity in pts refractory to induction therapy[2]. MGD024 is a second-generation, Fc-bearing CD123 x CD3 DART molecule designed for prolonged circulating half-life and intermittent delivery designed to diminish the propensity for cytokine release compared to flotetuzumab. The potentially improved tolerability and dosing convenience of MGD024 may provide a framework for introducing T-cell immunotherapy in early-stage AML or unfit pts. To explore whether MGD024 could complement AML standard of care (SOC), we investigated combination therapy in mouse models.

Materials and Methods: The DART molecules, flotetuzumab and MGD024, shared identical CD123 (humanized 7G3) and CD3 (humanized XR32) Fv arms, save for a mutation in the anti-CD3 arm of MGD024 that decreases its affinity for the CD3-epsilon chain. While flotetuzumab has no Fc domain, MGD024 includes an ala-ala-mutated human IgG1 Fc that extends its circulating half-life via the neonatal Fc receptor-mediated salvage pathway together with impairing binding to Fc-gamma receptors and complement. An IgG1-ala-ala Fc-bearing version of flotetuzumab (RES234M1.1) was also engineered to allow delivery at identical time intervals as MGD024 and avoid continuous infusion in experimental animals. MHC class I-*null*, NOD/SCID/IL2R-gamma-*null* mice were reconstituted with human PBMC (8x10^6 cells/mouse, retro orbital). Two human AML cell lines expressing low or high levels of CD123 (KG1a << MOLM-13) were implanted SC at 2.5 x 10^6 (KG1a) or 5 x 10^6 (MOLM-13) cells/mouse. Treatments (IV, IP or PO by gavage, as indicated) were initiated when tumor volumes reached ~150 mm^3, with volumes recorded weekly or twice weekly thereafter.

Results and Conclusions: Consistent with its decreased affinity for CD3, MGD024 demonstrated reduced in vitro potency in killing CD123-positive target cells compared to flotetuzumab or RES234M1.1, but proportionally greater reduction in cytokine release. MGD024, however, achieved maximal cytolytic activity as flotetuzumab or RES234M1.1, albeit at increased concentrations. Similarly, MGD024 showed reduced potency in vivo against CD123-positive tumors compared to RES234M1.1; nevertheless, tumor growth reduction of the same magnitude as that observed with RES234M1.1 was attained at higher doses of MGD024 (0.5–1 mg/kg IV 2QW MGD024 vs. 0.05-0.1 mg/kg IV 2QW RES234M1.1, depending on the model). Reduced cytokine release was also observed with MGD024 compared to RES234M1.1 in vivo. To explore MGD024 suitability for combination therapy, sub-active doses of cytarabine (CYT, 10 mg/kg IV 2QW or 7.5–10 mg/kg IP QD), venetoclax (VEN, range 10–80 mg/kg PO QD), or azacitidine (AZA, 2 mg/kg PO QD) were co-administered with suboptimal regimens of MGD024 (range 0.005–0.1 mg/kg IV 2QW, depending on the model). Complete or near complete tumor elimination was observed with the combination of suboptimal MGD024 and CYT or VEN. In contrast, AZA, at the dose tested, did not contribute to the antitumor effect of MGD024. CYT, VEN or AZA did not inhibit a fully active dose of MGD024, confirming no detrimental impact of the SOC agents at the doses employed on the effector cell population engaged by the DART molecule. All treatments were well tolerated, as indicated by body weigh profiles across treatment groups. These data support clinical exploration of the combination of MGD024 with SOC in patients with AML. An investigational new drug (IND) application of MGD024 in pts with selected relapsed or refractory hematologic malignancies is planned.

Introduction

- Acute myeloid leukemia (AML) remains an incurable disease, particularly in patients with refractory disease
- Redirected T-cell mediated killing via CD3-engaging bispecific molecules may offer an alternate therapeutic opportunity
- CD123, the IL-3 receptor alpha chain, is expressed by both leukemic blasts and leukemic stem cells[2] and is a suitable therapeutic target, including for redirected killing
- Flotetuzumab, a continuous infusion CD123 x CD3 DART molecule, has shown preliminary single-agent activity in refractory AML[1]
- MGD024 is a second-generation CD123 x CD3 DART investigational molecule similar to flotetuzumab but engineered for prolonged circulating half-life and reduced cytokine release, thus suitable for intermittent delivery
- To explore whether MGD024 could complement standard of care (SoC) regimens in AML, we investigated the effects of the combination with cytarabine (CYT), azacytidine (AZA) or venetoclax (VEN) in vitro and in mouse AML models

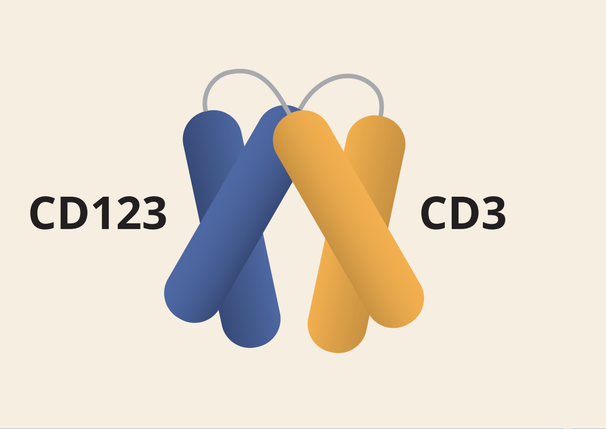

CD123
CD3

CD123 x CD3 Bispecific DART® Molecules: Flotetuzumab and MGD024

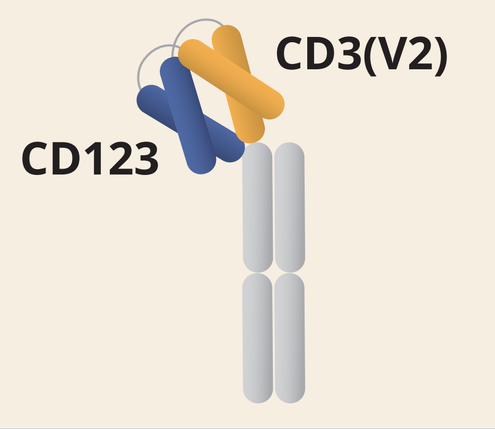

CD3(V2)
CD123

Specific Features	Common Molecular Features	Specific Features
▪ Basic DART molecule ▪ No Fc domain ▪ Administered as continuous IV infusion	▪ Bispecific diabodies stabilized at the COOH termini via a di-sulfide bond ▪ Humanized antibody components cross-reactive with cynomolgus monkey homologues with near identical affinities ▪ Proprietary heterodimerization sequences	▪ Affinity modulated CD3 arm ▪ Reduced propensity for cytokine induction ▪ Hu IgG1(ala-ala) *null* Fc ▪ Weekly administration (or longer interval)
Clinical Development	**Mechanisms of Action**	**Planned Clinical Development**
▪ Refractory AML https://clinicaltrials.gov/ct2/show/NCT04681105	▪ Co-engages T cells via CD3 with CD123, the low-affinity IL-3 receptor, expressed by AML and other heme malignancies ▪ TCR & MHC-independent tumor cell recognition: Virtually any T cell can kill cancer cells ▪ Monovalent binding to CD3: T-cell activation strictly dependent on target engagement	▪ AML, MDS ▪ CD123+ heme malignancies

Material & Methods

DART molecules

- Flotetuzumab and MGD024 shared identical CD123 (humanized 7G3 mAb) and CD3 (humanized XR32 mAb) Fv arms
- MGD024 includes a point mutation in the anti-CD3 heavy chain that decreases the affinity for CD3-ε and dissociates the induction of cytotoxicity from cytokine release. The mutated CD3-binding arm was obtained via phage display and recursive selection of CD123 x CD3 DART variants with reduced cytokine release and similar level of maximal cytolytic activity as the wild-type (WT) precursor
- MGD024 includes a *null* (ala-ala-mutated) human IgG1 Fc that extends its circulating half-life via the neonatal Fc receptor-mediated salvage pathway together with impairing binding to Fc-γ receptors and complement
- An IgG1-ala-ala Fc-bearing version of flotetuzumab (RES234M1.1, referred herein as CD123 x CD3 WT DART molecule) was engineered as a positive control and for compound delivery to experimental animals at identical time intervals as MGD024, thus avoiding continuous infusion
- All DART molecules cross-react with the corresponding cynomolgus monkey antigens with affinities similar to those for the human antigens
- DART molecules were expressed in CHO cells and purified by using standard protein A-based protocols

Cell lines

- Human AML cell lines used ranged from 5,600 (KG1a) to 16,000 (MV4-11) and 21,000 (Molm-13) CD123 antibody-binding sites per cell
- The murine myeloid leukemia cell line (C1498) was stably transduced to express human CD123 (HuCD123-C1498 cells) with a density of 23,000 CD123 antibody-binding sites per cell

Mice

- MHC class I-*null*, NOD/SCID/IL2R-γ-*null* mice were reconstituted with human PBMC (8 x 10^6 cells/mouse) by retro orbital (ro) or intravenous (iv) injection
- C57BL/6 mice were engineered to express the human CD3-ε chain epitope (HuCD3-k/i mice) recognized by the DART molecules as a knock-in of the corresponding murine CD3 components. HuCD3-k/i mice express normal numbers of T and B cells and are fully immune competent

Animal modeling

- MHC class I-*null*, NOD/SCID/IL2R-γ-*null* mice were implanted sub-cutaneously with human AML cell lines at 5 x 10^6 cells/mouse in a 1:1 suspension with Matrigel. Treatments were initiated when tumor volumes reached ~150 mm^3, with volumes recorded weekly or twice weekly thereafter
- HuCD3-k/i mice were inoculated iv with murine HuCD123-C1498 leukemia cells at 2.5 x 10^5/mouse. Treatment initiated on day 4

MGD024 Characterization

MGD024 Maintains Cytolysis with Decreased Cytokine Release

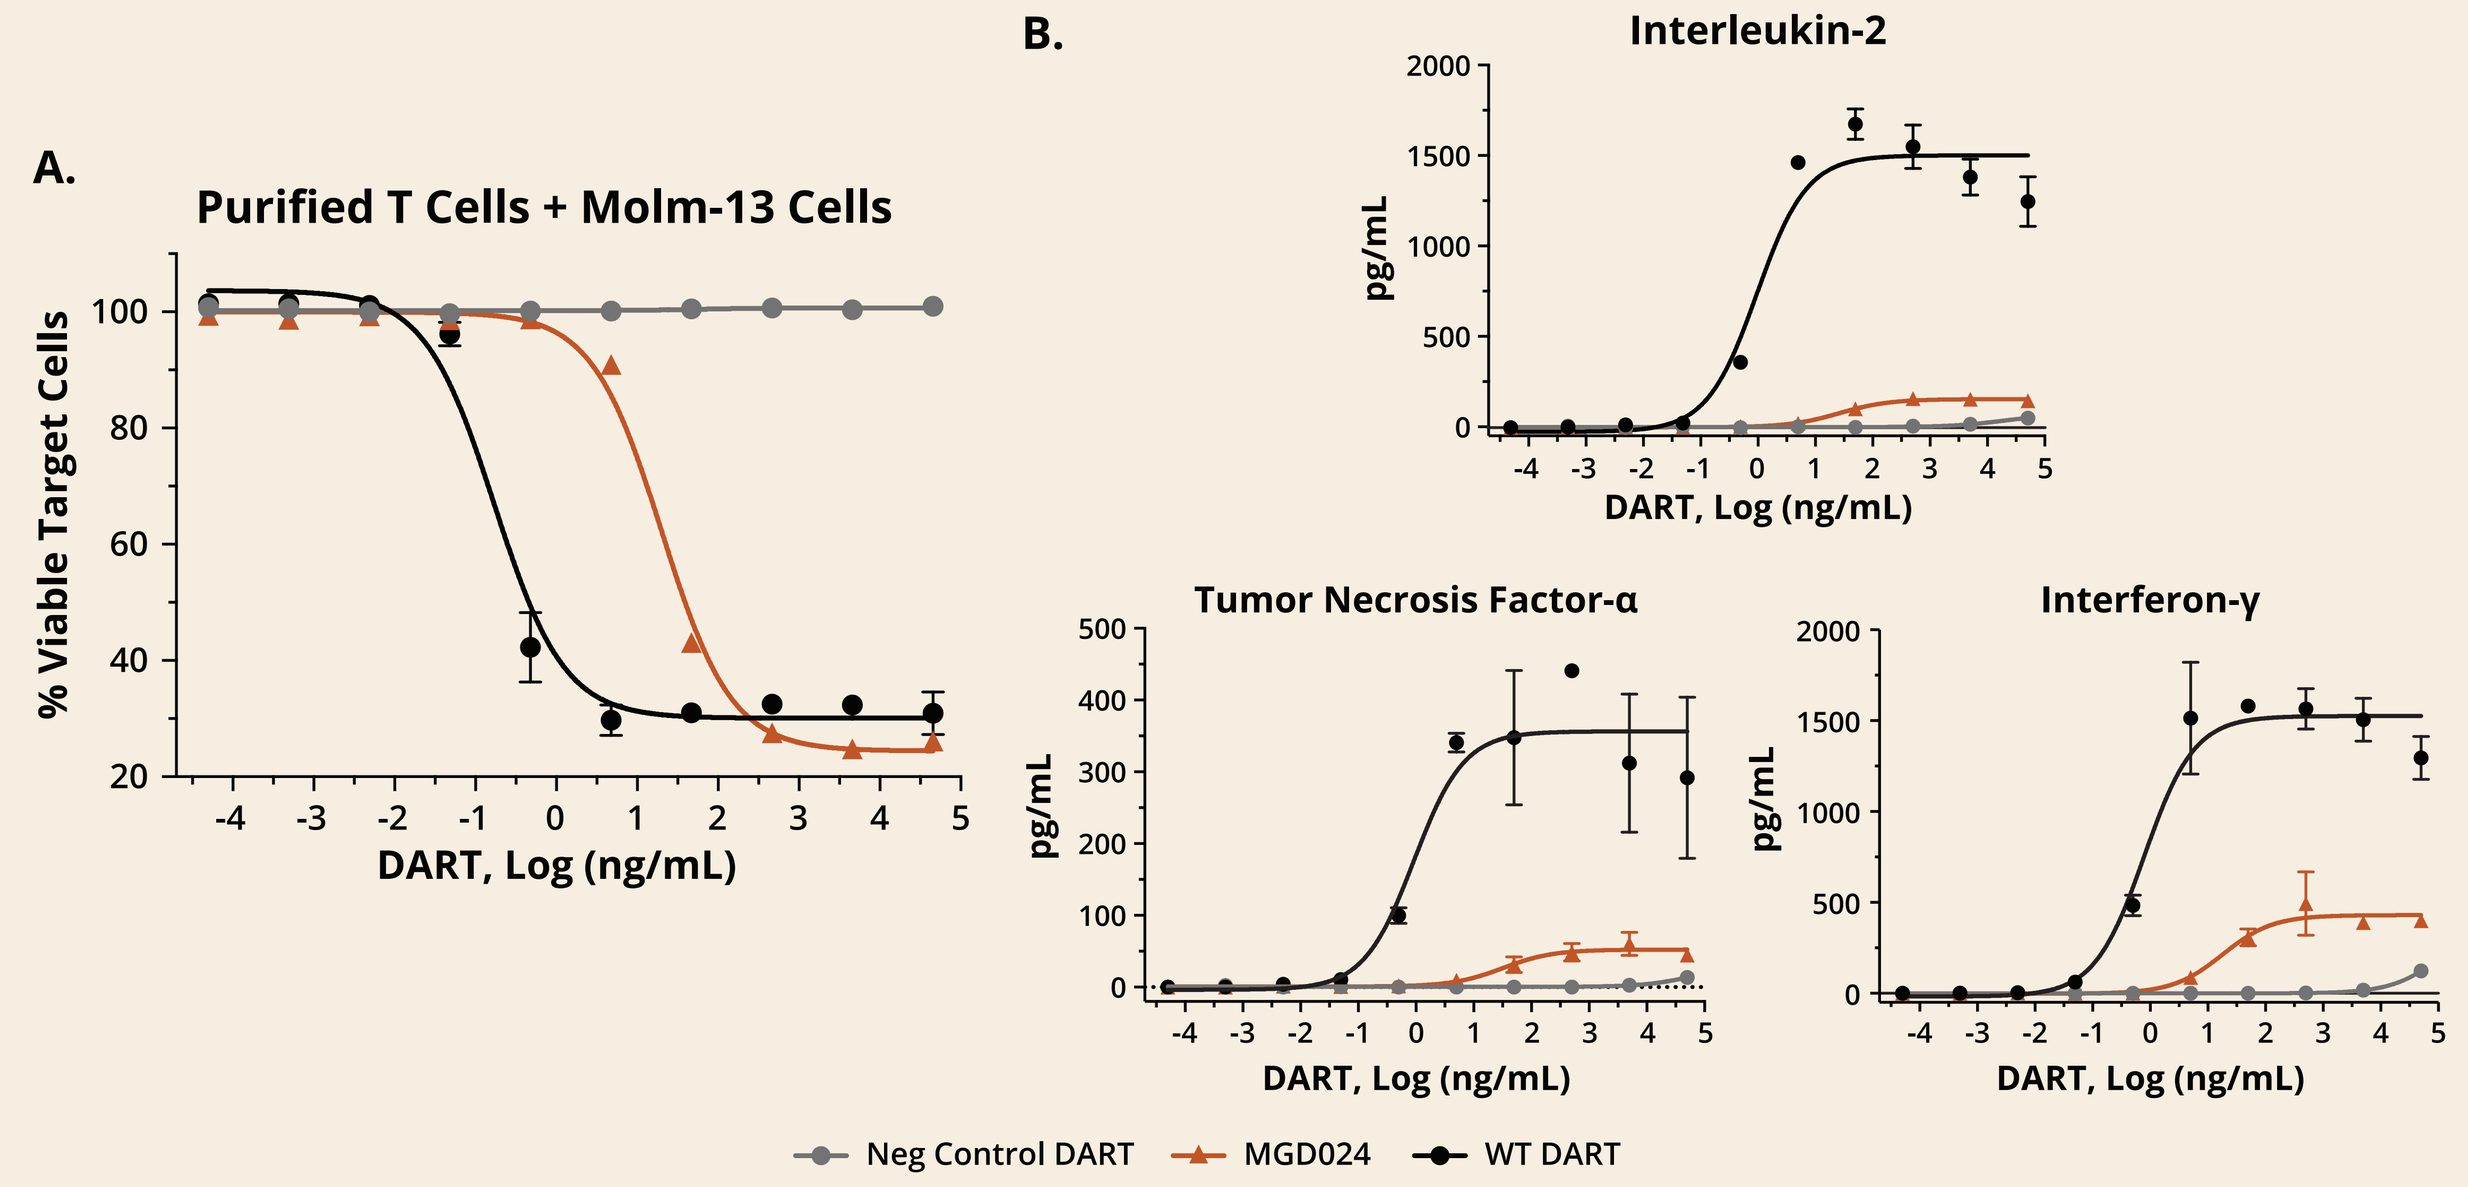

A.
Purified T Cells + Molm-13 Cells
% Viable Target Cells
DART, Log (ng/mL)
B.
Interleukin-2
Tumor Necrosis Factor-α
Interferon-γ
pg/mL
DART, Log (ng/mL)
Neg Control DART
MGD024
WT DART

- Same maximal cytolysis (E_{max}) as the parental WT DART molecule with decreased cytokine release

A. Purified T cells were incubated with PHK26-labeled CD123+ MOLM-13 cells at an effector:target ratio of 1:1. After 48 h incubation, target cell viability was assessed by DAPI staining and analyzed by flow cytometry.
B. Supernatant from the 48-h culture was analyzed for cytokine via the cytokine bead array (CBA) assay.

MGD024 Maintains In Vivo Anti-tumor Activity as the Parental WT DART Molecule

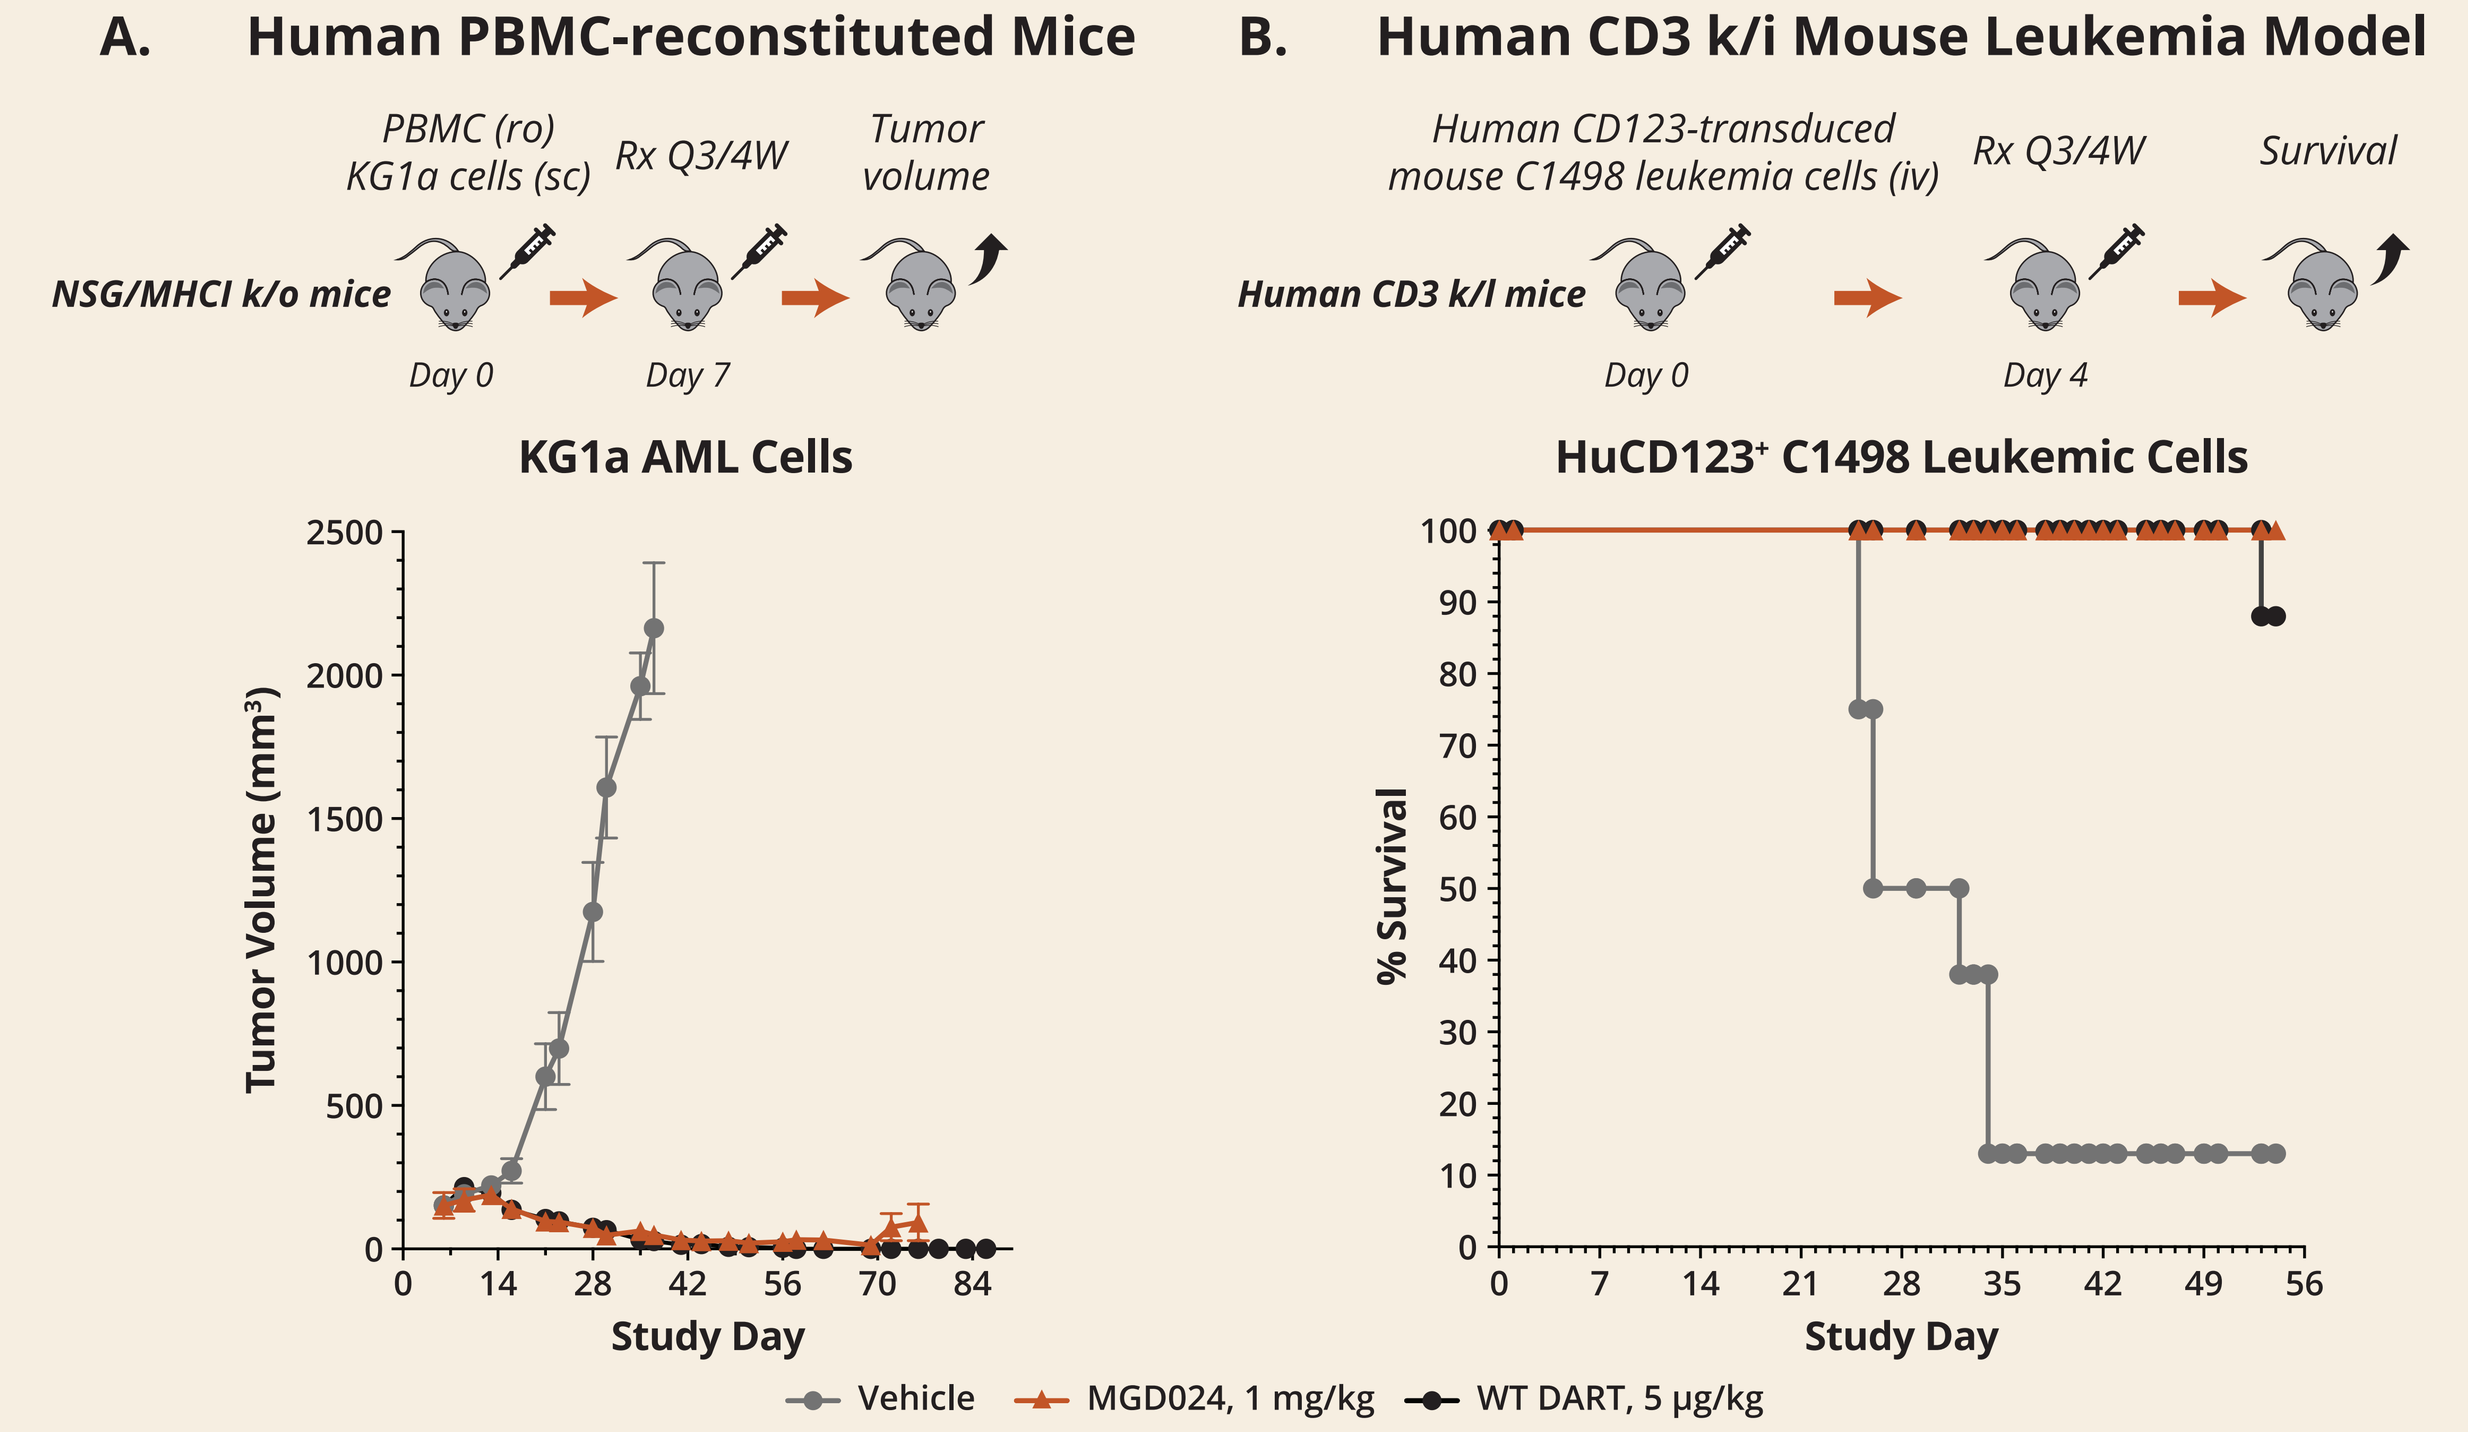

A. Human PBMC-reconstituted Mice
PBMC (ro)
KG1a cells (sc)
Rx Q3/4W
Tumor volume
NSG/MHCI k/o mice
Day 0
Day 7
KG1a AML Cells
Tumor Volume (mm³)
Study Day
B. Human CD3 k/i Mouse Leukemia Model
Human CD123-transduced mouse C1498 leukemia cells (iv)
Rx Q3/4W
Survival
Human CD3 k/I mice
Day 0
Day 4
HuCD123⁺ C1498 Leukemic Cells
% Survival
Study Day
Vehicle
MGD024, 1 mg/kg
WT DART, 5 µg/kg

- Per design, MGD024 shows decreased potency but comparable activity as the parental molecule

A. Human PBMC-reconstituted MHC class I-*null*, NOD/SCID/IL2R-γ-*null* mice were implanted with KG1a cells as described in Material and Methods. Following randomization based on tumor volume (N=8/group), treatment iv 3QW with MGD024 or the CD123 x CD3 WT DART molecule was initiated on day 7 post-implantation and maintained through day 43.
B. HuCD3-k/i mice were inoculated iv with murine HuCD123-C1498 leukemia (2.5 x 10^5 cells/mouse) as described in Material and Methods. Following randomization (N=8/group), treatment iv 2QW with MGD024 or the CD123 x CD3 WT DART molecule was initiated on day 4 post-inoculation and maintained throughout the experiment. Mice meeting predefined protocol criteria for euthanasia were identified in a treatment-blinded fashion.

MGD024: Extended PK and Good Tolerability in Cynomolgus Monkeys

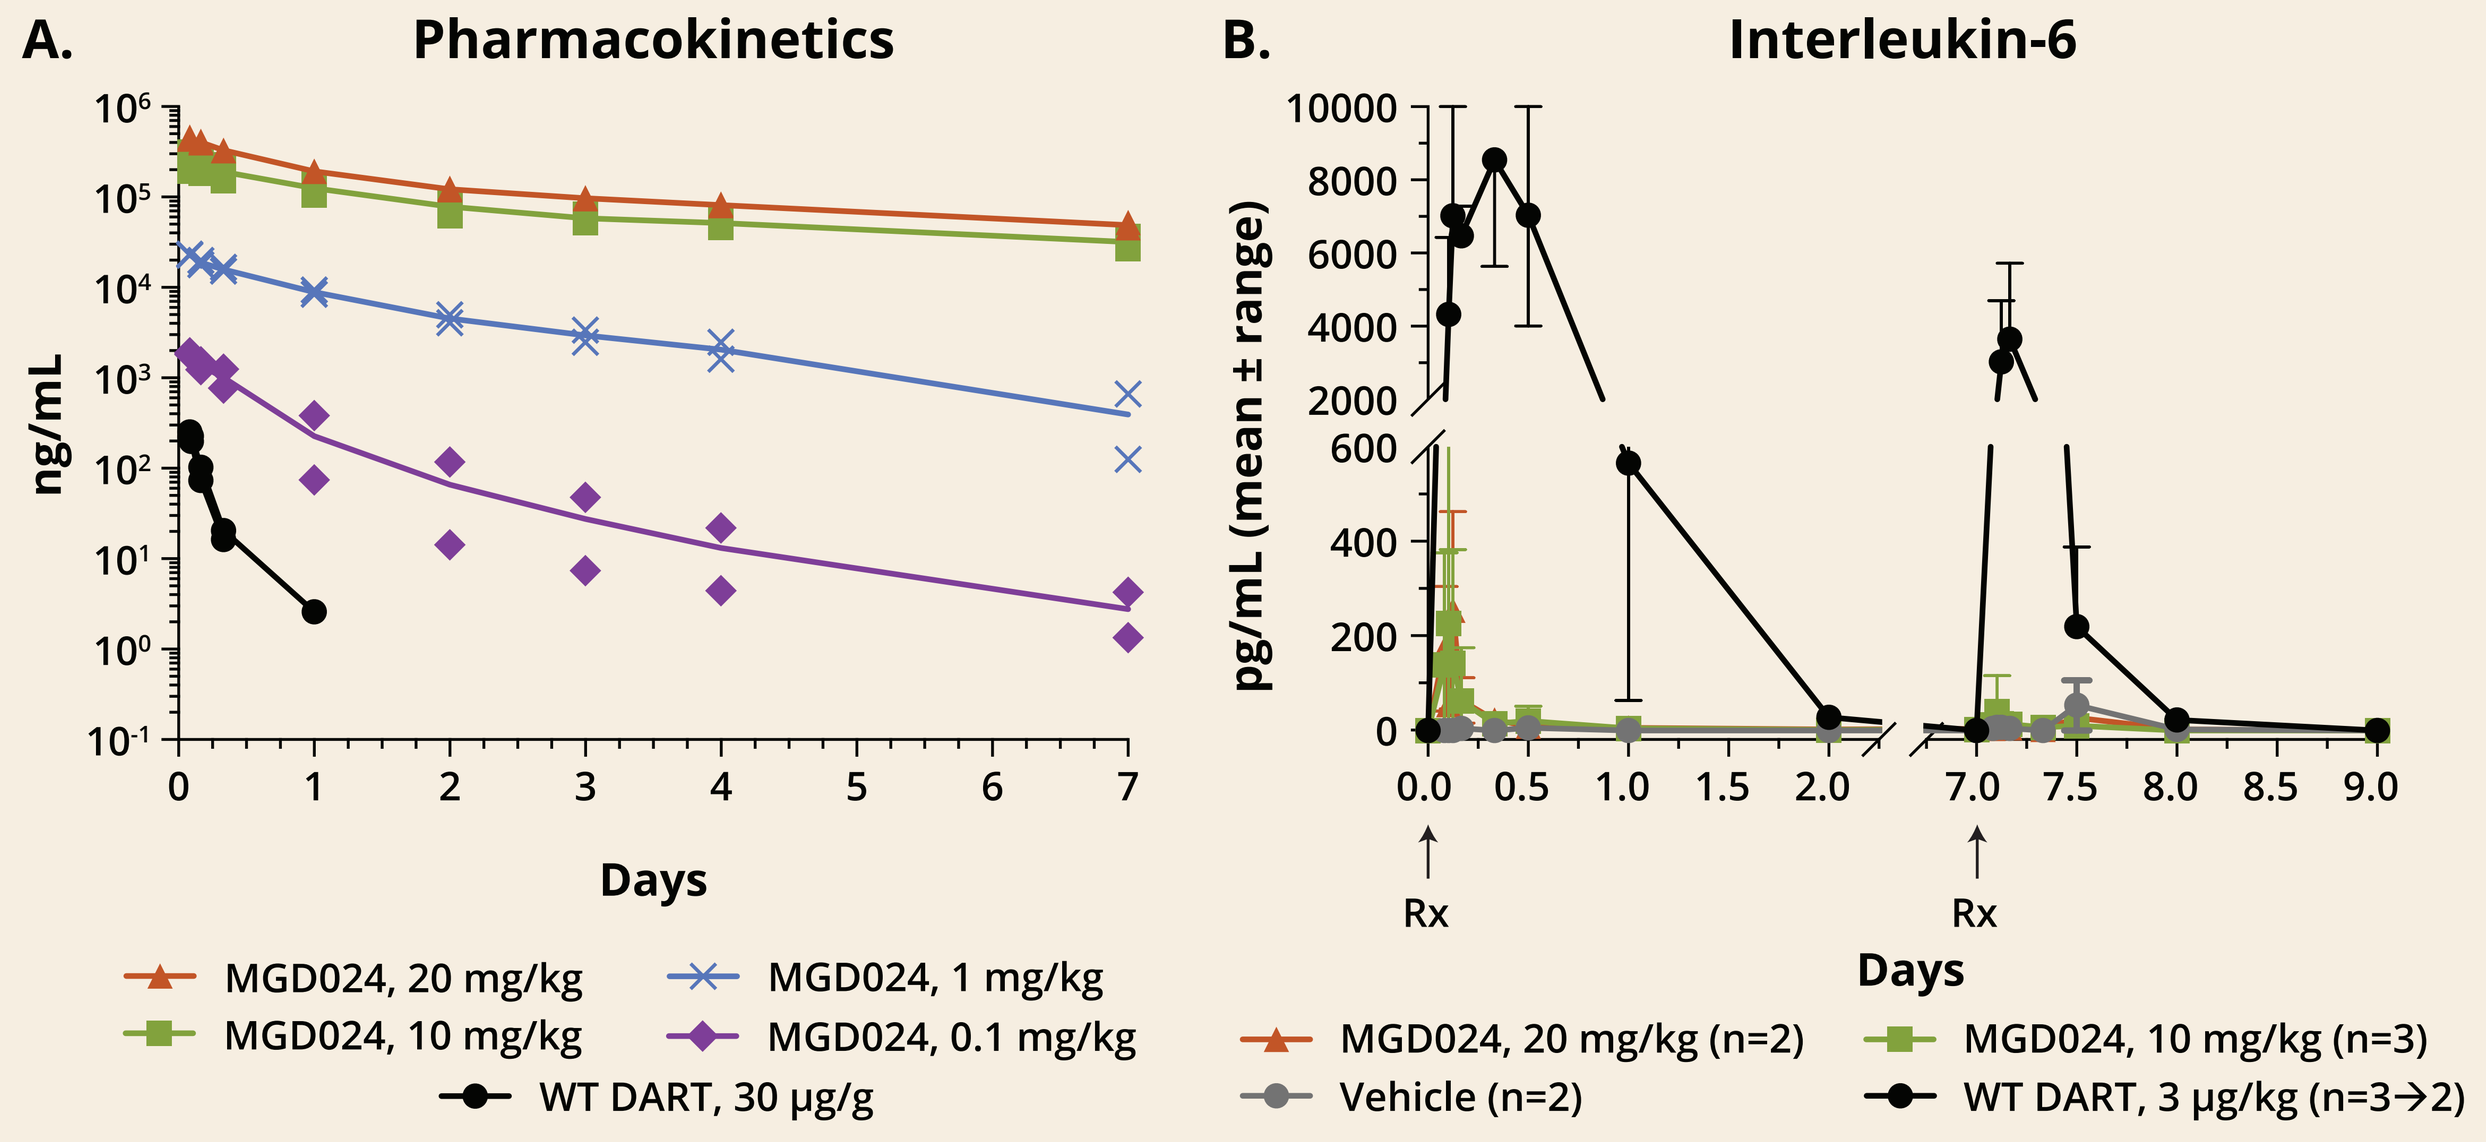

A. Pharmacokinetics
ng/mL
Days
MGD024, 20 mg/kg
MGD024, 10 mg/kg
MGD024, 1 mg/kg
MGD024, 0.1 mg/kg
WT DART, 30 µg/g
B. Interleukin-6
pg/mL (mean ± range)
Rx
Days
MGD024, 20 mg/kg (n=2)
MGD024, 10 mg/kg (n=3)
Vehicle (n=2)
WT DART, 3 µg/kg (n=3→2)

- Linear exposure at doses >1 mg/kg
- Minimal, asymptomatic interleukin-6 elevations

MGD024 doses of 0.1, 1, 10, or 20 mg/kg and CD123 x CD3 WT DART molecule doses of 0.003 or 0.030 mg/kg were administered to groups of 2 or 3 cynomolgus monkeys on Day 1 and Day 8. Serum samples were collected predose and at 2, 2.5, 3, 4, 8, 12, 24, and 48 hours after the start of infusion.
A. Circulating drug concentrations of the CD123 x CD3 WT DART molecule include data only from the 0.030 mg/kg dose level.
B. Circulating concentrations of interleukin-6 are shown for the 10 and 20 mg/kg MGD024 dose levels only and the 0.003 mg/kg CD123 x CD3 WT DART molecule dose level. The circulating levels of other cytokines, including IL-2, IL 4, IL-5, IL 8, IL-10, IL-15, IFN-γ, and TNF-α, were undetectable or similarly reduced (data not shown).

MGD024 Combinatorial Activity with SoC Agents

Effect of SoC Agents on Resting T-cell Viability

Clinical Plasma Concentrations

	Dose	Steady-state C_{max} Range
Cytarabine, low-dose[3] mw 243.2	20 mg/m²/day, continuous IV	0.015 – 0.1 µM
Azacitidine[4] mw 244.2	75 mg/m², SC or IV	750 ± 403 ng/mL, 1.4 – 4.8 µM
Venetoclax[5] mw 868.4	400 mg/day, PO	2.1 ± 1.1 µg/mL, 1.2 – 3.7 µM

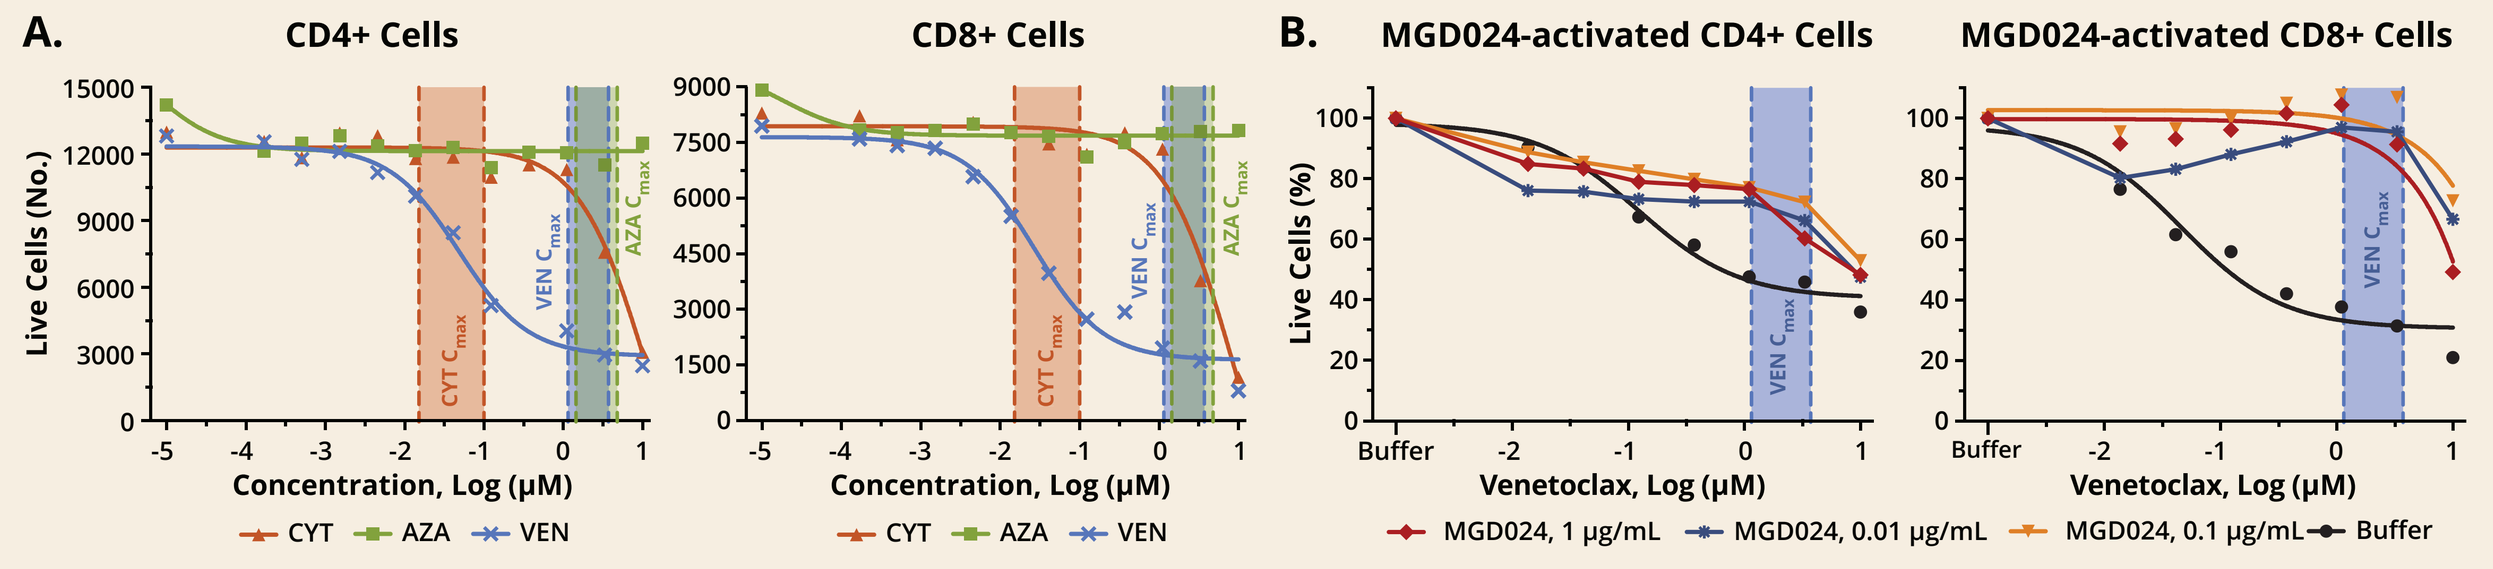

A.
CD4+ Cells
CD8+ Cells
Live Cells (No.)
Concentration, Log (µM)
CYT
AZA
VEN
B.
MGD024-activated CD4+ Cells
MGD024-activated CD8+ Cells
Live Cells (%)
Venetoclax, Log (µM)
MGD024, 1 µg/mL
MGD024, 0.01 µg/mL
MGD024, 0.1 µg/mL
Buffer

- No effect on T-cell viability by cytarabine or azacitidine at clinically relevant concentrations
- Resting CD4 and CD8 T-cell viability is negatively affected by venetoclax
- Activated CD8 T cells are resistant to venetoclax

A. Steady-state molar plasma concentrations (C_{max}) for low-dose cytarabine (CYT), azacytidine (AZA) and venetoclax (VEN) were obtained from published reports. Resting PBMC were treated in vitro for 3 days at the indicated range of concentrations and viable cells enumerated via flow cytometry. The color-coded SoC micromolar C_{max} concentration ranges are superimposed.
B. PBMC (1 x 10^6/mL) were incubated for 72 h with MOLM13 cells at 10:1 ratio in the absence or presence of MGD024. After harvesting and removal of dead cells by gradient centrifugation, cells were seeded in 96-well plates (10^5 cells/well) and treated with venetoclax for 24 h. CD4 and CD8 T cell viability was evaluated by flow cytometry.

Effect of SoC Agents on MGD024-mediated Cytolytic Activity and T-cell Expansion in vitro

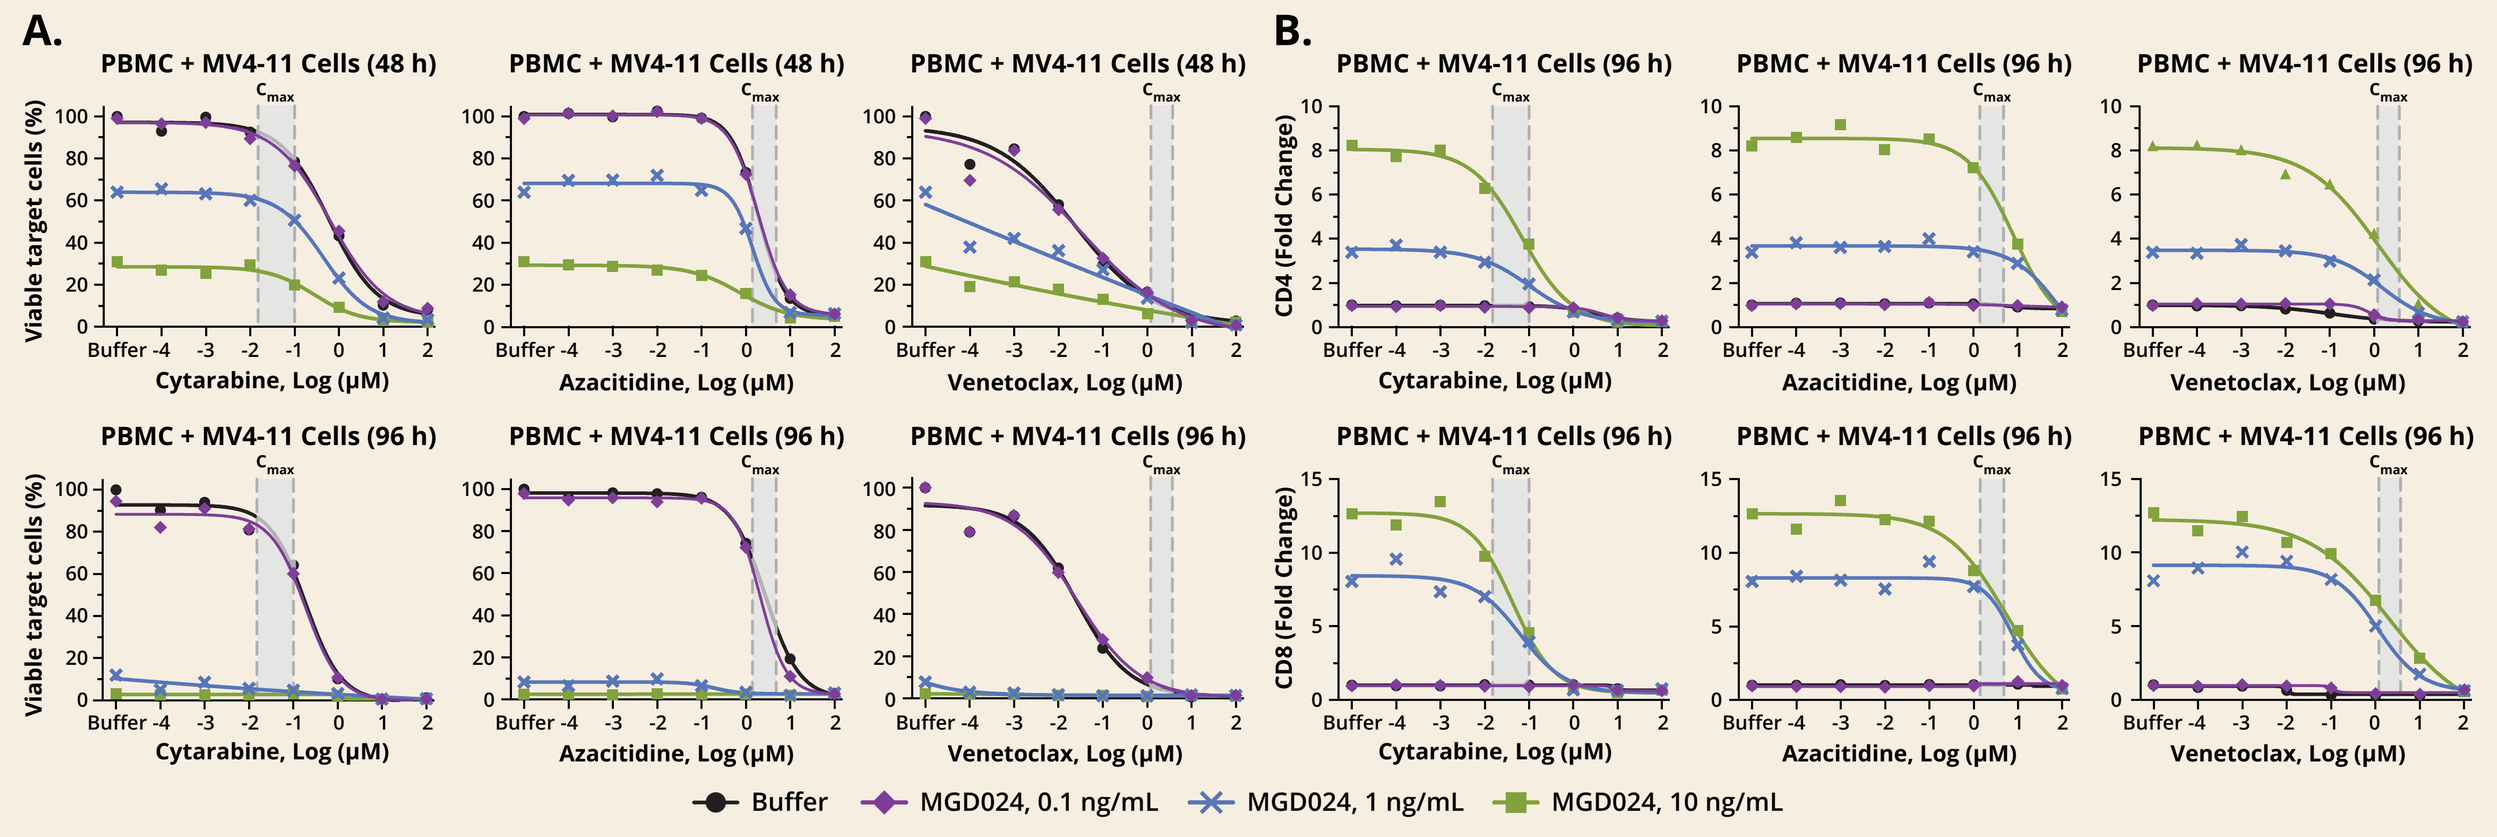

A.
PBMC + MV4-11 Cells (48 h)
PBMC + MV4-11 Cells (96 h)
Viable target cells (%)
Cytarabine, Log (µM)
Azacitidine, Log (µM)
Venetoclax, Log (µM)
B.
CD4 (Fold Change)
CD8 (Fold Change)
Buffer
MGD024, 0.1 ng/mL
MGD024, 1 ng/mL
MGD024, 10 ng/mL

- Concomitant exposure to MGD024 and SoC agents enhances AML cell line cytolysis
- CYT's and AZA's effect requires ≥ Cmax concentrations, while VEN's effect is seen across the whole range tested
- Decreased MGD024-induced proliferation observed with SoC at ≥C_{max}
- No or modest negative impact at concentration below C_{max}, including VEN

PBMCs (1 x 10^4/well in 200 µL) were mixed in 96-well plates with an equal number of MV4-11 AML cells (1:1 ratio) and treated with CYT, AZA, or VEN (0.1 nM – 100 µM) in the presence or absence of MGD024 (0.1, 1.0, or 10 ng/mL). Plates were incubated at 37C and harvested for analysis of target-cell depletion (48h and 96h) or T-cell expansion (96 h) by flow cytometry.
A. MV4-11 cell killing activity is shown as viable cells normalized by the number of viable cell count in untreated controls.
B. CD4 and CD8 T-cell expansion are shown as fold-changes over untreated controls.

Pre-exposure to VEN, but not CYT or AZA, Inhibits MGD024 Activity in vitro

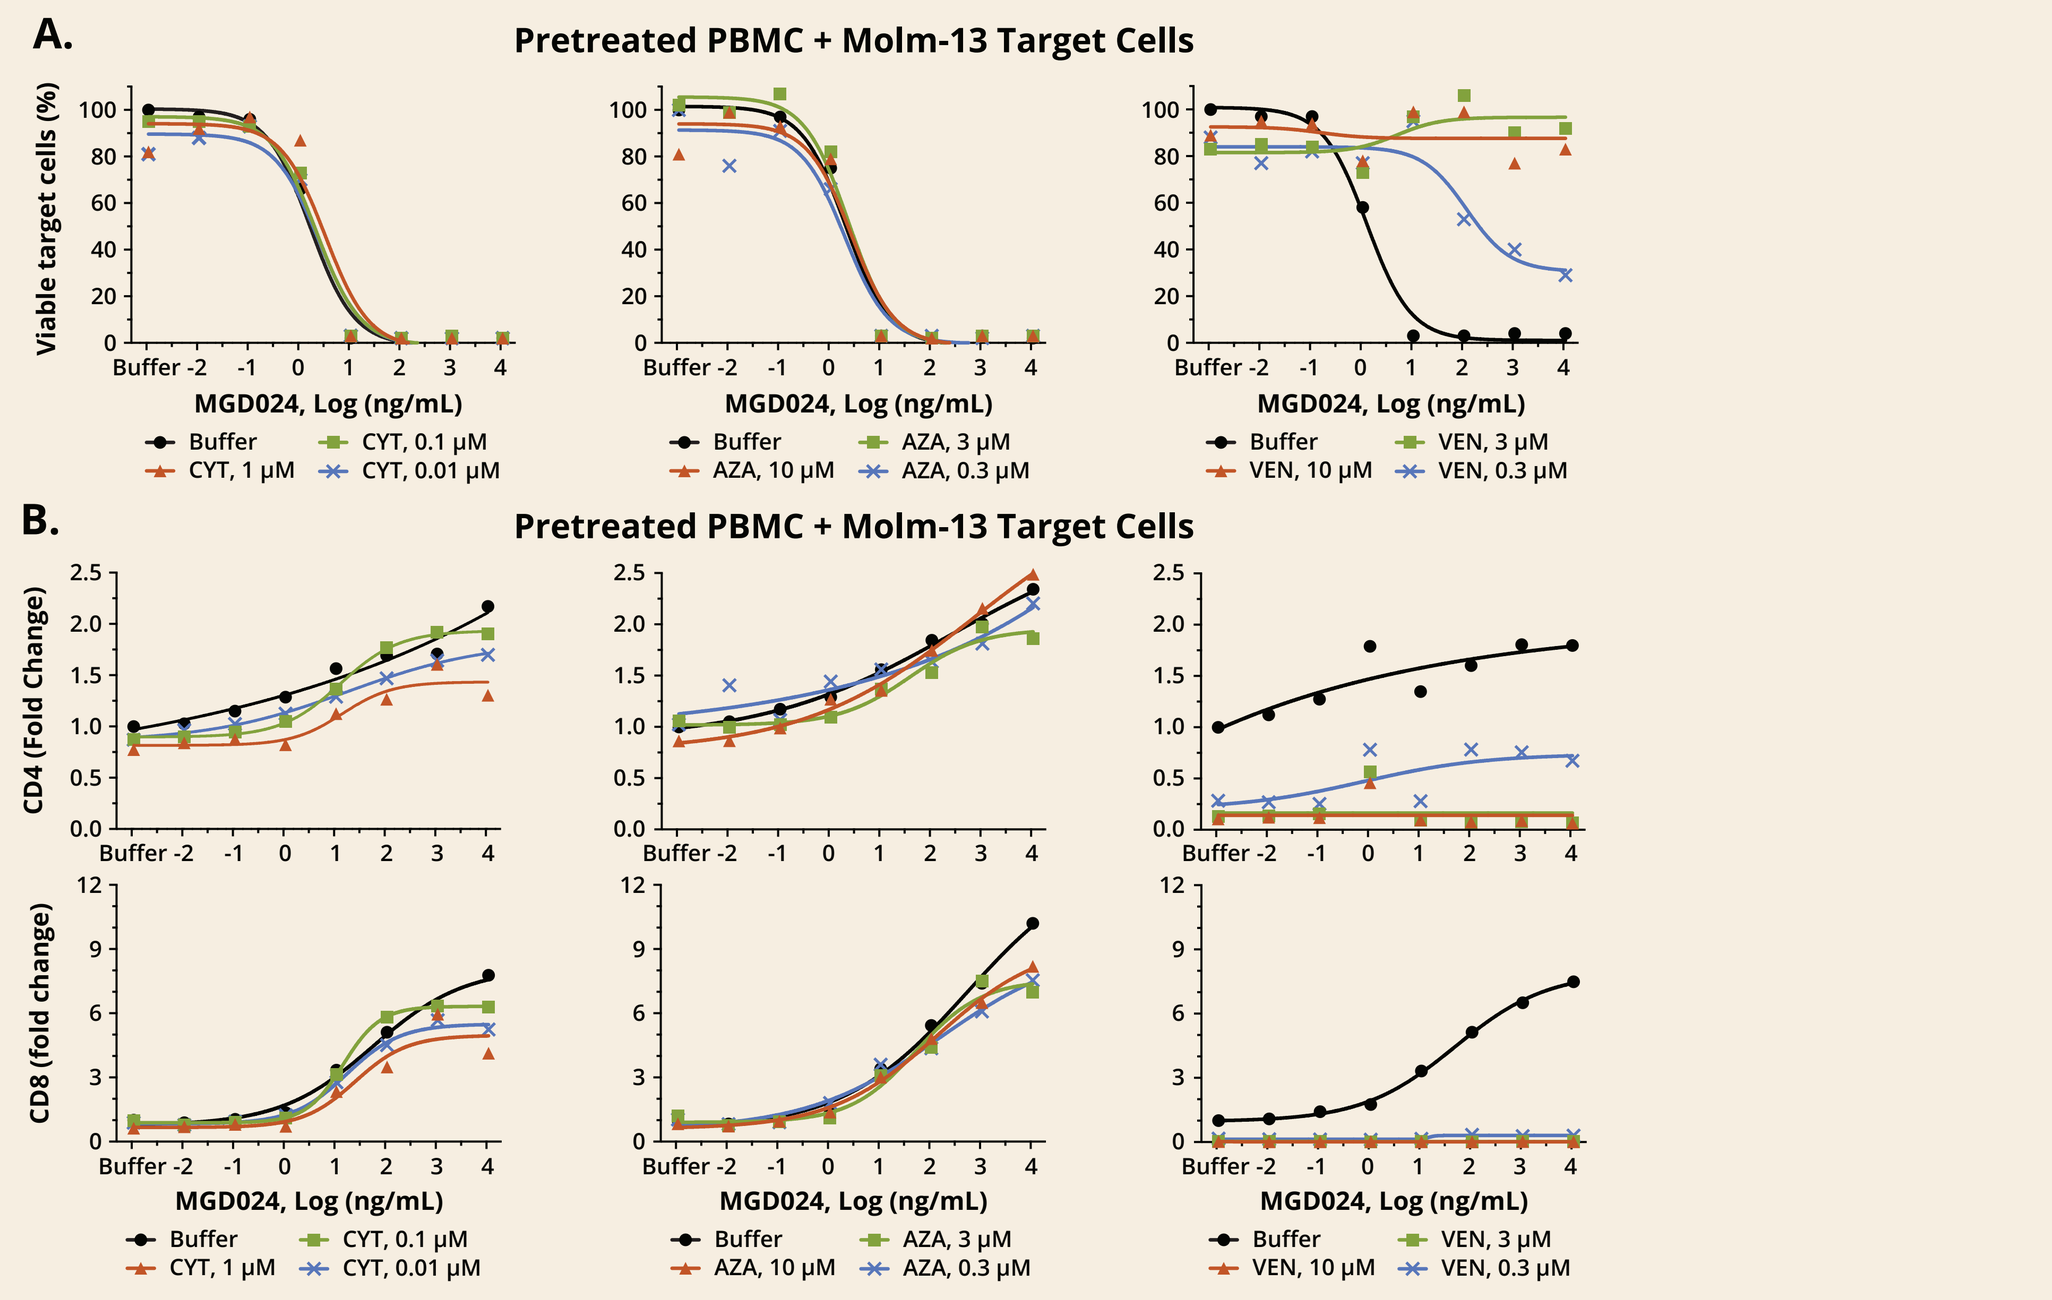

A.
Pretreated PBMC + Molm-13 Target Cells
Viable target cells (%)
MGD024, Log (ng/mL)
B.
Pretreated PBMC + Molm-13 Target Cells
CD4 (Fold Change)
CD8 (fold change)
Buffer
CYT, 1 µM
CYT, 0.1 µM
CYT, 0.01 µM
AZA, 10 µM
AZA, 3 µM
AZA, 0.3 µM
VEN, 10 µM
VEN, 3 µM
VEN, 0.3 µM

- Pretreatment of effector cells with VEN, but not CYT or AZA, inhibits MGD024-mediated cytolysis
- Pre-exposure to VEN, but not CYT or AZA, inhibits MGD024-induced T-cell expansion

PBMCs (1 x 10^6/mL) were treated with CYT (0.01, 0.1, 1 µM), AZA (0.3, 3, 10 µM), VEN (0.3 , 3, 10, µM) for 3 days at 37C in culture flasks, washed, resuspended in the original volume and distributed on 96-well plates containing PKH26-labeled Molm-13 cells (1 x 10^4 cells/well) as targets. Medium or MGD024 at final concentrations ranging from 0.001 ng/mL to 10 µg/mL were added to replicate wells. After 96 hours of incubation at 37C, wells were harvested and analyzed for target-cell depletion and T-cell expansion by flow cytometry.
A. Molm-13 cell killing activity is shown as the fraction of viable cells normalized by the number of viable cell count in untreated controls.
B. CD4 and CD8 T-cell expansion are shown as fold-changes over untreated controls.

Suboptimal CYT Enhances MGD024-mediated Anti-tumor Activity In Vivo

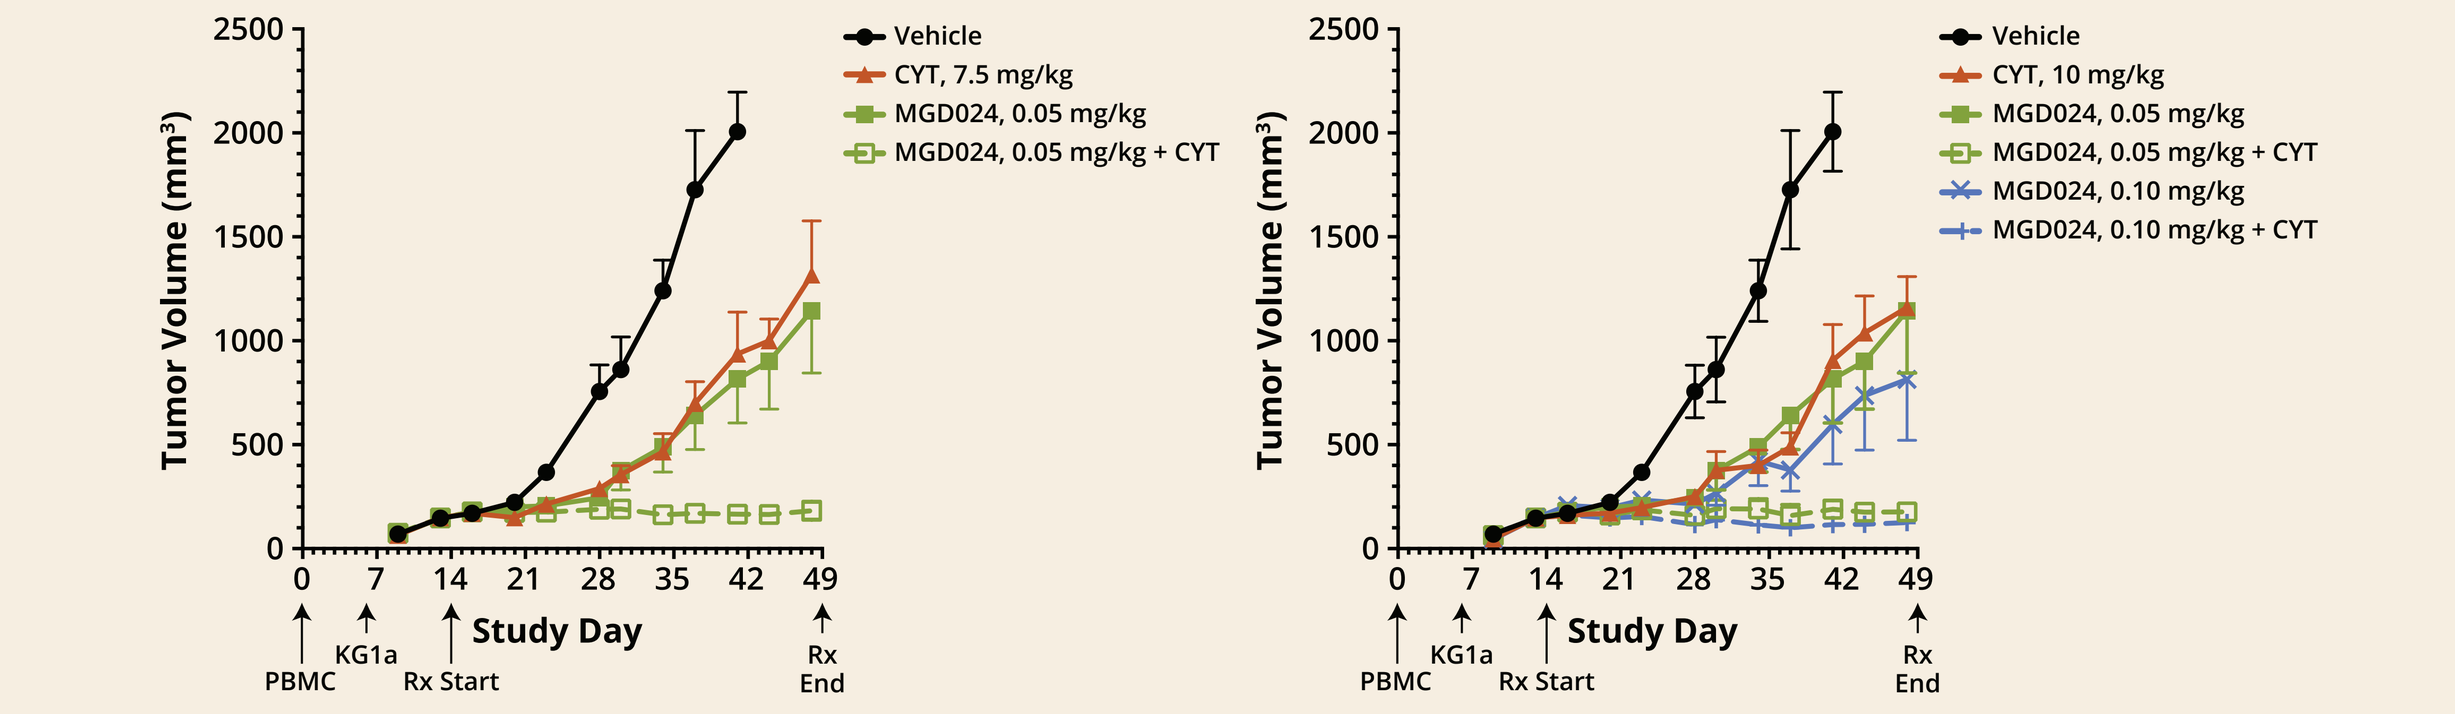

Tumor Volume (mm³)
Study Day
PBMC
KG1a
Rx Start
Rx End
Vehicle
CYT, 7.5 mg/kg
MGD024, 0.05 mg/kg
MGD024, 0.05 mg/kg + CYT
CYT, 10 mg/kg
MGD024, 0.10 mg/kg
MGD024, 0.10 mg/kg + CYT

MHC class I-*null*, NOD/SCID/IL2R-γ-*null* mice were reconstituted ro with human PBMC as described in Material & Methods. KG1a cells (5 x 10^6 cells/mouse) were implanted sc 1:1 in Matrigel on Study Day 6. On study Day 14, mice were randomized based on tumor volume (N=6/group) and treatment started with vehicle, MGD024 iv 2QW or cytarabine (CYT) ip QD (M-F).

AZA Does Not Enhance or Inhibit MGD024 Anti-tumor Activity In Vivo

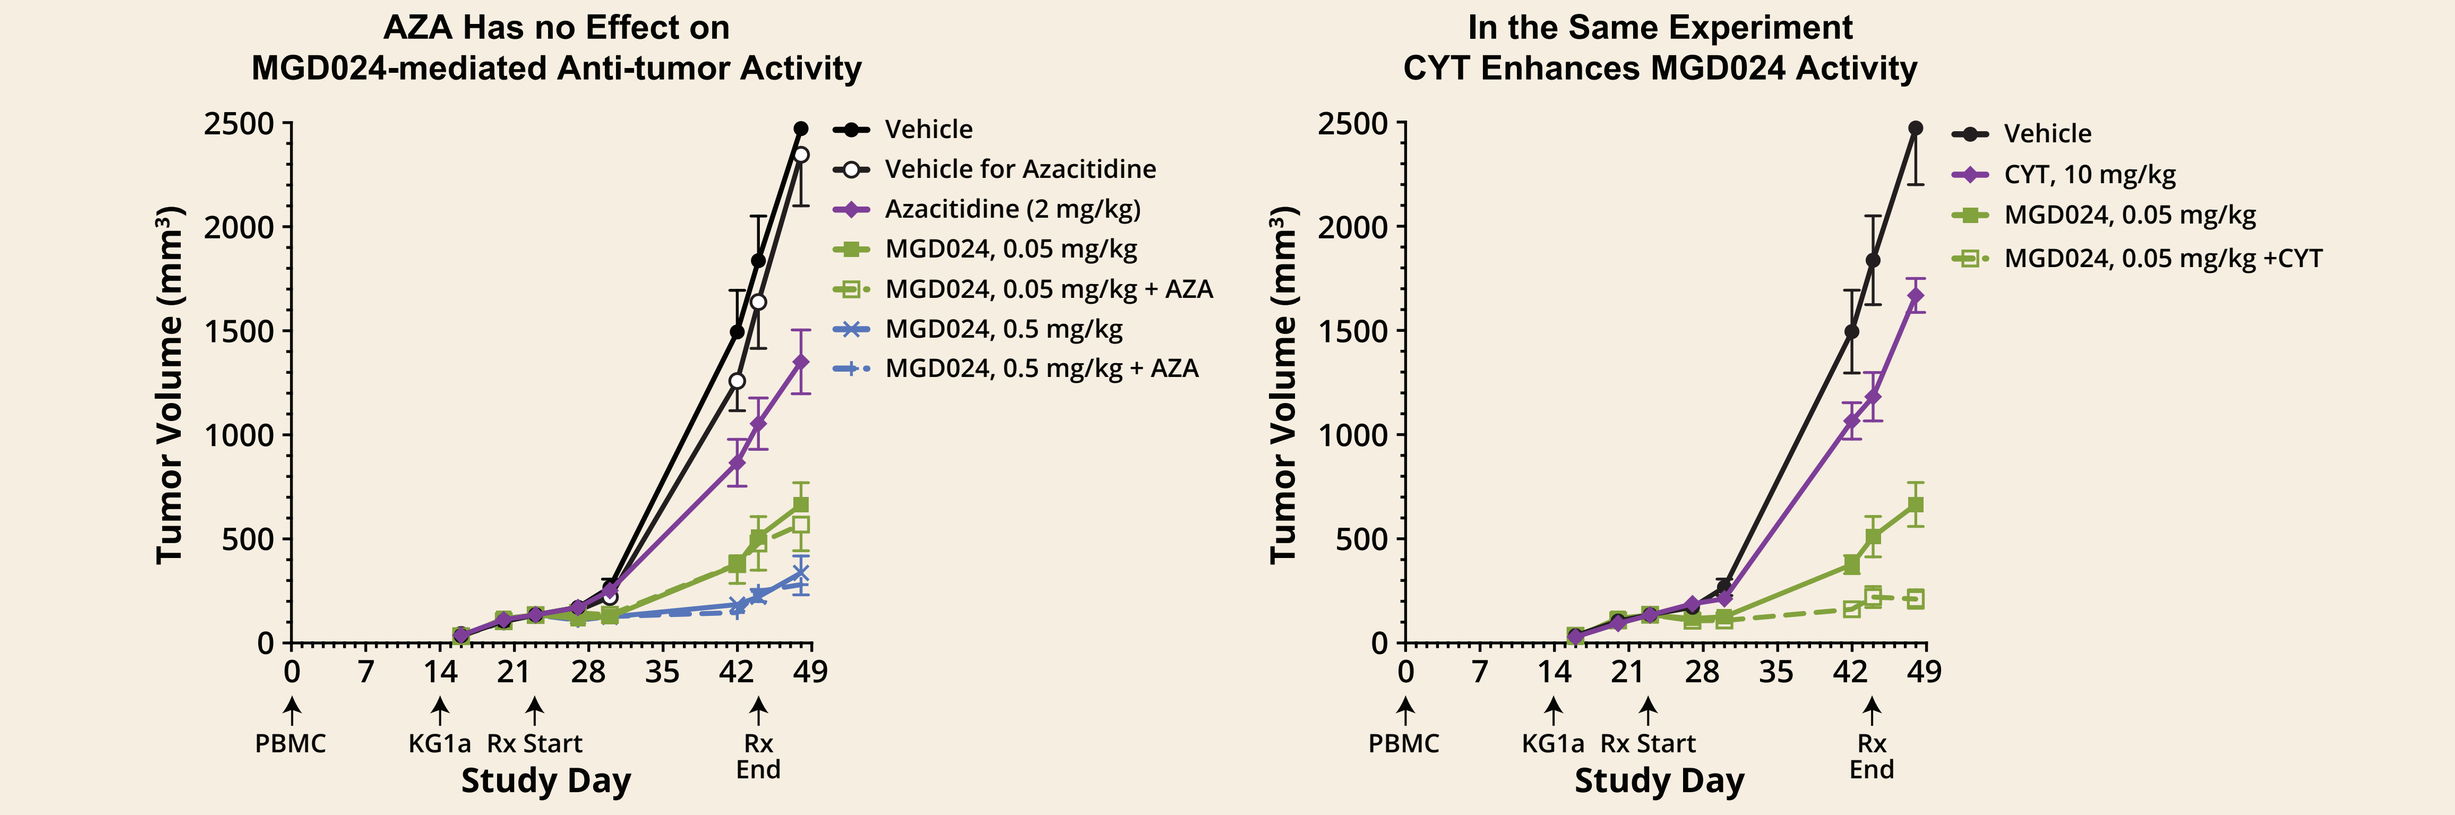

AZA Has no Effect on MGD024-mediated Anti-tumor Activity
In the Same Experiment CYT Enhances MGD024 Activity
Tumor Volume (mm³)
Study Day
PBMC
KG1a
Rx Start
Rx End
Vehicle
Vehicle for Azacitidine
Azacitidine (2 mg/kg)
MGD024, 0.05 mg/kg
MGD024, 0.05 mg/kg + AZA
MGD024, 0.5 mg/kg
MGD024, 0.5 mg/kg + AZA
CYT, 10 mg/kg
MGD024, 0.05 mg/kg +CYT

MHC class I-*null*, NOD/SCID/IL2R-γ-*null* mice were reconstituted ro with human PBMC as described in Material & Methods. KG1a cells (2.5 x 10^6 cells/mouse) were implanted sc 1:1 in Matrigel on Study Day 15. On Study Day 23 mice were randomized based on tumor volume (N=8/group) and treatments initiated through Study Day 44. MGD024 or vehicle control were administered iv 2QW. Azacitidine (AZA) or cytarabine (CYT) were administered ip QD (M-F).

Combinatorial Anti-tumor Activity of MGD024 Co-administered with Suboptimal VEN

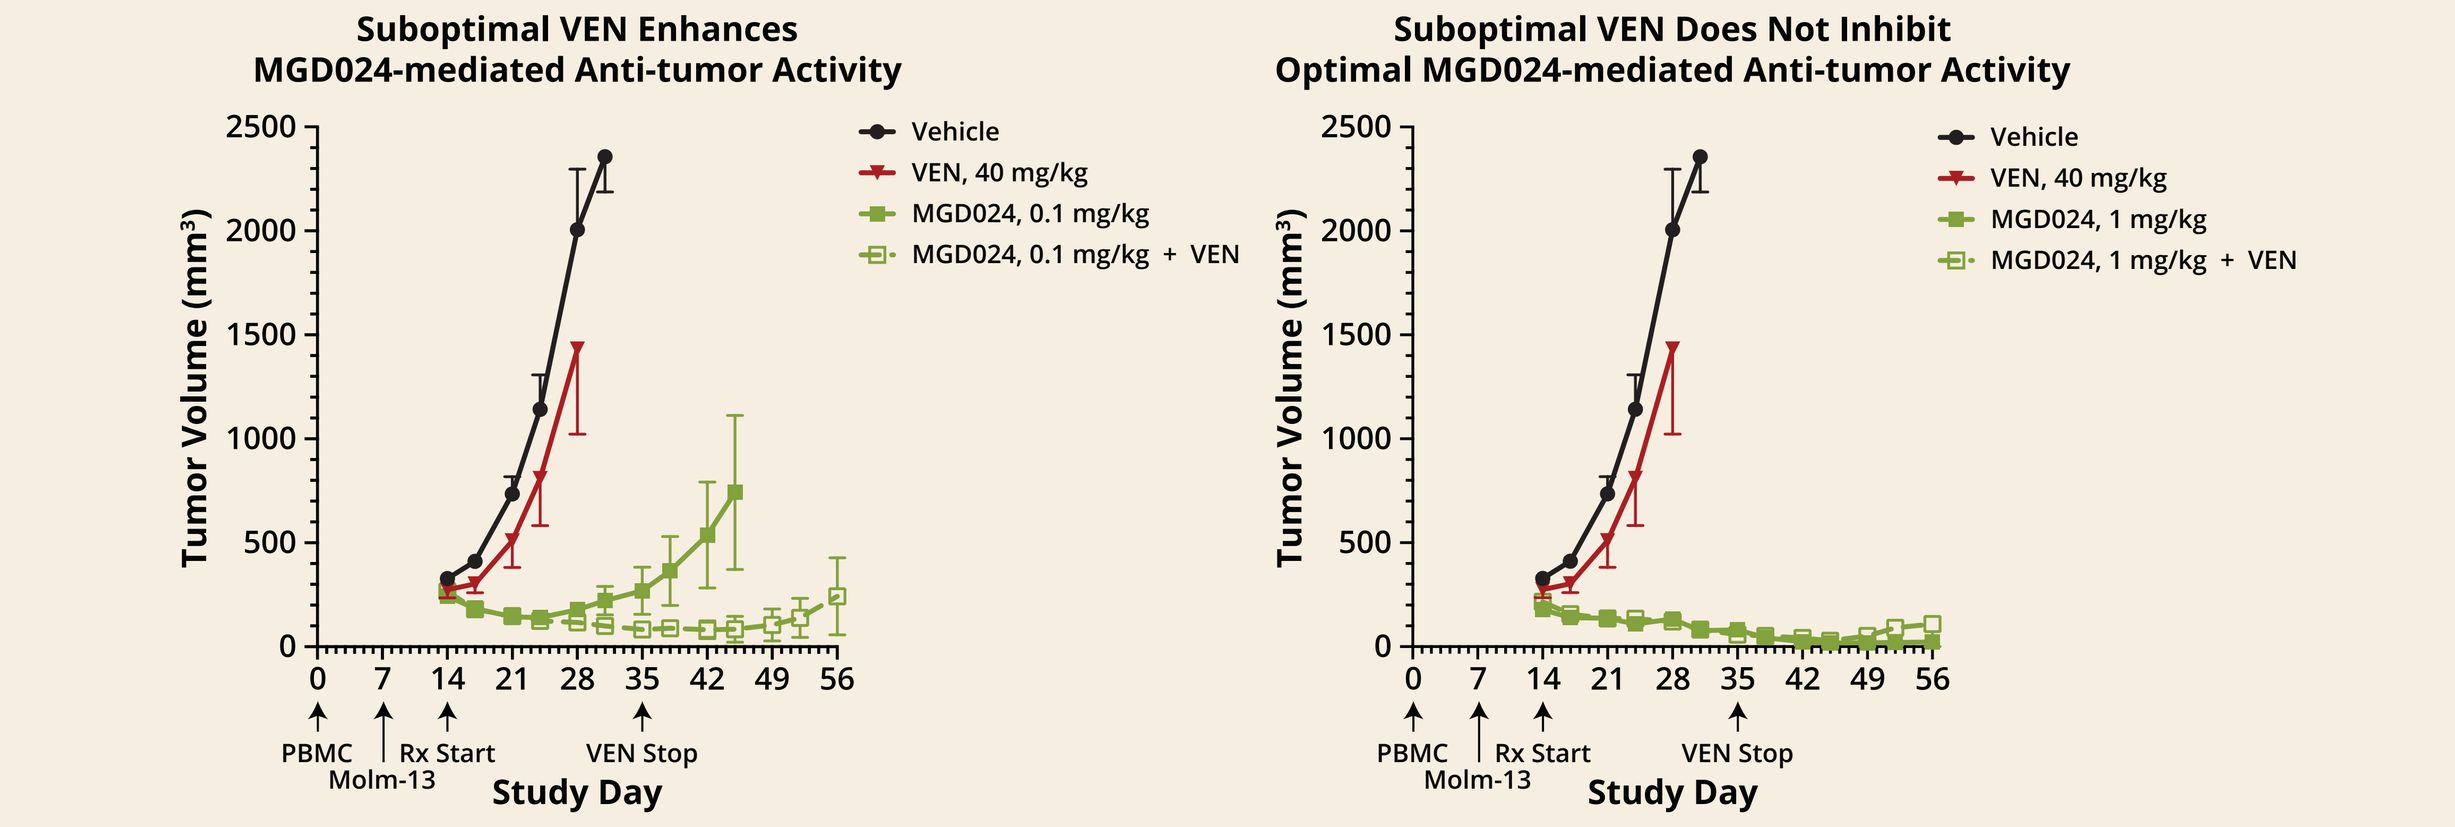

Suboptimal VEN Enhances MGD024-mediated Anti-tumor Activity
Suboptimal VEN Does Not Inhibit Optimal MGD024-mediated Anti-tumor Activity
Tumor Volume (mm³)
Study Day
PBMC
Molm-13
Rx Start
VEN Stop
Vehicle
VEN, 40 mg/kg
MGD024, 0.1 mg/kg
MGD024, 0.1 mg/kg + VEN
MGD024, 1 mg/kg
MGD024, 1 mg/kg + VEN

MHC class I-*null*, NOD/SCID/IL2R-γ-*null* mice were reconstituted ro with human PBMC as described in Material & Methods. Molm-13 cells (5 x 10^6 cells/mouse) were implanted sc 1:1 in Matrigel on Study Day 7. On Study Day 14 mice were randomized based on tumor volume (N=8/group) and treatments initiated through Study Day 56. MGD024 or vehicle control were administered iv 2QW. Venetoclax (VEN) was administered po via gavage QD (M-F) for 3 weeks (Study Days 14–35). The typical mouse venetoclax oral dose ranges between 100 and 200 mg/kg QD for 3 weeks.

Conclusions

- MGD024, a second generation CD123 x CD3 DART molecule, showed:
 - Anti-leukemic activity in vitro and in murine tumor models
 - Good tolerability in cynomolgus monkeys with reduced cytokine release
 - A pharmacokinetic profile consistent with dosing patient on a weekly basis or longer interval
- The combination of MGD024 with SoC agents demonstrated the following features:

SoC Agents	In vitro Effect on:						In Vivo Effect on MGD024 Anti-tumor Activity
	T-cell Viability		MGD024-induced T-cell Expansion		Target Cell Viability in the Presence of MGD024		
	Resting	Activated (MGD024)	Concomitant Exposure	Pre-treatment	Concomitant Exposure	Pre-treatment	
Cytarabine (Low Dose)	No effect	N.D.	Some reduction	No effect	Additive cytolysis	No effect	Enhanced
Azacitidine	No effect	N.D.	Some reduction	No effect	Additive cytolysis	No effect	No effect
Venetoclax	Reduced	No effect (CD8)	Some reduction	Reduced	Additive cytolysis	Reduced cytolysis	Enhanced

N.D., not done

- Low-dose cytarabine has the greatest potential for combinatorial anti-tumor activity with MGD024
- Venetoclax reduces resting T-cell viability, but spares MGD024-activated CD8 cells in vitro and demonstrated combinatorial anti-tumor activity in vivo
- Azacitidine, while devoid of combinatorial activity, does not affect the anti-tumor activity of MGD024

These data support clinical investigation of MGD024 as single agent and/or in combination with SoC in patients with CD123-positive hematological malignancies.

References

1. Testa et al., *Cancers* (Basel). 2019, 11:1358. **2.** Uy et al., *Blood.* 2021, 137:751. **3.** Spriggs D. et al., *Blood.* 1985, 65: 1087-1089; **4.** Vidaza Prescribing Information, Rev. 03/2020; **5.** Venclexta Prescribing Information, Rev. 20/2021.

Presented at the 2021 ASH Annual Meeting and Exposition Meeting, December 11–14, 2020

©2021 MacroGenics, Inc. All rights reserved.