Targeting B7-H3 in Squamous Cell Carcinoma of the Head and Neck: Preclinical Proof-of-Concept with the Investigational Anti-B7-H3 Antibody-drug Conjugate, MGC018

Juniper A. Scribner, Francine Z. Chen, Ying Li, Michael Chiechi, Jeff Hooley, Thomas Son, Scott Koenig, Paul A. Moore, Ezio Bonvini, Chet Bohac, Deryk Loo

MacroGenics, Inc., Brisbane, CA and Rockville, MD

950

Presenter Contact Information: scribnerj@macrogenics.com

Abstract

Introduction: Squamous cell carcinoma of the head and neck (SCCHN) is the seventh most common cancer worldwide, accounting for ~4% of all cancers in the United States. Although PD-1-directed therapy has efficacy in SCCHN, 85–95% of patients progress following initial response. B7-H3 is a member of the B7 family of immunomodulatory molecules, is overexpressed in SCCHN, and correlates with disease severity and poor clinical outcome. Furthermore, consistent with its putative coinhibitory function, B7-H3 expression in SCCHN is inversely correlated with the number of tumor infiltrating CD8+ T-cells. We are developing therapeutics targeting B7-H3, including enoblituzumab, an Fc-engineered anti-B7-H3 monoclonal antibody, and MGC018, a duocarmycin-based B7-H3 ADC, both of which are currently being evaluated in clinical studies. Here the potential of MGC018 was explored in preclinical models as a proof of concept for targeting B7-H3 in SCCHN.

Methods: Immunohistochemistry studies were performed to define the expression of B7-H3 in SCCHN tissue microarrays (TMA). Single and repeat-dose in vivo efficacy studies were conducted in CD-1 nude mice with cell line-derived SCCHN human tumor xenografts to explore the relationship between C_{max}, exposure and antitumor activity, and to define the minimal efficacious dose in these models. Based on the results in these cell-derived xenograft (CDX) studies, in vivo efficacy studies were extended to a panel of SCCHN patient-derived xenograft (PDX) models, which more closely mimic the biological characteristics of the patient tumor and exhibit heterogenous expression of B7-H3.

Results: Analysis of B7-H3 expression on a SCCHN TMA confirmed and extended previously reported expression of B7-H3 in SCCHN. Of the SCCHN samples evaluated, 90% (36/40) of the tumor samples were positive for B7-H3: 35% (14/40) had H-scores greater than 200, with the remaining 22 samples equally distributed between the H-score range of 101–200 and 1–100 (~28% each). MGC018 demonstrated specific, dose-dependent in vitro cytotoxicity toward SCCHN human tumor cell lines. The in vitro cytotoxicity translated to potent antitumor activity in vivo against SCCHN CDX models, with a single administration of 3 mg/kg resulting in complete responses in 7/7 mice in the FaDu model. In the PDX setting (H-scores 120–283), repeat dose administration every week or two weeks with MGC018 at 3 mg/kg/dose, led to regressions and/or stable disease in 10/19 models, and a delay in tumor growth in 5 additional models.

Conclusion: B7-H3 is frequently overexpressed in SCCHN. At clinically relevant dose levels, MGC018 demonstrated potent antitumor activity in vivo toward SCCHN CDX models and the majority of SCCHN PDX models examined. These results support SCCHN as a potential indication that may be responsive to ADC-based treatments directed toward B7-H3.

Introduction

MGC018: A Clinical-stage Anti-B7-H3 ADC Therapeutic

B7-H3

- Member of the B7-family of immune regulators
- Overexpressed on solid cancer, including SCCHN, with high tumor-versus-normal tissue binding differential
- Overexpression in SCCHN correlated with disease severity and poor outcome

MGC018

- Comprised of a humanized antibody targeting B7-H3 (MGA017)
- Conjugated to a duocarmycin-based DNA alkylating payload via native cysteines
- Cleavable peptide linker — facilitates bystander effect
- Potent antitumor activity in mouse models toward B7-H3-expressing human tumor xenografts at dose levels used in clinical studies, despite model limitations
 - Unlike humans and non-human primates, rodents express carboxylesterase CES1c that cleaves the ADC payload and limits exposure[1,2]
- Phase 1/2 clinical study in advanced solid cancers in progress (NCT03729596)

Duocarmycin-based Linker Payload

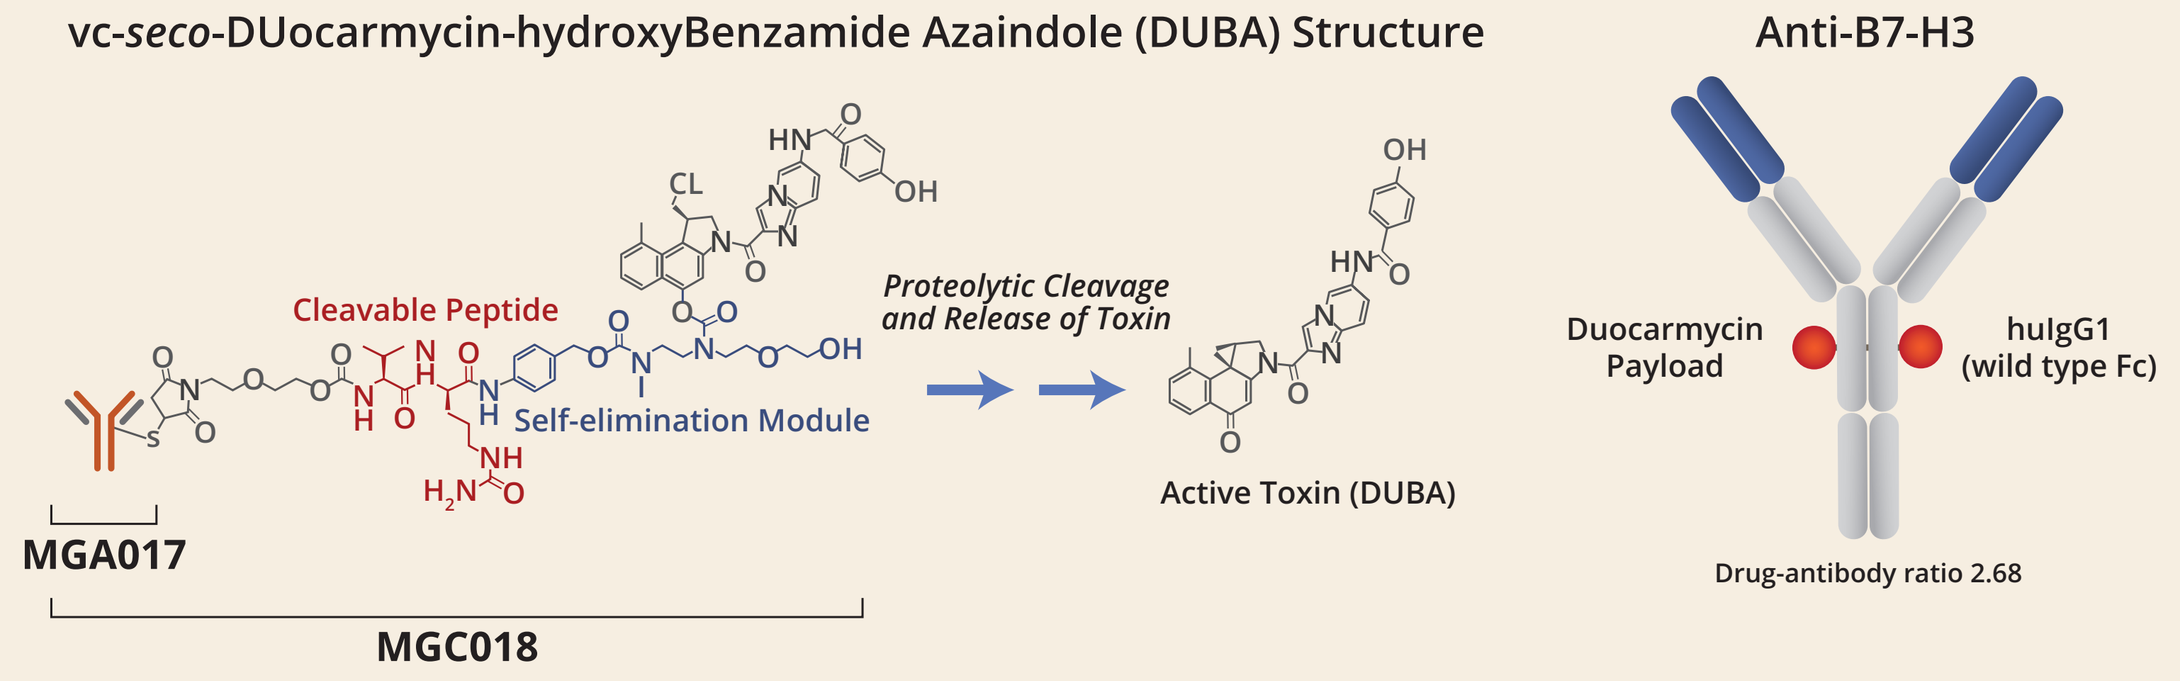

vc-seco-DUocarmycin-hydroxyBenzamide Azaindole (DUBA) Structure
Anti-B7-H3
Cleavable Peptide
Self-elimination Module
Proteolytic Cleavage and Release of Toxin
Active Toxin (DUBA)
Duocarmycin Payload
huIgG1 (wild type Fc)
Drug-antibody ratio 2.68
MGA017
MGC018

Head and Neck Cancer

- The majority of head and neck cancers are squamous cell carcinomas
 - Arising from flat squamous cells in the thin layer of tissue on the surface of structures in the head and neck
- Head and neck cancer accounts for ~4% of all cancers in the United States[3]
- It was estimated that in 2020, 65,630 people would develop head and neck cancer and 14,500 people would die from the disease[3]
- The stage at diagnosis predicts survival rates
 - The overall 5 year survival rate ranges from 58-71% depending on the type of cancer; however, the rate drops if the cancer has spread to surrounding tissues and/or regional lymph nodes (45-73%) or distant parts of the body (33-48%)[3]

Objectives

- Explore the expression profile and prevalence of B7-H3 in SCCHN
- Evaluate the therapeutic potential of MGC018 in preclinical models of SCCHN

Results

B7-H3 Is Overexpressed in SCCHN

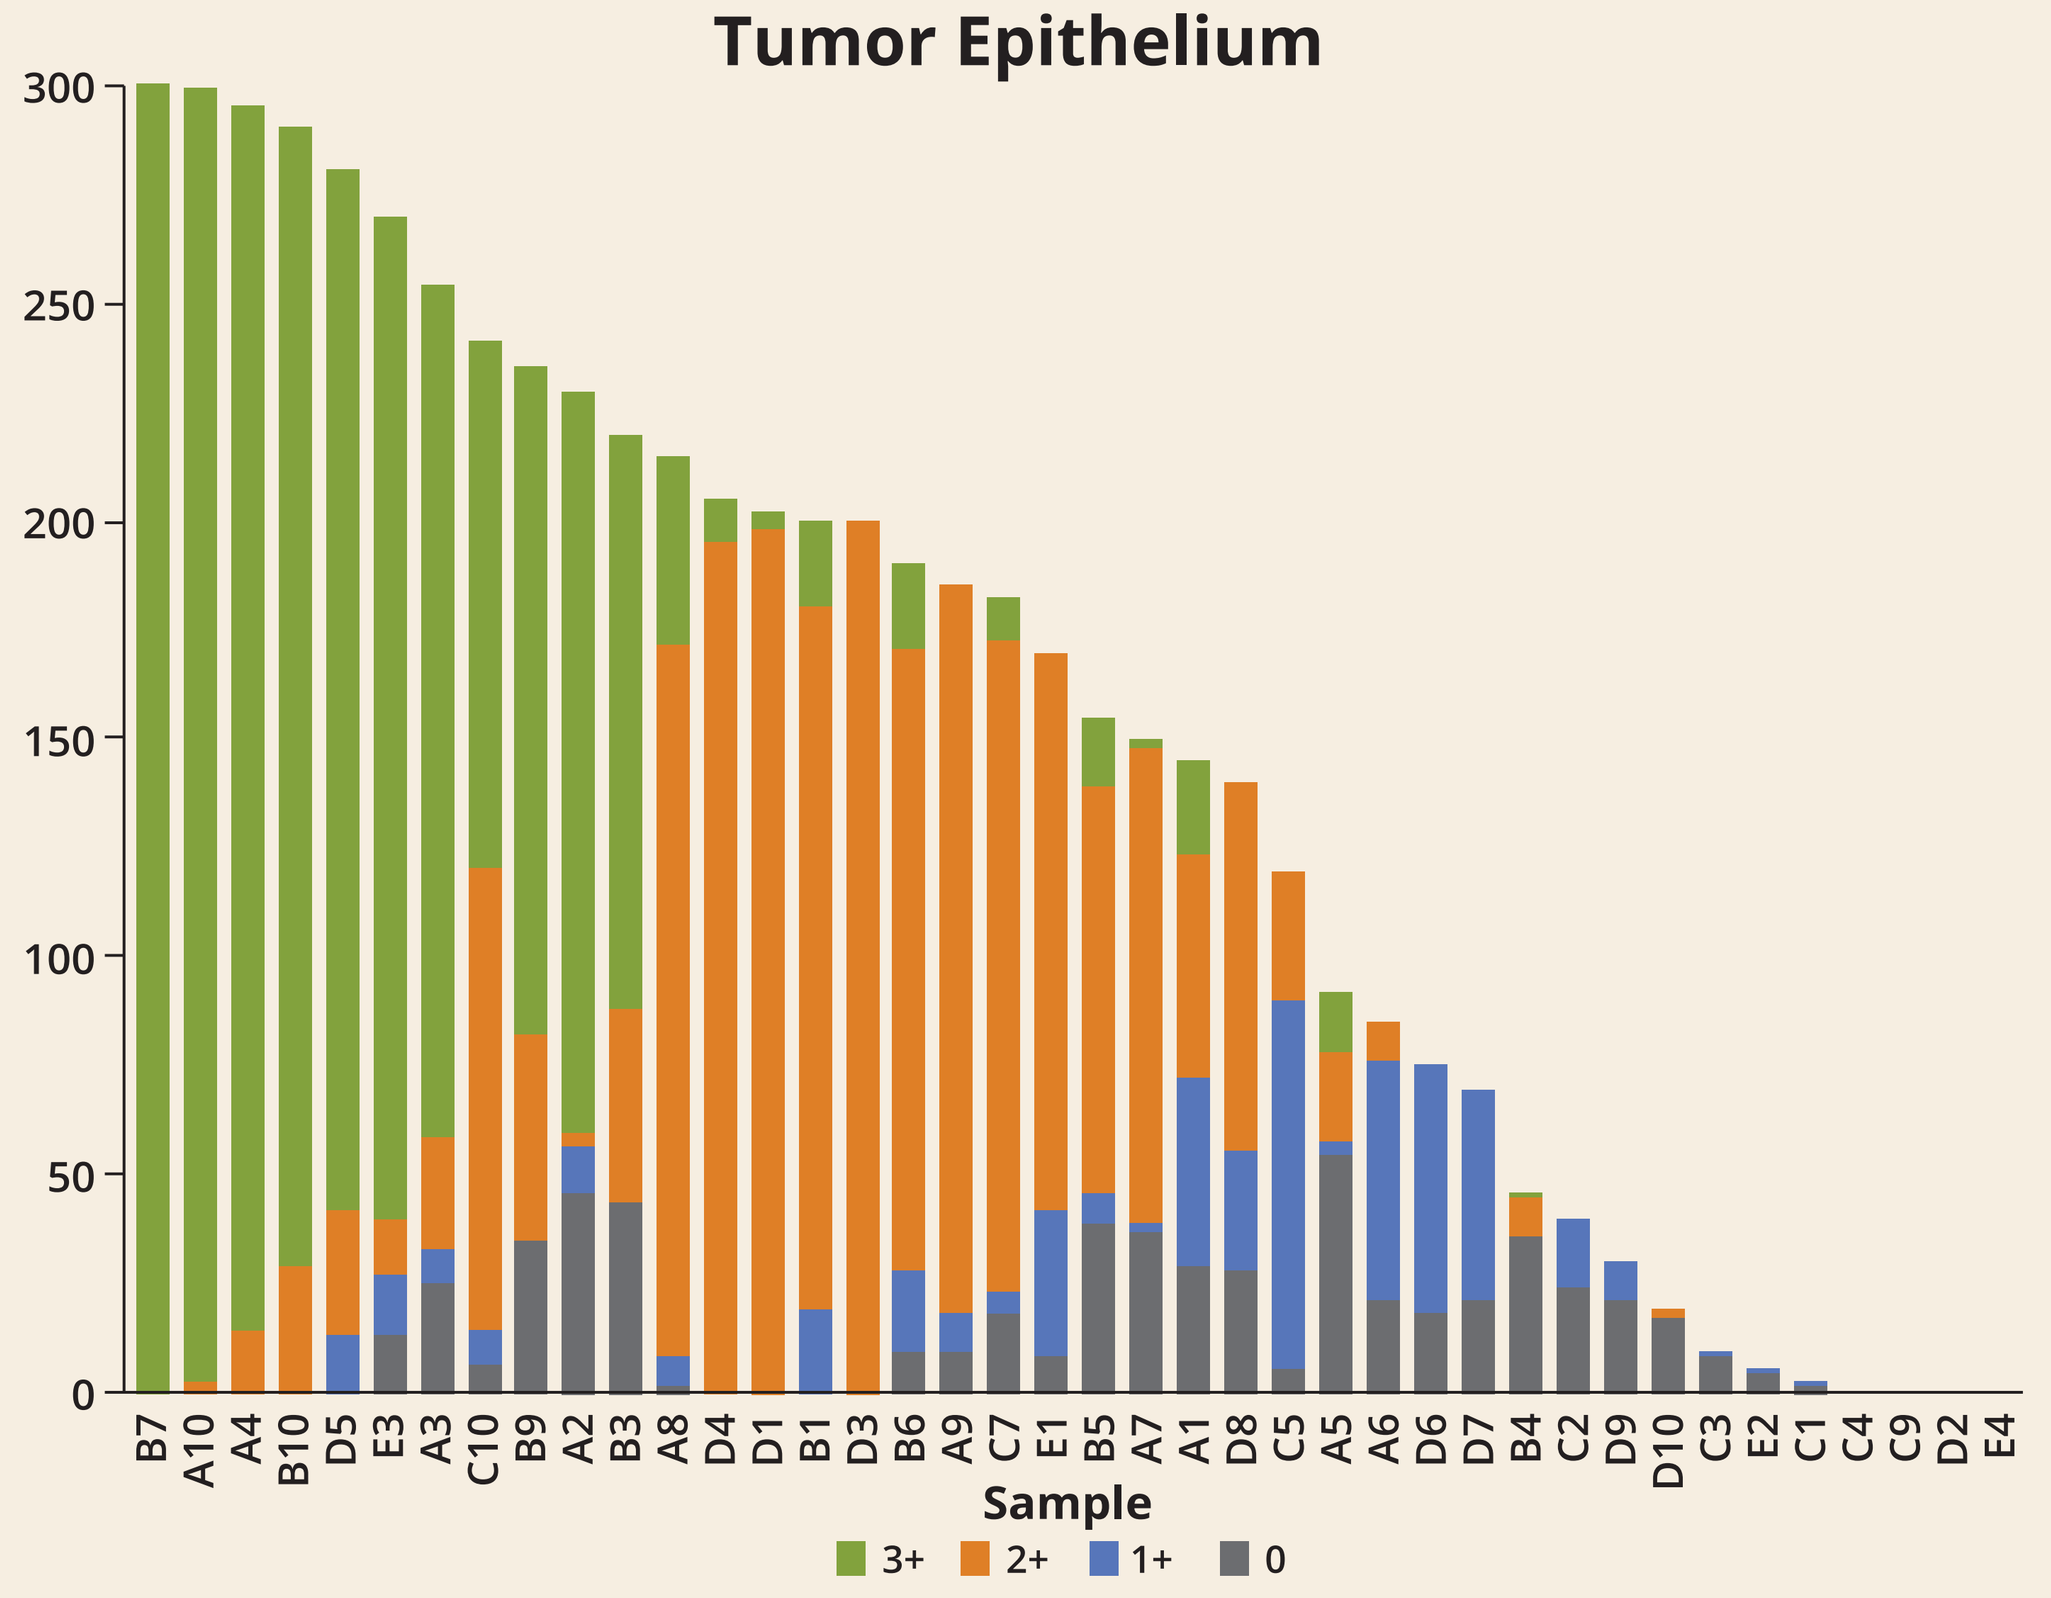

Tumor Epithelium
H-score
Sample
3+
2+
1+
0

- 90% of human tumor samples positive for B7-H3, with average H-score of 162
- 63% of human tumor samples have H-scores > 100, 35% > 200

Membrane expression evaluated with SP206 rabbit pAb on FFPE tissue microarray. H-score: Semiquantitative method that incorporates the intensity level (0, 1+, 2+, or 3+) and percentage of cells staining; ranges from 0 (no staining) to 300 (maximal staining), using the formula [1(% cells 1+) + 2(% cells 2+) + 3(% cells 3+)].

B7-H3 Tumor Expression and Heterogeneity Is Generally Recapitulated in PDX Models

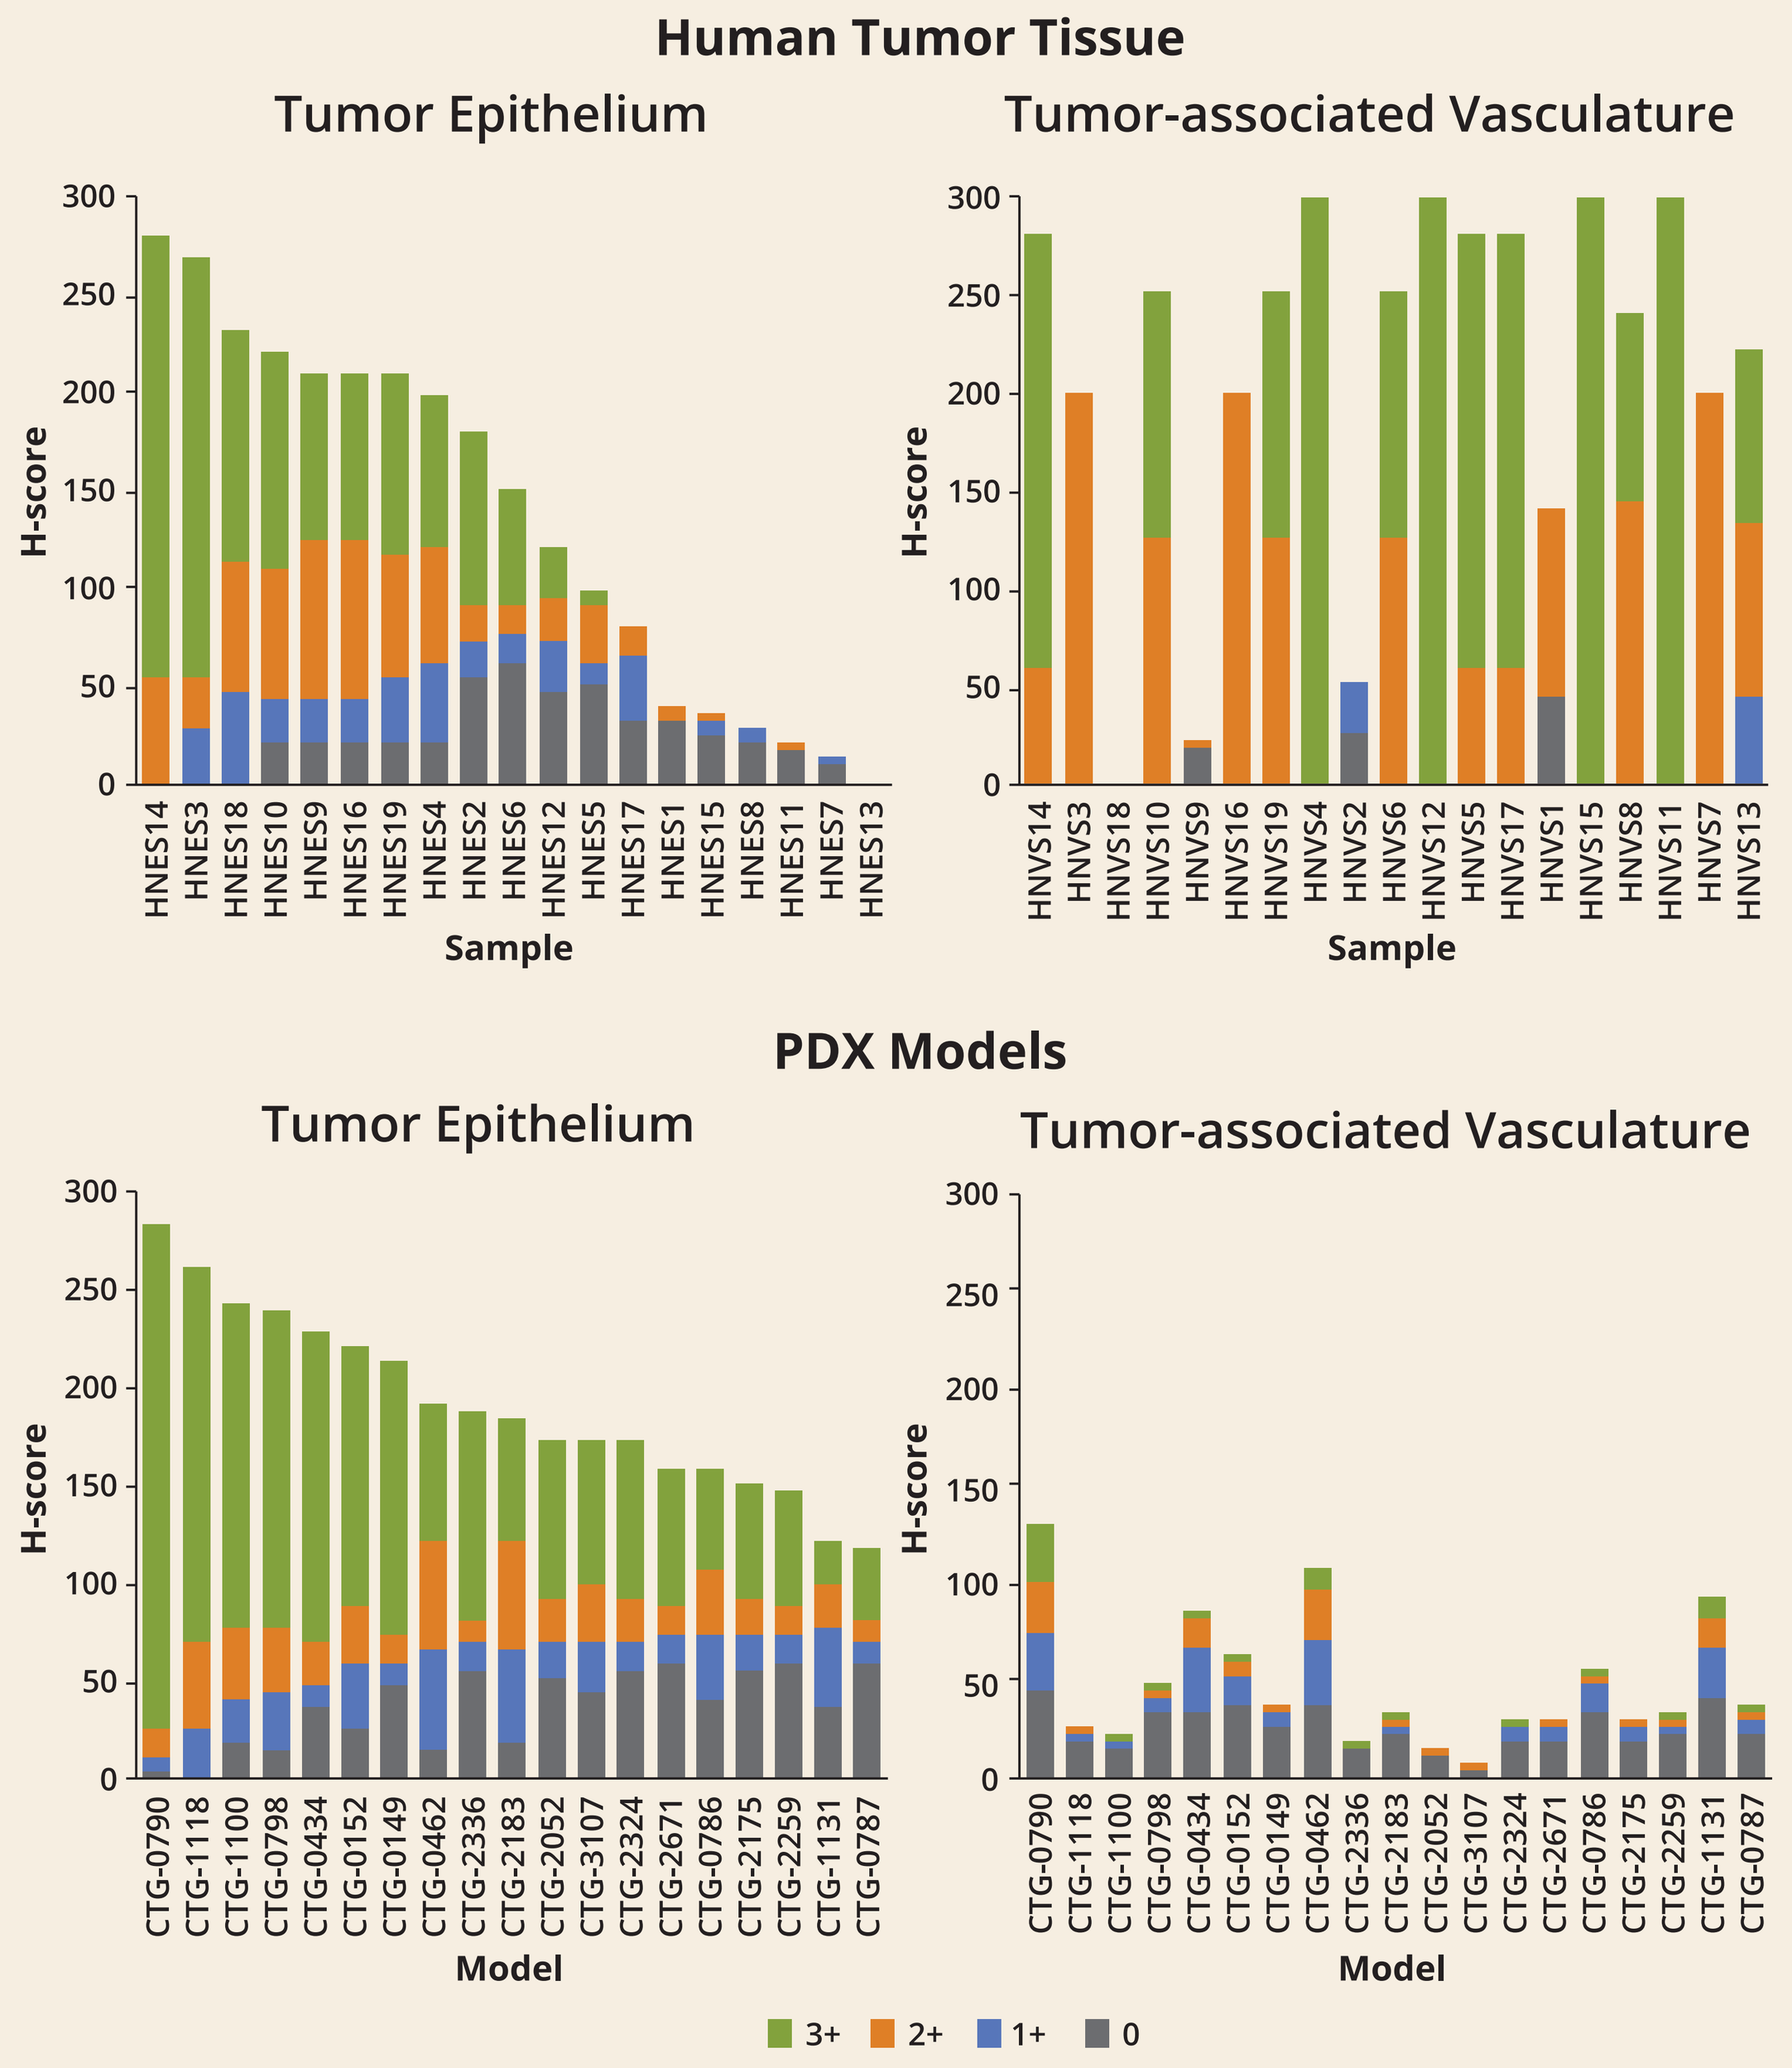

Human Tumor Tissue
Tumor Epithelium
Tumor-associated Vasculature
PDX Models
H-score
Sample
Model
3+
2+
1+
0

- Heterogeneous expression of B7-H3 observed on both tumor epithelium and tumor-associated vasculature of human tumor samples
 - 100% of samples were positive for B7-H3 on tumor epithelium and/or tumor-associated vasculature
- Expression of B7-H3 in PDX models is similar to human tumor samples, but with lower vascular expression

Membrane expression evaluated with R&D Systems Goat anti-B7-H3 pAb on FFPE tissues.

MGC018 Mediates Apoptosis of FaDu SCCHN Tumor Cells

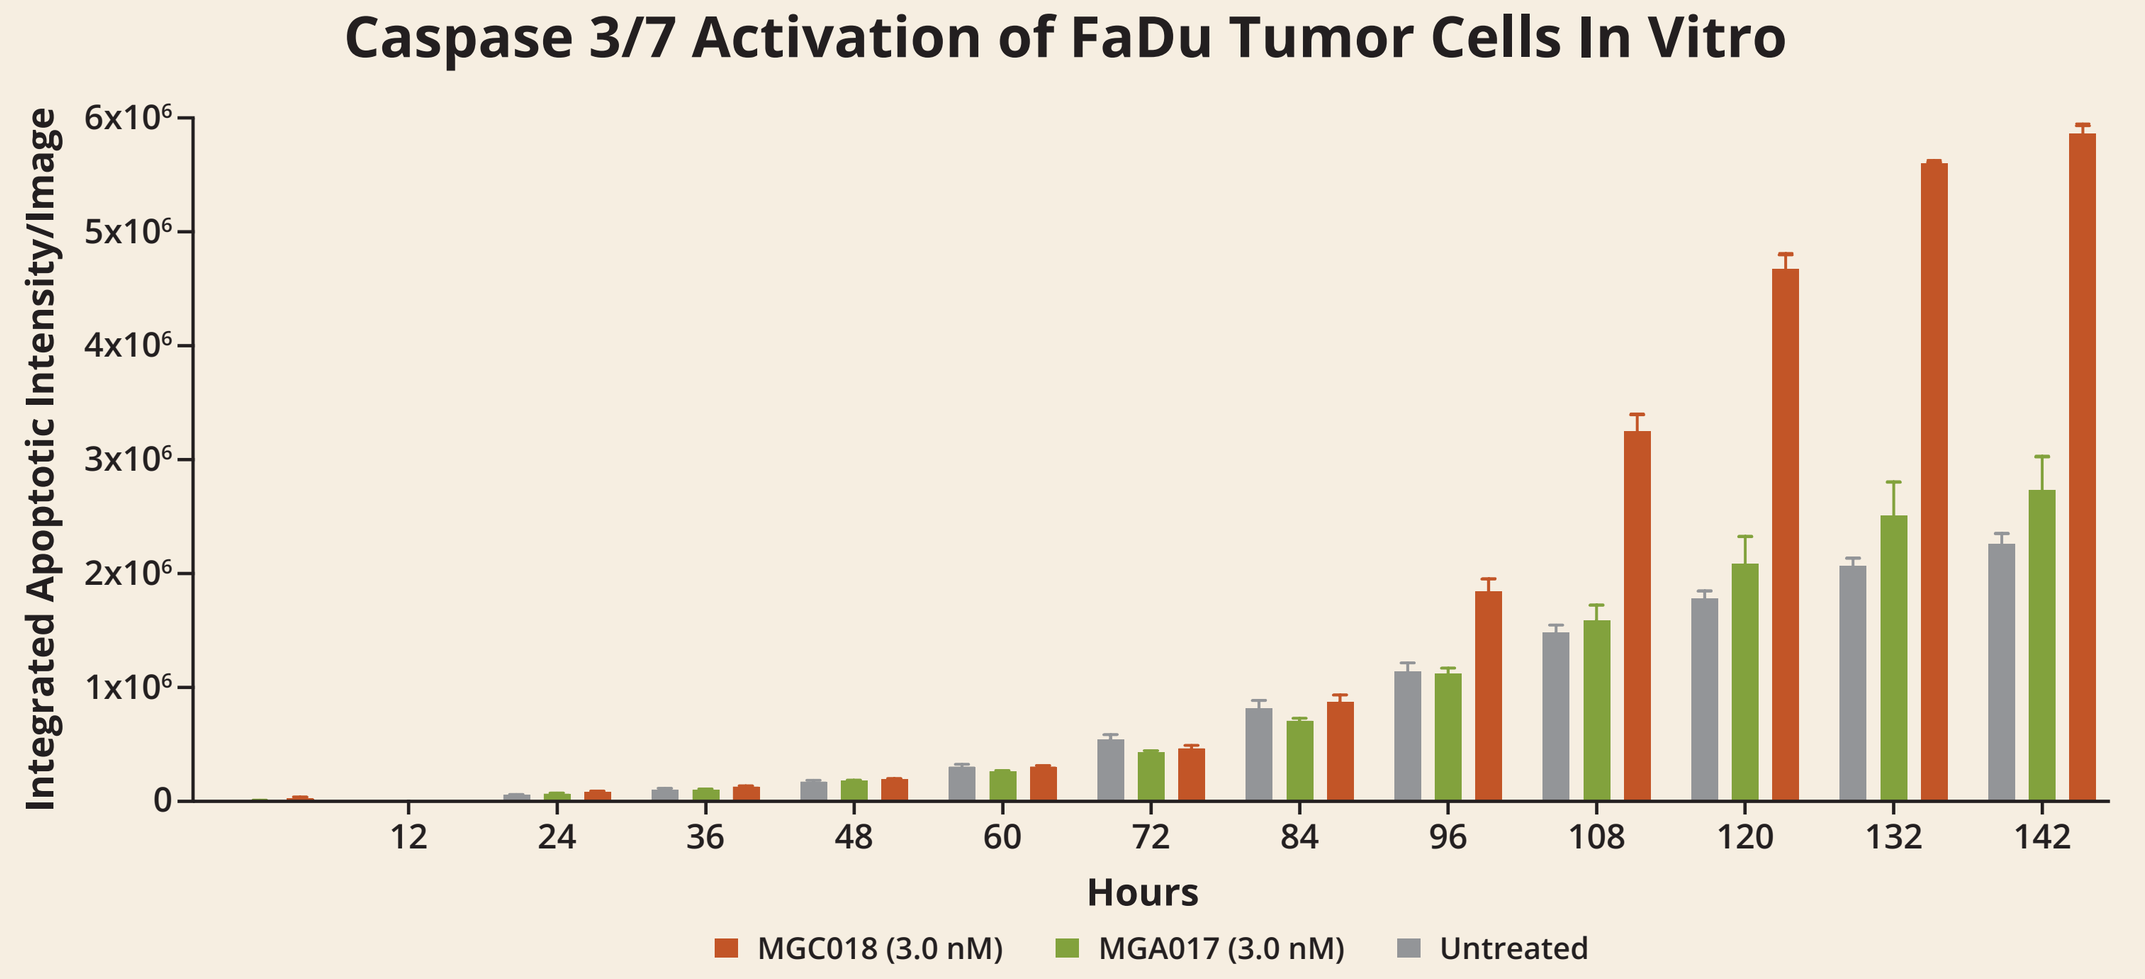

Caspase 3/7 Activation of FaDu Tumor Cells In Vitro
Integrated Apoptotic Intensity/Image
Hours
MGC018 (3.0 nM)
MGA017 (3.0 nM)
Untreated

MGC018 Leads to Cell Cycle Arrest of FaDu SCCHN Tumor Cells

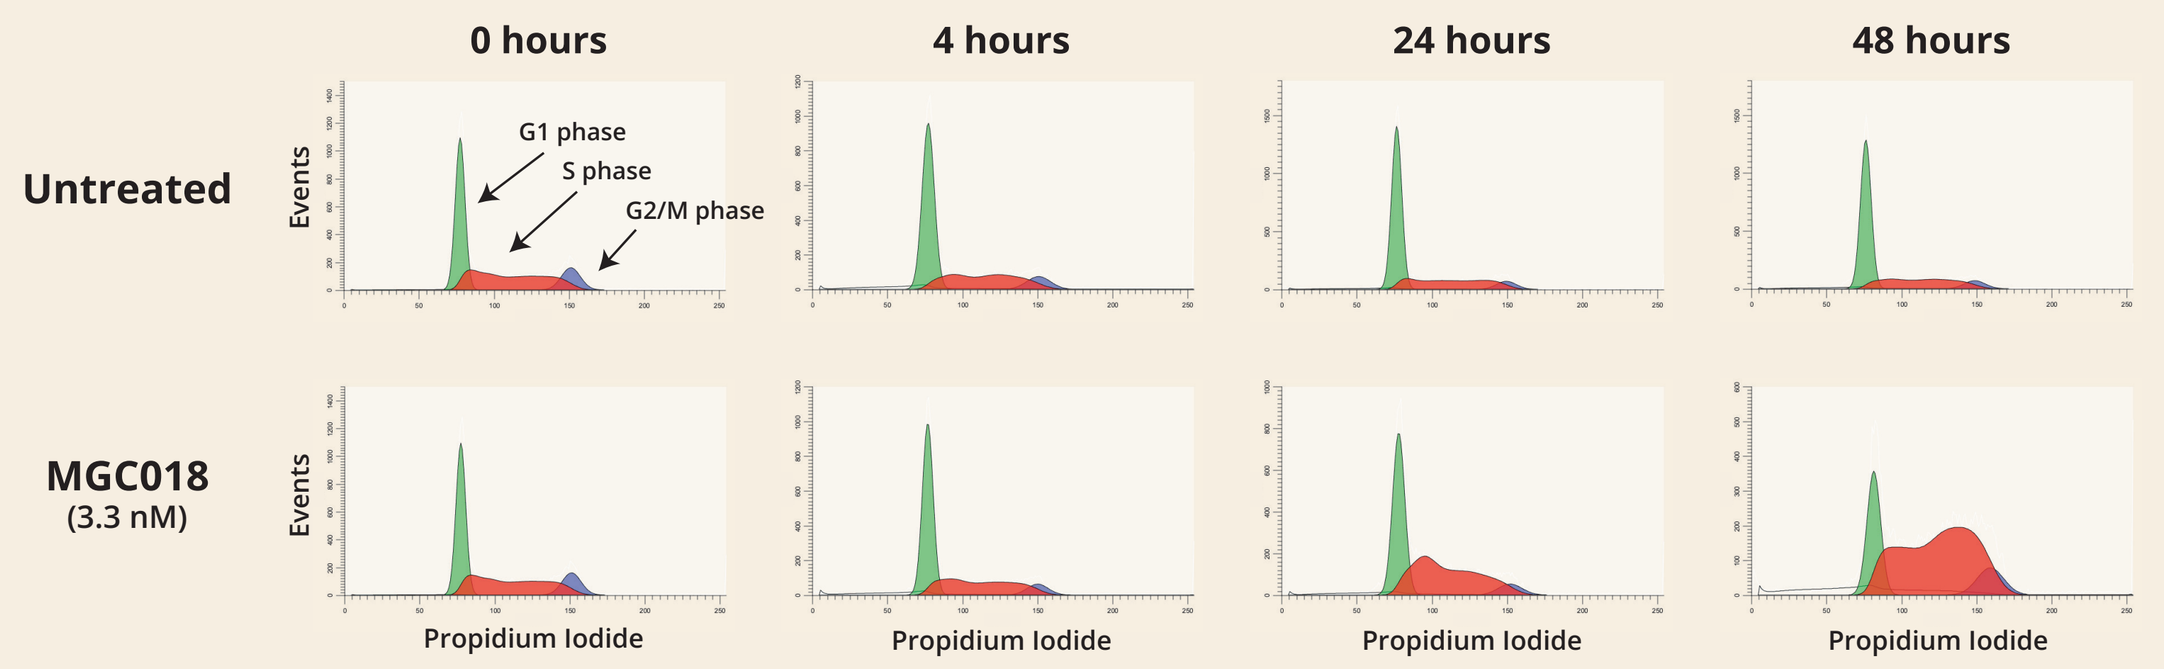

0 hours
4 hours
24 hours
48 hours
Untreated
MGC018 (3.3 nM)
G1 phase
S phase
G2/M phase
Events
Propidium Iodide

- Cell cycle arrest was observed in S and G2/M phase, consistent with the mechanism of action of duocarmycin

MGC018 Mediates Antitumor Activity Toward CDX Models of SCCHN

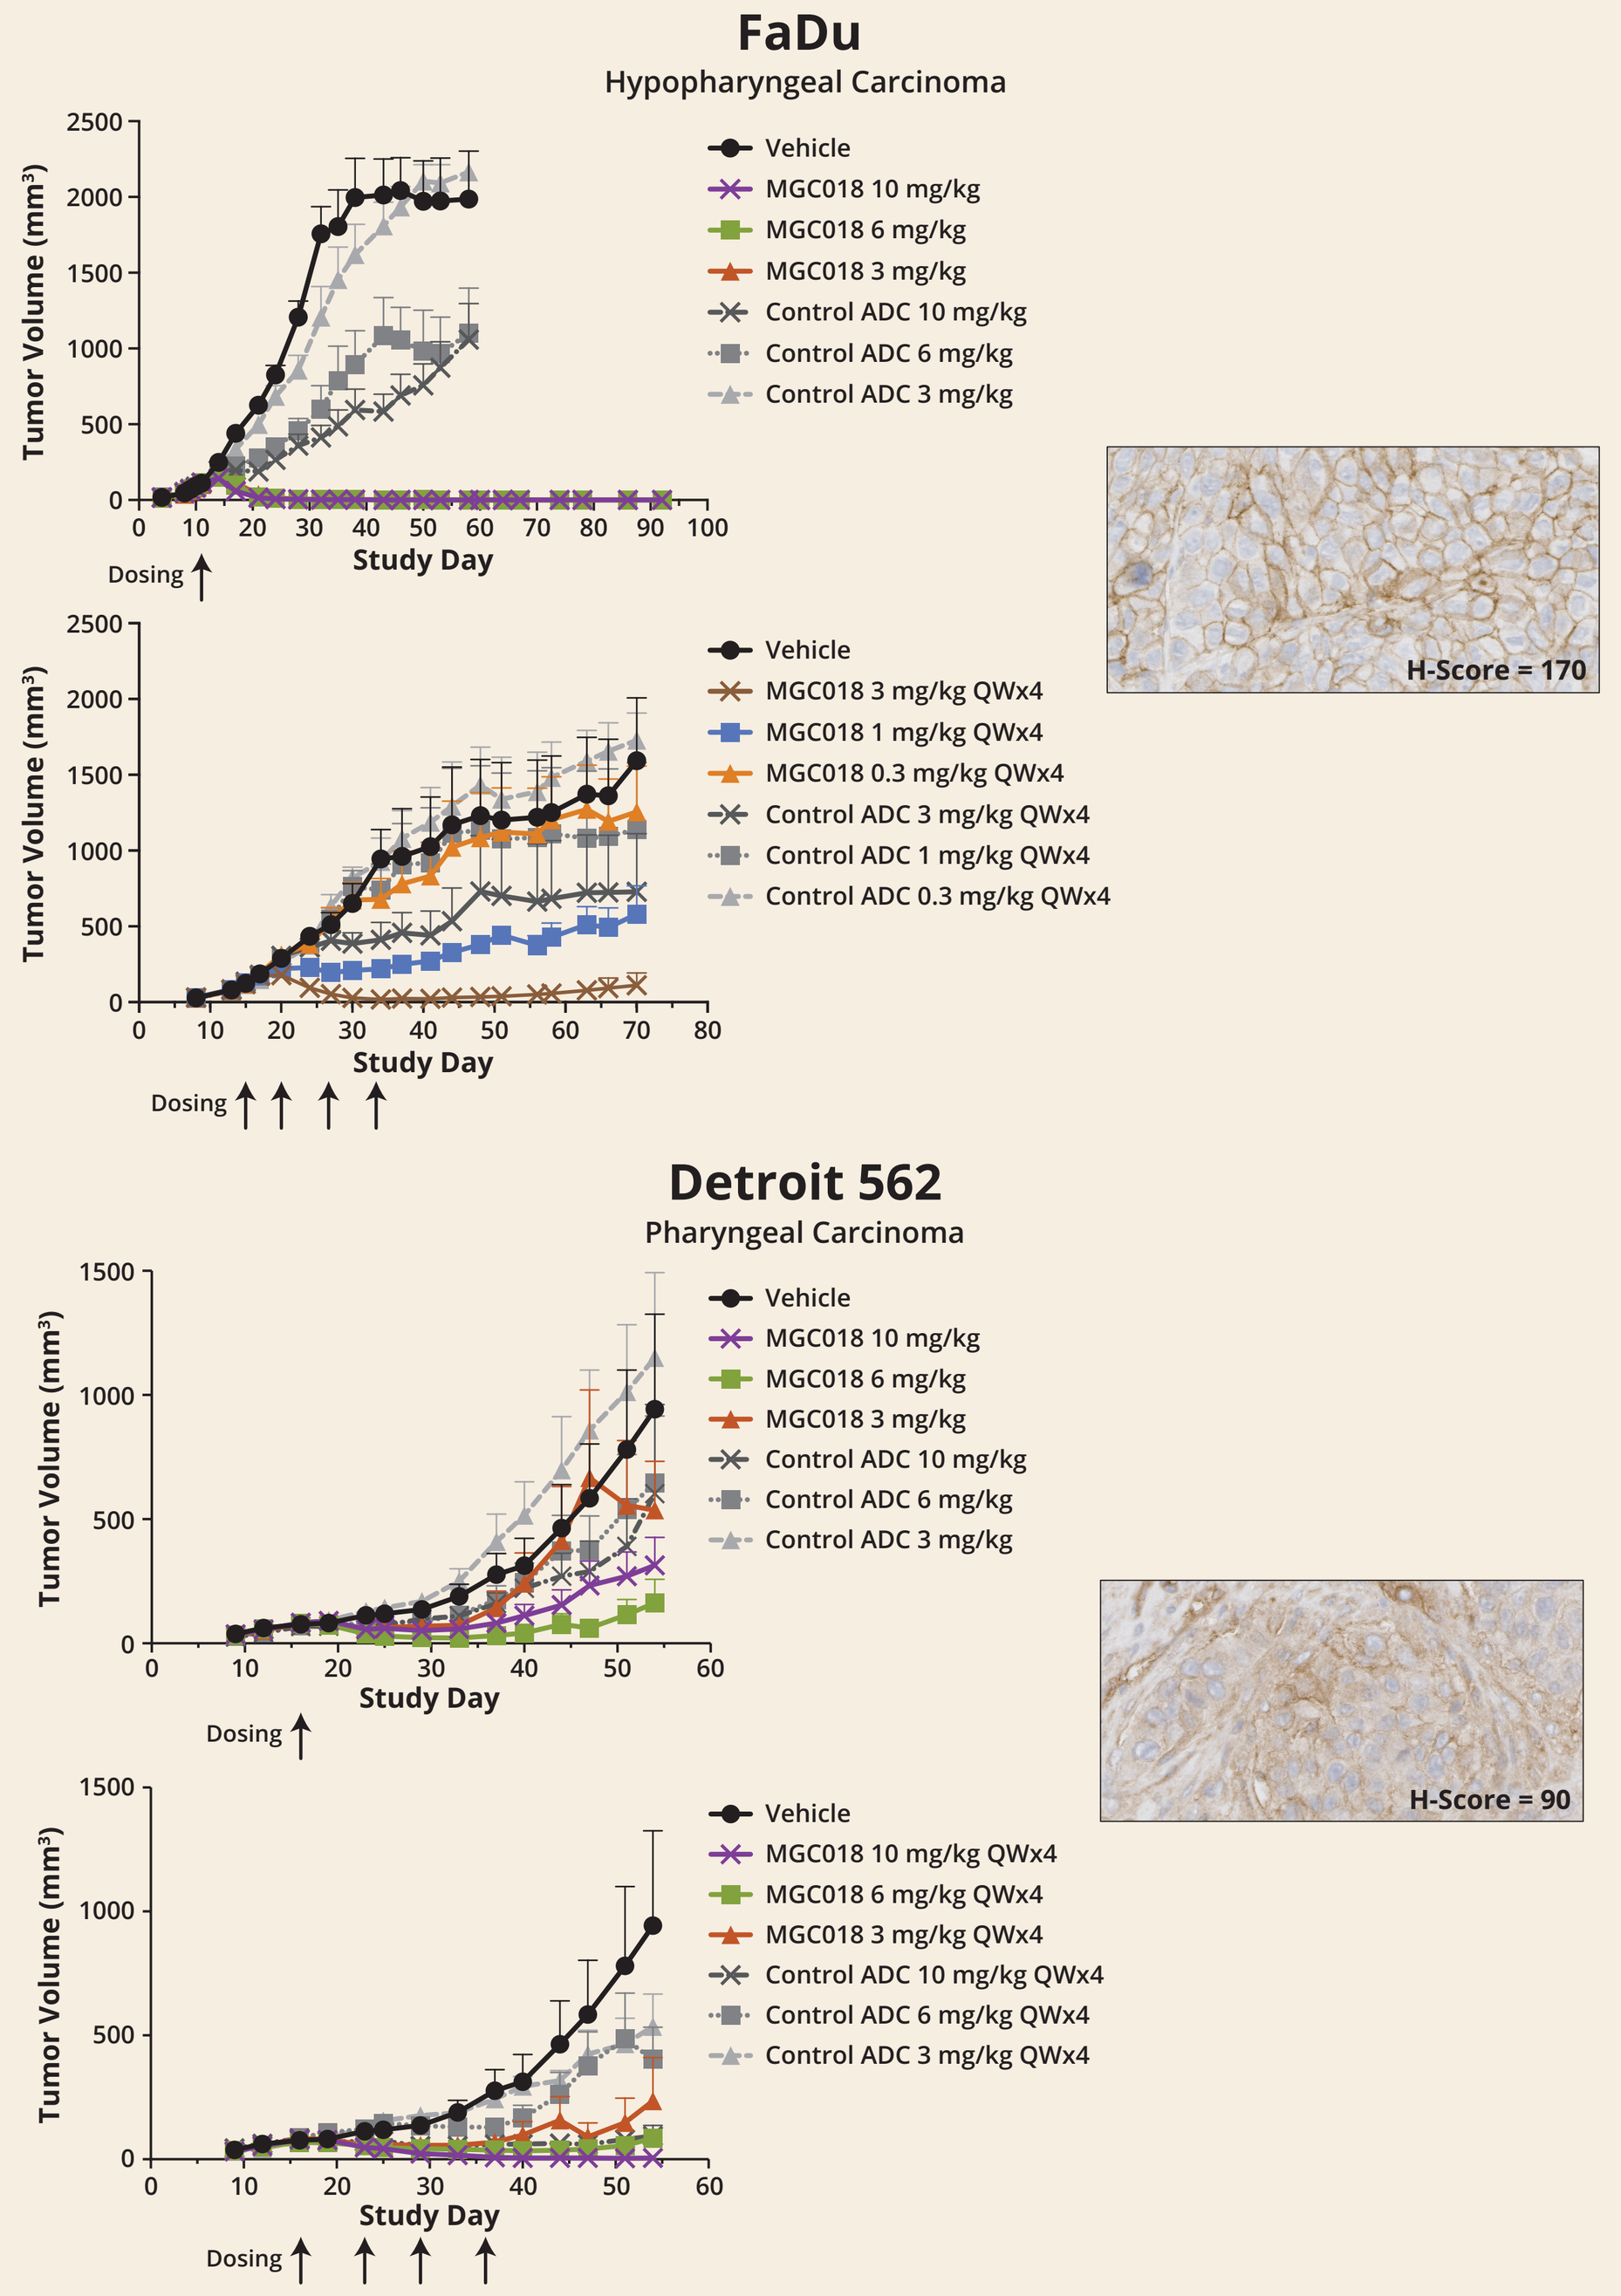

FaDu
Hypopharyngeal Carcinoma
Detroit 562
Pharyngeal Carcinoma
Vehicle
MGC018 10 mg/kg
MGC018 6 mg/kg
MGC018 3 mg/kg
Control ADC 10 mg/kg
Control ADC 6 mg/kg
Control ADC 3 mg/kg
MGC018 3 mg/kg QWx4
MGC018 1 mg/kg QWx4
MGC018 0.3 mg/kg QWx4
Control ADC 3 mg/kg QWx4
Control ADC 1 mg/kg QWx4
Control ADC 0.3 mg/kg QWx4
MGC018 10 mg/kg QWx4
MGC018 6 mg/kg QWx4
Control ADC 10 mg/kg QWx4
Control ADC 6 mg/kg QWx4
Tumor Volume (mm³)
Study Day
Dosing
H-Score = 170
H-Score = 90

- MGC018 exhibited antitumor activity toward FaDu and Detroit 562 xenograft models of SCCHN

MGC018 Mediates Antitumor Activity Toward PDX Models of Head and Neck Cancer

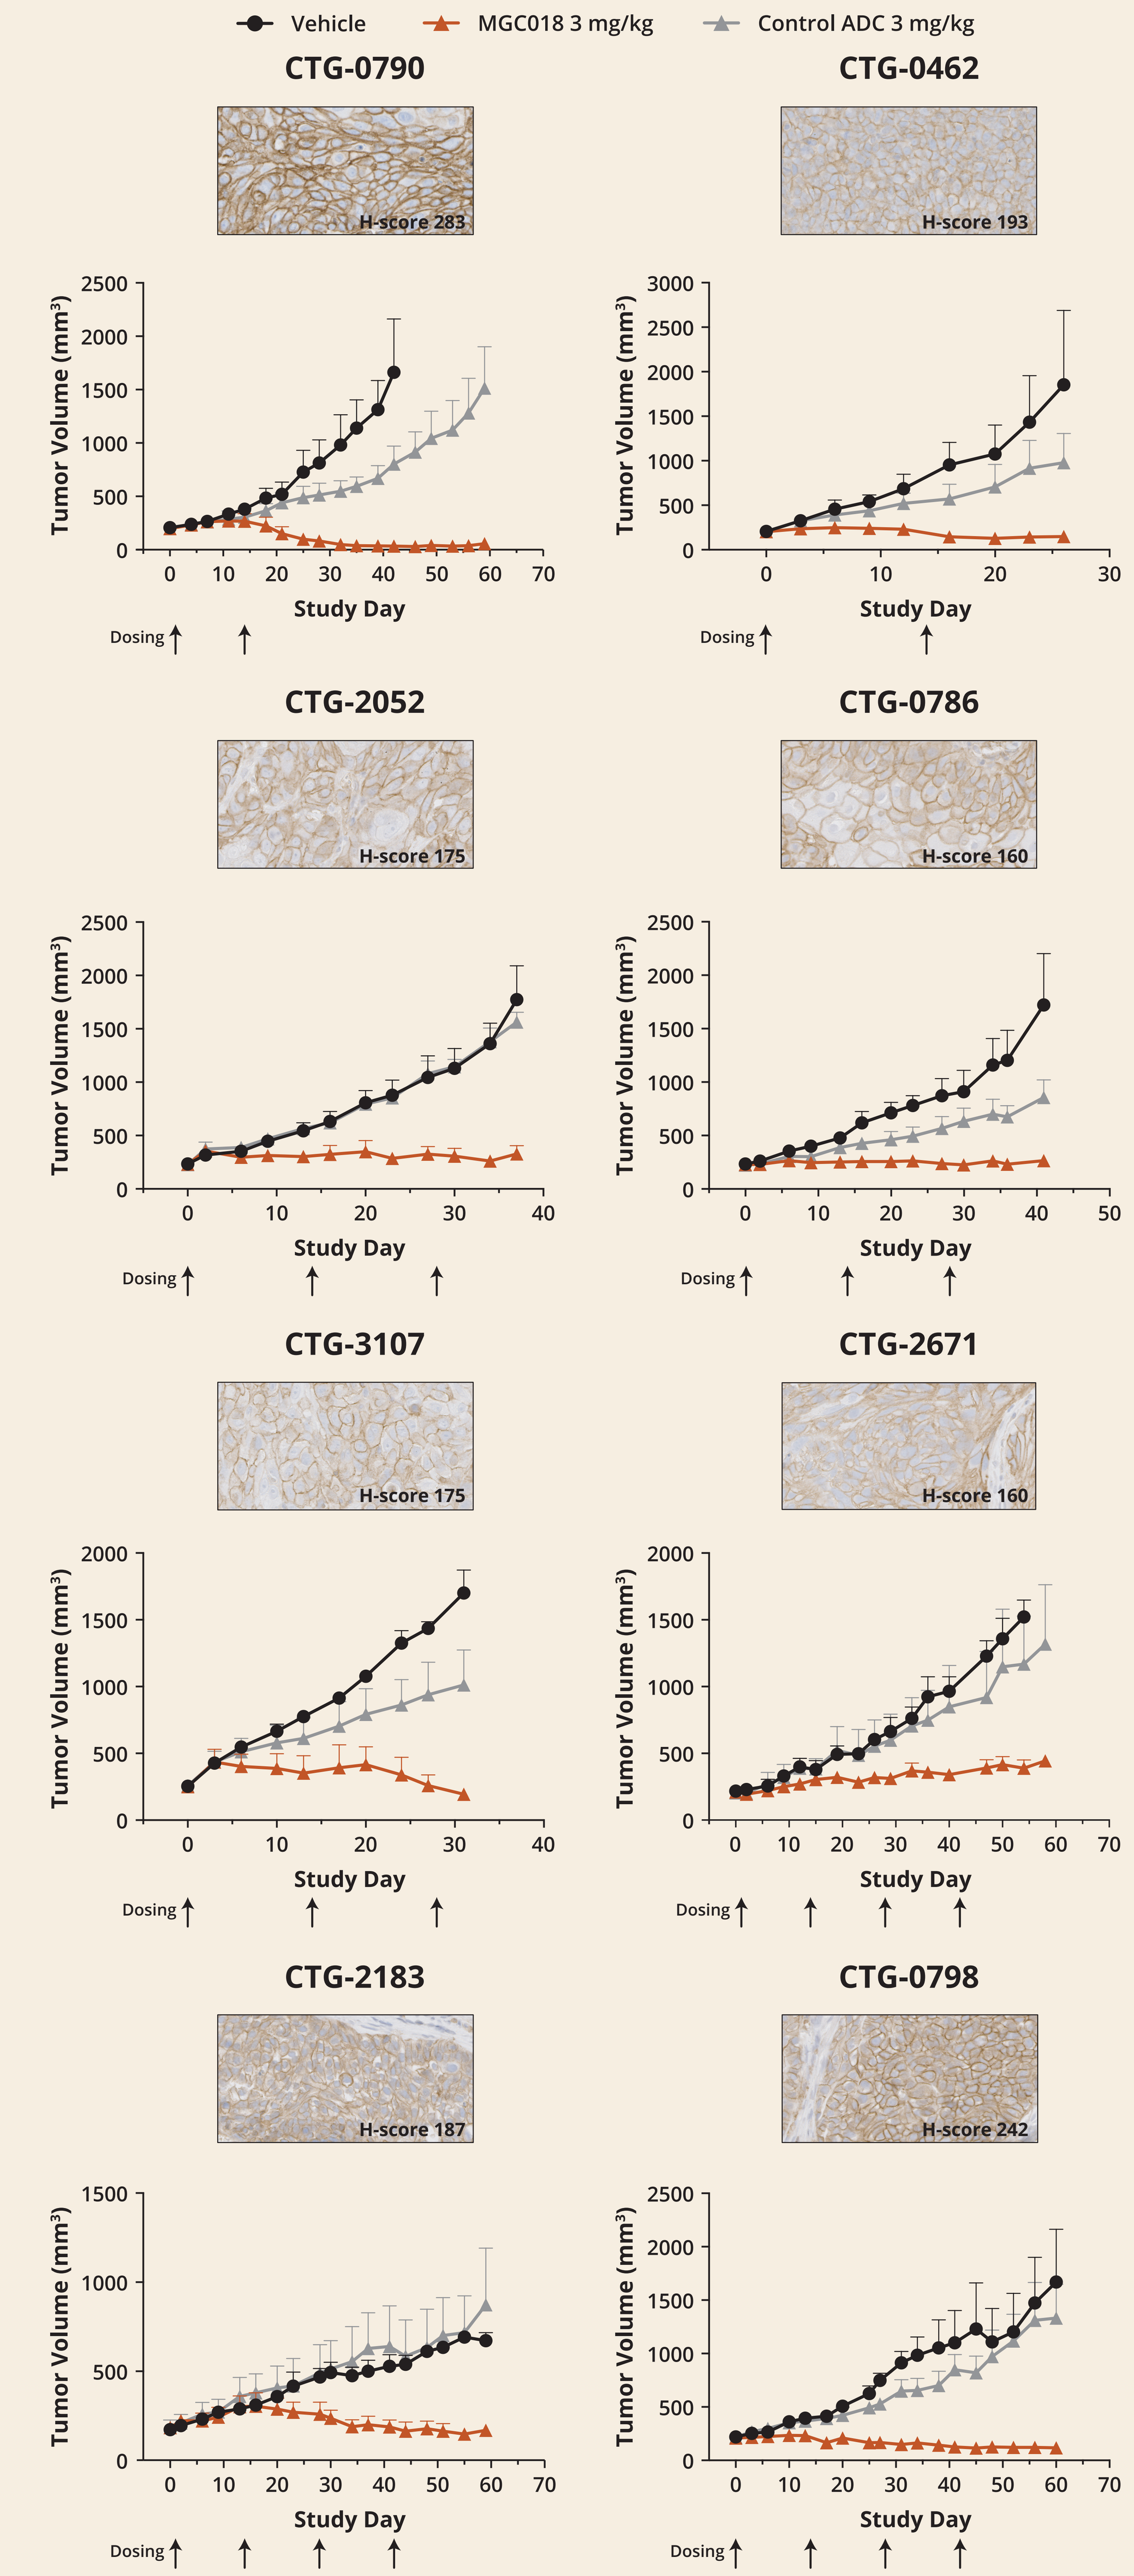

Vehicle
MGC018 3 mg/kg
Control ADC 3 mg/kg
CTG-0790
H-score 283
CTG-0462
H-score 193
CTG-2052
H-score 175
CTG-0786
H-score 160
CTG-3107
H-score 175
CTG-2671
H-score 160
CTG-2183
H-score 187
CTG-0798
H-score 242
Tumor Volume (mm³)
Study Day
Dosing

- MGC018 exhibited antitumor activity toward head and neck cancers expressing a range of B7-H3
- Additional factors beyond B7-H3 expression appear to influence sensitivity in these PDX models
- MGC018 was well tolerated in all models, with no impact on body weight

PDX Model Characteristics

Model	Tumor Status	Harvest Site	Histology	Disease Stage	Treatment History	HPV Status	H-Score	IV Dosing	% T/C	Antitumor Activity
CTG-0149	Metastatic	Skin	Squamous cell carcinoma	III	Naïve	16-/18-	213	Q2W x 3	99	Inactive
CTG-0152	Metastatic	Skin	Squamous cell carcinoma	III	Pretreated	16-/18-	222	Q2W x 3	58	Inactive
CTG-0434	Primary	Larynx	Squamous cell carcinoma	III	Not available	16+/18-	230	Q2W x 4	72	Inactive
CTG-0462	Metastatic	Liver	Carcinoma	IV	Pretreated	16+/18-	193	Q2W x 2	8	**Highly Active**
CTG-0786	Primary	Tongue	Squamous cell carcinoma	II	Naive	16-/18-	160	Q2W x 3	15	**Active**
CTG-0787	Primary	Buccal	Squamous cell carcinoma	II	Not available	16-/18-	120	Q2W x 2	53	Inactive
CTG-0790	Primary	Mandible	Squamous cell carcinoma	III	Naïve	16-/18-	283	Q2W x 2	2	**Highly Active**
CTG-0798	Metastatic	Lung	Ductal adenocarcinoma	IV	Pretreated	16-/18-	242	Q2W x 4	7	**Highly Active**
CTG-1100	Primary	Tongue	Carcinoma	II	Not available	16+/18-	243	Q2W x 4	1	**Highly Active**
CTG-1118	Local metastatic	Neck	Squamous cell carcinoma	III	Pretreated	16-/18-	263	Q2W x 4	38	**Active**
CTG-1131	Metastatic	Gluteus	Squamous cell carcinoma	IV	Pretreated	16-/18-	123	Q2W x 2	46	Inactive
CTG-2052	Primary	Mouth	Squamous cell carcinoma	IV	Naive	16-/18-	175	Q2W x 3	19	**Active**
CTG-2175	Primary	Larynx	Squamous cell carcinoma	III	Pretreated	N/A	153	Q2W x 4	26	**Active**
CTG-2183	Primary	Larynx	Squamous cell carcinoma	IV	Naive	16-/18-	187	Q2W x 4	25	**Active**
CTG-2259	Primary	Tongue	Squamous cell carcinoma	IV	Naive	16-/18-	150	Q2W x 3	29	**Active**
CTG-2324	Local metastatic	Neck	Squamous cell carcinoma	IV	Pretreated	16-/18-	173	Q2W x 4	109	Inactive
CTG-2336	Primary	Larynx	Squamous cell carcinoma	IV	Naive	N/A	190	Q2W x 3	18	**Active**
CTG-2671	Primary	Larynx	Squamous cell carcinoma	III	Naive	16-/18-	160	Q2W x 4	26	**Active**
CTG-3107	Metastatic	Lung	Squamous cell carcinoma	IV	Pretreated	N/A	175	Q2W x 3	11	**Active**

PDX studies performed by Champions Oncology. Antitumor activity evaluated according to National Cancer Institute (NCI) standards: Mean percent Tumor divided by Control; T/C < 10% highly active, T/C ≤ 42% is active, T/C > 42% is inactive.

An adaptive repeat-dose protocol was employed for the PDX studies due to rapid clearance of MGC018 in these rodent models.

Conclusions

- B7-H3 is frequently overexpressed in SCCHN
 - 90% (36/40) of human tumor samples evaluated showed B7-H3 membrane staining on the tumor epithelium with an average H-score of 162
 - Detailed analysis revealed that B7-H3 was heterogeneously expressed on both the tumor epithelium and tumor-associated vasculature
- MGC018 demonstrated antitumor activity in vivo toward SCCHN xenografts
 - Highly active toward FaDu and Detroit 562 CDX models
 - Active or highly active toward 13/19 (68%) of PDX models

These results support SCCHN as a suitable indication for an ADC-based treatment directed toward B7-H3

References

1. Elgersma et al., *Mol. Pharmaceutics* 12; 2015.
2. Scribner et al., *Mol. Cancer Ther.* 19(11); 2020.
3. American Cancer Society's publication, Cancer Facts & Figures 2020, and the National Cancer Institute.

Acknowledgements

DUBA linker payload conjugated by and licensed from Byondis B.V., Nijmegen, the Netherlands.

Presented at the American Association for Cancer Research 2021 Virtual Annual Meeting, April 10–15, 2021

©2021 MacroGenics, Inc. All rights reserved.