Phase 1 Dose Escalation Study of MGC018, an anti-B7-H3 Antibody-Drug Conjugate (ADC), In Patients with Advanced Solid Tumors

Abstract #2631

NCT03729596

Sekwon Jang[1], John Powderly[2], Alexander Spira[3], Iwona Lugowska[4], Girish Mallesera[5], Andrew Weickhardt[6], Piotr Wysocki[7], Jakub Zolniereck[8], Ouiam Bakkacha[9], Deryk Loo[9], Chet Bohac[9], Jeanny Aragon-Ching[1], Eugene Shenderov[10], Emmanuel Antonarakis[10], Manish Sharma[11]

[1]Inova Schar Cancer Institute, Fairfax, VA; [2]Carolina BioOncology, Huntersville, NC; [3]Virginia Cancer Specialists, Fairfax, VA; [4]Narodowy Instytut Onkologii im. Marii Skłodowskiej-Curie Państwowy Instytut Badawczy, Warsaw, Poland; [5]Calvary Mater Newcastle, Waratah, Australia; [6]Austin Health–Olivia Newton John Cancer Center, Heidelberg, Australia; [7]Samodzielny Publiczny Zakład Opieki Zdrowotnej Szpital Uniwersytecki w Krakowie, Krakow, Poland; [8]Magodent Sp. z o.o. Szpital Elbląska Oddział Onkologii Klinicznej/ Chemioterapii, Warsaw, Poland; [9]MacroGenics, Inc., Rockville, MD; [10]The Johns Hopkins Kimmel Cancer Center, Baltimore, MD; [11]START Midwest, Grand Rapids, MI

bohacg@macrogenics.com

Background

MGC018: B7-H3 Directed ADC with Duocarmycin-based Linker Payload

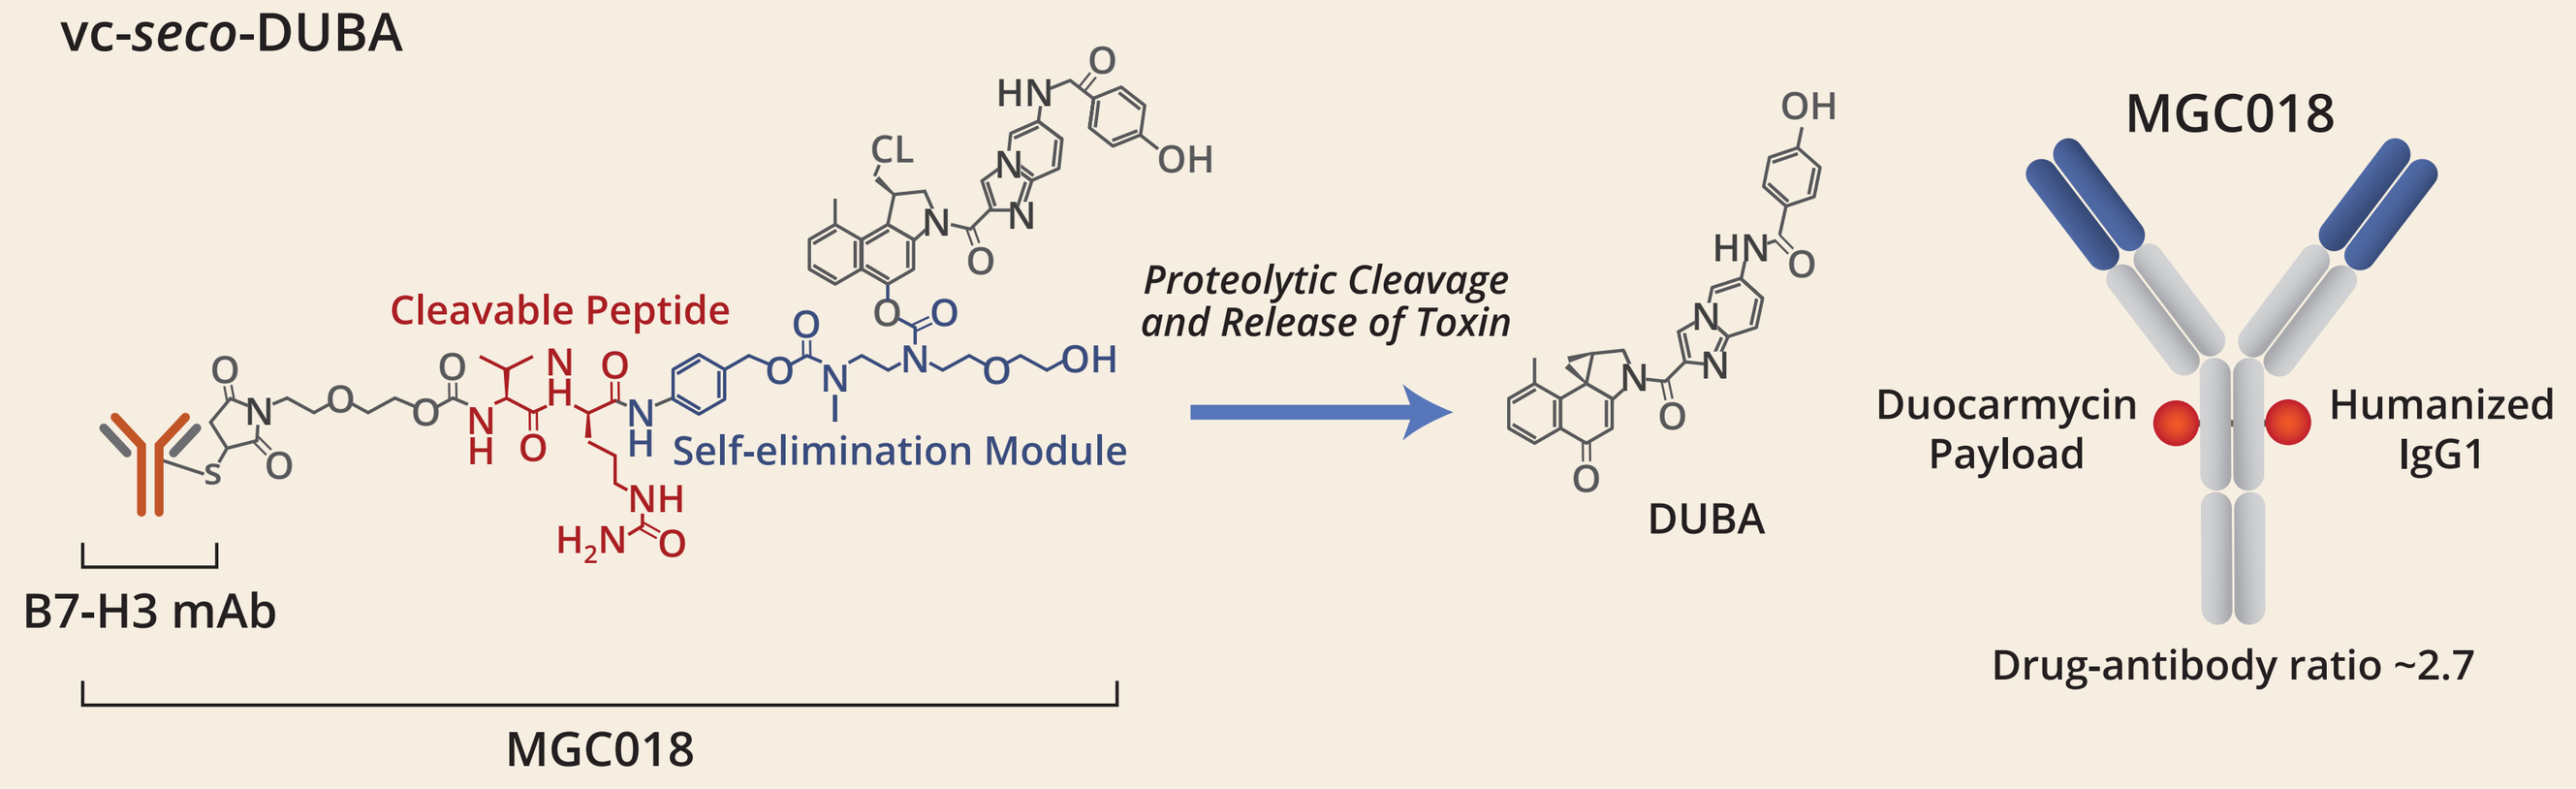

vc-*seco*-DUBA
Cleavable Peptide
Self-elimination Module
Proteolytic Cleavage and Release of Toxin
DUBA
B7-H3 mAb
MGC018
MGC018
Duocarmycin Payload
Humanized IgG1
Drug-antibody ratio ~2.7

- MGC018 is an anti-B7-H3 antibody-drug conjugate (ADC) with a duocarmycin payload
- vc-seco-DUocarmycin-hydroxyBenzamide Azaindole (DUBA) is a DNA alkylating agent
- DUBA cytotoxic activity is cell-cycle independent
- DUBA retains potency in multidrug-resistant cell lines
- Cleavable peptide linker – facilitates bystander effect
- Induces immunogenic cell death in preclinical models

DUBA Linker Payload provided and conjugated by *Byondis*.

Rationale for Targeting B7-H3 with MGC018

- B7-H3 is highly expressed in multiple solid tumors, with limited expression in normal tissue
- B7-H3 may play immune suppressive and tumor-autonomous roles that favor cancer growth
- B7-H3 is expressed by vascular endothelium and stroma in tumor microenvironment
- MGC018 is a novel ADC that delivers a potent duocarmycin-based DNA alkylating payload via native cysteines
- Duocarmycin DNA-targeted activity is directed to both dividing and non-dividing cells

Study Design and Objectives

- 3+3+3 Dose escalation design
- Six planned dose escalation cohorts: 0.5 to 5.0 mg/kg IV every three weeks
- Tumor response by investigator per RECIST v1.1 and PSA every 6 weeks (Dose Escalation)
- Tumor response by investigator every 9 weeks and PSA every 3 weeks (Cohort Expansion)
- Cohort Expansion at recommended Phase 2 dose (RP2D) to assess safety and tumor response (metastatic castration resistant prostate cancer (mCRPC), non-small cell lung cancer (NSCLC), squamous cell carcinoma of the head and neck (SCCHN), triple-negative breast cancer (TNBC), melanoma)

Primary Objective

- Safety and maximum tolerated dose (or maximum administered dose)

Secondary Objectives

- Pharmacokinetics and immunogenicity
- Antitumor activity

Key Eligibility Criteria

Inclusion

- Patients with histologically proven, relapsed or refractory, unresectable locally advanced or metastatic solid tumors of any histology for whom no therapy with demonstrated clinical benefit is available (Dose Escalation)
- ≥ 18 years old; ECOG PS ≤ 2; life expectancy ≥ 12 weeks in Dose Escalation (24 weeks in Cohort Expansion); measurable disease as per RECIST v1.1
- Tumor tissue available to evaluate B7-H3 immunohistochemistry (B7-H3 expression not required for eligibility)

Exclusion

- Abnormal laboratory parameters (hematologic, renal, and/or liver function); clinically significant cardiovascular or pulmonary disease; patients with history of CNS metastasis must have completed treatment, be asymptomatic, and not have had CNS progression within 6 months; no history of leptomeningeal disease or spinal cord compression
- Chemotherapy, biologic, investigational agents or mediastinal/pelvic radiation within 4 weeks; small molecule targeted or kinase inhibitors within 14 days; prior therapy with B7-H3 targeted agent (prior radioligand within 6 months in mCRPC Cohort Expansion)

Cohort Expansion Disease-Specific Criteria

- mCRPC that has progressed during or following 1 prior line of chemotherapy for metastatic disease, and if approved and available, no more than 2 prior lines of an anti-hormonal agent (e.g., abiraterone, enzalutamide) with a PSA value of at least 2 ng/mL and meeting at least one of the following:
 - Progression in measurable disease (RECIST v1.1)
 - Appearance of 2 or more new bone lesions according to Prostate Cancer Working Group 2 (PCWG2)
 - Rising PSA defined as at least 2 sequential rises in PSA (≥ 1 week apart) over a reference value (the last PSA [PSA ≥ 2 ng/mL] measured before the first rise in PSA) (as defined by the PCWG2)

Dose Escalation Results

Enrollment Status

Dose Escalation Complete (N=29)

- Maximum administered dose 4 mg/kg every 3 weeks
- RP2D defined as 3 mg/kg every 3 weeks

Cohort Expansion in mCRPC: Enrollment Ongoing

- *28 enrolled (of 40)*
- *Disease classification available for 20 of 28 patients: Bone only (7), mixed soft tissue + bone (9), soft tissue only (4)*

B7-H3 IHC Data

- 21 of 24 Patients with available tissue had samples evaluable for B7-H3 expression
- H-score: Median 190 (range 17–279); mean 175.7
- Vasculature score: Median 2+ (range 0–3+)

Data cutoff 03 May 2021.

Related Adverse Events ≥ 20%, All Grades

Fatigue, nausea, infusion related reaction, skin disorders, and neutropenia were most common

System Organ Class Preferred Term	0.5 mg/kg (N=3)	1.0 mg/kg (N=6)	2.0 mg/kg (N=7)	3.0 mg/kg (N=7)	4.0 mg/kg (N=6)	All (N=29)
AT LEAST ONE EVENT	3 (100%)	5 (83.3%)	6 (85.7%)	7 (100%)	6 (100%)	27 (93.1%)
Blood and lymphatic system disorders	0	1 (16.7)	2 (28.6)	3 (42.9)	2 (33.3)	8 (27.6)
Neutropenia	0	1 (16.7)	2 (28.6)	3 (42.9)	2 (33.3)	8 (27.6)
Gastrointestinal disorders	0	5 (83.3)	2 (28.6)	2 (28.6)	3 (50.0)	12 (41.4)
Nausea	0	2 (33.3)	2 (28.6)	1 (14.3)	3 (50.0)	8 (27.6)
General disorders and administration site conditions	2 (66.7)	2 (33.3)	2 (28.6)	4 (57.1)	5 (83.3)	15 (51.7)
Fatigue	1 (33.3)	1 (16.7)	2 (28.6)	4 (57.1)	3 (50.0)	11 (37.9)
Chills	1 (33.3)	0	2 (28.6)	0	4 (66.7)	7 (24.1)
Pyrexia	1 (33.3)	1 (16.7)	2 (28.6)	0	2 (33.3)	6 (20.7)
Injury, poisoning and procedural complications	0	0	2 (28.6)	5 (71.4)	2 (33.3)	9 (31.0)
Infusion-related reaction	0	0	2 (28.6)	5 (71.4)	2 (33.3)	9 (31.0)
Skin and subcutaneous tissue disorders	0	3 (50.0)	5 (71.4)	5 (71.4)	4 (66.7)	17 (58.6)
Skin hyperpigmentation	0	3 (50.0)	1 (14.3)	3 (42.9)	2 (33.3)	9 (31.0)
Palmar-plantar erythrodysaesthesia syndrome	0	0	3 (42.9)	3 (42.9)	2 (33.3)	8 (27.6)

Includes events with causality ratings of 'Possible', 'Probable' or 'Definite'. Subjects are counted once for each Preferred Term reported.
Data were extracted on 03May2021.

Grade ≥ 3 Related Adverse Events

Cytopenias were most common

System Organ Class Preferred Term	0.5 mg/kg (N=3)	1.0 mg/kg (N=6)	2.0 mg/kg (N=7)	3.0 mg/kg (N=7)	4.0 mg/kg (N=6)	All (N=29)
AT LEAST ONE EVENT	2 (66.7%)	2 (33.3%)	6 (85.7%)	4 (57.1%)	5 (83.3%)	19 (65.5%)
Blood and lymphatic system disorders	0	0	2 (28.6)	2 (28.6)	2 (33.3)	6 (20.7)
Neutropenia	0	0	2 (28.6)	2 (28.6)	2 (33.3)	6 (20.7)
Lymphopenia	0	0	1 (14.3)	1 (14.3)	1 (16.7)	3 (10.3)
General disorders and administration site conditions	0	0	0	0	2 (33.3)	2 (6.9)
Fatigue	0	0	0	0	2 (33.3)	2 (6.9)
Investigations	1 (33.3)	2 (33.3)	3 (57.1)	2 (28.6)	2 (33.3)	10 (34.5)
Lymphocyte count decreased	0	1 (16.7)	2 (28.6)	1 (14.3)	0	4 (13.8)
Neutrophil count decreased	0	1 (16.7)	1 (14.3)	0	0	2 (6.9)
Platelet count decreased	0	0	1 (14.3)	1 (14.3)	0	2 (6.9)
Lipase increased	1 (33.3)	0	0	0	1 (16.7)	2 (6.9)
White blood cell count decreased	0	1 (16.7)	0	0	1 (16.7)	2 (6.9)
Metabolism and nutrition disorders	0	0	2 (28.6)	0	0	2 (6.9)
Hypophosphataemia	0	0	2 (28.6)	0	0	2 (6.9)
Skin and subcutaneous tissue disorders	0	0	3 (42.9)	1 (14.3)	0	4 (13.8)
Palmar-plantar erythrodysaesthesia syndrome	0	0	1 (14.3)	1 (14.3)	0	2 (6.9)
Rash maculo-papular	0	0	2 (28.6)	0	0	2 (6.9)

Data were extracted on 03May2021.

Treatment-Emergent Adverse Events

Manageable Safety Profile

Patients Experiencing at Least One Adverse Event	0.5 mg/kg (N=3)	1.0 mg/kg (N=6)	2.0 mg/kg (N=7)	3.0 mg/kg* (N=7)	4.0 mg/kg (N=6)	All (N=29)
Adverse Event	3 (100%)	6 (100%)	7 (100%)	7 (100%)	6 (100%)	29 (100%)
Treatment-Related Adverse Event[1]	3 (100)	5 (83.3)	6 (85.7)	7 (100)	6 (100)	27 (93.1)
Adverse Event ≥ Grade 3[2]	3 (100)	4 (66.7)	7 (100)	5 (71.4)	5 (83.3)	24 (82.8)
Treatment-Related Adverse Event ≥ Grade 3[2]	2 (66.7)	2 (33.3)	6 (85.7)	4 (57.1)	5 (83.8)	19 (65.5)
Serious Adverse Event	1 (33.3)	1 (16.7)	3 (42.9)	2 (28.6)	2 (33.3)	9 (31.0)
Dose-limiting Toxicity	0	0	1 (14.3)[3]	0	1 (16.7)[4]	2 (6.9)
Event that Resulted in Study Discontinuation	1 (33.3)	2 (33.3)	3 (42.9)	4 (57.1)	2 (33.3)	10 (34.5)
Event that Resulted in MGC018 Withdrawal	1 (33.3)	1 (16.7)	3 (42.9)	4 (57.1)	2 (33.3)	11 (37.9)
Event that Resulted in MGC018 Dose Reduction	0	0	1 (14.3)	2 (28.6)	2 (33.3)	5 (17.2)
Event that Resulted in MGC018 Interruption	1 (33.3)	0	1 (14.3)	5 (71.4)	5 (83.3)	12 (41.4)
Fatal Adverse Event (pneumonitis/pneumonia)	1 (33.3)	0	0	0	0	1 (3.4)
Adverse Event of Special Interest (AESI) – Infusion Reaction	0	0	2 (28.6)	5 (71.4)	2 (33.3)	9 (31.0)

[1]Includes events with causality assessments of 'Possible', 'Probable' or 'Definite'. [2]Based on CTCAE criteria version 4.0.3. [3]Grade 4 neutropenia resolved to baseline. [4]G3 fatigue > 72 hours. *Amendment during 3.0 mg/kg dose level applied to allow dose modification.
Data were extracted on 03May2021.

Best Percent Change of Target Lesions in Evaluable Population[1]

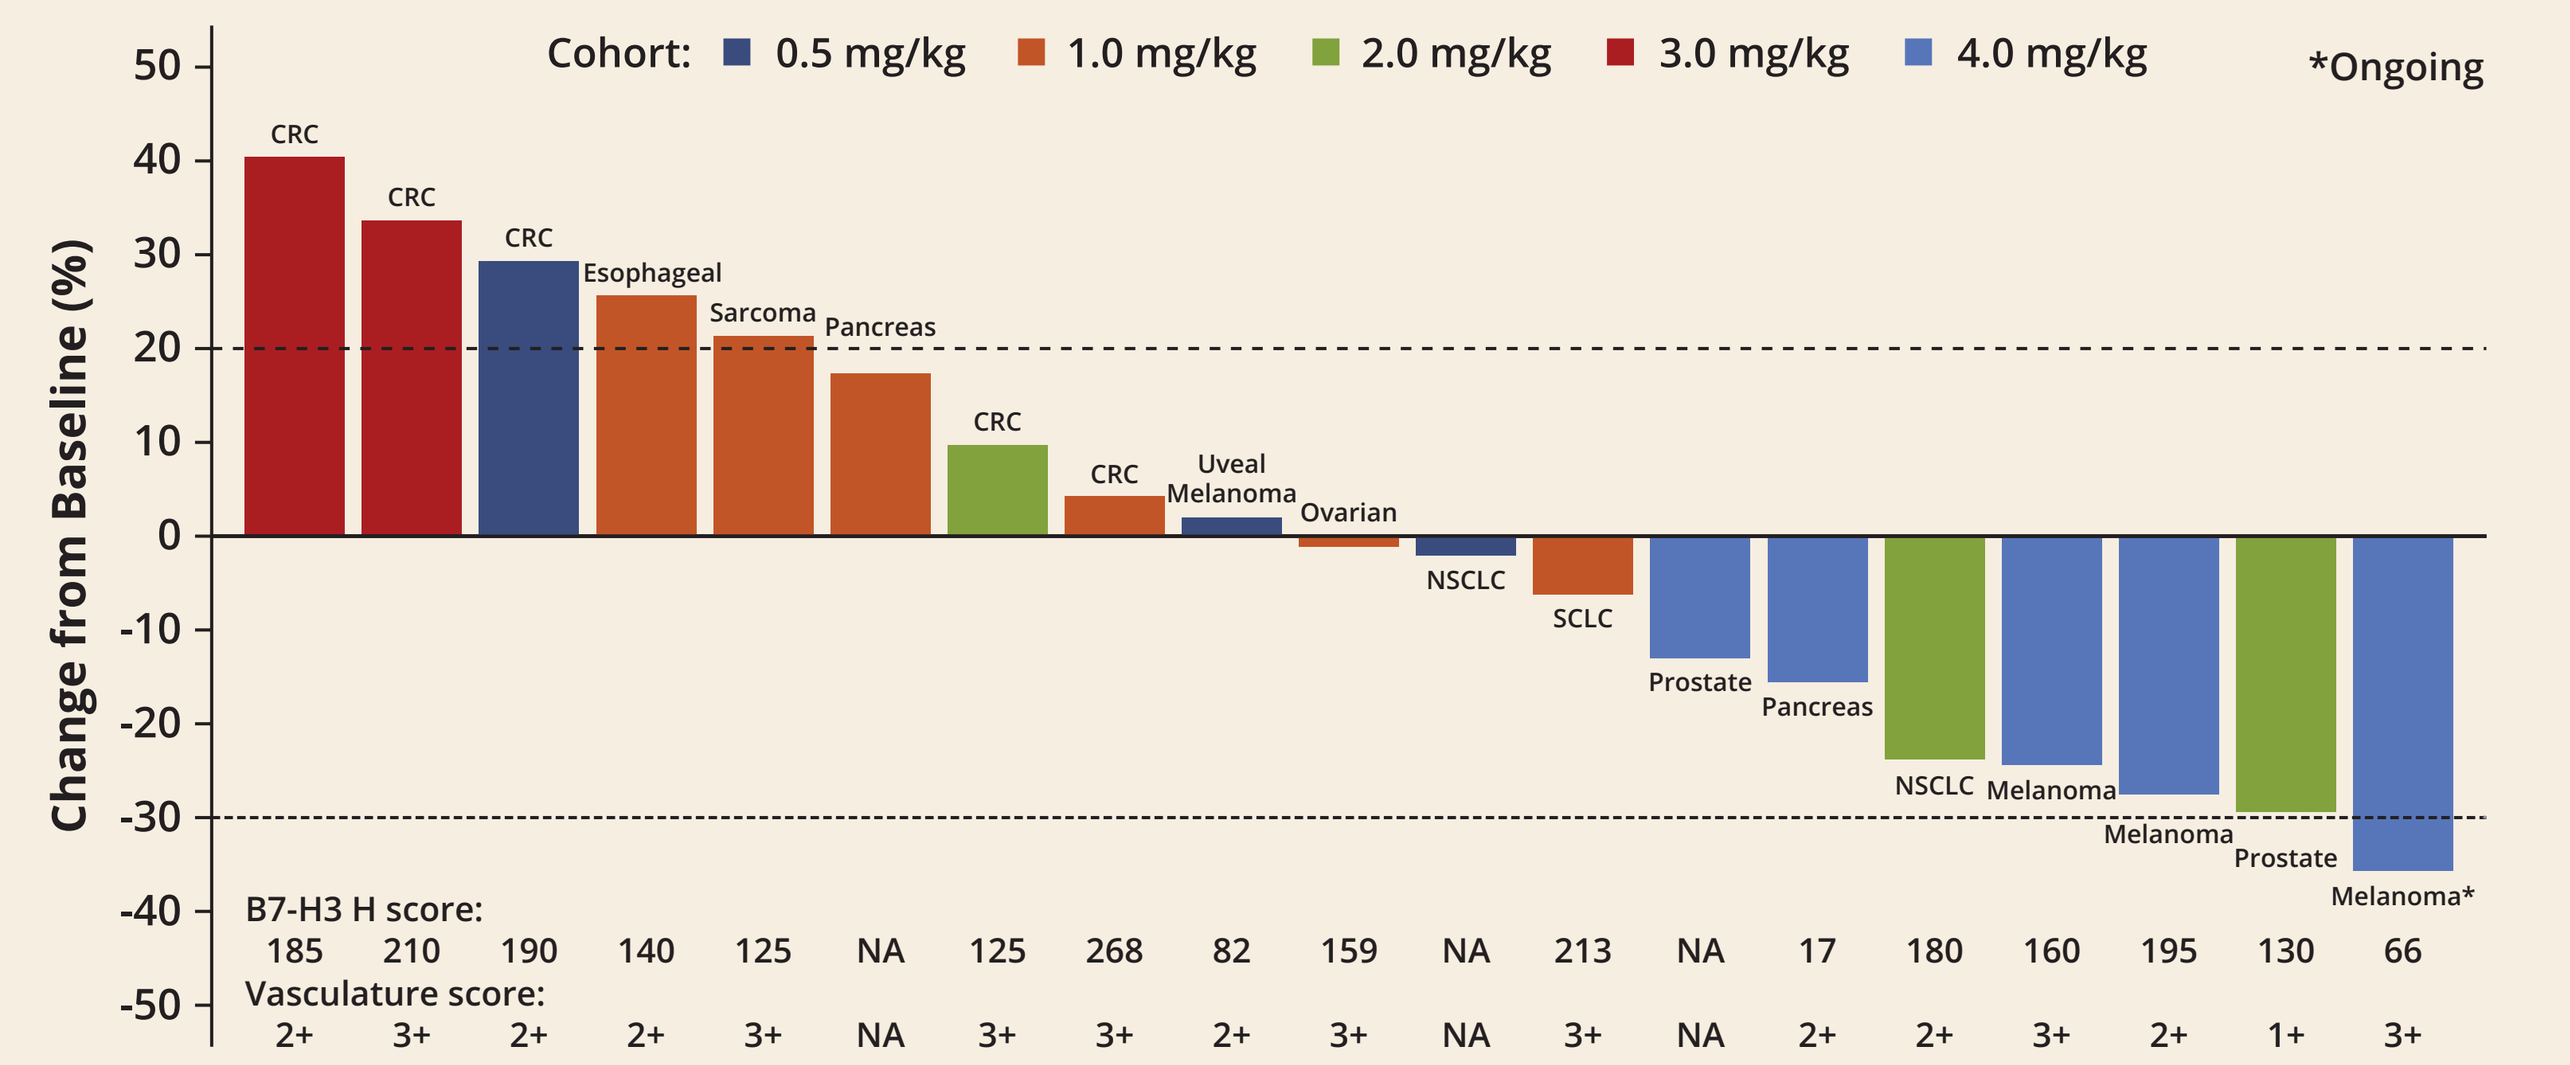

Cohort: 0.5 mg/kg 1.0 mg/kg 2.0 mg/kg 3.0 mg/kg 4.0 mg/kg
*Ongoing
Change from Baseline (%)
CRC CRC CRC Esophageal Sarcoma Pancreas CRC CRC Uveal Melanoma Ovarian NSCLC SCLC Prostate Pancreas NSCLC Melanoma Melanoma Prostate Melanoma*
B7-H3 H score:
185 210 190 140 125 NA 125 268 82 159 NA 213 NA 17 180 160 195 130 66
Vasculature score:
2+ 3+ 2+ 2+ 3+ NA 3+ 3+ 2+ 3+ NA 3+ NA 2+ 2+ 3+ 2+ 1+ 3+

[1]Patients who received at least one dose and had at least one post-baseline tumor evaluation. CRC = Colorectal cancer. NA = Not available.
Data were extracted on 03MAY2021.

Percent Change of Target Lesions

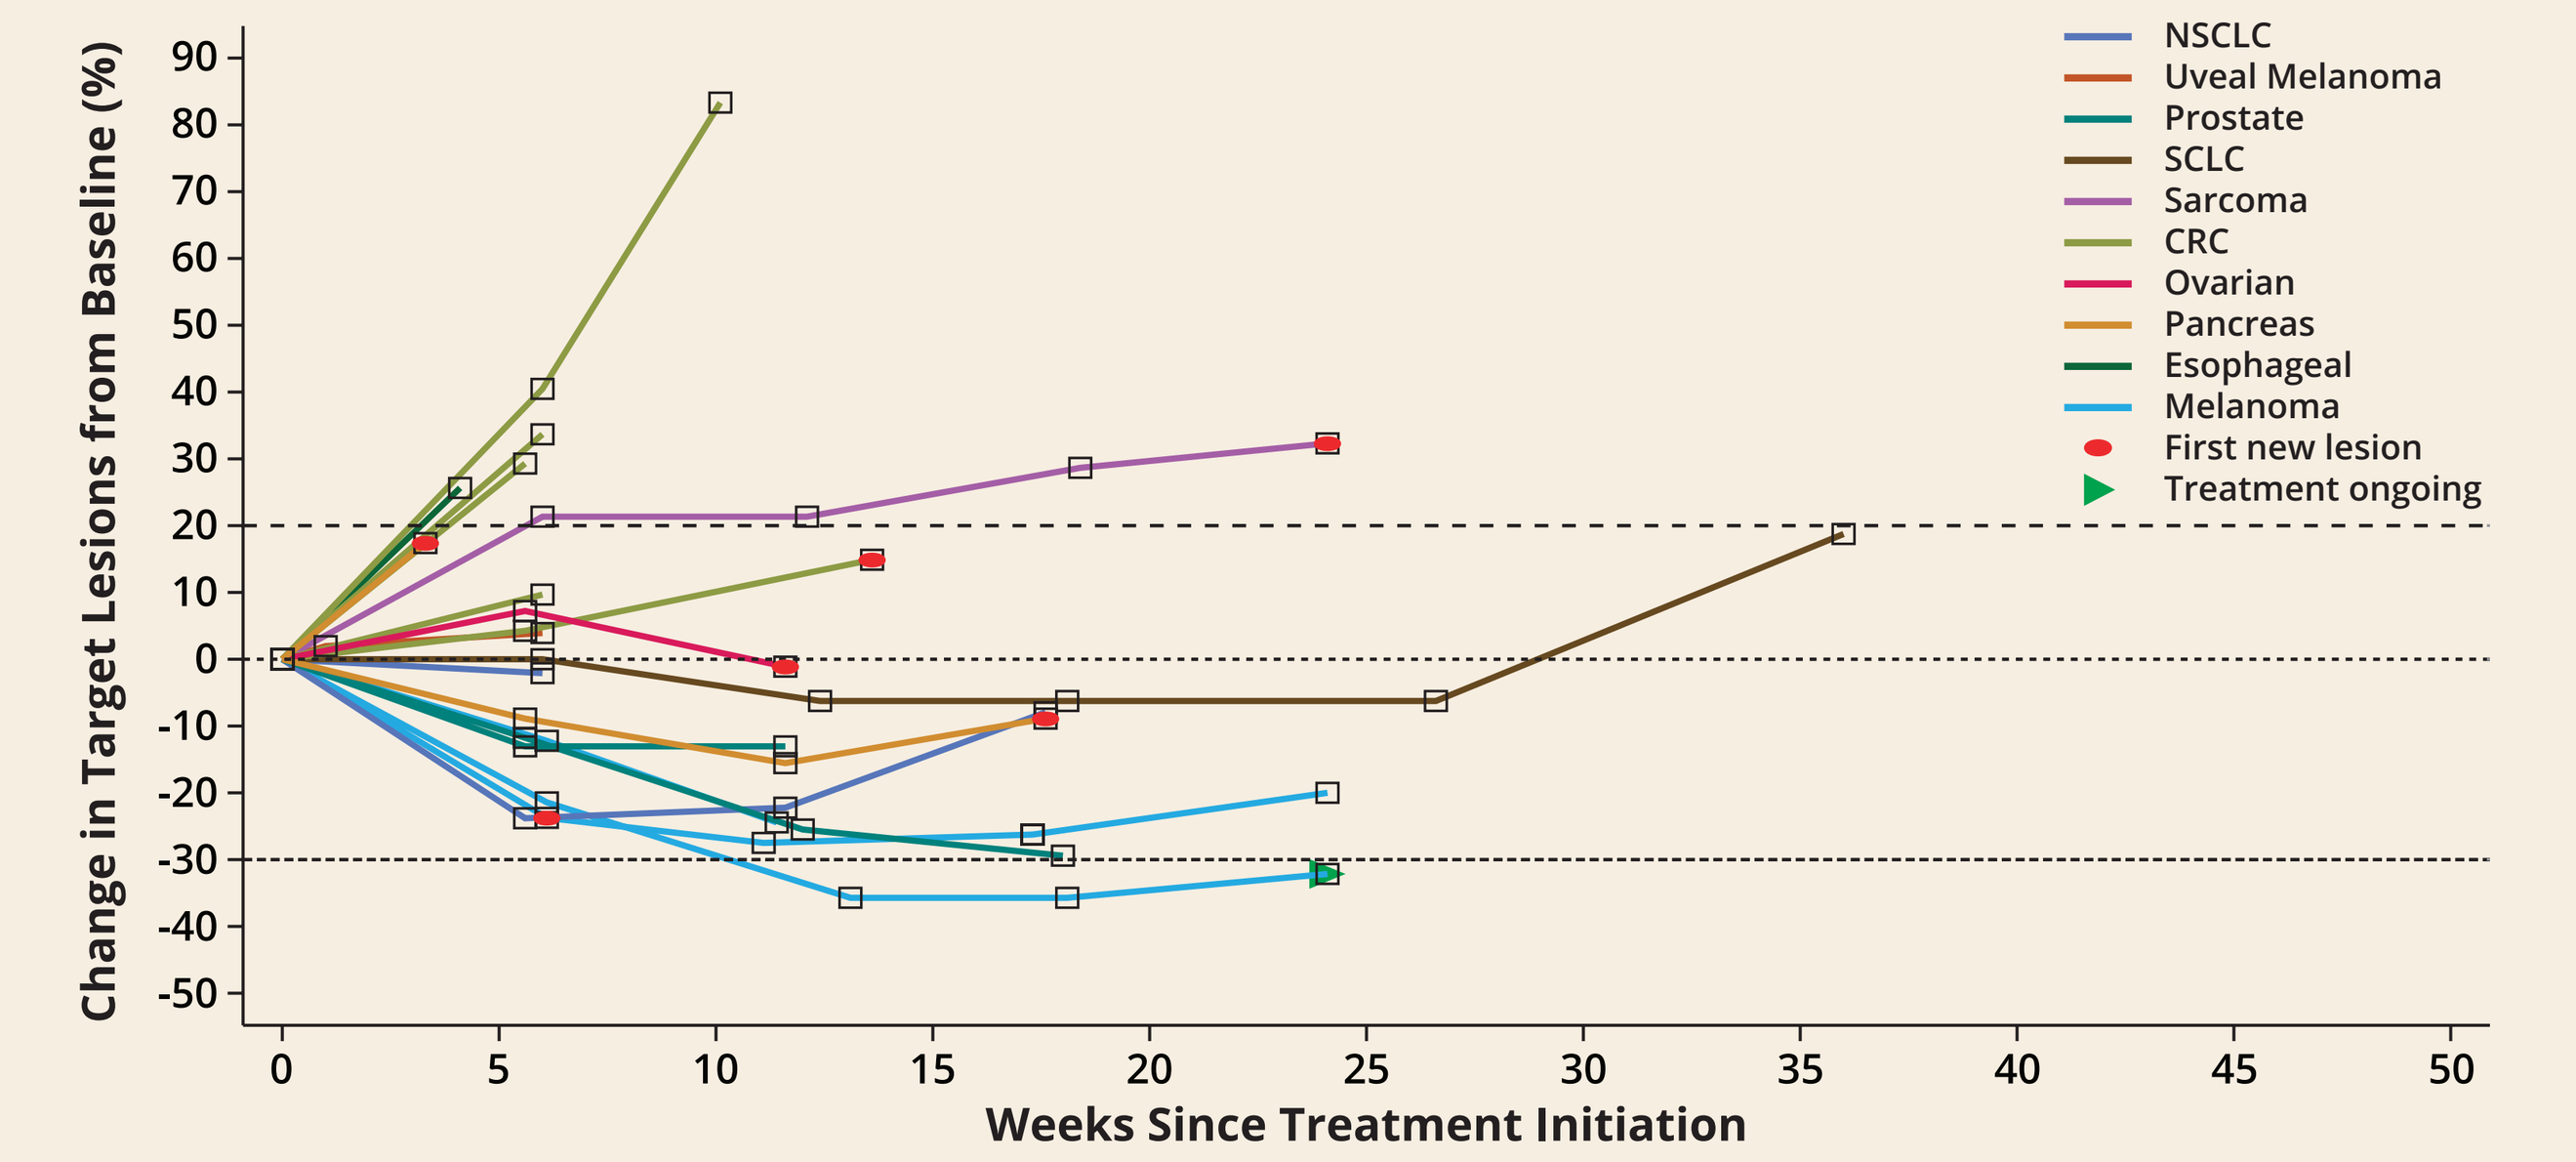

Change in Target Lesions from Baseline (%)
Weeks Since Treatment Initiation
NSCLC
Uveal Melanoma
Prostate
SCLC
Sarcoma
CRC
Ovarian
Pancreas
Esophageal
Melanoma
First new lesion
Treatment ongoing

Data were extracted on 03May2021.

Activity in Melanoma Patients

Patient	Prior Radiation/ Surgery	B7-H3 H Score (Vasculature Score)	Line of Therapy	Treatment	Duration of Therapy	Reason for MGC018 Discontinuation	Best Response in Target Lesions
Patient #1 3 target lesions (lung, liver, chest wall) Non-target pelvic nodes and perirectal/lung lesions	Radiation and Surgery	160 (3+)	1 2 2 2 3 **4**	Nivolumab Ipilimumab Nivolumab Relatlimab Carboplatin/Taxol **MGC018**	10/18 – 07/19 08/19 – 09/19 08/19 – 02/20 11/19 – 02/20 04/20 (1 dose) **05/20 – 07/20**	**SAE hematuria/ thrombocytopenia**	**-24%**
Patient #2 4 target lesions (2 lung, hilar node, soft tissue) Non-target bilateral lung lesion	Surgery	195 (2+)	1 2 **3**	Pembrolizumab Ipilimumab + Nivolumab **MGC018**	03/20 – 07/20 07/20 – 08/20 **10/20 – 03/21**	**PD**	**-28%**
Patient #3 2 target lesions (skin) Non-target multiple lower extremity lesions	Radiation and Surgery	66 (3+)	1 2 3 4 **5**	Ipilimumab Pembrolizumab Ipilimumab Nivolumab **MGC018**	02/15 – 05/15 08/15 – 10/17 06/18 – 08/18 10/18 – 09/20 **10/20 – 04/21**	**N/A, ongoing**	**cPR (-36%)**

PD = progressive disease; cPR = confirmed partial response; N/A = not applicable.
Data were extracted on 03May2021.

mCRPC Patients with < 50% PSA Reduction

Patient (Dose)	B7-H3 H Score (Vasculature Score)	Line of Therapy	Treatment	Duration of Therapy (# months)	MGC018 Best Response	MGC018 PSA Change	Reason for MGC018 Discontinuation
Patient #6 **2 mg/kg** Bone only disease	245 (2+)	1 2 3 **4**	Docetaxel Sipuleucel-T Abiraterone **MGC018**	4 2 18 **1.5**	**SD**	**+70%**	**Stasis dermatitis**
Patient #7 **2 mg/kg** Bone only disease	N/A (insufficient tumor)	1 2 3 4 **5**	Enzalutamide Docetaxel Abiraterone Radium 223 **MGC018**	Unknown 4 12 2 **1.5**	**SD**	**+25%**	**Palmar plantar erythrodysesthesia/ maculopapular rash**
Patient #8 **4 mg/kg** Bone only disease	No biopsy available	1 2 **3**	Docetaxel Enzalutamide **MGC018**	5 8 **2**	**SD**	**-12%**	**Patient decision**
Patient #9 **4 mg/kg** Periaortic lymph node Bone disease	Not performed	1 2 3 4 **5**	Enzalutamide Docetaxel Cabazitaxel Tesetaxel **MGC018**	24 3 3 1 **.25**	**Not evaluable**	**Unknown**	**DLT Grade 3 fatigue > 72 hours**

Data were extracted on 03May2021.

Update on mCRPC PSA Responders from ASCO 2020

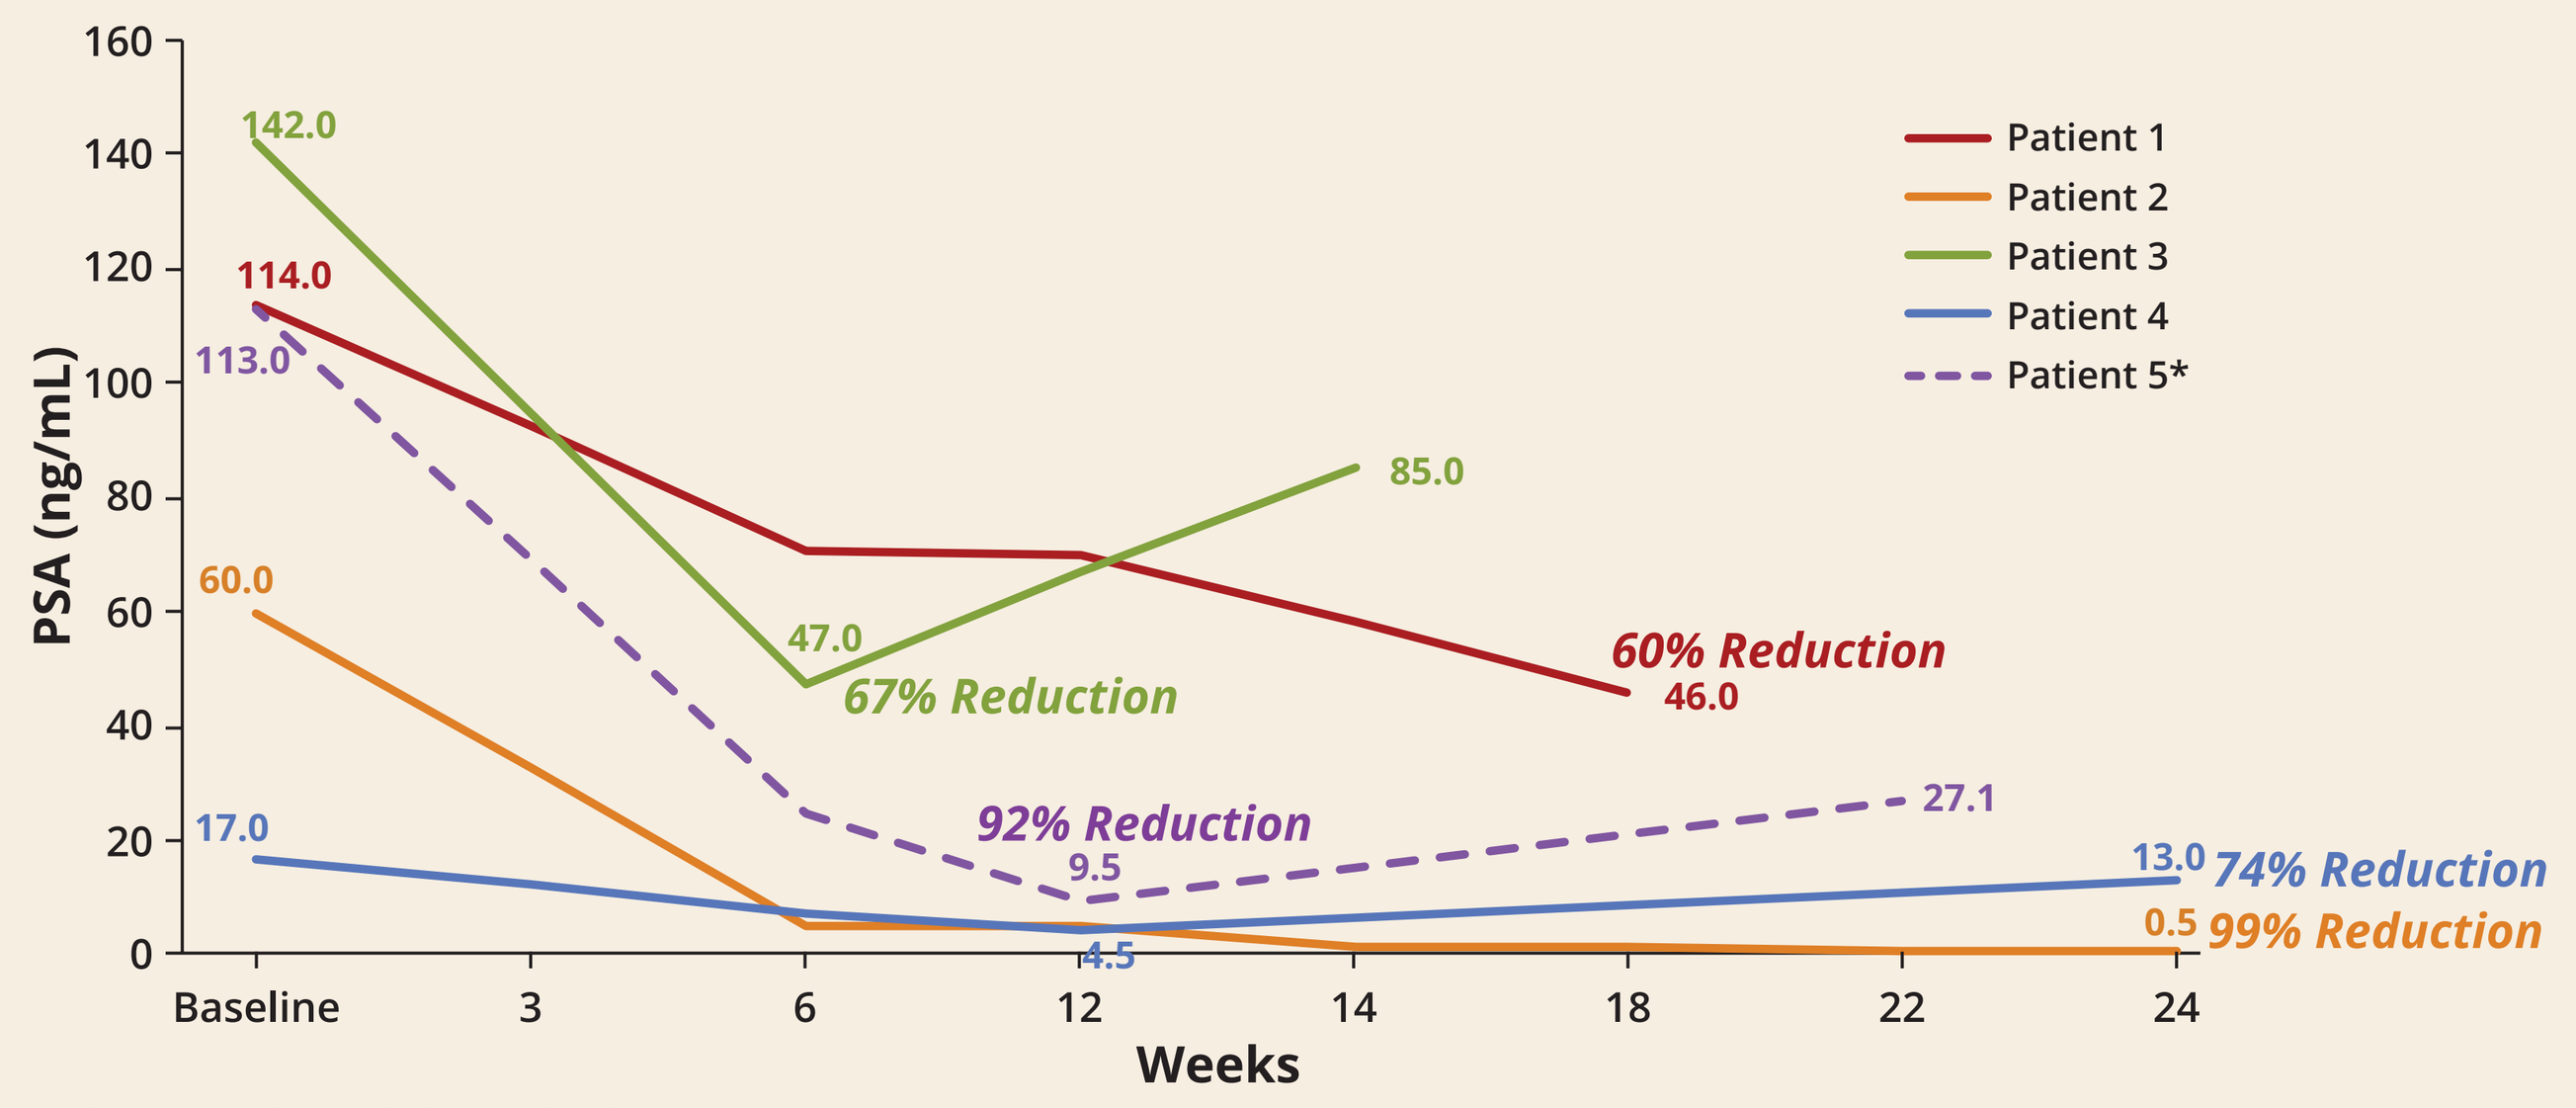

PSA (ng/mL)
Weeks
Baseline
142.0
114.0
113.0
60.0
17.0
47.0
67% Reduction
85.0
60% Reduction
46.0
92% Reduction
9.5
4.5
27.1
13.0
74% Reduction
0.5
99% Reduction
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5*

* Patient 5 data scaled (1/10) for charting purposes.

Patient (Dose)	B7-H3 H Score (Vasculature Score)	Line of Therapy	Treatment	Duration of Therapy (# months)	MGC018 Best Response	MGC018 PSA Reduction	MGC018 Time to Progression (# months)	Reason for MGC018 Discontinuation
Patient #1 **2 mg/kg** One target lesion (lymph node); non-target abdominal adenopathy and bone lesions	130 (1+)	1 2 3 4 5 6	Docetaxel Enzalutamide Prostvac Abiraterone Nivolumab **MGC018**	4 24 5 6 6 **4**	**SD (-29%)**	**-60%**	**Unknown (>4)**	**Patient decision due to numerous clinic visits**
Patient #2 **3 mg/kg** Bone only disease	N/A (insufficient invasive tumor)	1 2 3 4 **5**	Docetaxel Abiraterone Enzalutamide Radium 223 **MGC018**	6 4 12 6 **6**	**SD**	**-99%**	**6**	**New skull lesions on CT scan obtained for head injury; skull lesions not seen on baseline bone scan: no baseline head CT**
Patient #3 **3 mg/kg** Bone only disease	250 (2+)	1 2 3 4 **5**	Docetaxel Provenge Enzalutamide Abiraterone **MGC018**	8 2 6 9 **3**	**SD**	**-67%**	**Not yet progressed (7)**	**Palmar plantar erythrodysesthesia**
Patient #4 **3 mg/kg** Bone only disease	279 (2+)	1 2 **3**	Abiraterone Nivo + Rucaparib **MGC018**	Unknown Unknown **5**	**SD**	**-74%**	**Not yet progressed (7)**	**Pericardial effusion**
Patient #5 **3 mg/kg** Bone only disease	215 (1+)	1 2 3 4 5 **6**	Docetaxel Provenge Enzalutamide Abiraterone Docetaxel **MGC018**	4 12 7 7 4 **3**	**SD**	**-92%**	**Initiated subsequent therapy (6)**	**Increasing PSA**

Data were extracted on 03May2021.

Best Percent Change in PSA: Dose Escalation and Cohort Expansion

>50% PSA Reduction in 11/22 (50%) mCRPC Expansion Patients; (16/31 [52%] in Escalation + Expansion)

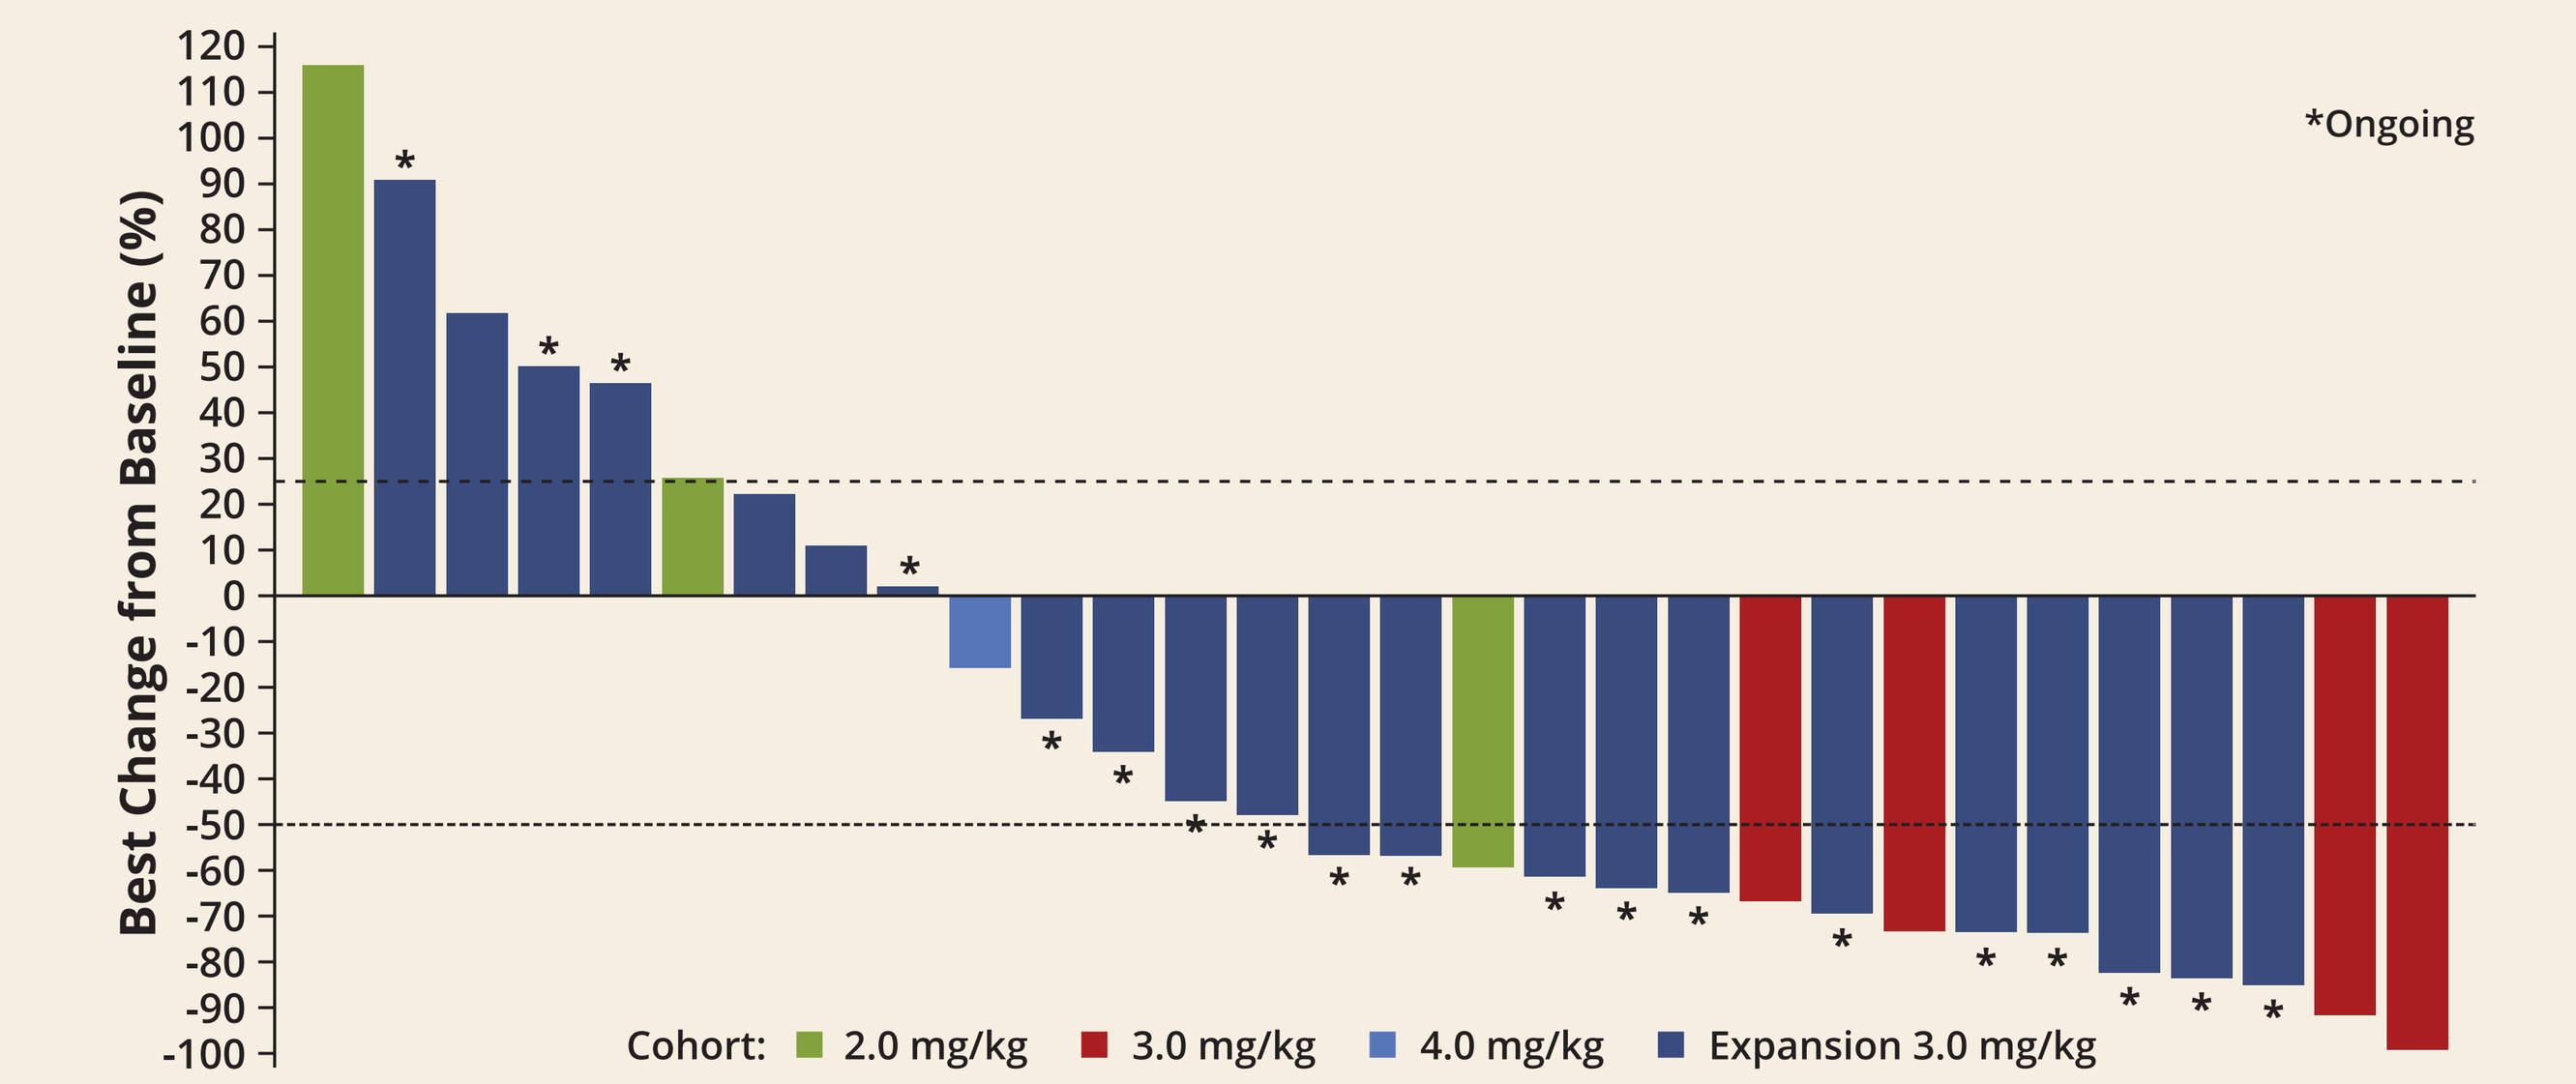

Best Change from Baseline (%)
*Ongoing
Cohort: 2.0 mg/kg 3.0 mg/kg 4.0 mg/kg Expansion 3.0 mg/kg

Patients who received at least one dose.
Data were extracted on 03MAY2021.

Conclusions

- Acceptable safety profile with manageable hematologic and skin toxicity
 - Recommended Phase 2 dose = 3 mg/kg
- Anti-tumor activity observed in 3 dose escalation melanoma patients
 - Reductions in target lesion sums of 24%, 28%, and 36% (cPR, remains on treatment 6 mos.)
- Preliminary mCRPC cohort expansion results as of data cutoff (03 May 2021)
 - > 50% PSA reduction in 11/22 (50%) patients
 - Of 13 patients with measurable disease, 6 not yet evaluable, 7 had first 9-week imaging
 - Of the 7 patients, 4 had reductions in target lesion sums of 13%, 21%, 27%, and 35% (uPR)
 - 12 of 13 patients remain on treatment
- Ongoing enrollment in mCRPC, TNBC, NSCLC, SCCHN, and melanoma

Presented at the 2021 ASCO Annual Meeting, June 4–8, 2021