Tumor Antigen Expression-dependent Activation of the CD137 Costimulatory Pathway by Bispecific DART® Proteins

Liqin Liu, Chia-Ying K. Lam, Vatana Long, Lusiana Widjaja, Yinhua Yang, Robert Burns, Alex Berezhnoy, Kalpana Shah, Doug Smith, Joanna Pan, Syd Johnson, Ezio Bonvini and Paul Moore

MacroGenics, Inc., Rockville, MD and South San Francisco, CA

3642

http://ir.macrogenics.com/events.cfm

Abstract

Introduction: CD137 (4-1BB) is a costimulatory molecule expressed by activated T and NK cells that, upon interaction with its CD137 ligand, supports cell activation, proliferation and survival. Activation via CD137 holds great promise for cancer immunotherapy; however, current CD137 agonistic interventions are associated with systemic safety concerns. To develop a therapeutic modality that reduces the potential for systemic CD137 effects, we applied the DART® bispecific platform to generate proteins that can induce tumor-antigen dependent T-cell activation.

Methods: DART molecules were constructed containing anti-CD137 variable regions together with either anti-HER2 or anti-EphA2 variable regions. DART binding properties were evaluated by flow cytometry; signaling responses assessed using a NF-κB luciferase reporter cell line expressing CD137. Costimulatory activity was characterized with primary human T cells in the presence or absence of tumor target antigen-expressing cells.

Results: Flow cytometry analysis demonstrated that both HER2 x CD137 and EphA2 x CD137 DART molecules bind their respective target antigens. Co-culturing of a CD137/NF-κB reporter cell line with tumor lines expressing HER2 or EphA2 revealed tumor antigen-dependent CD137 pathway activation by HER2 x CD137 and EphA2 x CD137 DART molecules, respectively. To evaluate the effects of HER2 x CD137 and EphA2 x CD137 DART molecules on T-cell responses, costimulation T-cell assays were performed. In the presence of the relevant antigen-positive cell line, each respective DART molecule was able to promote T-cell proliferation and cytokine release in a HER2 or EphA2-dependent manner. No T-cell costimulation was observed by either DART molecule in the absence of antigen-expressing tumor cells. Furthermore, the level of tumor antigen-dependent costimulation supported by the DART molecules correlated with the level of tumor target expression. Consistent with the preferential induction of CD137 by the CD8 T-cell subset, CD137-based DART proteins induced a substantial increase in the fraction of $CD8^+$ central memory and effector memory T cells in the presence of the proper tumor antigen-expressing cells.

Conclusions: HER2 x CD137 and EphA2 x CD137 DART proteins promote T-cell costimulation in a tumor antigen-dependent manner and may provide an opportunity to target the CD137 costimulatory pathway for cancer immunotherapy, while limiting systemic T-cell activation and related side effects.

Introduction

- CD137 (4-1BB) is a potent costimulatory molecule expressed by activated T, NK and dendritic cells
 - ↑ Expression on TILs following MHC-peptide/TCR engagement
 - ↑ Expression on NK cells by mAb-opsonized tumor cells
 - Expressed by vascular endothelium in tumors
- CD137 interaction with CD137 ligand or agonistic Ab results in:
 - ↑ Immune cell proliferation and antitumor cytolytic activity
 - Countering of immune cell exhaustion and apoptosis
 - ↑ Expression of endothelial molecules and chemokines that facilitates ↑ CD8 T cells homing to tumor site
 - ↑ ADCC by NK cells; synergy with rituximab, trastuzumab or cetuximab in mouse models
- Current CD137 agonistic interventions may present systemic safety concerns
 - Urelumab associated with dose-dependent hepatitis (some fatal)
- Localized CD137 activation in tumor microenvironment may diminish probability of systemic effects
- To limit systemic effects, tumor antigen specific, CD137-targeted bispecific DART proteins were designed to potentially limit CD137 activation to the tumor microenvironment

Proposed Mechanism of Action

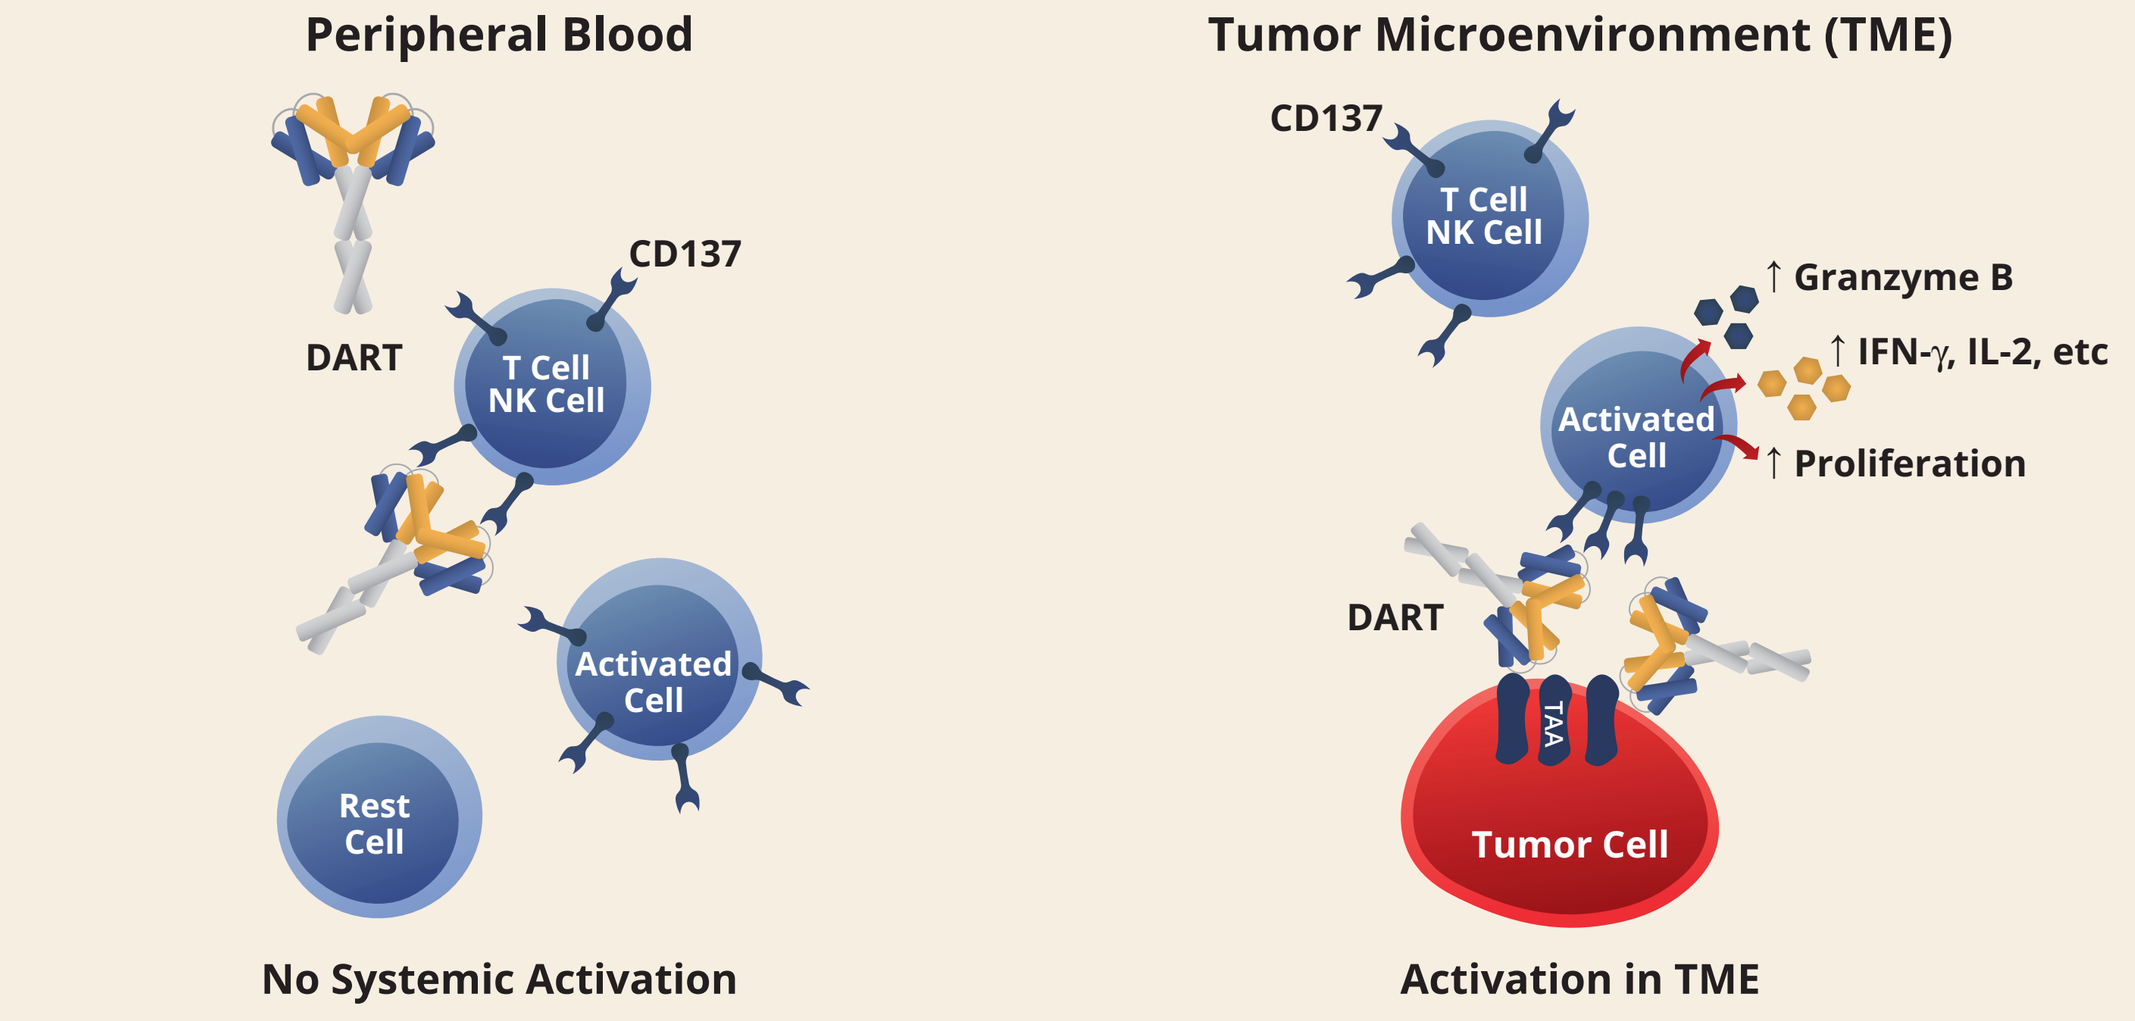

Peripheral Blood
Tumor Microenvironment (TME)
CD137
DART
T Cell NK Cell
Activated Cell
Rest Cell
↑ Granzyme B
↑ IFN-γ, IL-2, etc
↑ Proliferation
TAA
Tumor Cell
No Systemic Activation
Activation in TME

- CD137 expression restricted to activated immune cells
- Bivalent TAA x CD137 DART engagement of CD137 insufficient to provide costimulatory signal

- CD137 expression restricted to activated immune cells
- Clustering of TAA x CD137 on tumor cells supports multivalent engagement of CD137 and costimulation

TAA: tumor associated antigen

- MacroGenics CD137 agonistic antibody (MG24) incorporated in DART construction was selected based on prerequisite for agonistic activity dependent on clustering

Restricted CD137 Activation Properties of MG24 Compared to Urelumab

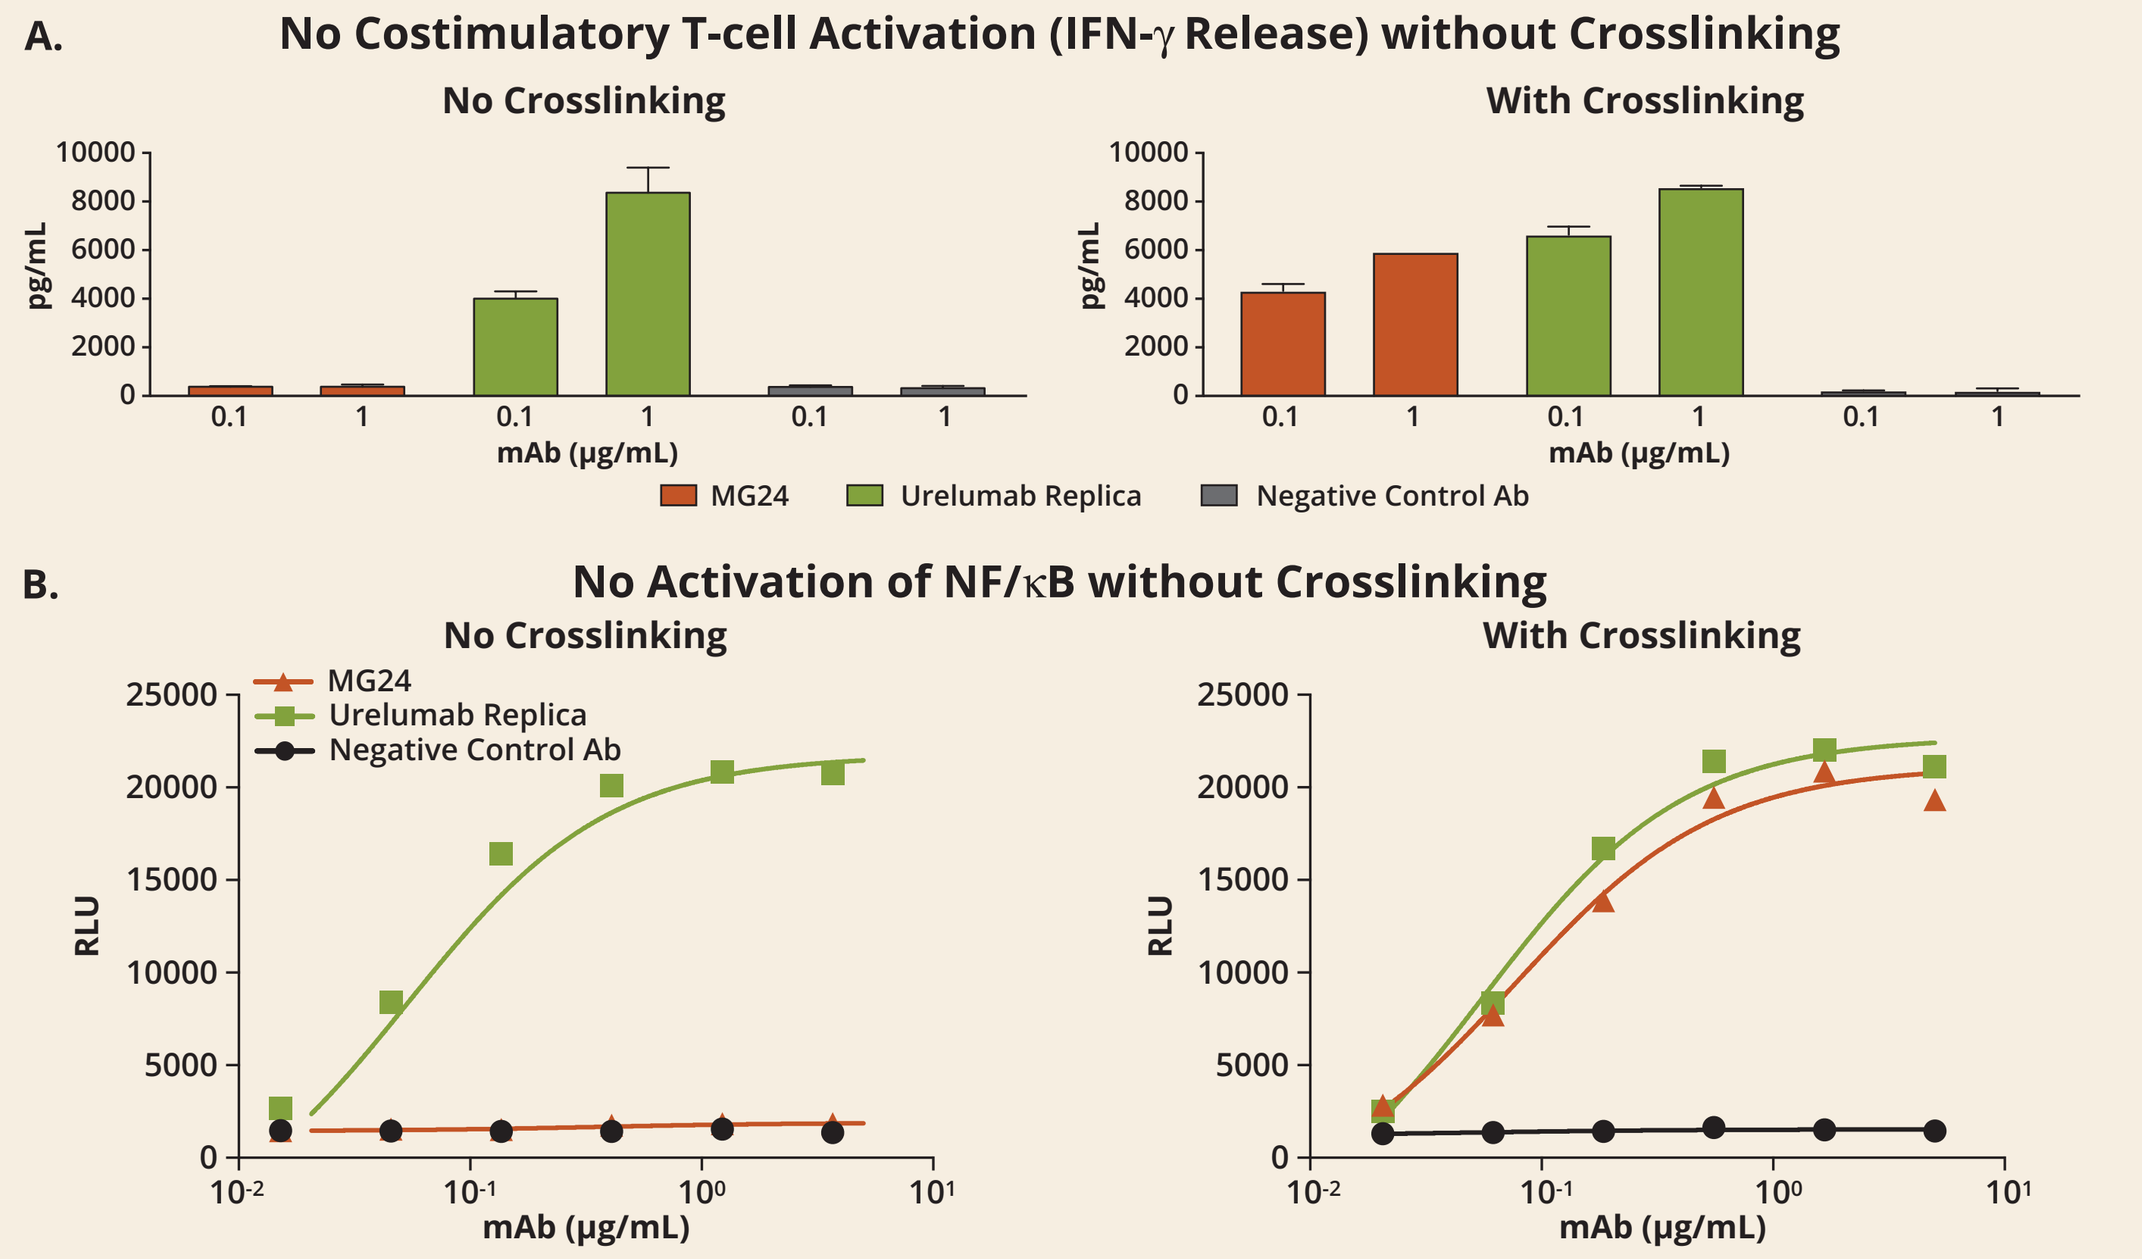

A.
No Costimulatory T-cell Activation (IFN-γ Release) without Crosslinking
No Crosslinking
With Crosslinking
pg/mL
mAb (μg/mL)
MG24
Urelumab Replica
Negative Control Ab
B.
No Activation of NF/κB without Crosslinking
No Crosslinking
With Crosslinking
RLU
mAb (μg/mL)

A. T-cell cytokine assay containing either no secondary crosslinking antibody (left panel) or 4 x αhFc crosslinking reagent (right panel).
B. CD137 reporter assay (Jurkat-NF-κB-Luc) containing either no crosslinking reagent (left panel) or 4 x αhFc crosslinking reagent (right panel). The luminescent signal (RLU) was used to as a relative measure of CD137 pathway activation.

Results

Tumor-targeted CD137 DART Proteins Bind Each Antigen

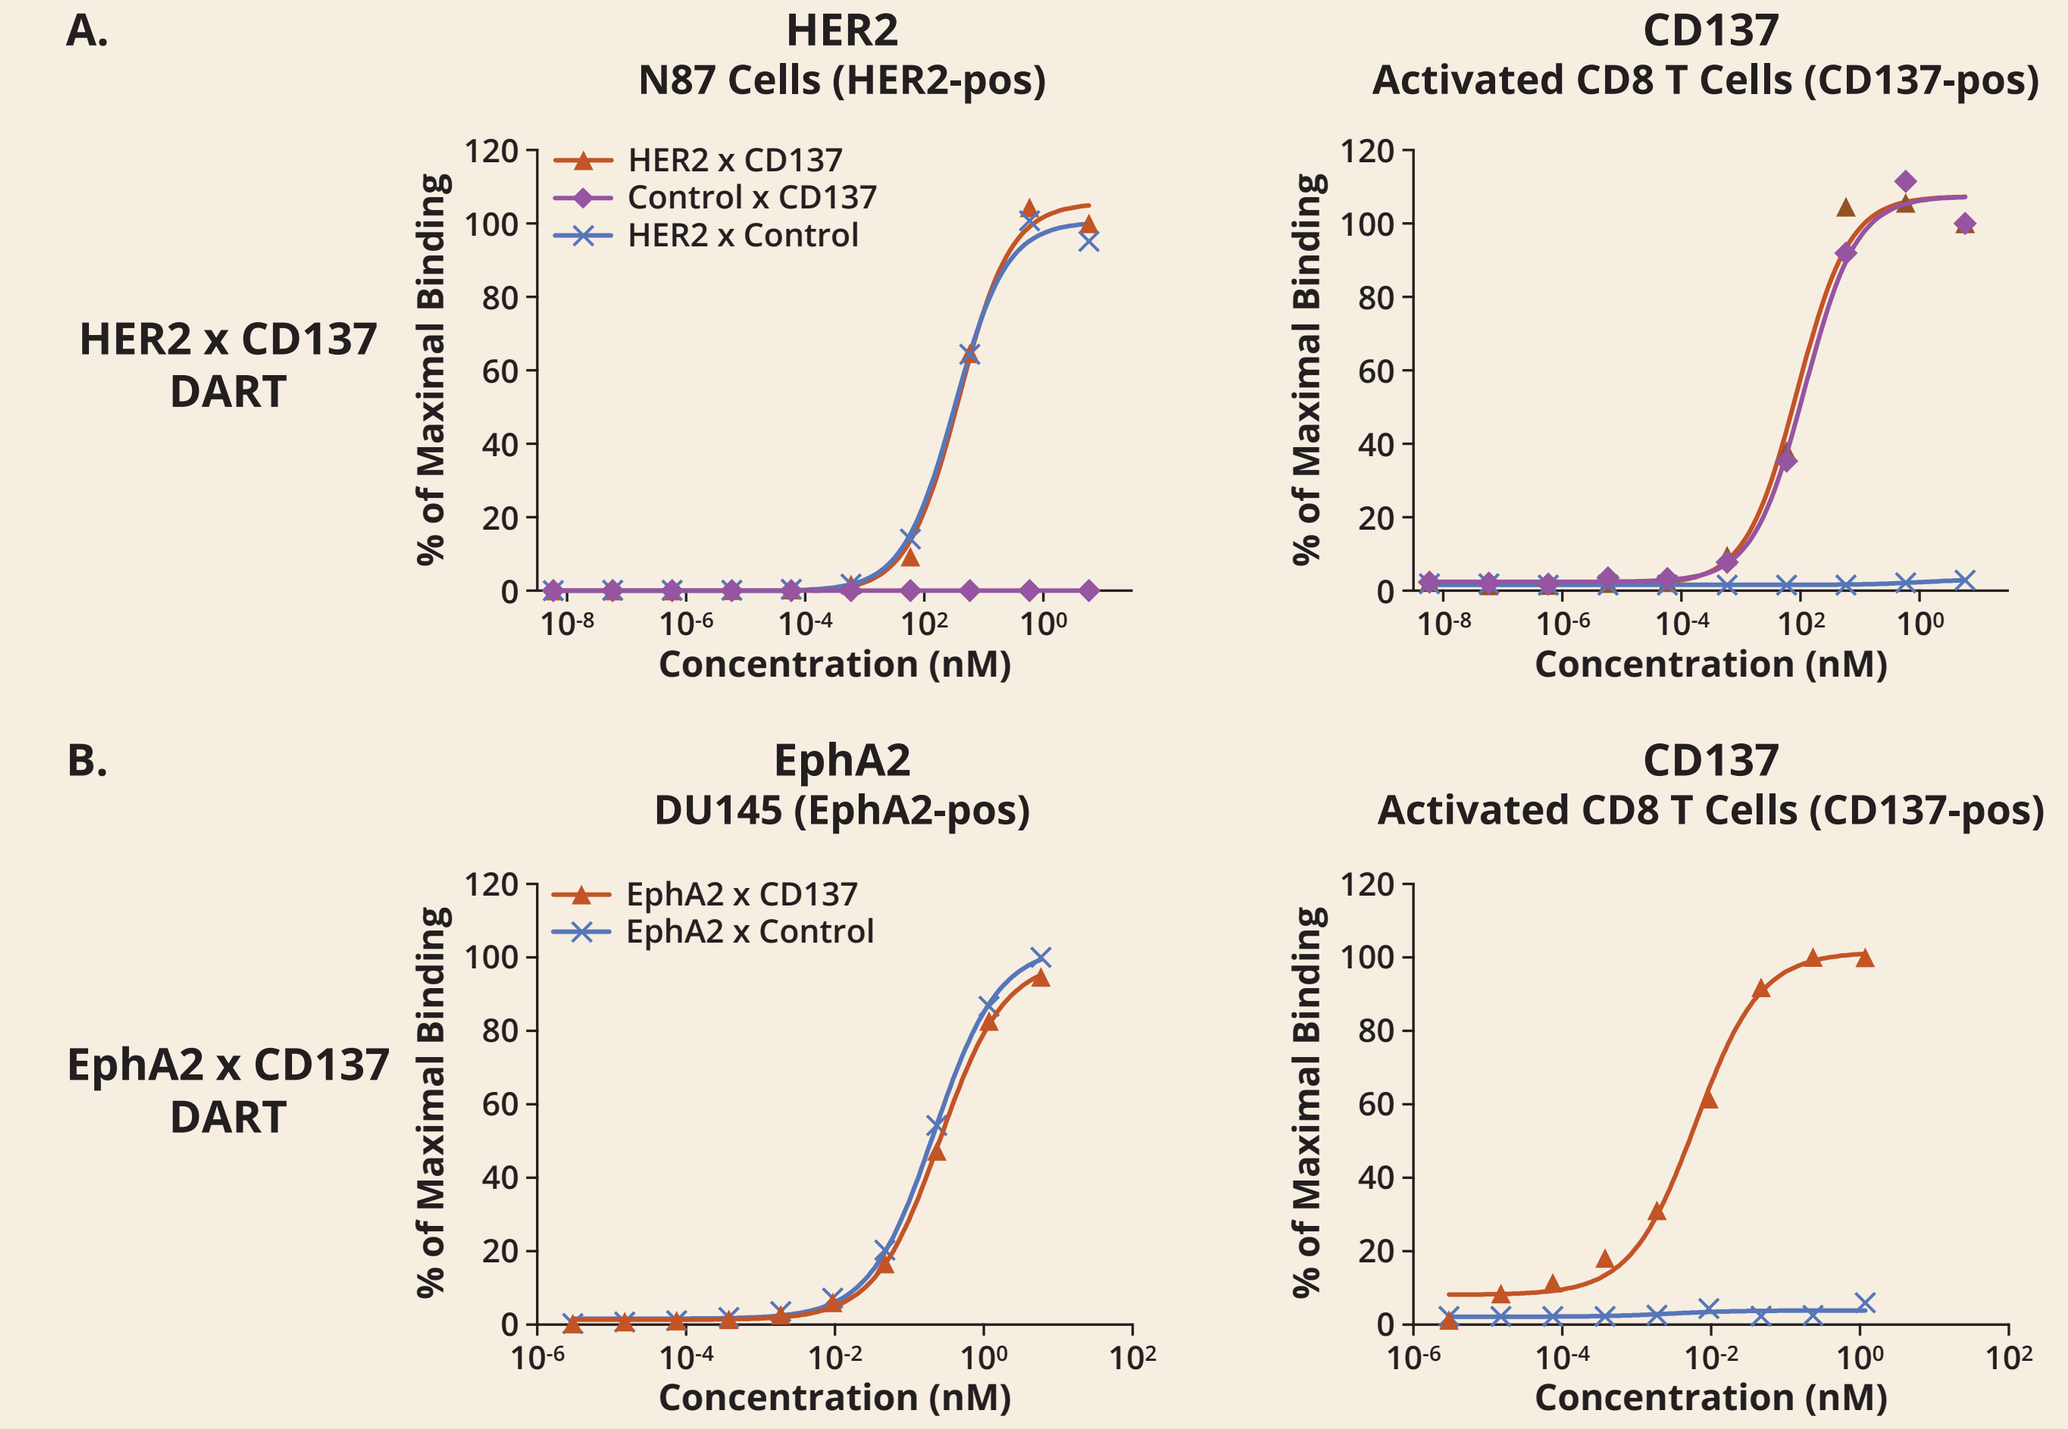

A.
HER2
N87 Cells (HER2-pos)
CD137
Activated CD8 T Cells (CD137-pos)
HER2 x CD137 DART
HER2 x CD137
Control x CD137
HER2 x Control
% of Maximal Binding
Concentration (nM)
B.
EphA2
DU145 (EphA2-pos)
CD137
Activated CD8 T Cells (CD137-pos)
EphA2 x CD137 DART
EphA2 x CD137
EphA2 x Control

HER2 x CD137 **(A)** or EphA2 x CD137 **(B)** DART binding titration on tumor target cell lines expressing either HER2 or EphA2, or primary activated T cells expressing CD137, determined by flow cytometry analysis. HER2 x CD137 or EphA2 x CD137 DART was constructed with anti-CD137 variable region together with either anti-HER2 or anti-EphA2 variable regions.

HER2 x CD137 DART-mediated Signaling Is Dependent on HER2 Expression Level

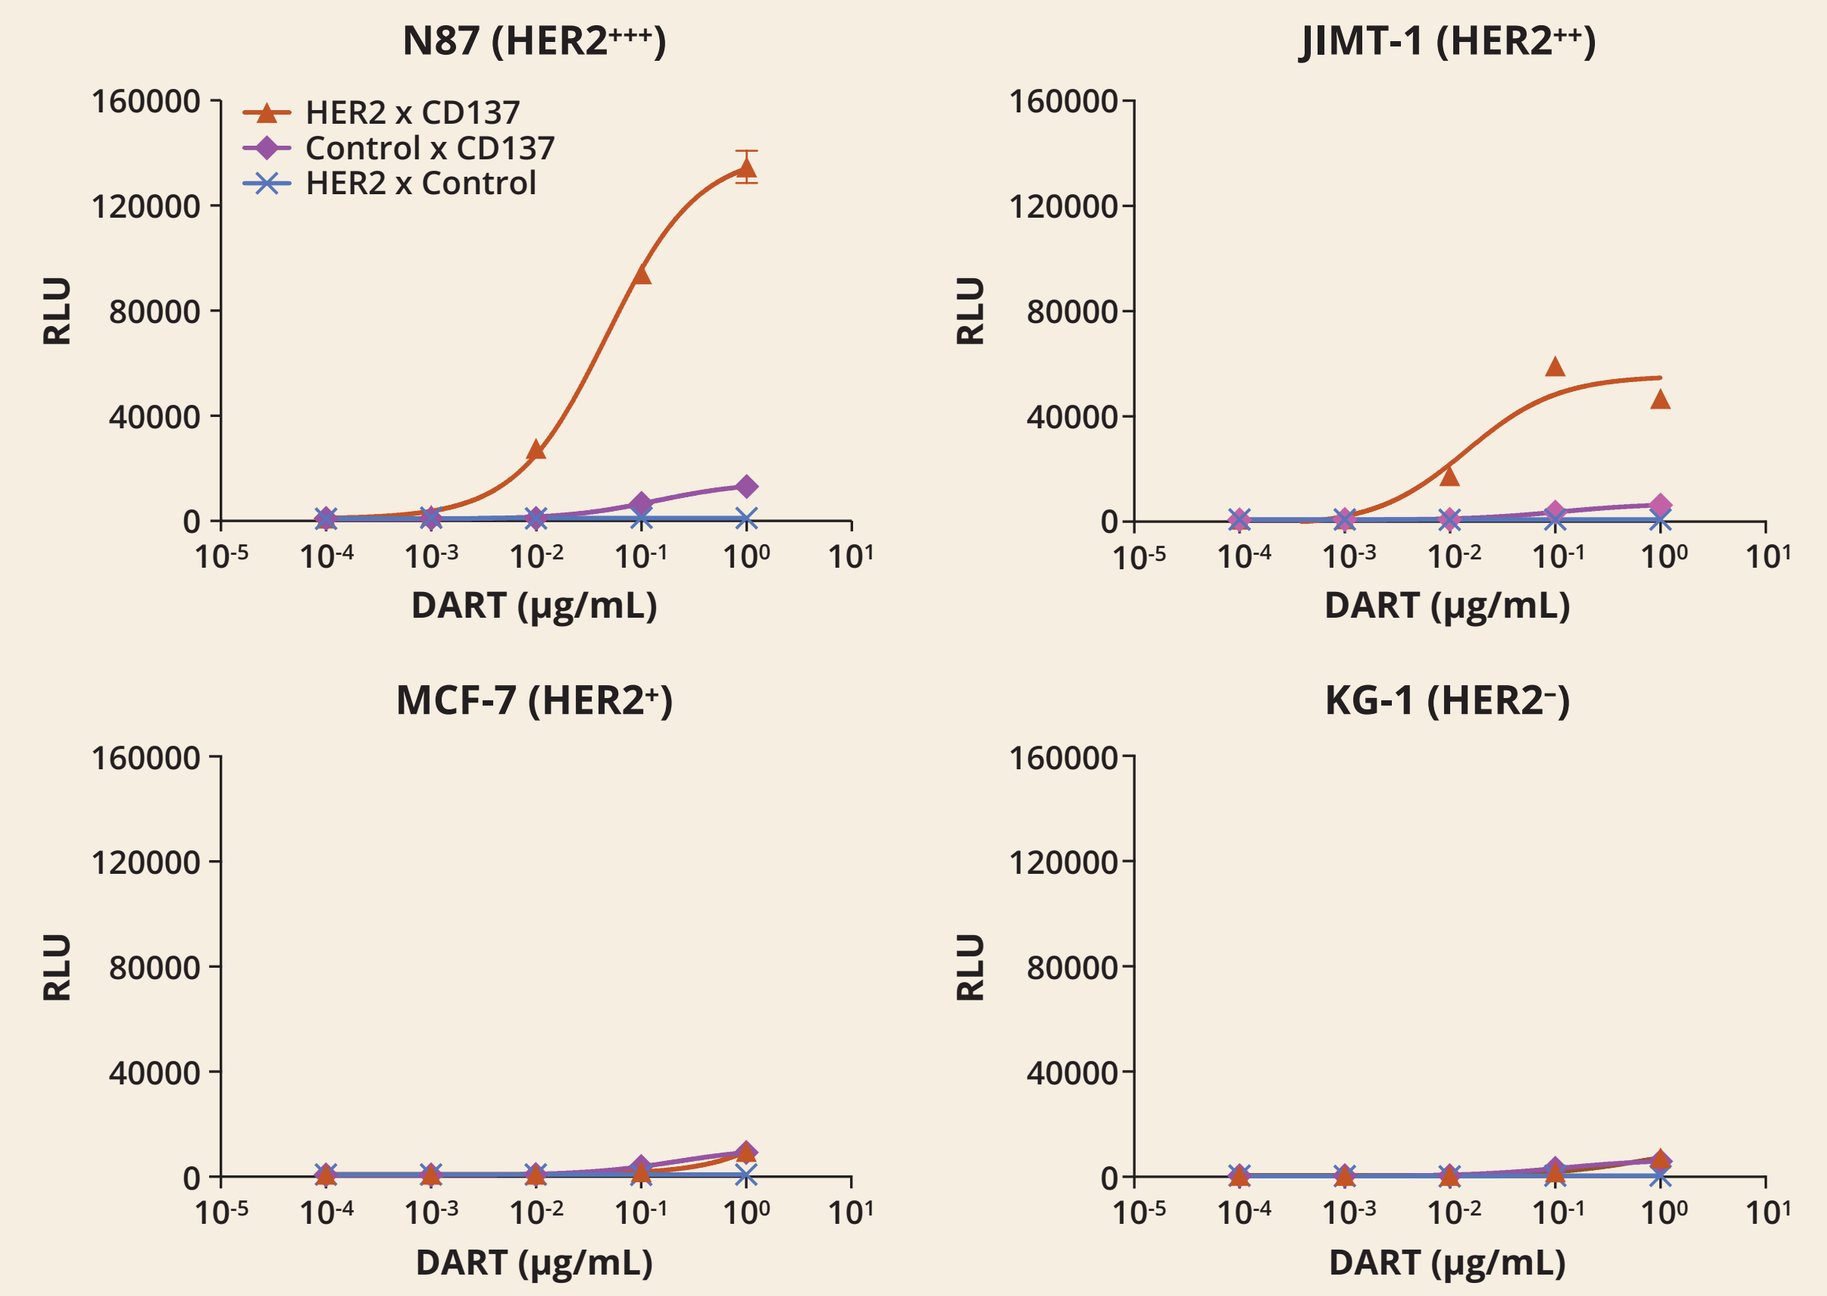

N87 (HER2+++)
JIMT-1 (HER2++)
MCF-7 (HER2+)
KG-1 (HER2−)
HER2 x CD137
Control x CD137
HER2 x Control
RLU
DART (μg/mL)

NF-κB activation by HER2 x CD137 DART or monospecific control DART protein for either HER2 (HER2 x Control) or CD137 (Control x CD137) in CD137 reporter cell line (Jurkat-NF-κB-Luc) co-cultured with either N87 ($HER2^{+++}$), JIMT-1 ($HER2^{++}$), MCF-7 ($HER2^{+}$) cells or KG-1 ($HER2^{-}$) cells. The luminescent signal (RLU) was used to as a relative measure of CD137 pathway activation.

HER2-dependent Enhancement of T-cell Proliferation by HER2 x CD137 DART

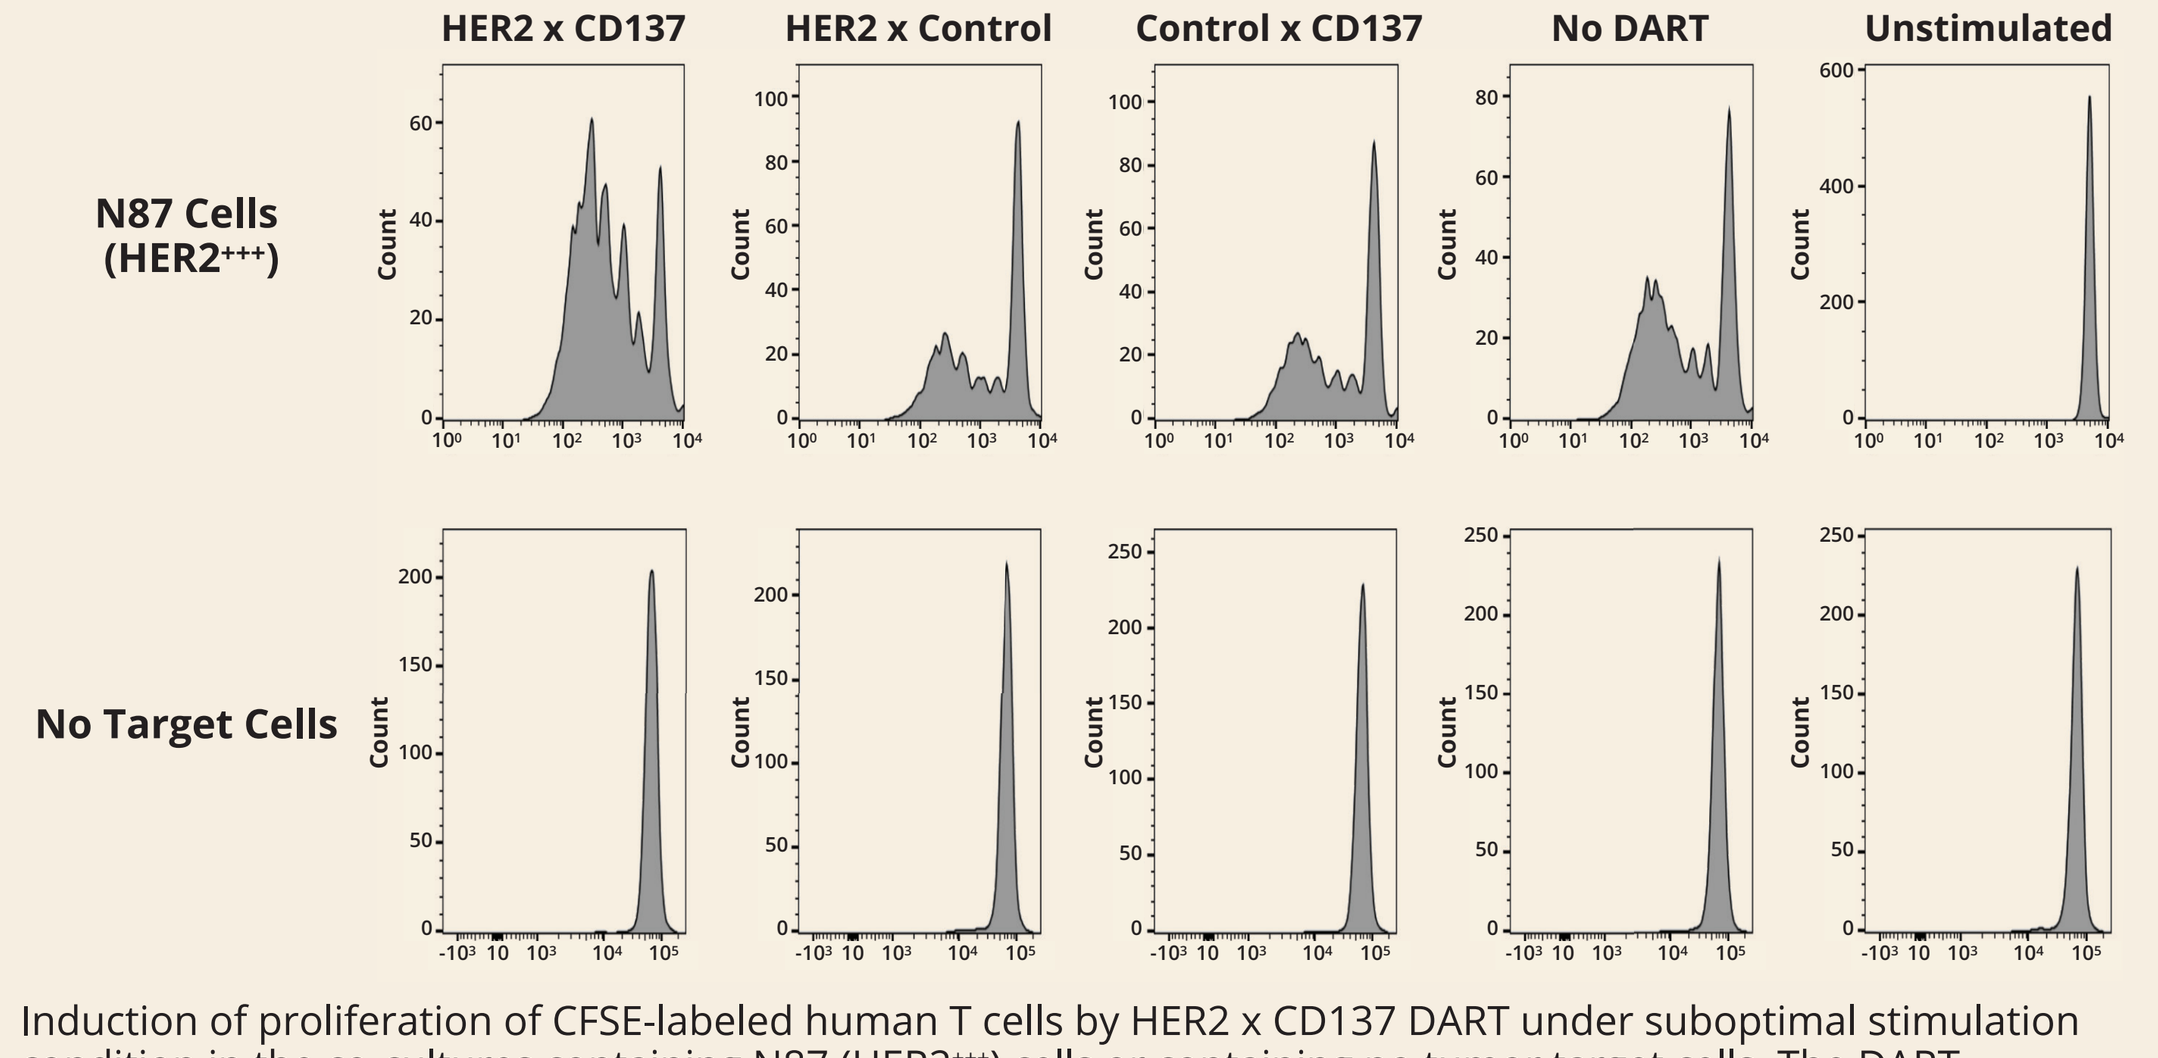

HER2 x CD137
HER2 x Control
Control x CD137
No DART
Unstimulated
N87 Cells (HER2+++)
No Target Cells
Count

Induction of proliferation of CFSE-labeled human T cells by HER2 x CD137 DART under suboptimal stimulation condition in the co-cultures containing N87 ($HER2^{+++}$) cells or containing no tumor target cells. The DART concentration tested was 0.1 μg/mL.

Antigen-dependent Enhancement of T-cell Activation by HER2 x CD137 and EphA2 x CD137 DART Molecules

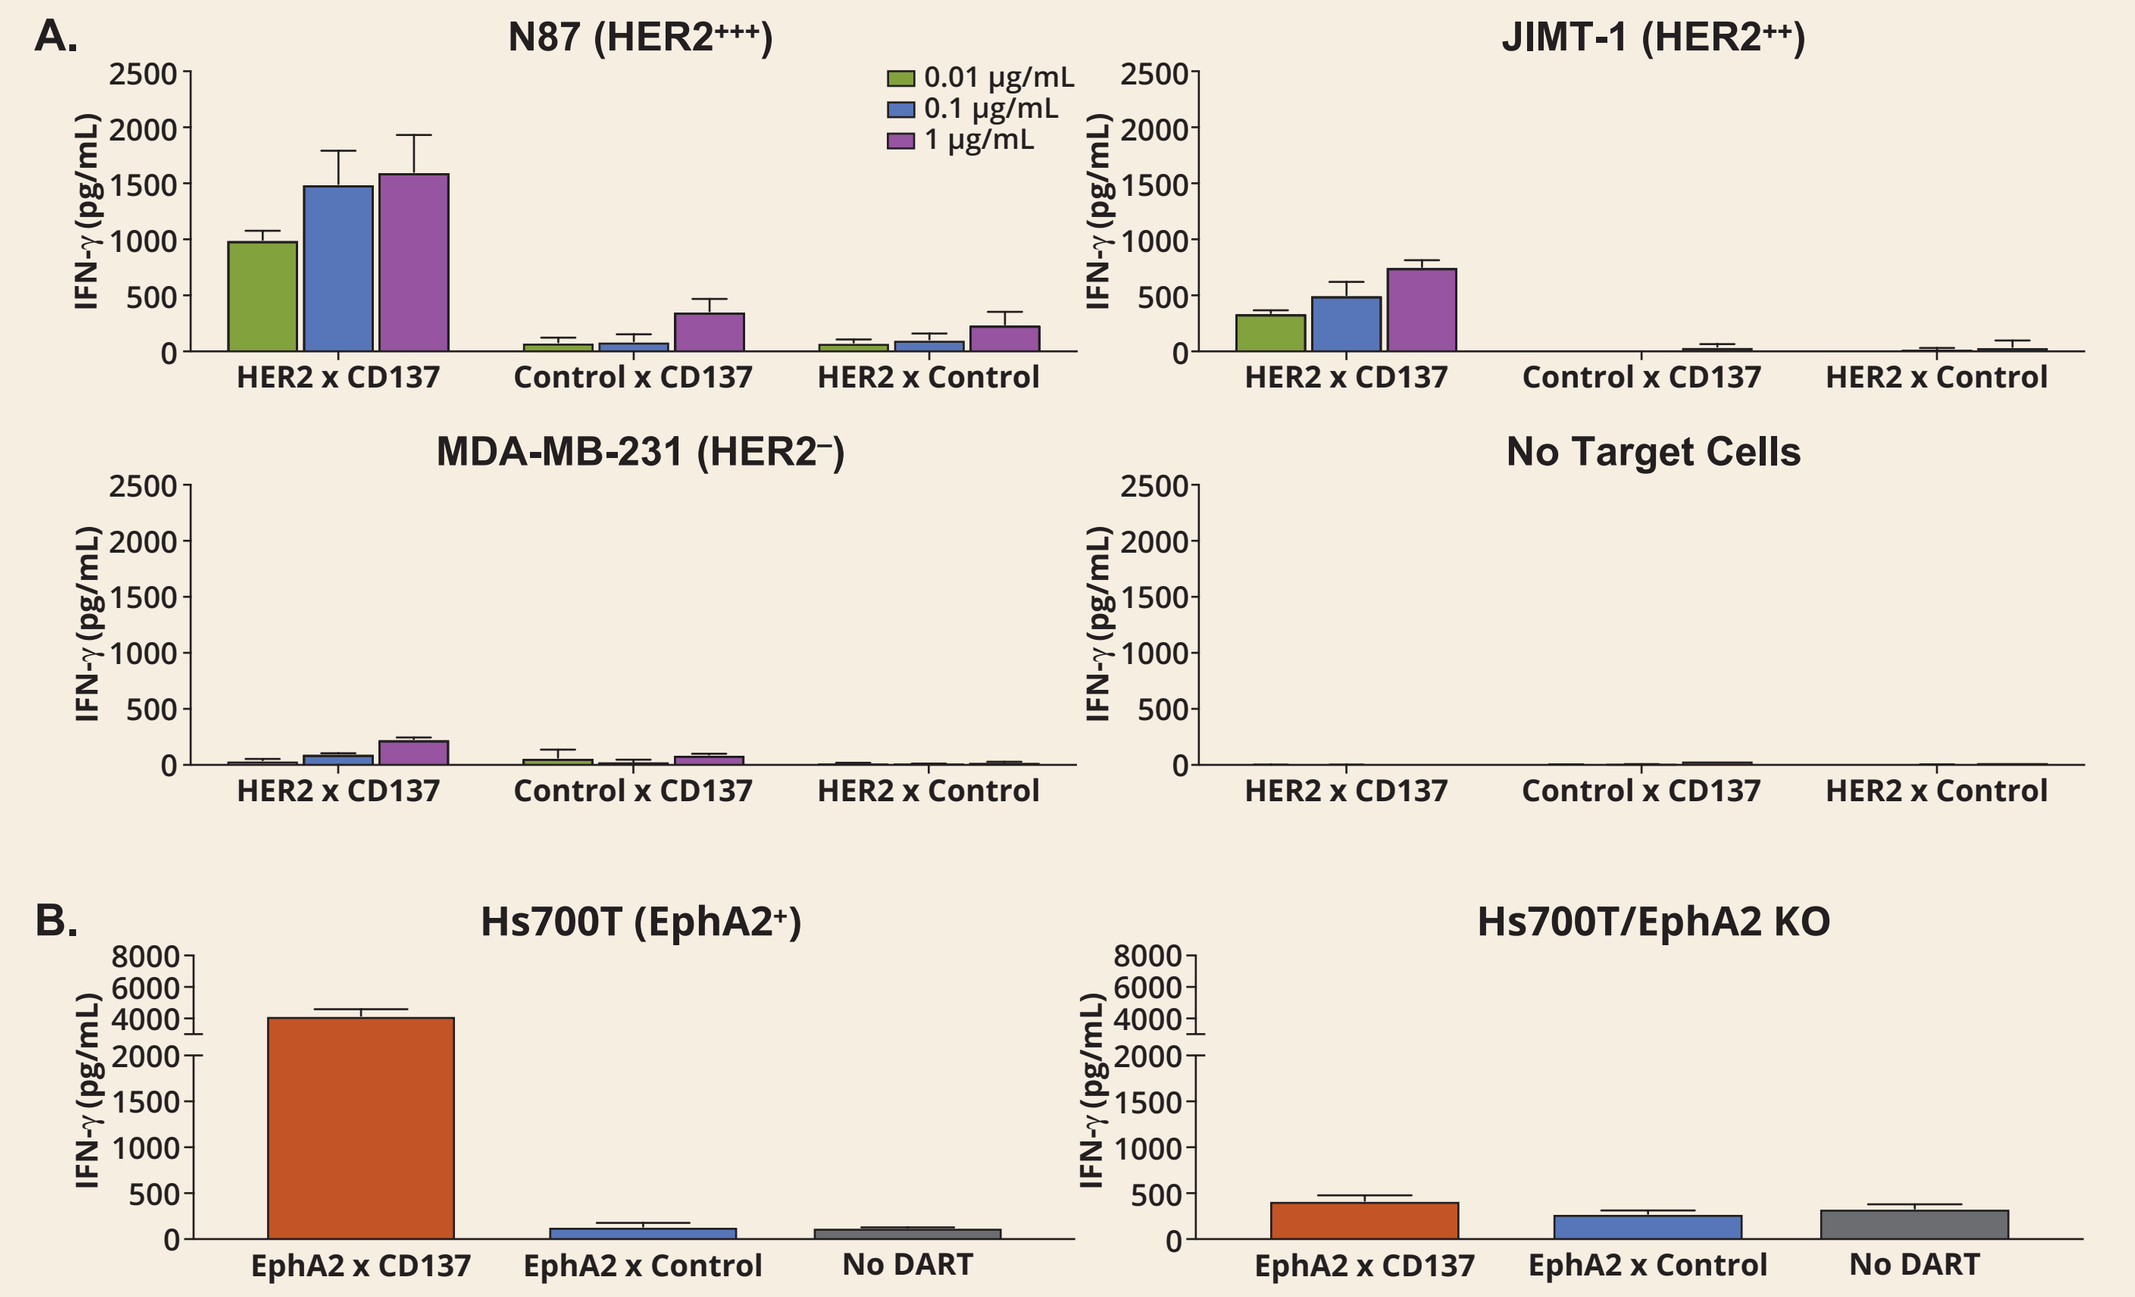

A.
N87 (HER2+++)
JIMT-1 (HER2++)
MDA-MB-231 (HER2−)
No Target Cells
0.01 μg/mL
0.1 μg/mL
1 μg/mL
IFN-γ (pg/mL)
HER2 x CD137
Control x CD137
HER2 x Control
B.
Hs700T (EphA2+)
Hs700T/EphA2 KO
EphA2 x CD137
EphA2 x Control
No DART

A. IFN-γ production by T cells treated with HER2 x CD137 DART or monospecific control DART for either HER2 (HER2 x Control) or CD137 (Control x CD137) in the presence of N87 ($HER2^{+++}$) cells, JIMT-1 ($HER2^{++}$) cells, MDA-MB-231 ($HER2^{-}$) cells or in the absence of target cells following a 72-h culture under suboptimal stimulation condition.
B. IFN-γ production by T cells treated with EphA2 x CD137 DART or monospecific EphA2 x Control DART in the presence of Hs700T ($EphA2^{+}$) cells or EphA2-neg Hs700T/EphA2 KO cells. The EphA2 experiment did not have "no tumor target cells" panel following a 72-h culture under suboptimal stimulation condition. The DART concentration evaluated was 0.1 μg/mL.

HER2 x CD137 DART-mediated Expansion of Cytolytic T Cells Depends on HER2 Expression Level

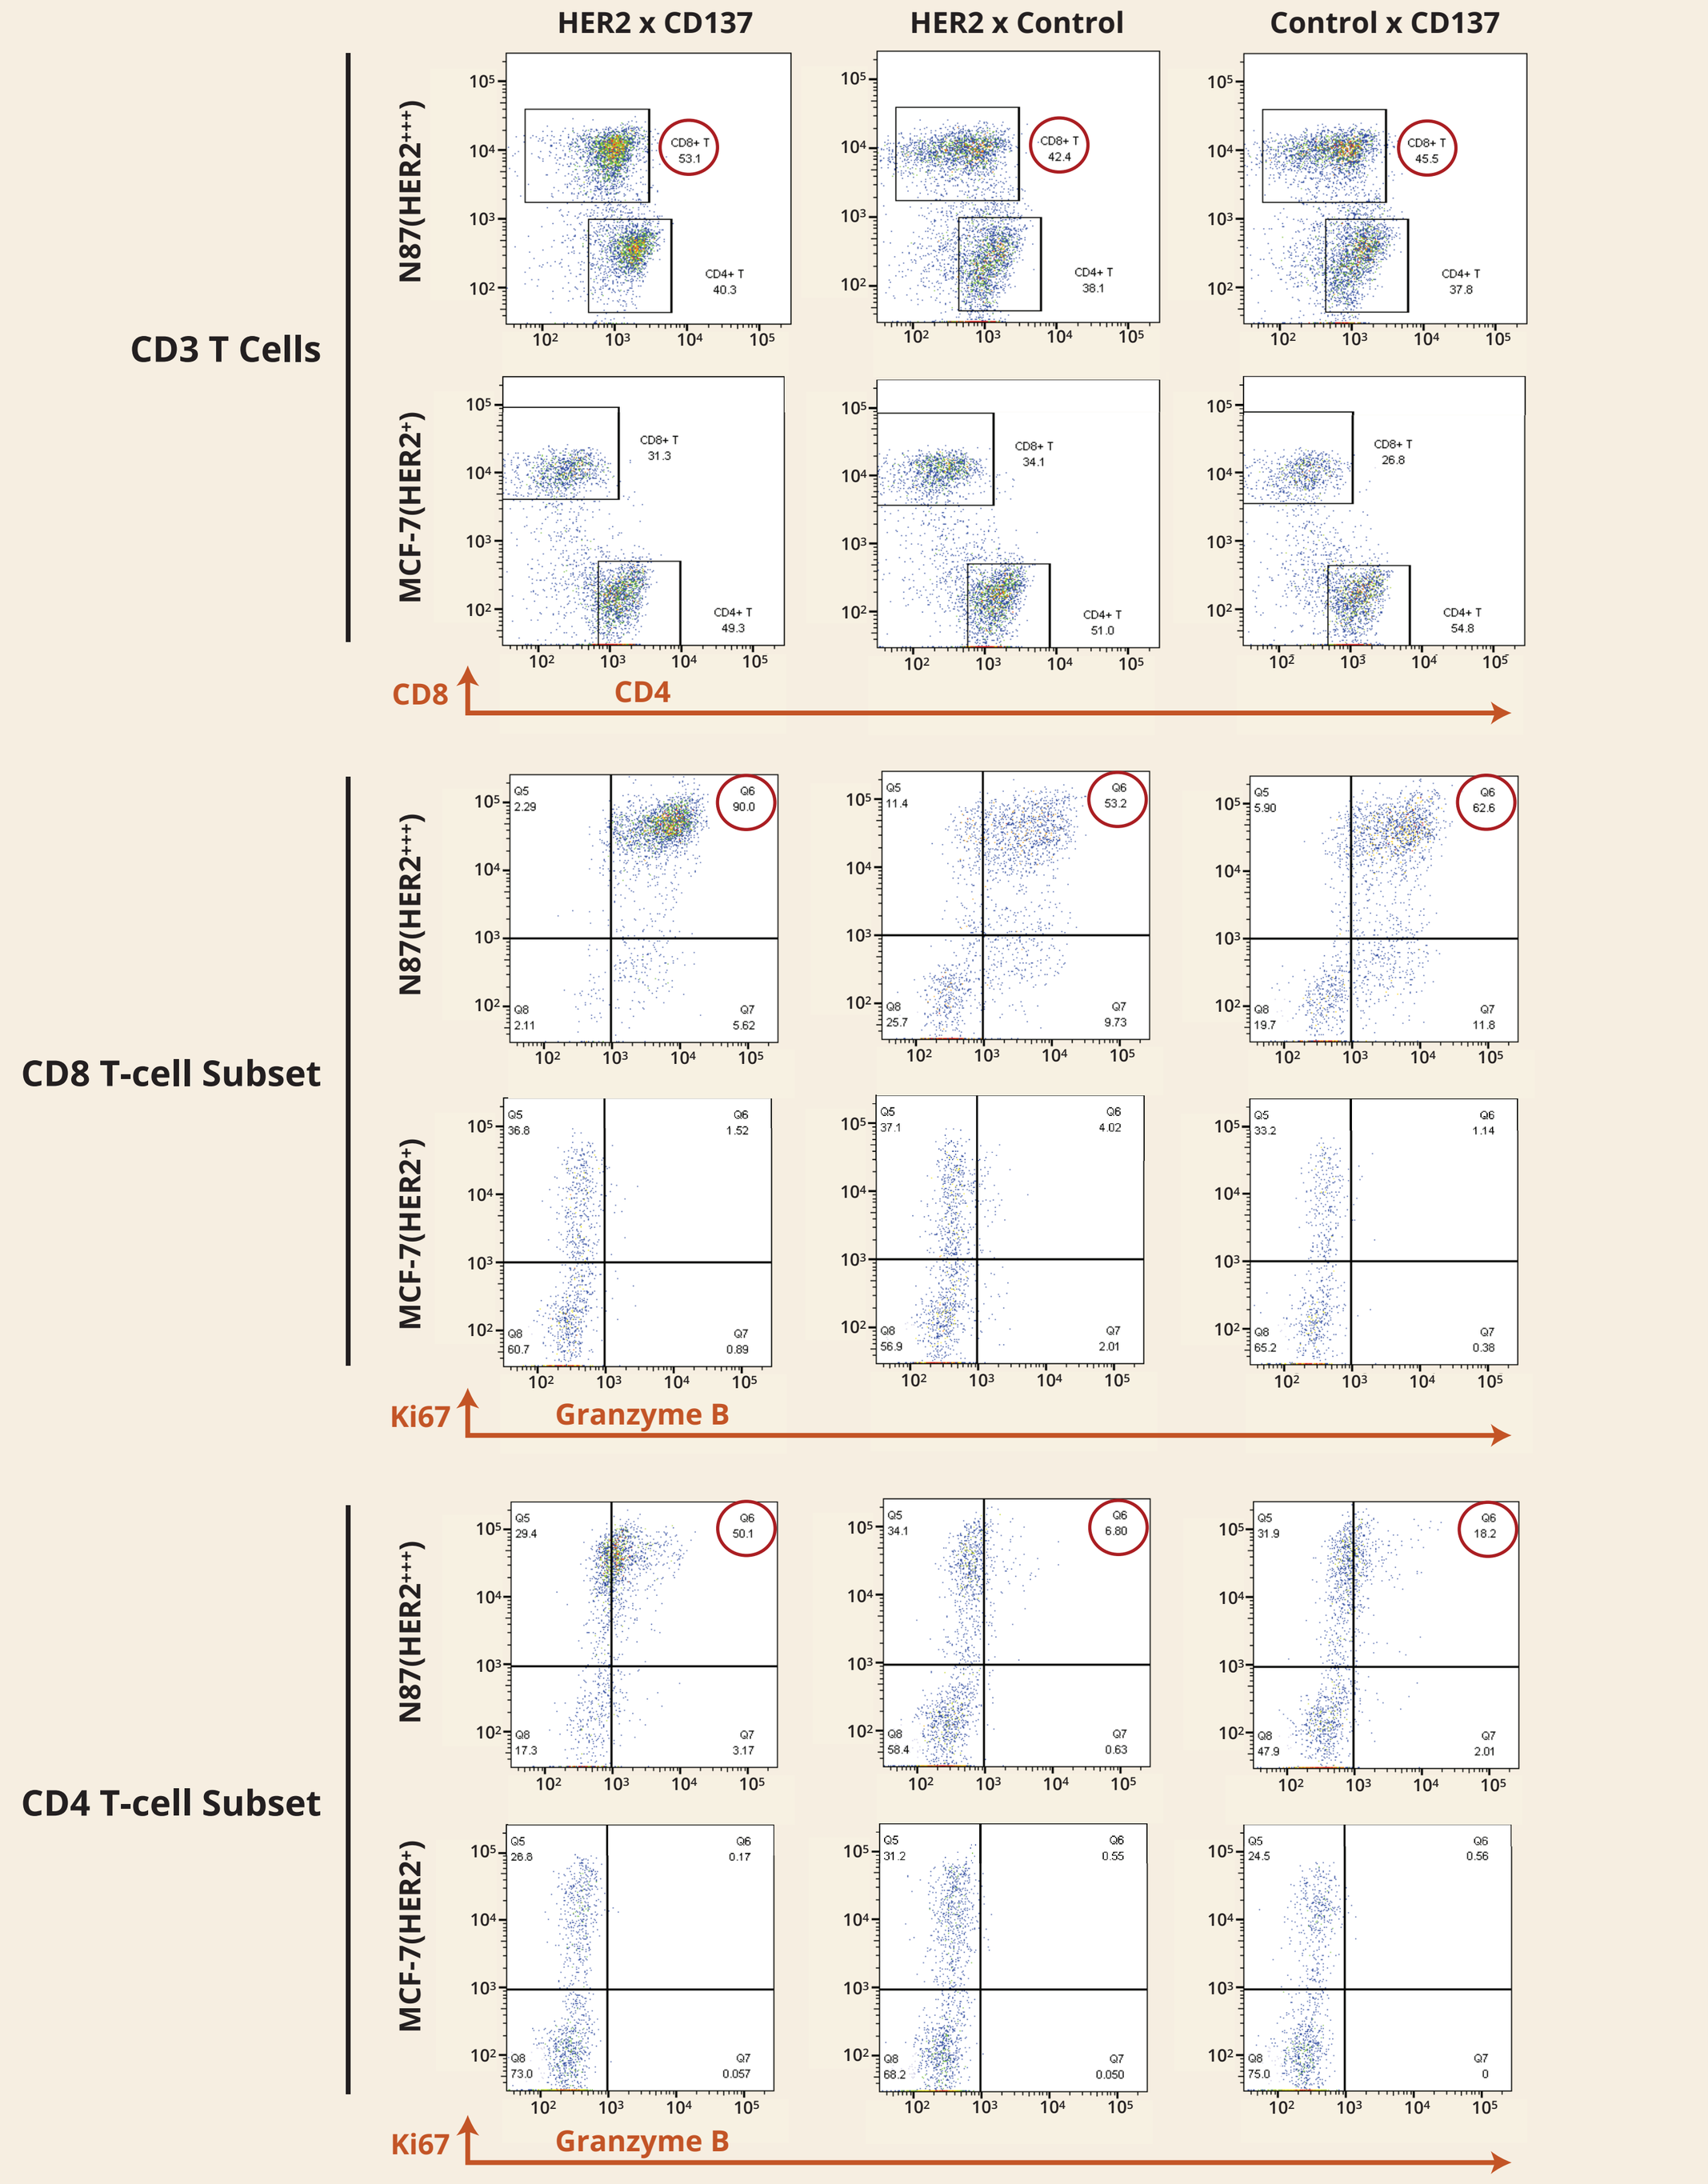

HER2 x CD137
HER2 x Control
Control x CD137
CD3 T Cells
CD8 T-cell Subset
CD4 T-cell Subset
N87(HER2+++)
MCF-7(HER2+)
CD8
CD4
Ki67
Granzyme B

Percentage of $CD8^+$ and $CD4^+$ cells (top panels) and $Ki67^+$ (proliferating), granzyme B^+ (cytolytic) cells in the gated $CD8^+$ or $CD4^+$ subset (middle and bottom panels, respectively) following co-culture of human T cells with N87 ($HER2^{+++}$) or MCF-7 ($HER2^{+}$) cells for 4 days under suboptimal stimulation condition and in the presence of HER2 x CD137 DART or monospecific control DART for HER2 (HER2 x Control) or CD137 (Control x CD137). The DART concentration tested was 0.1 μg/mL.

EphA2 x CD137 DART Induces EphA2-dependent Increase in the CD8 Memory T-cell Subsets

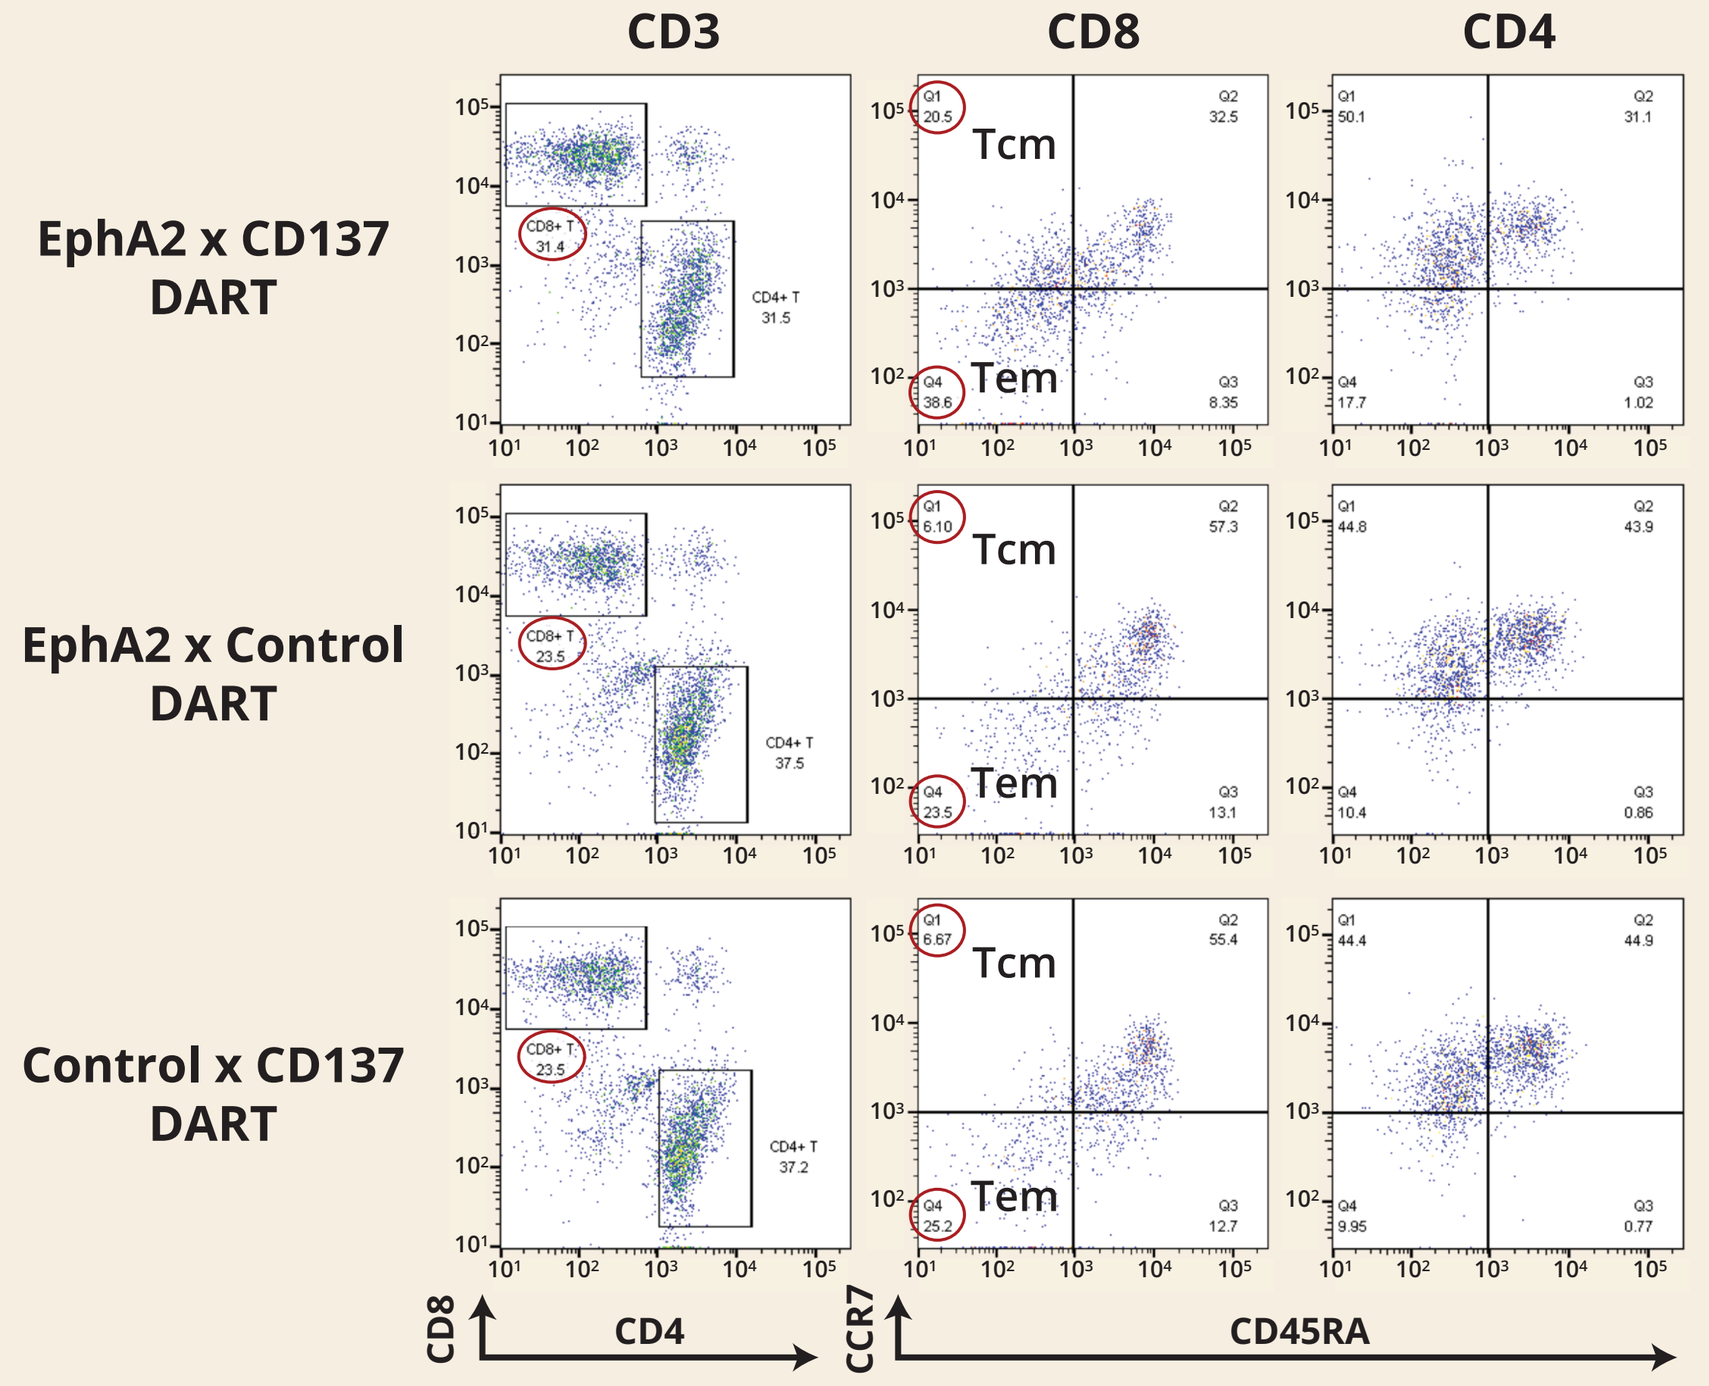

CD3
CD8
CD4
EphA2 x CD137 DART
EphA2 x Control DART
Control x CD137 DART
Tcm
Tem
CD8
CD4
CCR7
CD45RA

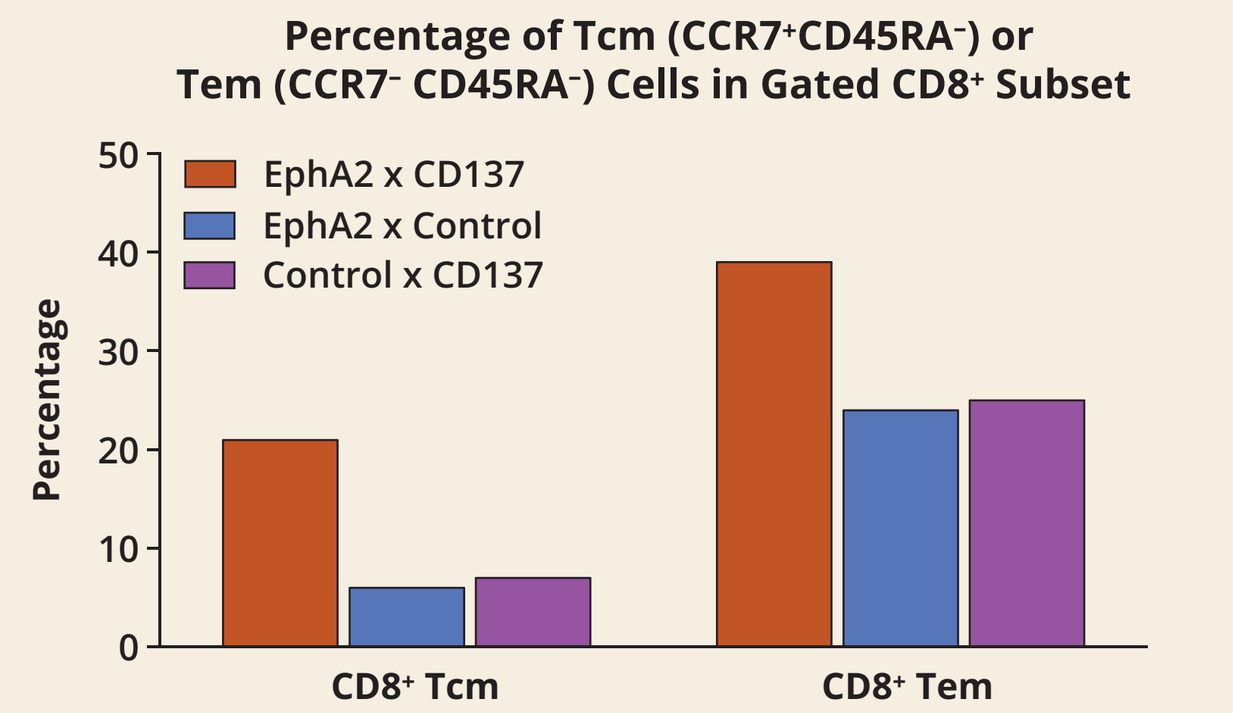

Percentage of Tcm (CCR7+CD45RA−) or Tem (CCR7− CD45RA−) Cells in Gated CD8+ Subset
EphA2 x CD137
EphA2 x Control
Control x CD137
Percentage
CD8+ Tcm
CD8+ Tem

Percentage of Tcm ($CCR7^+CD45RA^-$) or Tem ($CCR7^-CD45RA^-$) cells in the gated $CD4^+$ or $CD8^+$ subset following co-culture of T cells with EphA2-pos Colo205 cells for 5 days under suboptimal stimulation condition and in the presence EphA2 x CD137 DART or monospecific control DART for EphA2 (EphA2 x Control) or CD137 (Control x CD137). The DART concentration tested was 0.1 μg/mL.

Conclusions

- Tumor-specific, CD137-targeted DART proteins, HER2 x CD137 and EphA2 x CD137:
 - Demonstrate good binding activity to each target antigen
 - Promote activation of CD137 signaling (NF-κB pathway)
 - Induce strong costimulatory functions, including enhanced T-cell cytokine release, T-cell proliferation, cytolytic T-cell expansion and expansion of CD8 central memory and effector memory cells in a tumor antigen-dependent fashion
- Tumor-specific, CD137-targted DART proteins may offer an opportunity to induce tumor cell anchored CD137 activation, limiting systemic effects and related adverse events

References

1. Kohrt HE, et al., European Journal of Cancer 54 (2016) 112-119.
2. Ignacio Melero, et. Al., European Journal of Immunology 46 (2016) 513-522.
3. Ye, Q. et al., Clin Canc Res 20(1) (2014) 44-55.
4. Palazon, A. et al., Cancer Discovery 2 (2012) 608 – 623.
5. Kohrt, H. et al, J Clin Invest 122 (3) (2012) 1066-75.

Acknowledgment

We thank Jim Tamura, Qin Wang, Marc Bailly, Qihong Xu, Arash Adami and Judy Downey for their help in generating the poster.

Presented at the American Association for Cancer Research Annual Meeting 2017, April 1–5, 2017, Washington, DC

©2017 MacroGenics, Inc. All rights reserved.